P-2133

Resistance Analysis of Emergent Nsp12 Substitutions Observed in Immunocompromised COVID-19 Patients Treated With Remdesivir

Lauren Rodriguez, Dong Han, Nadine Peinovich, Jasmine Moshiri, Jason K Perry, Jiani Li, Charlotte Hedskog

Gilead Sciences, Inc., Foster City, CA, USA

Lauren Rodriguez
lauren.rodriguez14@gilead.com
333 Lakeside Dr
Foster City, CA 94404, USA

Copies of this poster obtained through QR (Quick Response) code are for personal use only and may not be reproduced without written permission of the authors

Conclusions

- Across 6 case studies of immunocompromised patients with COVID-19, emergent amino acid substitutions in SARS-CoV-2 Nsp12 were uncommon
- While most observed emergent Nsp12 substitutions were associated with no change in in vitro remdesivir susceptibility, the substitutions E136A, V166L + I536V, and C464Y resulted in half-maximal effective concentration fold change values relative to wildtype ranging from 2.8 to 4.0
- Structural analysis of the emergent Nsp12 substitutions associated with low-level reductions in remdesivir susceptibility revealed clustering around the Nsp12 palm domain motif D, which is associated with the dynamics of nucleoside triphosphate incorporation into replicating viral RNA
- Despite prolonged SARS-CoV-2 infection and remdesivir treatment in immunocompromised patients, remdesivir demonstrated a high barrier to resistance, highlighting its utility in this population

Plain Language Summary

- Remdesivir is a drug that is used to treat COVID-19, the disease caused by the virus SARS-CoV-2
- When a person with a weakened immune system has COVID-19, they may be sick with COVID-19 for longer or need longer treatment with remdesivir than those with a healthy immune system
- A virus can sometimes become resistant to a drug if it develops mutations that make the drug less effective
- Mutations previously reported in 6 studies of people with COVID-19 and weakened immune systems were tested to find out whether the mutations led to resistance against remdesivir
- Viral mutations were rare and had little or no effect on the effectiveness of remdesivir
- Overall, these findings suggest that the virus that causes COVID-19 is unlikely to become resistant to remdesivir

References: **1.** VEKLURY® (remdesivir) for injection, for intravenous use [package insert]. Gilead Sciences, Inc.; 2025. **2.** Gordon CJ, et al. *J Biol Chem*. 2020;295:6785-97. **3.** Beigel JH, et al. *N Engl J Med*. 2020;383:1813-26. **4.** Gottlieb RL, et al. *N Engl J Med*. 2022;386:305-15. **5.** Ahmed A, et al. *Pediatrics*. 2024;153:e2023063775. **6.** Sise ME, et al. *Clin Infect Dis*. 2024;79:1172-81. **7.** Hedskog C, et al. *J Infect Dis*. 2023;228:1263-73. **8.** Hedskog C, et al. *Viruses*. 2024;16:546. **9.** Rodriguez L, et al. *Antimicrob Agents Chemother*. 2024;69:e0123824. **10.** Andreatta K, et al. *Open Forum Infect Dis*. 2023;10:ofad500.575. **11.** Rodriguez L, et al. *Antimicrob Agents Chemother*. 2026;70:e0191225. **12.** Kemp SA, et al. *Nature*. 2021;592:277-82. **13.** Nooruzzaman M, et al. *Nat Commun*. 2024;15:7999. **14.** Bay AC, et al. *IDCases*. 2024;38:e02118. **15.** Heyer A, et al. *Cell Rep Med*. 2022;3:100735. **16.** Sama B, et al. *Antiviral Res*. 2024;232:106034. **17.** Ichikawa T, et al. *Antiviral Res*. 2025;242:106264. **18.** Xie X, et al. *Nat Commun*. 2020;11:5214. **19.** Zhang Y, et al. *Antiviral Res*. 2021;185:104974. **20.** Malone BF, et al. *Nature*. 2023;614:781-7. **21.** Chen J, et al. *Cell*. 2020;182:1560-73.e13. **22.** Stevens LJ, et al. *Sci Transl Med*. 2022;14:eabo0718.

Acknowledgments: This study was funded by Gilead Sciences, Inc. Medical writing and editorial support were provided by Jane Abolafia, PhD, of Lumanity Communications Inc., and were funded by Gilead Sciences, Inc.

Correspondence: Lauren Rodriguez, lauren.rodriguez14@gilead.com

Disclosures: **LR**, **DH**, **NP**, **JM**, **JL**, and **CH** are stockholders and employees of Gilead Sciences, Inc. **JKP** is a stockholder of Gilead Sciences, Inc., and was an employee of Gilead Sciences, Inc., at the time of this study.

Introduction

- Remdesivir (RDV) is a nucleotide analog prodrug with potent antiviral activity against multiple viruses, including SARS-CoV-2[1]
 - RDV inhibits the highly conserved SARS-CoV-2 RNA-dependent RNA polymerase Nsp12[2]
- RDV has demonstrated efficacy and safety in multiple clinical trials and is approved for the treatment of COVID-19 in hospitalized and nonhospitalized patients[1,3-6]
- While uncommon, treatment-emergent Nsp12 substitutions have been identified in RDV clinical trials[5,7-11]
 - Virologic analyses of these substitutions are consistent with a high barrier to RDV resistance
- Here we present virologic, structural, and clinical analyses of Nsp12 substitutions reported in 6 case studies of immunocompromised patients with COVID-19 who were treated with RDV[12-17]

Objective

- To evaluate the antiviral activity of RDV against SARS-CoV-2 Nsp12 substitutions observed in immunocompromised patients with COVID-19 in published literature

Methods

- A comprehensive literature review was performed to identify case studies reporting RDV treatment–emergent Nsp12 substitutions among immunocompromised patients with COVID-19 receiving RDV for 3 to 20 consecutive days over 1 to 6 separate treatment courses
- The antiviral activity of RDV was evaluated against the identified emergent Nsp12 amino acid substitutions by phenotypic testing of site-directed mutants using a SARS-CoV-2 replicon system[18,19]
- Substitution prevalence was calculated using all publicly available SARS-CoV-2 sequences deposited in the Global Initiative on Sharing All Influenza Data (GISAID) database since the onset of the COVID-19 pandemic
- Emergent Nsp12 amino acid substitutions were mapped onto a cryogenic electron microscopy (cryo-EM) model of the SARS-CoV-2 replication-transcription complex[20,21]

Results

Identification of Published Nsp12 Substitutions in Immunocompromised Patients With COVID-19 Treated With RDV

- Six case studies were identified that reported Nsp12 substitutions in immunocompromised patients with COVID-19 treated with RDV (**Table 1**)[12-17]

Table 1. Nsp12 Substitutions Reported Among Immunocompromised Patients With COVID-19 Treated With RDV

Case Study	Nsp12 Amino Acid Substitutions
Kemp 2021[12]	G446C G446C + V729I
Nooruzzaman 2024[13]	E136A C464Y T643I E796K E796K + V792I C799Y C464Y + C799Y
Bay 2024[14]	C464F
Heyer 2022[15]	V166L I536V V166L + I536V
Sama 2024[16]	A777S
Ichikawa 2025[17]	E796D

RDV, remdesivir.

