P-2059

ABI-1179-101

Safety and Tolerability of GS-1179, a Novel, Oral, Long-Acting HSV Helicase-Primase Inhibitor for Recurrent Genital Herpes: Results From a Phase 1b Study

Andrew Edwards[1], Rebecca Dunsmoor-Su[2], Martha Sue Fanning[3], Claire Thurlow[4], Wayne Hayter[5], Victoria Siriett[6], James Stanley[7], King Cheung[8], Tim Humphrey[9], Cory Sellwood[10], Joseph Sasadeusz[11,12], Grace Wang[13], Jieming Liu[13], Katie Zomorodi[13], Yiannis Koullias[14], Yoshihiko Murata[14], Shuguang Chen[14], Tina Chakrabarti[14], Kathryn M Kitrinos[13], Edward J Gane[15], Mark Bloch[16,17], Christine Johnston[18,19]

[1]Momentum Clinical Research Kapiti, Waikanae Beach, New Zealand; [2]Seattle Clinical Research Center, Seattle, WA, USA; [3]AMR Clinical Kansas City, Kansas City, MO, USA; [4]Pacific Clinical Research Network – Tasman, Nelson, New Zealand; [5]Momentum Clinical Research Palmerston North, Palmerston North, New Zealand; [6]Pacific Clinical Research Network – Hamilton, Hamilton, New Zealand; [7]Momentum Clinical Research Rotorua, Rotorua, New Zealand; [8]Momentum Clinical Research Sunshine, Melbourne, VIC, Australia; [9]Pacific Clinical Research Network – Wellington, Upper Hutt, New Zealand; [10]New Zealand Clinical Research, Christchurch, New Zealand; [11]University of Melbourne at the Doherty Institute for Infection and Immunity, Melbourne, VIC, Australia; [12]The Royal Melbourne Hospital at the Doherty Institute for Infection and Immunity, Melbourne, VIC, Australia; [13]Assembly Biosciences, Inc., South San Francisco, CA, USA; [14]Gilead Sciences, Inc., Foster City, CA, USA; [15]University of Auckland, Auckland, New Zealand; [16]Momentum Clinical Research Darlinghurst, Sydney, NSW, Australia; [17]Kirby Institute, University of New South Wales, Sydney, NSW, Australia; [18]University of Washington Medical Center, Seattle, WA, USA; [19]Fred Hutchinson Cancer Center, Seattle, WA, USA

Cory Sellwood
cory.sellwood@nzcr.co.nz
Level 3, 264 Antigua St
Christchurch 8011, New Zealand
Tel: +64 3 372 9477

Copies of this poster obtained through QR (Quick Response) code are for personal use only and may not be reproduced without written permission of the authors

Conclusions

- When administered at 50, 20, or 10 mg once-weekly, GS-1179 was generally safe and well tolerated in participants seropositive for herpes simplex virus type 2 with recurrent genital herpes
- Treatment-emergent adverse events and laboratory abnormalities were reported at similar rates among participants receiving once-weekly GS-1179 up to 50 mg compared with those receiving placebo
- Most treatment-emergent adverse events and all treatment-emergent laboratory abnormalities were Grade 1 or 2
 - One serious treatment-emergent adverse event of abdominal pain occurred in a participant receiving GS-1179 10 mg; this event was not related to the study drug
- No treatment-emergent adverse events leading to premature treatment discontinuation or death occurred
- These data support the continued development of GS-1179 as a long-acting suppressive therapy for recurrent genital herpes

Plain Language Summary

- Recurrent genital herpes is a common infection caused by herpes simplex virus type 2 that can lead to episodes of sores or blisters in the genital area
- While current antiviral medicines are safe, people often experience breakthrough episodes while taking them
 - Better medicines with proven safety are needed to suppress breakthrough episodes in people with recurrent genital herpes
- GS-1179 is a new medicine that is being developed for recurrent genital herpes
- In this study, once-weekly GS-1179 was generally safe and well tolerated at 3 different doses in people with herpes simplex virus type 2 and recurrent genital herpes

References: **1.** Harfouche M, et al. *Sex Transm Infect*. 2025;101:214-23. **2.** Gupta R, et al. *Lancet*. 2007;370:2127-37. **3.** VALTREX (valacyclovir) tablets, for oral use [package insert]. GSK; 2025. **4.** Corey L, et al. *N Engl J Med*. 2004;350:11-20. **5.** Bermek O, Williams RS. *Open Biol*. 2021;11:210011. **6.** Yao Q, et al. *Nat Microbiol*. 2025;10:3191-201.

