Effectiveness of Remdesivir in Patients With Underlying Hepatic or Renal Comorbidity Hospitalized for SARS-CoV-2 Infection

P-1632

Patrick Godwin[1], John Jay Hawkshead[2], Valentina Shvachko[2], Thomas Oppelt[2], Chen-Yu Wang[2], Essy Mozaffari[2], Amos Lichtman[2], Mark Berry[2]

[1]Department of Medicine, University of Illinois College of Medicine, Chicago, IL, USA; [2]Gilead Sciences, Inc., Foster City, CA, USA

Copies of this poster obtained through QR (Quick Response) code are for personal use only and may not be reproduced without written permission of the authors.

Conclusions

- This retrospective analysis of US patients hospitalized for COVID-19 found that those with underlying renal or hepatic comorbidities had a decreased risk of 28-day all-cause in-hospital mortality when treated with remdesivir in the first 2 days of hospitalization compared with those who did not receive remdesivir in the first 2 days of hospitalization
 - Nearly 70% of patients included in the study received supplemental oxygen during the first 2 days of hospitalization
 - The reduction in mortality risk with early remdesivir initiation was observed among patients with renal or hepatic comorbidities who received supplemental oxygen during the first 2 days of hospitalization
 - Those who did not receive supplemental oxygen during the first 2 days of hospitalization also had a reduced risk of mortality with early remdesivir initiation; this reduction was statistically significant in patients with hepatic comorbidities but not statistically significant in patients with renal comorbidities
- These results indicate that early remdesivir initiation can improve survival among patients with renal or hepatic disease who are hospitalized for COVID-19

Plain Language Summary

- People who have kidney or liver disease have a higher chance of getting very sick from COVID-19
- Remdesivir is a medication used to treat people for COVID-19, but the real-world benefits of remdesivir in people with kidney or liver disease have not been specifically studied
- This study examined data from people in the United States with kidney or liver disease who were hospitalized for COVID-19 and found that those who received remdesivir in the first 2 days of being in the hospital had a lower risk of dying for up to 28 days compared with those who did not
- These findings suggest that starting remdesivir treatment soon after entering the hospital can help lower the risk of dying in people with kidney or liver disease

References: **1.** He M, et al. *Front Med* (*Lausanne*). 2023;10:1250631. **2.** Nagarajan R, et al. *Prev Chronic Dis*. 2022;19:210228. **3.** Mozaffari E, et al. *Clin Infect Dis*. 2023;77:1626-34. **4.** Mozaffari E, et al. *Clin Infect Dis*. 2024;79:S149-59. **5.** Mozaffari E, et al. *Clin Infect Dis*. 2024;79:S137-48. **6.** VEKLURY® (remdesivir) for injection, for intravenous use [package insert]. Gilead Sciences, Inc.; 2025.

Acknowledgments: This study was funded by Gilead Sciences, Inc. Medical writing and editorial support were provided by Catherine Bautista, PhD, of Lumanity Communications Inc., and were funded by Gilead Sciences, Inc.

Correspondence: Mark Berry, mark.berry1@gilead.com; 333 Lakeside Dr, Foster City, CA 94404, USA

Disclosures: **PG** served as an advisor/consultant and participated in advisory boards and speakers bureaus for Gilead Sciences, Inc. **JJH** and **EM** are former employees of Gilead Sciences, Inc. **VS**, **TO**, **C-YW**, **AL**, and **MB** are employees of Gilead Sciences, Inc. **VS**, **TO**, **C-YW**, **EM**, **AL**, and **MB** are stockholders of Gilead Sciences, Inc.