RDV Susceptibility Phenotype of Emergent Nsp12 Substitutions

- Compared with the SH01 wildtype (WT) reference replicon, Nsp12 amino acid substitutions E136A, V166L + I536V, and C464Y resulted in mean half-maximal effective concentration (EC_{50}) fold change values of 2.8 to 4.0; this difference was consistent with low-level reduced susceptibility to RDV (**Table 2**; **Figures 1** and **2**)
- Nsp12 amino acid substitutions V166L, G446C, G446C + V792I, C464F, C464Y + C799Y, I536V, T643I, A777S, E796K + V792I, and C799Y led to mean EC_{50} fold change values of 0.8 to 2.4 compared with the SH01 WT reference replicon, suggesting no impact on RDV susceptibility
- The EC_{50} fold change for the Nsp12 amino acid substitution E796K could not be determined due to lack of replication

Table 2. RDV EC_{50} Against SARS-CoV-2 Reference and Mutant Replicons

Substitution	Mean (SD) RDV EC_{50}, nM
SH01 WT	12.6 (2.2)
S759A + V792I[a]	123.7 (24.3)
V792I[a]	43.4 (8.7)
E136A	50.9 (5.0)
V166L	18.3 (1.4)
V166L + I536V	39.7 (3.2)
G446C	9.6 (0.1)
G446C + V792I	26.7 (1.6)
C464F	23.2 (0.9)
C464Y	35.5 (6.9)
C464Y + C799Y	19.5 (0.2)
I536V	13.5 (0.4)
T643I	11.9 (0.2)
A777S	8.2 (2.3)
E796D	29.5 (2.8)
E796K	–[b]
E796K + V792I	13.6 (0.2)
C799Y	16.5 (0.1)

[a]This substitution was included as a positive control.
[b]No replication was observed for this substitution.
EC_{50}, half-maximal effective concentration; RDV, remdesivir; WT, wildtype.

Figure 1. RDV EC_{50} Fold Change Against Emergent Nsp12 Amino Acid Substitutions Relative to WT

