P0514

Routinely Captured Measurements of Vital Signs Within Long-Term Care Facilities Indicate Substantial and Sustained Physiological Health Impacts Associated With COVID-19

Harriet Dickinson[1], Rikisha Gupta[2], Aaron Norfolk[3], Brittin Wagner[3], Kate Mathers[3], Santosh Davies[2], Stanley Wang[3], Anand Chokkalingam[2]

[1]Gilead Sciences, Inc., London, UK; [2]Gilead Sciences, Inc., Foster City, CA, USA; [3]PointClickCare®, Toronto, ON, Canada

Copies of this poster obtained through QR (Quick Response) are for personal use only and may not be reproduced without written permission of the authors.

Conclusions

- This study evaluated vital sign readings of long-term care residents before and after a COVID-19 diagnosis
- High rates of abnormal vital sign readings were observed before and for several weeks after COVID-19 diagnosis, which revealed a measurable and sustained physiological burden associated with the infection
- Even residents who were not hospitalised exhibited abnormal vital sign readings, which only resolved after several weeks
- These findings highlight the potential for COVID-19 to cause both acute and long-term adverse effects in residents of long-term care facilities
- Future research should investigate how vital sign abnormalities can inform the use of early treatment interventions to reduce the burden of COVID-19, particularly in long-term care facilities

Plain Language Summary

- This study looked at vital sign changes before and after COVID-19 diagnosis in older people living in long-term care facilities
- On average, vital signs in residents started worsening before their COVID-19 diagnosis and were the worst at the time of diagnosis
- Even residents who did not go to the hospital showed poor vital sign readings, and it took several weeks before their vital signs returned to normal levels
- These findings suggest that residents were measurably unwell before COVID-19 was diagnosed and continued to be unwell for several weeks after

Introduction

- Older adults and residents of long-term care facilities (LTCFs) are at a heightened risk of severe illness and mortality from COVID-19[1,2]
- Routinely collected vital sign measurements (eg, temperature, heart rate, respiratory rate, oxygen saturation, and blood pressure) can provide insight on patients' physiological health status before, during, and after a COVID-19 diagnosis[3]
- Changes in vital sign measurements in patients with COVID-19 are associated with clinical deterioration, increased disease severity, respiratory failure, and higher mortality rates[4-9]
- Studying vital signs at scale within LTCFs will help define the burden of COVID-19 in this vulnerable population and identify trends that may be masked by individual physiological responses and natural variability
- Timely identification of changes in vital signs may also facilitate early intervention that may prevent or mitigate severe disease[6]

Objective

- To evaluate vital sign changes before and after a COVID-19 diagnosis in a cohort of patients living in an LTCF in the United States

Methods

- A retrospective analysis was conducted using PointClickCare®, a database of electronic medical records from LTCFs in the United States
 - The study period was from 1 March 2020 to 30 June 2023
- Patients were excluded if they were aged <50 years, fitted with pacemakers, or undergoing mechanical ventilation at baseline 2 weeks prior to their COVID-19 diagnosis
- Patients with coinfections were allowed to participate in the study
- Baseline demographic and clinical characteristics were assessed 30 days prior to the COVID-19 diagnosis
- Residents were followed from their COVID-19 diagnosis to the earliest date of hospitalisation, death, loss of follow-up, or study end (ie, 30 June 2023)
- Vital signs evaluated included temperature (cutoff >38 °C), heart rate (cutoff >136 beats per minute), respiration (cutoff >18 breaths per minute), blood pressure (cutoff >62.5 mmHg), and oxygen saturation (with and without supplemental oxygen, cutoff <94%)
 - Vital sign cutoffs were chosen to be representative of residents in LTCFs and were determined using clinician input, scientific and grey literature, guidance for health care providers and LTCFs, and an analysis of data distribution within the PointClickCare® database

Results

- Overall, 471,348 residents with 526,977 COVID-19 diagnoses (episodes) were identified
- The median (SD) age at first diagnosis was 78.4 (11.4) years, 62% of residents were female, and 66% of residents were White (**Table 1**)

Table 1. Resident Demographic and Baseline Characteristics

Characteristic	Overall (n = 471,348)
Age, years, mean (SD)	78.4 (11.4)
Age, years, n (%)	
50-64	62,686 (13)
≥65	408,662 (87)
Sex, n (%)	
Male	180,862 (38)
Female	290,486 (62)
Race, n (%)	
American Indian or Alaska Native	2244 (<1)
Asian	9142 (2)
Black or African American	57,767 (12)
Hispanic or Latino	22,088 (5)
Native Hawaiian or Other Pacific Islander	848 (<1)
White	308,741 (66)
Other	70,518 (15)
Region, n (%)	
Midwest	145,212 (31)
Northeast	91,256 (19)
South	162,963 (35)
West	71,917 (15)
Days in facility, mean (SD)	953.4 (1027.5)