Introduction

- Individuals with COVID-19 who have underlying renal or hepatic comorbidities are at a higher risk of mortality than those without these comorbidities[1,2]
- Antiviral treatment with remdesivir (RDV) in individuals hospitalized for COVID-19 has been shown to reduce mortality across various high-risk populations[3-5]
- Real-world data on the effectiveness of early RDV initiation in individuals with renal or hepatic comorbidities are needed to inform clinical decision making

Objective

- To examine the effect of early RDV initiation on 28-day all-cause in-hospital mortality among patients in the United States who were hospitalized for COVID-19 and had renal or hepatic comorbidities

Methods

- A retrospective comparative effectiveness study was conducted using deidentified medical claims and hospital chargemaster data in the HealthVerity database (December 2021-April 2025) from US patients aged ≥18 years who were hospitalized with a primary diagnosis of COVID-19
 - Patients were excluded if they had evidence of RDV use within 90 days prior to hospitalization, were admitted from hospice, or were pregnant
 - The renal cohort included patients with evidence of kidney disease that required renal replacement therapy (eg, hemodialysis, peritoneal dialysis, and hemofiltration) during the baseline period (ie, 12 months before RDV initiation or corresponding matched date)
 - The hepatic cohort included patients with hepatic dysfunction (eg, alcoholic hepatic failure, acute and subacute hepatic failure, chronic hepatic failure, postprocedural hepatic failure, unspecified hepatic failure, biliary tract disease, other liver disease, and noninfectious hepatitis) during the baseline period
- Patients who received ≥1 dose of RDV in the first 2 days of hospitalization (early RDV) were compared with those who did not receive RDV in the first 2 days of hospitalization (comparator)
- Patients were followed from the third day after hospitalization until hospital discharge, transfer to hospice or death, the end of the study period, or the end of follow-up (30 days after hospitalization), whichever occurred first
- Coarsened exact matching (based on sex, age category, maximum level of supplemental oxygen use in the first 2 days of hospitalization, and calendar time of hospitalization) followed by propensity score matching (based on the same variables as well as baseline comorbidities [eg, metabolic disease and hypertension], insurance type, hospital size, hospital type, hospital location, sepsis at admission, acute respiratory distress syndrome or acute respiratory failure with hypoxia or pneumonia at admission, intensive care unit admission in the first 2 days of hospitalization, medication use [eg, corticosteroids and anticoagulants] in the 14 days prior to hospitalization through 1 day after hospitalization, and any hospitalization during the baseline period) was performed (1:1) to balance baseline demographic and clinical characteristics between the early RDV and comparator groups
- Propensity score–matched hazard ratios and 95% CIs for 28-day all-cause in-hospital mortality risk after RDV initiation were estimated using Cox proportional hazards models
 - Analyses were conducted for the renal and hepatic cohorts overall and stratified by the use of supplemental oxygen in the first 2 days of hospitalization (ie, no supplemental oxygen vs supplemental oxygen [low-flow oxygen, high-flow oxygen, noninvasive ventilation, invasive mechanical ventilation, or extracorporeal membrane oxygenation])

Results

Study Population

- Among 280,843,029 patients included in the HealthVerity dataset, 24,787 patients were included in the renal cohort (early RDV, 7125; comparator, 17,662) and 8377 patients were included in the hepatic cohort (early RDV, 2686; comparator, 5691)
 - 17,043 (69%) patients in the renal cohort and 5629 (67%) patients in the hepatic cohort received supplemental oxygen in the first 2 days of hospitalization
- After 1:1 propensity score matching, 13,824 patients were included in the renal cohort (6912 per group) and 5026 patients were included in the hepatic cohort (2513 per group; **Table 1**)
 - For both cohorts, the characteristics between the 2 study groups were generally similar after propensity score matching