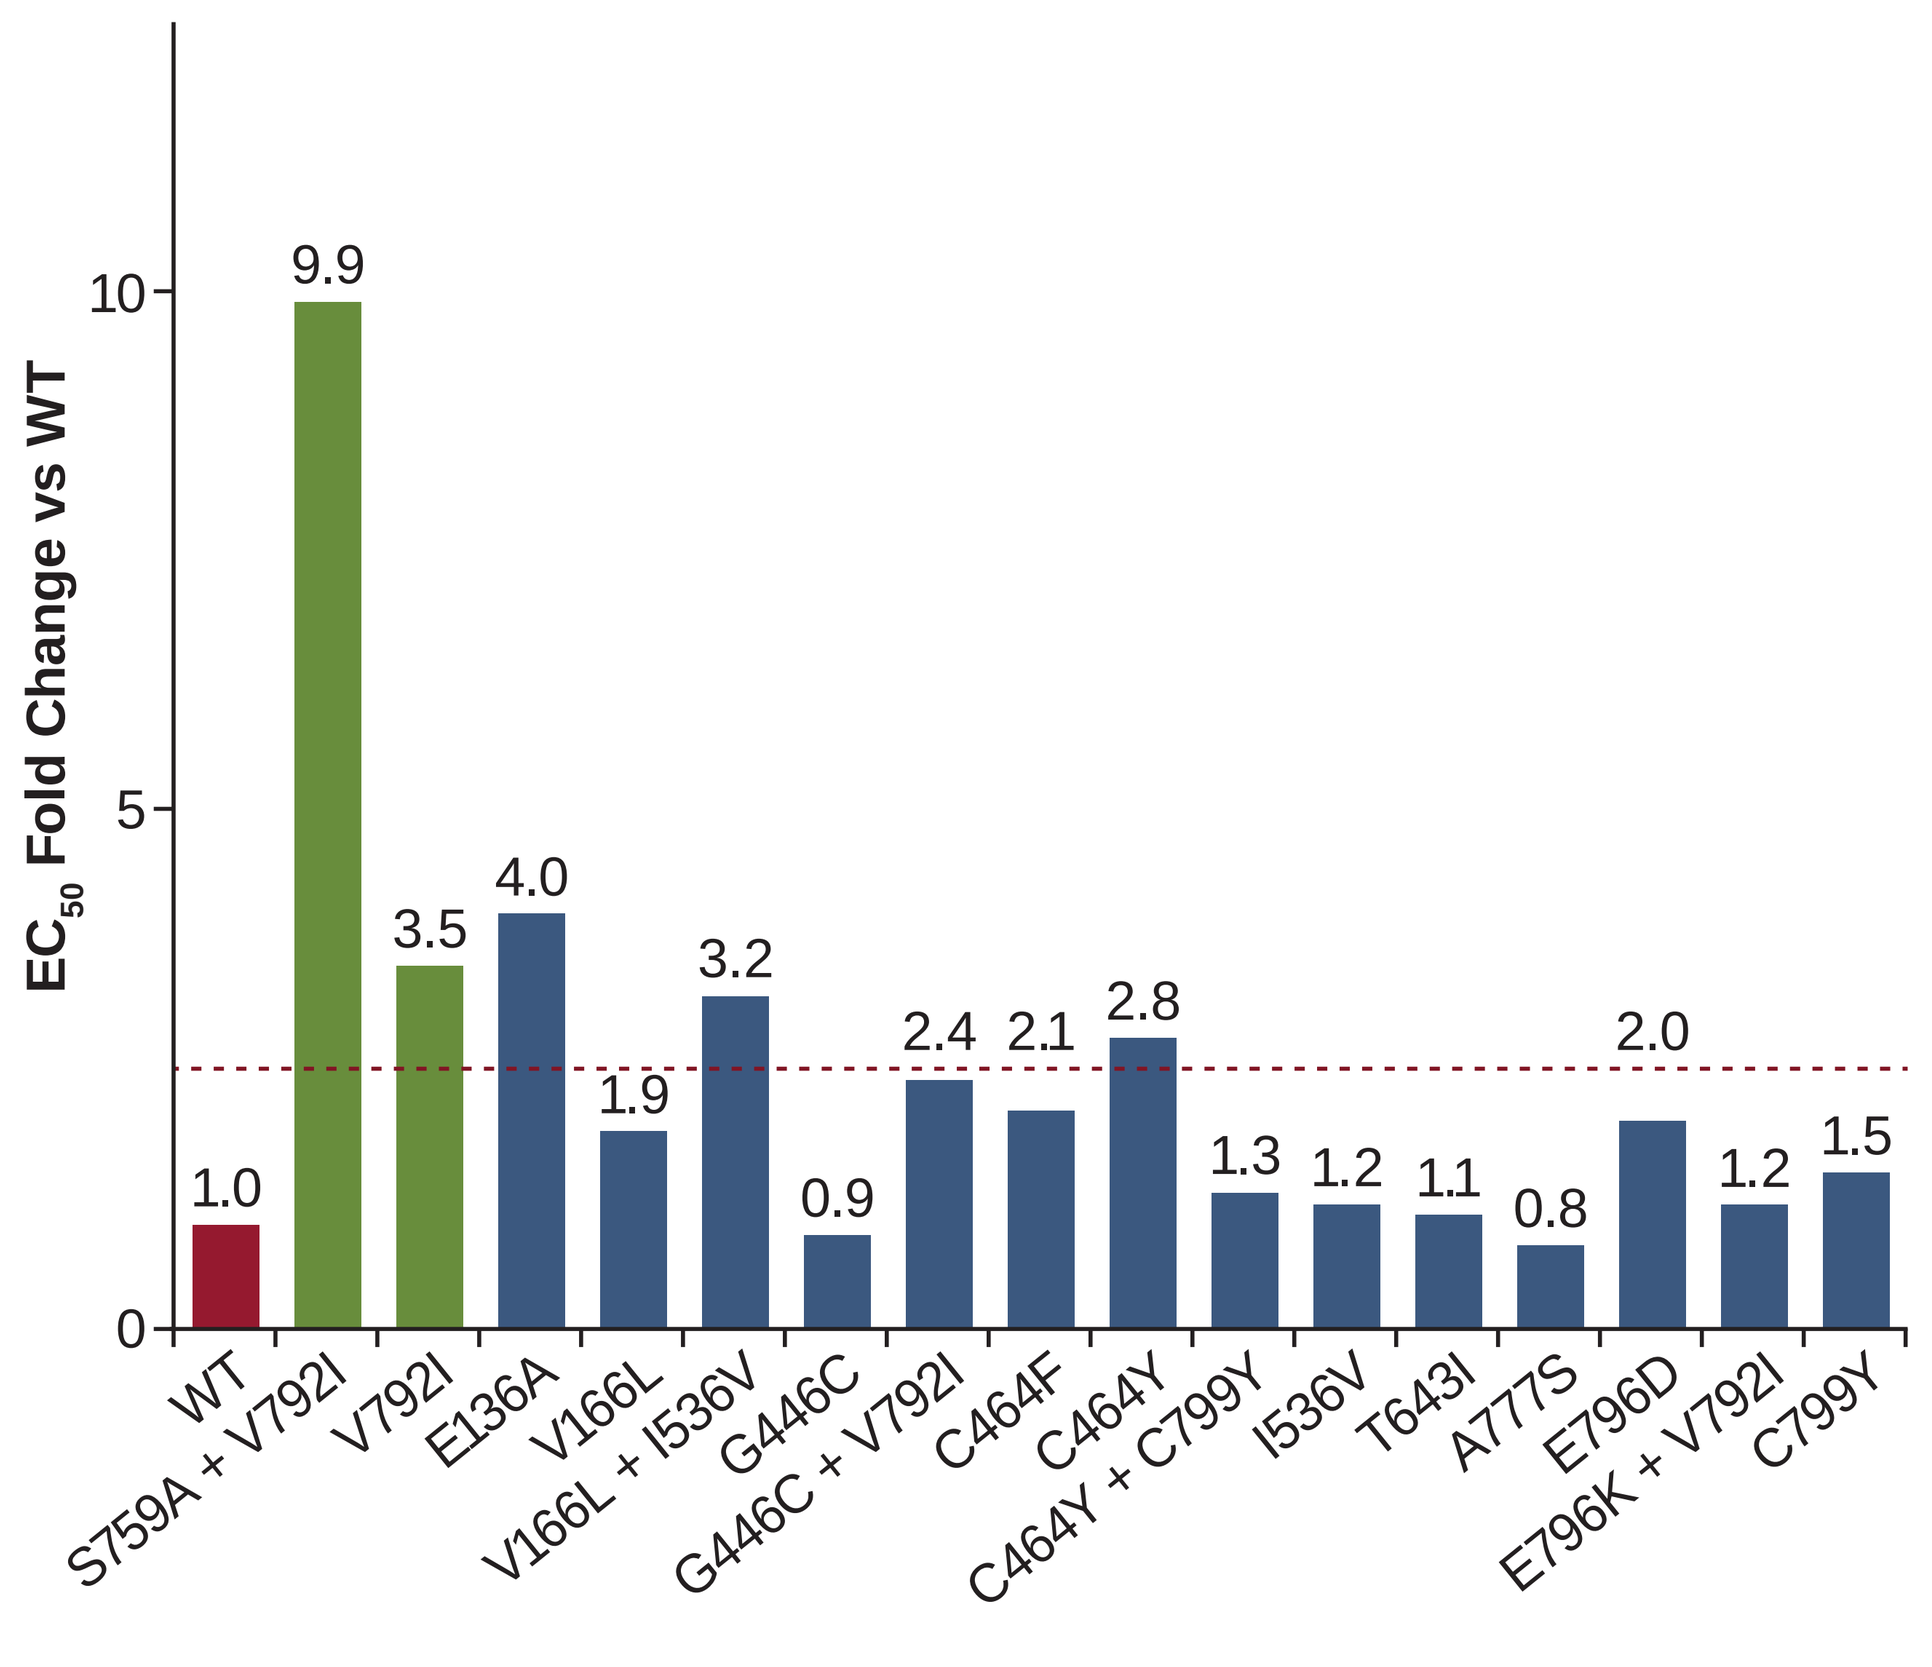

EC50 Fold Change vs WT
0
5
10
1.0
9.9
3.5
4.0
1.9
3.2
0.9
2.4
2.1
2.8
1.3
1.2
1.1
0.8
2.0
1.2
1.5
WT
S759A + V792I
V792I
E136A
V166L
V166L + I536V
G446C
G446C + V792I
C464F
C464Y
C464Y + C799Y
I536V
T643I
A777S
E796D
E796K + V792I
C799Y

EC_{50} fold change values <2.5 (dashed line) were within the variability of the assay, indicating no change in susceptibility to RDV compared with the WT reference strain.
EC_{50} fold change values relative to the SH01 reference strain from the replicon assay are shown.
Substitutions included as positive controls are shown in green.
No replication was observed for the E796K substitution.
EC_{50}, half-maximal effective concentration; RDV, remdesivir; WT, wildtype.