- Of all COVID-19 episodes, 12% were associated with an all-cause death within 4 months after diagnosis and 16% were associated with a hospital stay within 4 months after diagnosis (**Table 2**)

Table 2. All-Cause Death, Hospitalisation, and Reinfection in Residents With a COVID-19 Diagnosis

Group, n (%)	Residents (n = 471,348)	COVID-19 Episodes (n = 526,977)
All-cause death (within washout period[a])	64,053 (14)	64,053 (12)
Any SARS-CoV-2 reinfection[b]	52,627 (11)	55,629 (11)
Any hospitalisation (within washout period[a])	83,527 (18)	84,560 (16)

[a]The washout period was 4 months after diagnosis.
[b]A reinfection was defined as any COVID-19 diagnosis occurring >4 months after prior diagnosis.

- Most residents (88%) had data for ≥1 vital sign
- In the observable period 2 weeks before and 5 weeks after the initial COVID-19 diagnosis, 64% of COVID-19 episodes resulted in an elevated blood pressure, 31% resulted in supplemental oxygen or low oxygen status, 10% resulted in an elevated heart rate, 15% resulted in an elevated temperature, and 67% resulted in an elevated respiration rate (**Table 3**)

Table 3. Vital Sign Abnormalities in the 5-Week Period After Initial COVID-19 Diagnosis

Group, n (%)	Residents (n = 471,348)	COVID-19 Episodes (n = 526,977)
Elevated heart rate	51,082 (11)	52,104 (10)
Low oxygen saturation	83,235 (18)	85,540 (16)
Supplemental oxygen since baseline	109,598 (23)	112,433 (21)
Supplemental oxygen since baseline or low oxygen saturation	159,624 (34)	165,635 (31)
Elevated blood pressure	306,551 (65)	335,159 (64)
Elevated respiration rate	322,633 (68)	352,550 (67)
Elevated temperature	77,959 (17)	78,990 (15)

- The proportion of COVID-19 episodes with abnormal vital signs was highest at COVID-19 diagnosis and declined steadily until 4 weeks after diagnosis (**Figure 1**)

Figure 1. Percentage of COVID-19 Episodes With Abnormal Vital Sign Measurements Before and After COVID-19 Diagnosis[a]