Table 1. Propensity Score–Matched Cohort Characteristics

	Renal Cohort			Hepatic Cohort		
Characteristic	**Early RDV (n = 6912)**	**Comparator (n = 6912)**	***P* Value**	**Early RDV (n = 2513)**	**Comparator (n = 2513)**	***P* Value**
Sex, female, n (%)	3334 (48)	3335 (48)	1.00	1277 (51)	1281 (51)	0.93
Age category, n (%)						
18-<30 y	30 (<1)	31 (<1)	1.00	21 (1)	26 (1)	0.56
30-<40 y	137 (2)	137 (2)	1.00	90 (4)	98 (4)	0.60
40-<50 y	316 (5)	331 (5)	0.57	153 (6)	144 (6)	0.63
50-<60 y	778 (11)	778 (11)	1.00	412 (16)	412 (16)	1.00
60-<75 y	2447 (35)	2459 (36)	0.84	1133 (45)	1153 (46)	0.59
≥75 y	3204 (46)	3176 (46)	0.65	704 (28)	680 (27)	0.47
CCI score, mean (SD)	6.50 (3.39)	6.60 (3.41)	0.08	6.22 (3.80)	6.21 (3.71)	0.94
Insurance type, n (%)						
Private	723 (10)	716 (10)	0.87	369 (15)	385 (15)	0.55
Medicare	4070 (59)	4039 (58)	0.60	1267 (50)	1257 (50)	0.80
Medicaid	543 (8)	550 (8)	0.85	319 (13)	318 (13)	1.00
Missing	1576 (23)	1607 (23)	0.54	558 (22)	553 (22)	0.89
Hospital size, n (%)						
<100 beds	498 (7)	495 (7)	0.95	205 (8)	201 (8)	0.88
100-199 beds	1455 (21)	1491 (22)	0.47	536 (21)	554 (22)	0.56
200-299 beds	1001 (14)	1037 (15)	0.40	375 (15)	370 (15)	0.87
300-499 beds	2088 (30)	2090 (30)	0.99	710 (28)	709 (28)	1.00
≥500 beds	1664 (24)	1587 (23)	0.13	623 (25)	623 (25)	1.00
Missing	206 (3)	212 (3)	0.80	64 (3)	56 (2)	0.52
Hospital type, n (%)						
Nonteaching	4062 (59)	4092 (59)	0.62	1512 (60)	1536 (61)	0.51
Teaching	2699 (39)	2659 (38)	0.50	963 (38)	946 (38)	0.64
Missing	151 (2)	161 (2)	0.61	38 (2)	31 (1)	0.47
Hospital location, n (%)						
Rural	671 (10)	642 (9)	0.42	266 (11)	276 (11)	0.68
Urban	6207 (90)	6236 (90)	0.43	2235 (89)	2227 (89)	0.75
Missing	34 (<1)	34 (<1)	1.00	12 (<1)	10 (<1)	0.83
Sepsis at admission, n (%)	1148 (17)	1181 (17)	0.47	404 (16)	403 (16)	1.00
ARDS or ARF with hypoxia or pneumonia at admission, n (%)	2837 (41)	2840 (41)	0.97	1020 (41)	994 (40)	0.47
RDV use during hospitalization, n (%)	6912 (100)	501 (7)[a]	<0.01	2513 (100)	172 (7)[a]	<0.01
Supplemental oxygen use in the first 2 days of hospitalization, n (%)						
No supplemental oxygen	2076 (30)	2081 (30)	0.94	787 (31)	795 (32)	0.83
Any supplemental oxygen	4836 (70)	4831 (70)	0.94	1726 (69)	1718 (68)	0.83
ICU admission in the first 2 days of hospitalization, n (%)	2640 (38)	2611 (38)	0.62	985 (39)	955 (38)	0.40
Corticosteroid use,[b] n (%)	5381 (78)	5421 (78)	0.42	1951 (78)	1960 (78)	0.79
Oral antiviral use,[b] n (%)	14 (<1)	14 (<1)	1.00	17 (1)	18 (1)	1.00
Anticoagulant use,[b] n (%)	3947 (57)	3952 (57)	0.95	975 (39)	979 (39)	0.93
Convalescent plasma use,[b] n (%)	20 (<1)	15 (<1)	0.50	9 (<1)	12 (<1)	0.66
Immunomodulator use,[b] n (%)	228 (3)	239 (3)	0.64	108 (4)	107 (4)	1.00
Any hospitalization during the baseline period, n (%)	4851 (70)	4844 (70)	0.91	1315 (52)	1318 (52)	0.95

[a]RDV use during hospitalization but not during the first 2 days of hospitalization.
[b]Comedication use was identified by evidence of use at any time in the 14 days prior to hospitalization through 1 day after hospitalization.
ARDS, acute respiratory distress syndrome; ARF, acute respiratory failure; CCI, Charlson Comorbidity Index; ICU, intensive care unit; RDV, remdesivir.