Figure 2. In Vitro RDV Activity Against SARS-CoV-2 Nsp12 Reference and Mutant Replicons

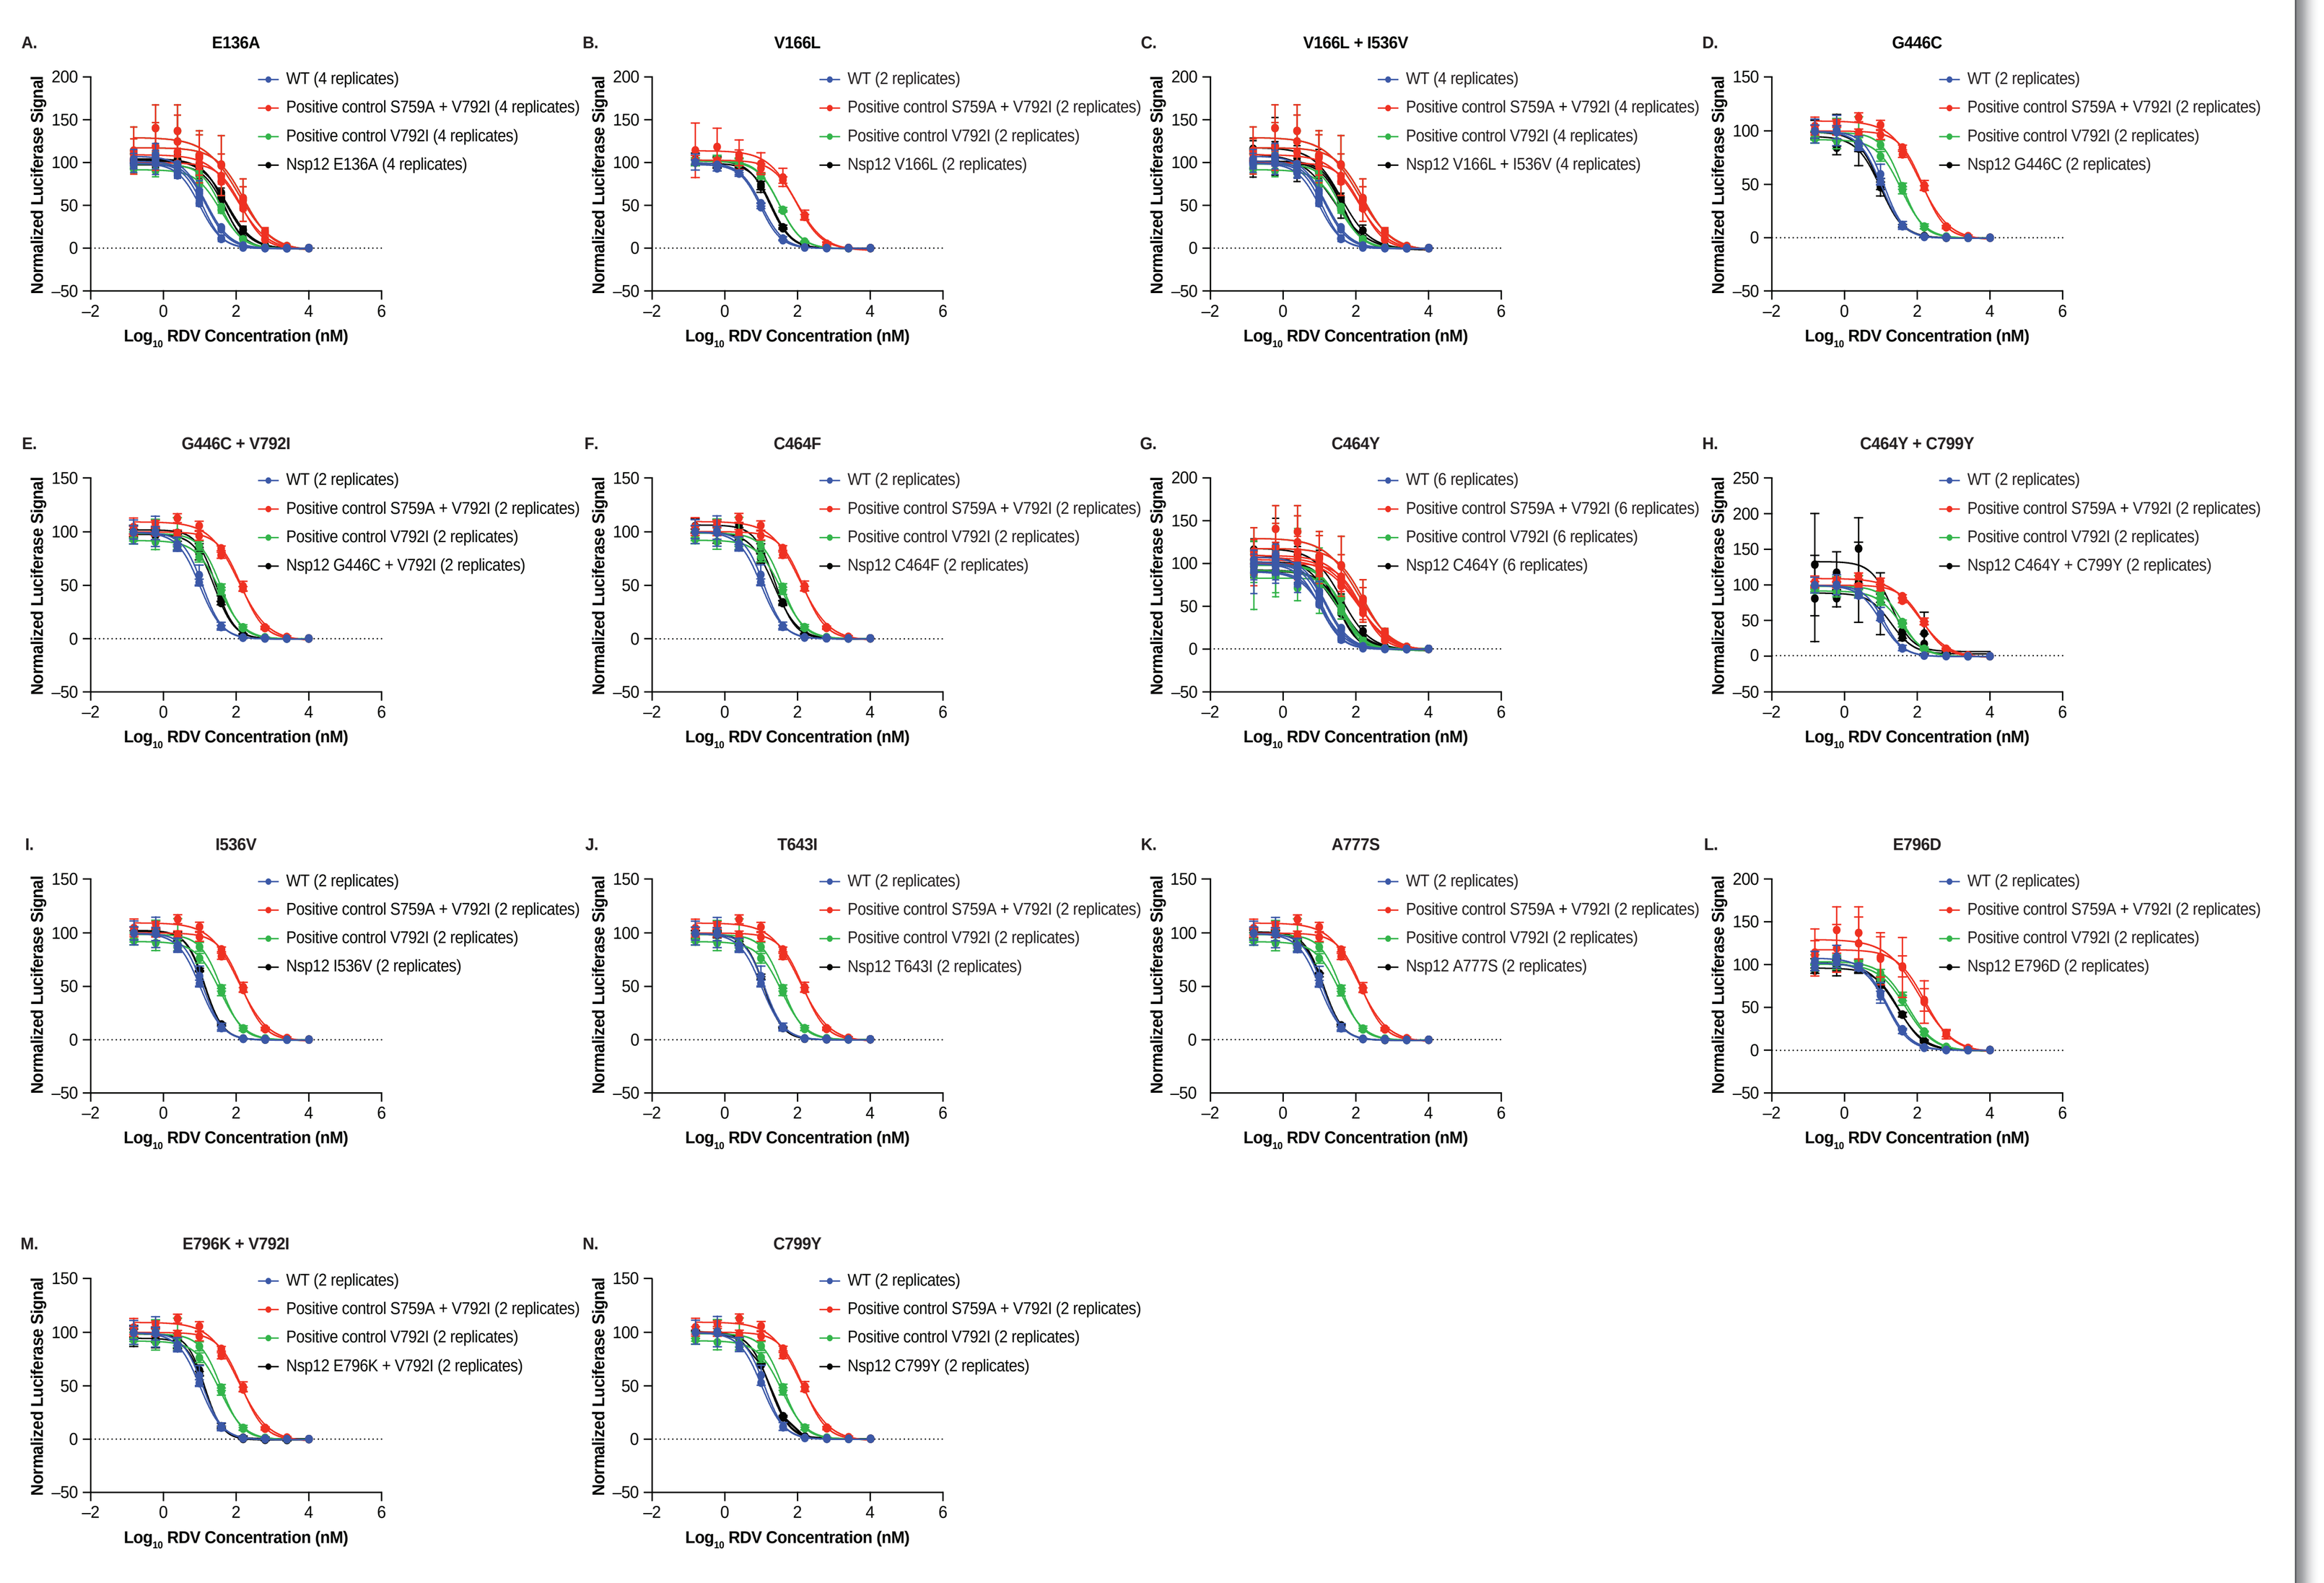

A. E136A
B. V166L
C. V166L + I536V
D. G446C
E. G446C + V792I
F. C464F
G. C464Y
H. C464Y + C799Y
I. I536V
J. T643I
K. A777S
L. E796D
M. E796K + V792I
N. C799Y
Normalized Luciferase Signal
Log10 RDV Concentration (nM)
WT (2 replicates)
Positive control S759A + V792I (2 replicates)
Positive control V792I (2 replicates)

The SARS-CoV-2 SH01 reference strain was included as a WT control.
Luciferase signal was normalized against the luciferase signal observed upon treatment with DMSO.
DMSO, dimethyl sulfoxide; RDV, remdesivir; WT, wildtype.

- In the publicly available GISAID database, the prevalence of the observed substitutions was <0.06% across 17 million sequences since the onset of the COVID-19 pandemic (**Table 3**)

Table 3. Prevalence of Substitutions in Publicly Available SARS-CoV-2 Sequences

Substitution	Substitution Prevalence, n (%)
E136A	64 (0.00037)
V166L	469 (0.00271)
G446C	346 (0.00200)
C464F	1262 (0.00729)
C464Y	305 (0.00176)
I536V	2897 (0.01673)
T643I	9520 (0.05498)
A777S	123 (0.00071)
E796D	263 (0.00152)
E796K	138 (0.00080)
C799Y	177 (0.00102)

Substitution prevalence was calculated using a total of 17,315,376 SARS-CoV-2 sequences deposited in the GISAID database as of September 4, 2026.
GISAID, Global Initiative on Sharing All Influenza Data.