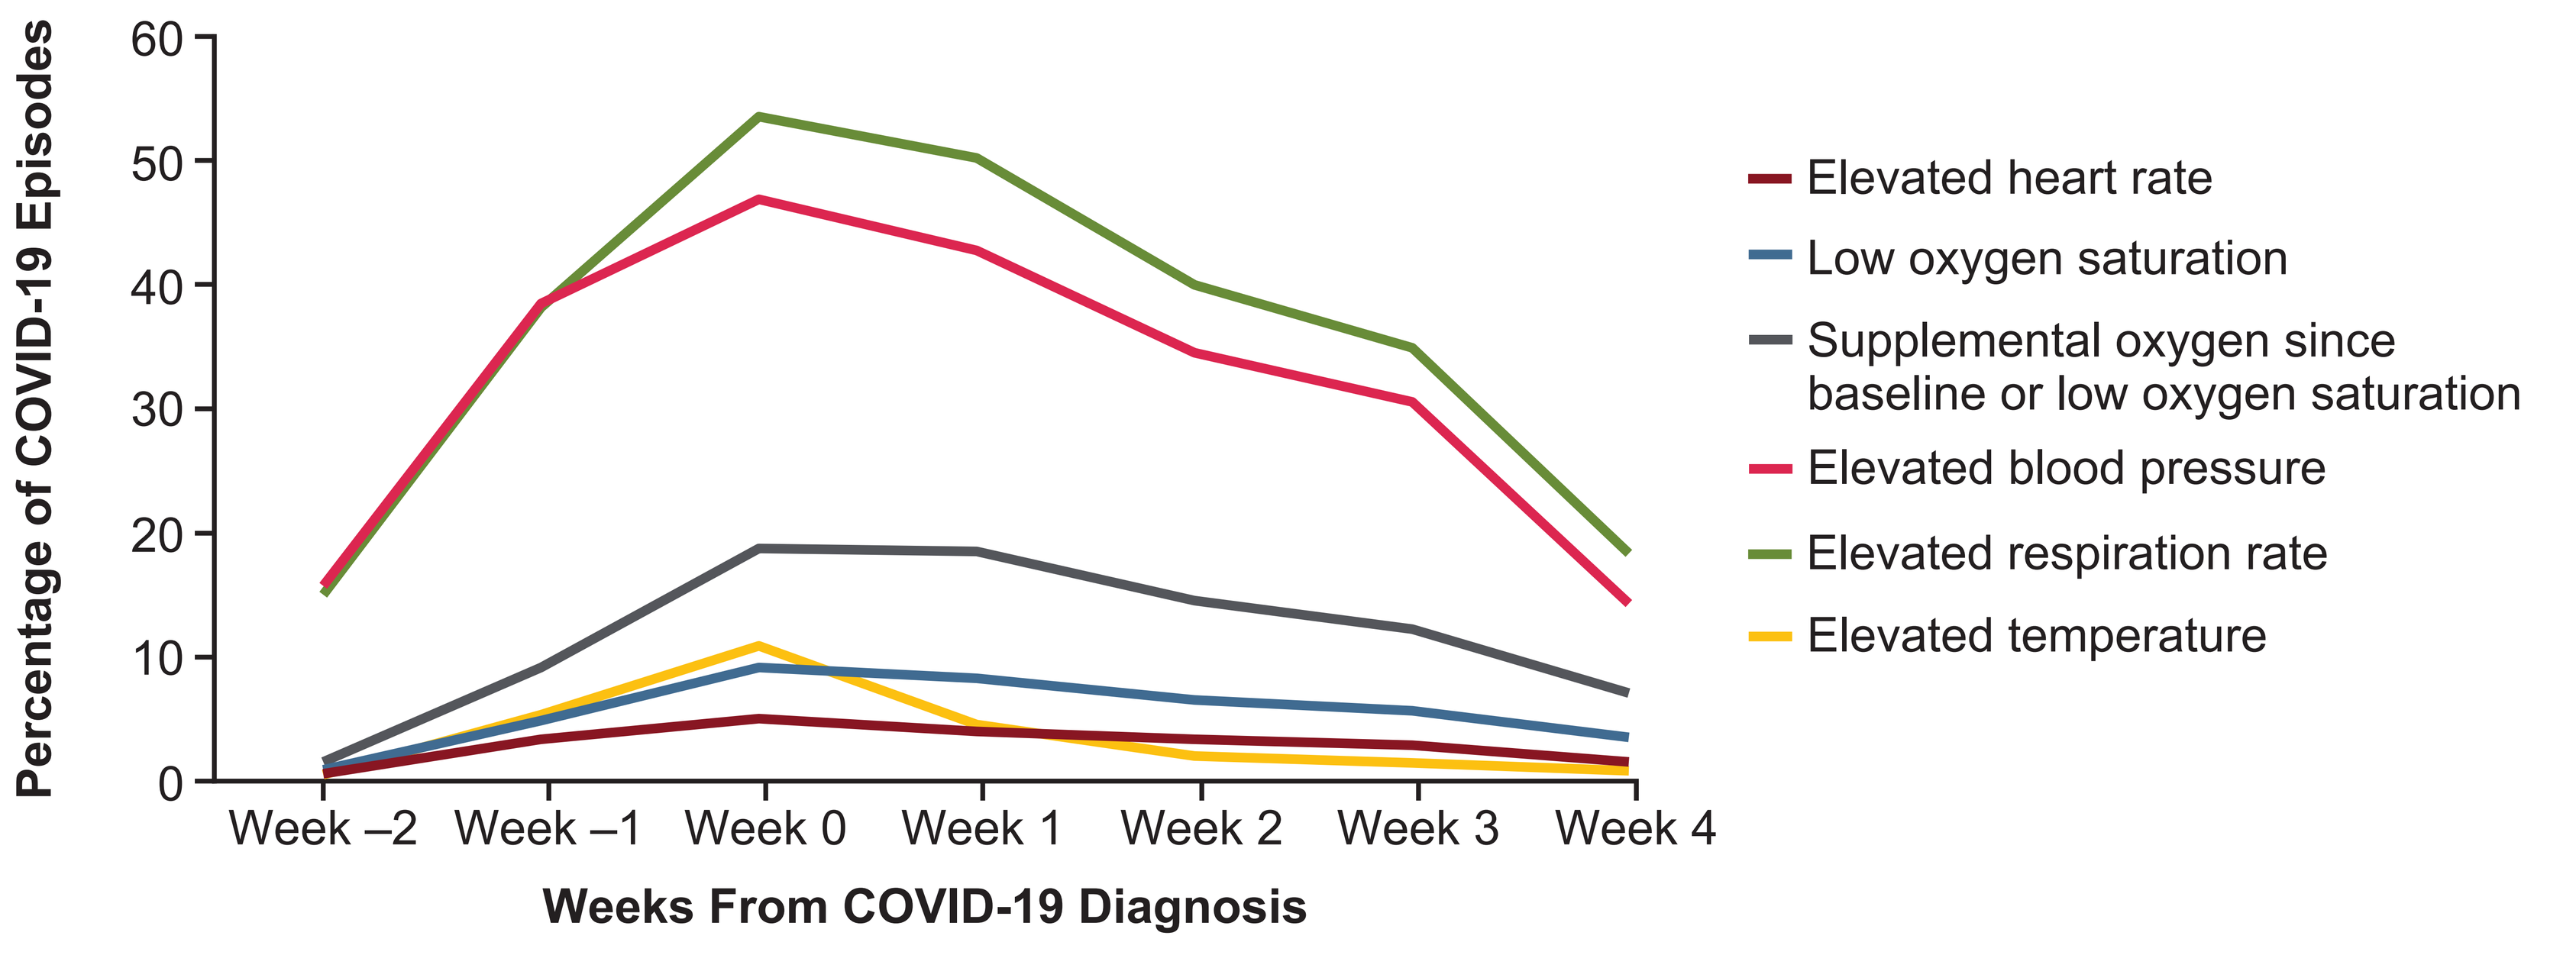
Percentage of COVID-19 Episodes
60
50
40
30
20
10
0
Week –2
Week –1
Week 0
Week 1
Week 2
Week 3
Week 4
Weeks From COVID-19 Diagnosis
Elevated heart rate
Low oxygen saturation
Supplemental oxygen since baseline or low oxygen saturation
Elevated blood pressure
Elevated respiration rate
Elevated temperature