Effect of Early RDV Initiation on 28-Day All-Cause In-Hospital Mortality in the Renal Cohort

- In the renal cohort, there was a propensity score–matched risk difference per 1000 patients of –13.17 for the early RDV group versus the comparator group (**Table 2**), with a 17% reduction in the risk of 28-day all-cause in-hospital mortality (*P* <0.01; **Figure 1**)
 - Among patients who did not receive supplemental oxygen, there was a 5% reduction in the propensity score–matched risk of 28-day all-cause in-hospital mortality with early RDV initiation, although this reduction was not statistically significant (*P* = 0.68)
 - Among patients who received supplemental oxygen, there was a 24% reduction in the propensity score–matched risk of 28-day all-cause in-hospital mortality with early RDV initiation (*P* <0.01)

Table 2. Propensity Score–Matched Risk Parameters for 28-Day All-Cause In-Hospital Mortality With RDV

	Renal Cohort		Hepatic Cohort	
Parameter	**Early RDV**	**Comparator**	**Early RDV**	**Comparator**
Patients	6912	6912	2513	2513
Number of events	467	558	126	165
Risk per 1000 patients	67.56	80.73	50.14	65.66
Risk ratio vs comparator (95% CI)	0.84 (0.74, 0.94)	Comparator	0.76 (0.61, 0.96)	Comparator
Risk difference per 1000 patients vs comparator (95% CI)	–13.17 (–22.04, –4.29)	Comparator	–15.52 (–28.82, –2.21)	Comparator

RDV, remdesivir.

Figure 1. Propensity Score–Matched 28-Day All-Cause In-Hospital Mortality Risk With Early RDV Initiation