Structural Analysis of Emergent Nsp12 Amino Acid Substitutions

- Emergent Nsp12 substitutions were mapped onto a cryo-EM structure of the SARS-CoV-2 polymerase complex (**Figure 3**)[20,21]
- The Nsp12 amino acid substitutions E136A, V166L, and C464Y were found to be either on the Nsp12 palm domain motif D or in direct contact with it
 - The palm domain has been previously shown to impact the relative favorability of incorporating the active triphosphate form of RDV versus adenosine triphosphate (ATP)[22]

Figure 3. Map of Emergent Nsp12 Amino Acid Substitutions on the Cryo-EM Structure of the SARS-CoV-2 Polymerase Complex

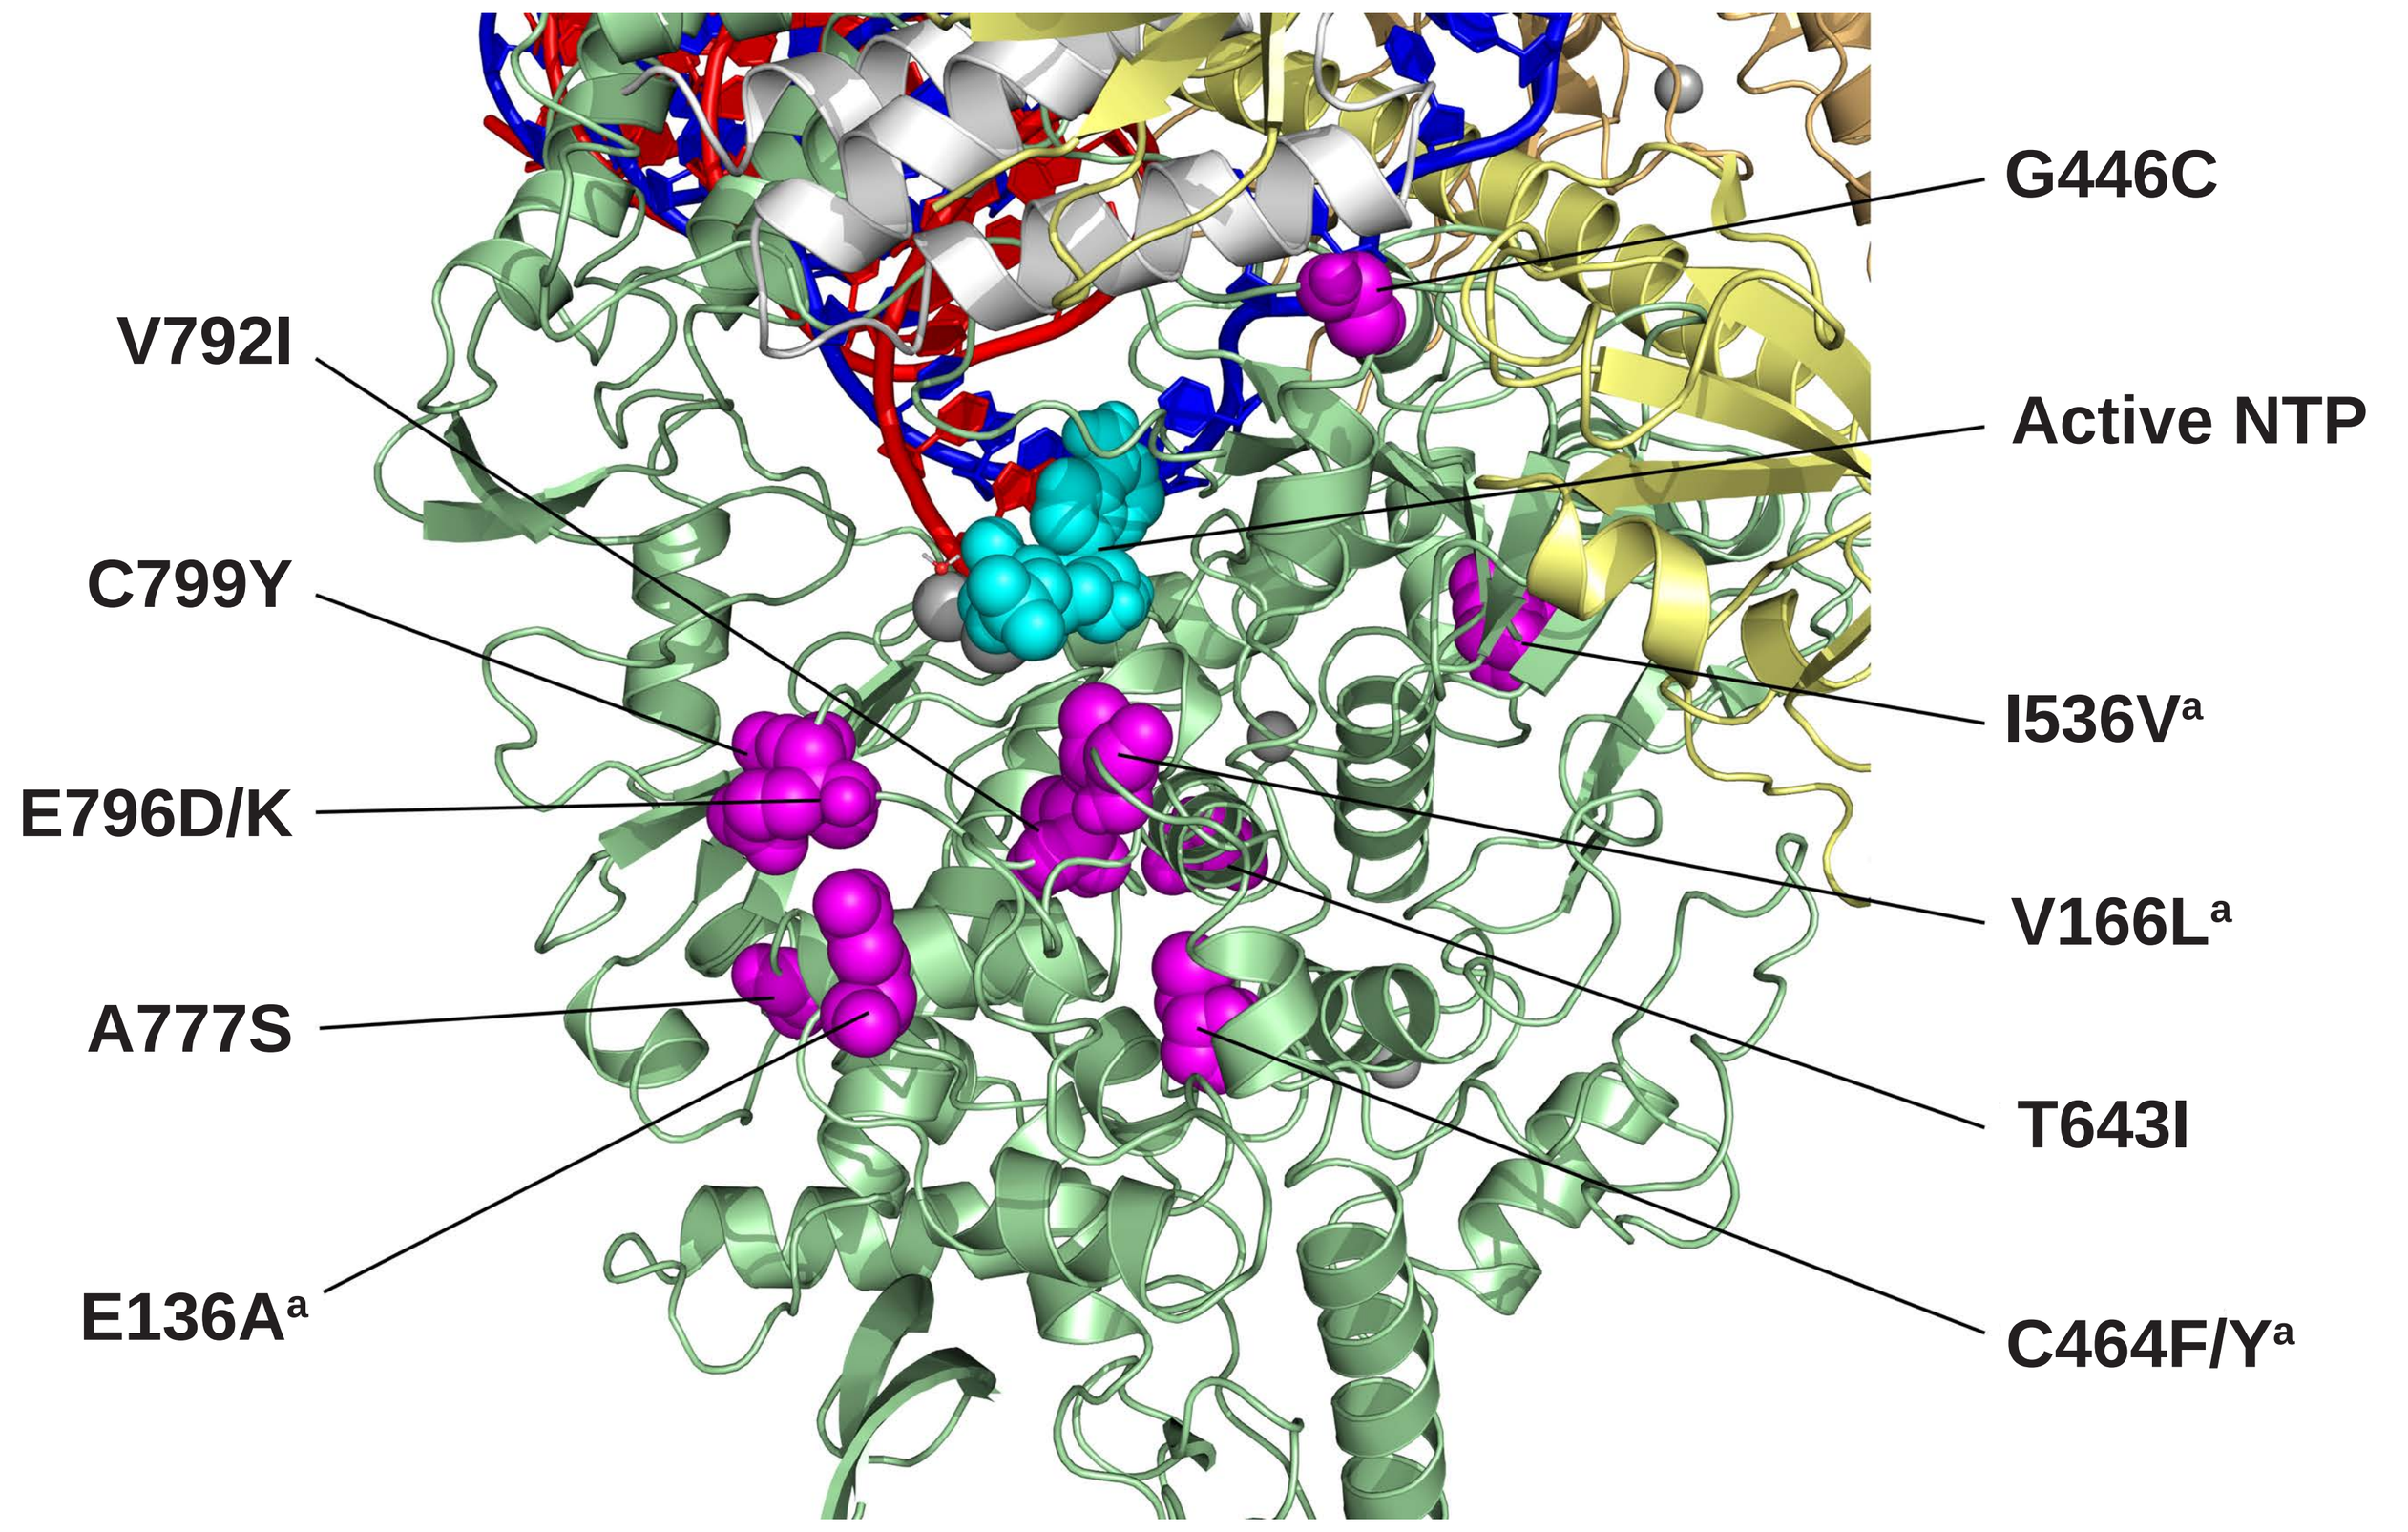

G446C
V792I
Active NTP
C799Y
I536Vª
E796D/K
V166Lª
A777S
T643I
E136Aª
C464F/Yª

Nsp12 protein is shown in green, with emergent amino acid substitutions shown in magenta. Nsp7 and Nsp8 are shown in white and yellow, respectively. Template and nascent RNA strands are shown in dark blue and red, respectively. Active NTP is shown in light blue.
[a]Nsp12 amino acid substitutions E136A, V166L + I536V, and C464Y demonstrated low-level reduced susceptibility to RDV based on phenotypic testing of site-directed mutants using a SARS-CoV-2 replicon system.
Cryo-EM, cryogenic electron microscopy; NTP, nucleoside triphosphate; RDV, remdesivir.