[a]In residents who survived ≥5 weeks without death or hospitalisation.

- The mean (SD) continuous duration of any abnormal readings in residents who survived 5 weeks without hospitalisation or death was 2.3 (2.7) days; elevated respiration rate had the longest continuous duration (mean [SD], 2.4 [2.7] days; **Table 4**)

Table 4. Continuous Duration of Abnormal Vital Signs in Residents Who Survived 5 Weeks Without Hospitalisation or Death After COVID-19 Diagnosis

Symptom	Continuous Duration, Days, Mean (SD)
Any	2.3 (2.7)
Elevated heart rate	1.8 (2.1)
Low oxygen saturation	1.6 (1.8)
Supplemental oxygen since baseline	2.3 (3.1)
Supplemental oxygen since baseline or low oxygen saturation	2.3 (3.1)
Elevated blood pressure	2.3 (2.6)
Elevated respiration rate	2.4 (2.7)
Elevated temperature	1.5 (1.3)

Limitations

- These results should be interpreted with consideration of the individual physiological differences between patients and may not reflect the experience of an individual patient
- Not all patients had vital sign readings, and it was not possible to ascertain vaccination status
- Standards of care and referral pathways for COVID-19 may also be subject to change, limiting generalisability

References: **1.** Gordon AL, et al. *Age Ageing*. 2020;49:701-5. **2.** Aalto UL, et al. *Eur Geriatr Med*. 2022;13:705-9. **3.** Elliott M. *Br J Nurs*. 2021;30:956-62. **4.** Rivas E, et al. *Front Med (Lausanne)*. 2023;10:1243050. **5.** Pimentel MAF, et al. *Resuscitation*. 2020;156:99-106. **6.** Ikram AS, Pillay S. *BMC Emerg Med*. 2022;22:68. **7.** Lippi G, et al. *Pol Arch Intern Med*. 2020;130:304-9. **8.** Choron RL, et al. *J Intensive Care Med*. 2021;36:484-93. **9.** Azami P, et al. *BMC Cardiovasc Disord*. 2024;24:240.

Acknowledgements: This study was funded by Gilead Sciences, Inc. Medical writing and editorial support were provided by Katherine Townsend, PhD, of Lumanity Communications Inc., and were funded by Gilead Sciences, Inc.

Correspondence: Harriet Dickinson, harriet.dickinson@gilead.com

Disclosures: **HD**, **RG**, **SD**, and **AC** are employees of and may own stock or stock options in Gilead Sciences, Inc. **AN**, **BW**, **KM**, and **SW** are employees of PointClickCare®.

35th Annual Congress of the European Society of Clinical Microbiology and Infectious Diseases (ESCMID); 11-15 April 2025; Vienna, Austria