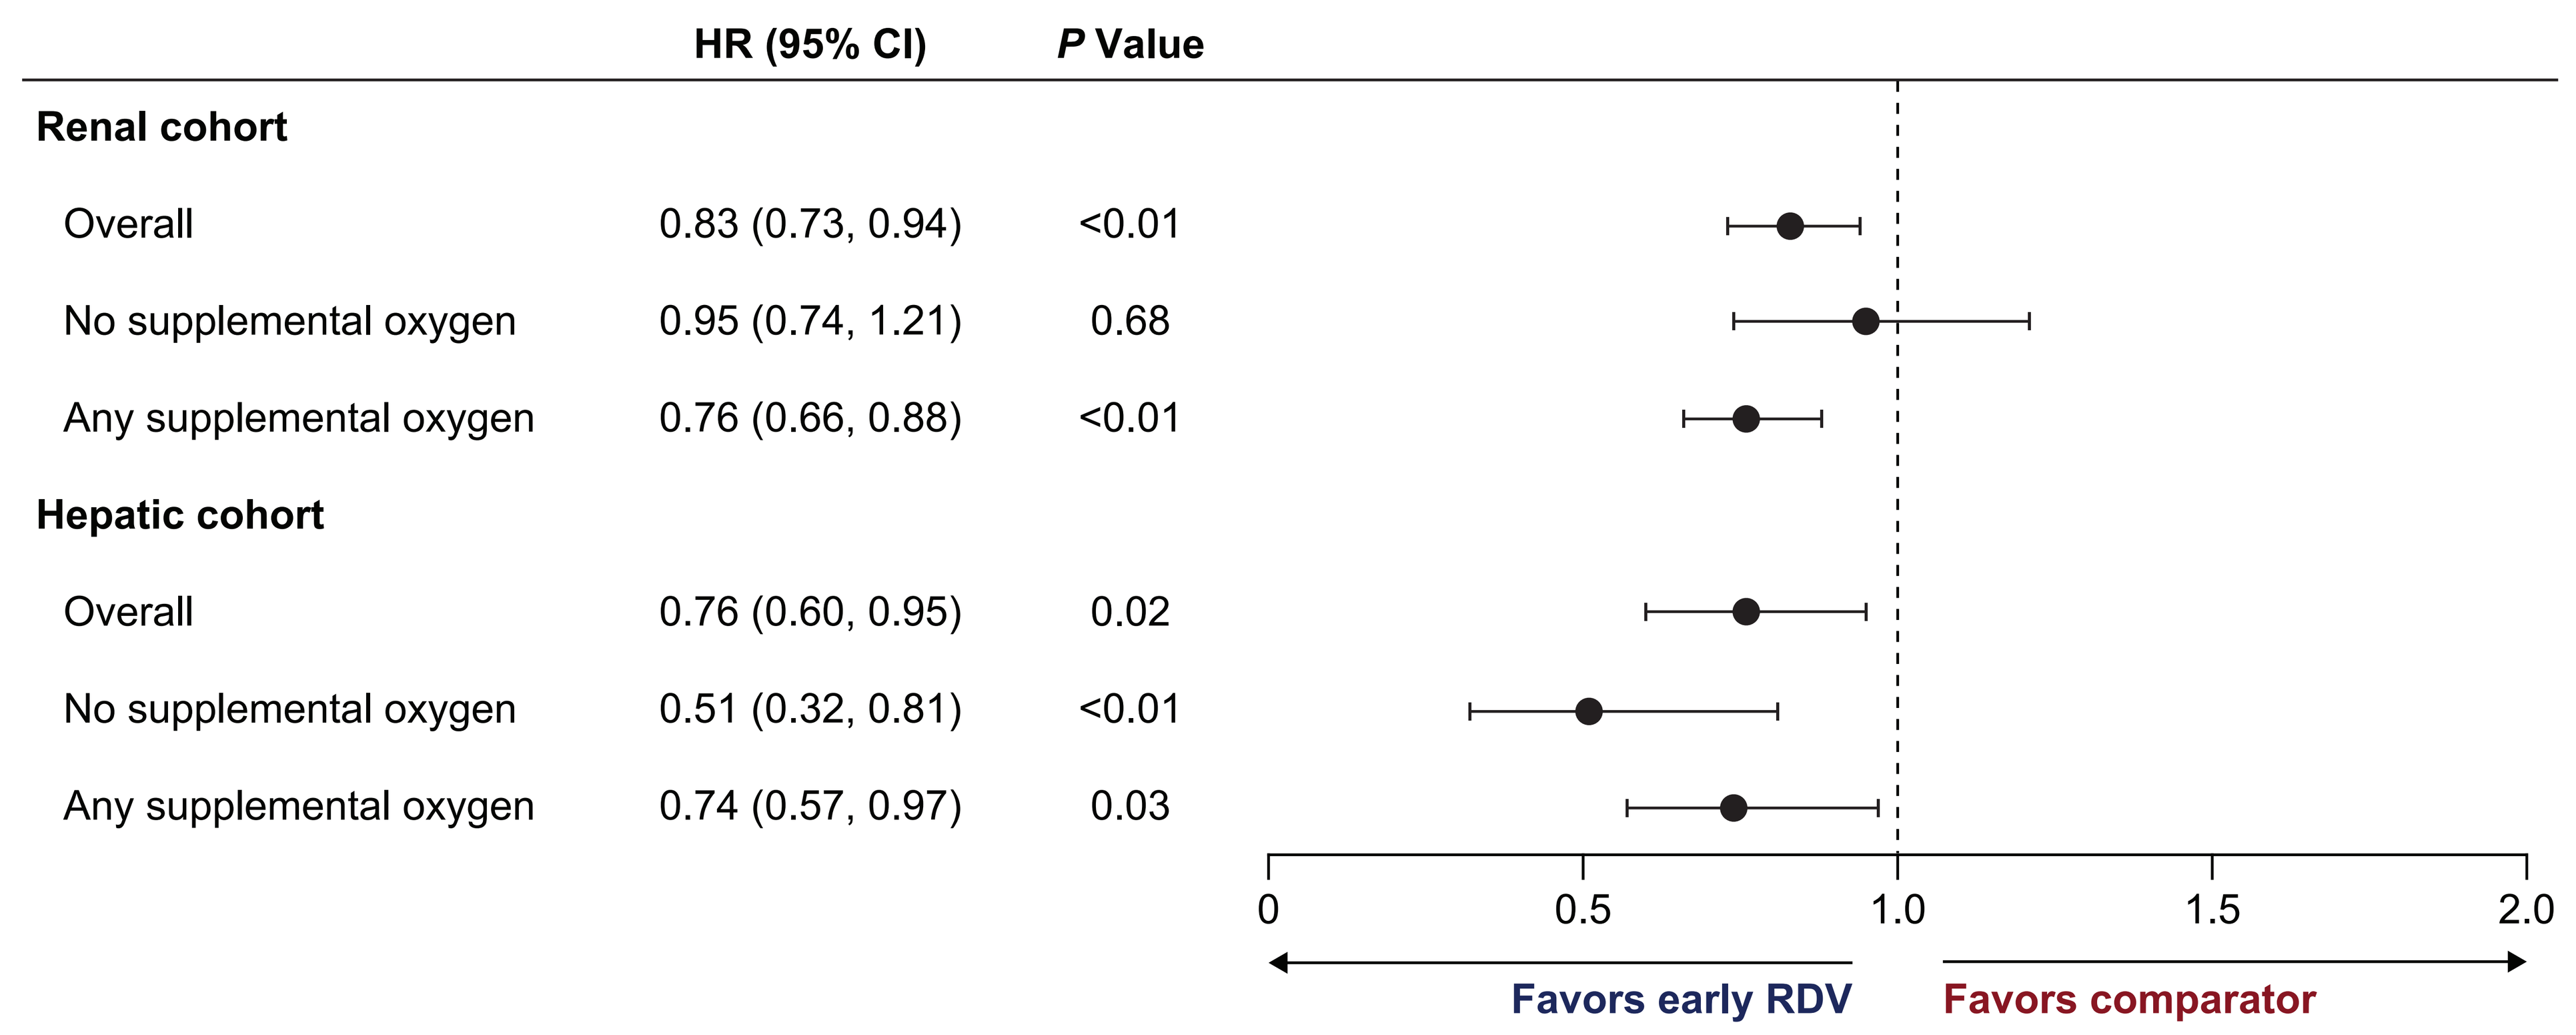
HR (95% CI)
P Value
Renal cohort
Overall
0.83 (0.73, 0.94)
<0.01
No supplemental oxygen
0.95 (0.74, 1.21)
0.68
Any supplemental oxygen
0.76 (0.66, 0.88)
<0.01
Hepatic cohort
Overall
0.76 (0.60, 0.95)
0.02
No supplemental oxygen
0.51 (0.32, 0.81)
<0.01
Any supplemental oxygen
0.74 (0.57, 0.97)
0.03
0
0.5
1.0
1.5
2.0
Favors early RDV
Favors comparator

HR, hazard ratio; RDV, remdesivir.

Effect of Early RDV Initiation on 28-Day All-Cause In-Hospital Mortality in the Hepatic Cohort

- In the hepatic cohort, there was a propensity score–matched risk difference per 1000 patients of –15.52 for the early RDV group versus the comparator group (**Table 2**), with a 24% reduction in the risk of 28-day all-cause in-hospital mortality (*P* = 0.02; **Figure 1**)
 - Among patients who did not receive supplemental oxygen and those who did receive supplemental oxygen, there was a 49% (*P* <0.01) and 26% (*P* = 0.03) reduction, respectively, in the propensity score–matched risk of 28-day all-cause in-hospital mortality with early RDV initiation

Limitations

- Patients with more time between symptom onset and hospital admission may be less likely to initiate RDV; since the time of symptom onset was not captured in the HealthVerity database, this may be a source of unmeasured confounding
- Limited or no data were available regarding patients' vaccination status or the severity of their underlying renal or hepatic comorbidities
- During the study period, the US Food and Drug Administration expanded the RDV label (July 2023) to include use in patients with COVID-19 with severe renal impairment, including those on dialysis,[6] which may have influenced treatment patterns in this population

IDWeek; October 19-22, 2025; Atlanta, GA, USA