Clinical Characteristics of Immunocompromised Patients With Emergent Nsp12 Amino Acid Substitutions Conferring Reduced RDV Susceptibility

- Three patients had emergent Nsp12 amino acid substitutions conferring low-level reductions in RDV susceptibility[13,15] (**Figure 4**)
 - These patients had hematologic malignancies and prolonged SARS-CoV-2 infections of 38 to 90 days
 - These patients were treated with RDV for 6 to 12 consecutive days, and substitutions with reduced susceptibility were first observed on the second-to-last day of RDV treatment or within 11 days after the end of the first course of RDV treatment

Figure 4. Disease Progression and Emergence Timing of Nsp12 Amino Acid Substitutions With Reduced RDV Susceptibility

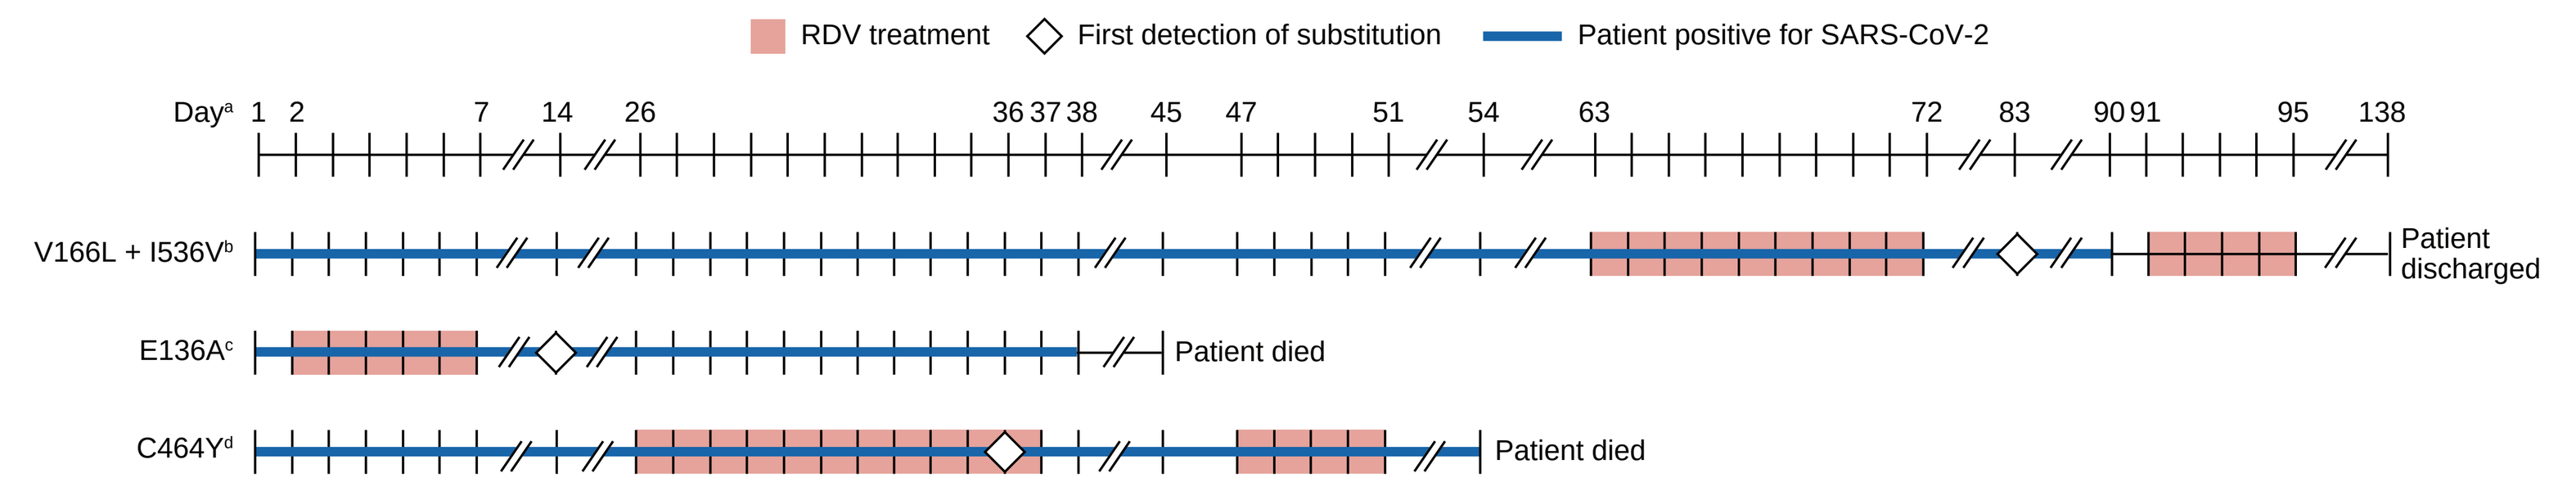

RDV treatment
First detection of substitution
Patient positive for SARS-CoV-2
Dayª 1 2 7 14 26 36 37 38 45 47 51 54 63 72 83 90 91 95 138
V166L + I536Vᵇ
Patient discharged
E136Aᶜ
Patient died
C464Yᵈ
Patient died

[a]Timeline for V166L + I536V shows days posthospitalization. Timelines for E136A and C464Y show days postdiagnosis.
[b]Patient had a history of follicular lymphoma, which was in remission. Patient received the immunosuppressive drug obinutuzumab 3 months prior to admission. In addition to the RDV treatments shown, patient received convalescent plasma on Days 95, 97, and 99.[15]
[c]Patient had a history of autoimmune hemolytic anemia. Patient was admitted to the hospital for treatment of IgM-κ monoclonal gammopathy. In the prior 6 months, patient had received the immunosuppressive drugs rituximab and ibrutinib as well as steroids. In addition to the RDV treatment shown, patient received dexamethasone (Days 2-7).[13]
[d]Patient had a history of high-grade myeloproliferative disease and had received an allogeneic stem cell transplant 78 days prior to SARS-CoV-2 diagnosis. In the prior 6 months, patient had received the immunosuppressive drugs fludarabine/melphalan/TBI 400 cGy, alemtuzumab, and tacrolimus. In addition to the RDV treatments shown, patient received sotrovimab (~Day 1), dexamethasone (Days 26-53), tocilizumab (Day 31), and high-flow oxygen supplementation through Day 45.[13]
cGy, centigray; IgM-κ, immunoglobulin M kappa; RDV, remdesivir; TBI, total body irradiation.

IDWeek; October 21–24, 2026; Washington, DC, USA