Acknowledgments: We express our gratitude to all the participants, investigators, and site staff who participated in the study. This study was funded by Assembly Biosciences, Inc. Medical writing and editorial support were provided by Jane Abolafia, PhD, of Lumanity Communications Inc., and were funded by Gilead Sciences, Inc.

Correspondence: Cory Sellwood, cory.sellwood@nzcr.co.nz

Disclosures: **RD-S** provides consultative services for Bayer, Eli Lilly, and Novo Nordisk. **CS** received funding from Assembly Biosciences, Inc., and Gilead Sciences, Inc., to attend scientific meetings. **J Sasadeusz** received research funding from Aicuris, Moderna, Takeda, and Vedanta Biosciences; and served on advisory boards for Gilead Sciences, Inc., Merck, and Takeda. **GW**, **JL**, **KZ**, and **KMK** are stockholders and employees of Assembly Biosciences, Inc. **YK**, **YM**, **SC**, and **TC** are stockholders and employees of Gilead Sciences, Inc. **EJG** is a member of scientific advisory boards for Aligos, Assembly Biosciences, Inc., AusperBio, Epigenics, Gilead Sciences, Inc., GSK, IntegerBio, nChroma, OrsoBio, Precision BioSciences, Tune Therapeutics, Vir Biotechnology, and Virion Therapeutics. **MB** received funding from Assembly Biosciences, Inc., Gilead Sciences, Inc., and ViiV Healthcare to attend scientific meetings; and received funding from Gilead Sciences, Inc., GSK, MSD, and ViiV Healthcare for lectureships and medical advisory boards. **CJ** is a consultant for Aicuris, Assembly Biosciences, Inc., Gilead Sciences, Inc., GSK, Oximio, and Pfizer; and received research funding from Assembly Biosciences, Inc., GSK, Moderna, and Pfizer. **AE**, **MSF**, **CT**, **WH**, **VS**, **J Stanley**, **KC**, and **TH** have no disclosures to report.

Introduction

- Herpes simplex virus type 2 (HSV-2) affects an estimated 520 million people aged 15 to 49 years globally and is the most common cause of recurrent genital herpes (RGH)[1,2]
- Nucleoside analogs, which inhibit the viral polymerase, can be administered episodically during recurrences or daily as suppressive therapy for RGH[3]
 - As nucleoside analogs are only partially effective at preventing breakthrough recurrences and transmission, there remains an unmet need for novel, potent HSV-2 therapies for individuals with RGH[3,4]
- Herpes simplex virus (HSV) helicase-primase inhibitors target the viral helicase-primase complex, which is essential for viral replication[5,6]
- GS-1179 is an investigational, long-acting HSV helicase-primase inhibitor in development as a suppressive therapy for RGH
- A Phase 1b study evaluated the safety and tolerability of once-weekly (QW) GS-1179 50, 20, or 10 mg compared with placebo

Objective

- To investigate the safety and tolerability of GS-1179 in participants who are seropositive for HSV-2 with RGH

Methods

- This randomized, blinded, placebo-controlled study (ClinicalTrials.gov Identifier: NCT06698575) enrolled participants who were seropositive for HSV-2 with RGH (**Figure 1**)
 - Eligible participants had 4 to 9 clinical RGH episodes within the past 12 months if not currently on suppressive therapy, or within the 12 months prior to initiation of suppressive therapy
 - Exclusion criteria included current coinfection with HIV, hepatitis B virus, hepatitis C virus, acute hepatitis A virus, or acute hepatitis E virus and any clinically significant medical, behavioral, or mental condition, or pharmacologic or surgical treatment that may interfere with safety assessments or the absorption, distribution, and/or elimination of the study drug or make the individual unsuitable for study participation
- Participants were randomized 20:5 to receive QW GS-1179 (50 mg [cohort B1], 20 mg [cohort B2], or 10 mg [cohort B3]) or placebo for 29 days
 - Dose levels were determined based on emergent safety, tolerability, and available pharmacokinetic data from completed cohorts in a concurrent single-ascending dose study of GS-1179
- Participants were monitored through Day 57 for safety evaluations, including treatment-emergent adverse event (TEAE) reporting, laboratory testing, and physical examinations

Figure 1. Study Design

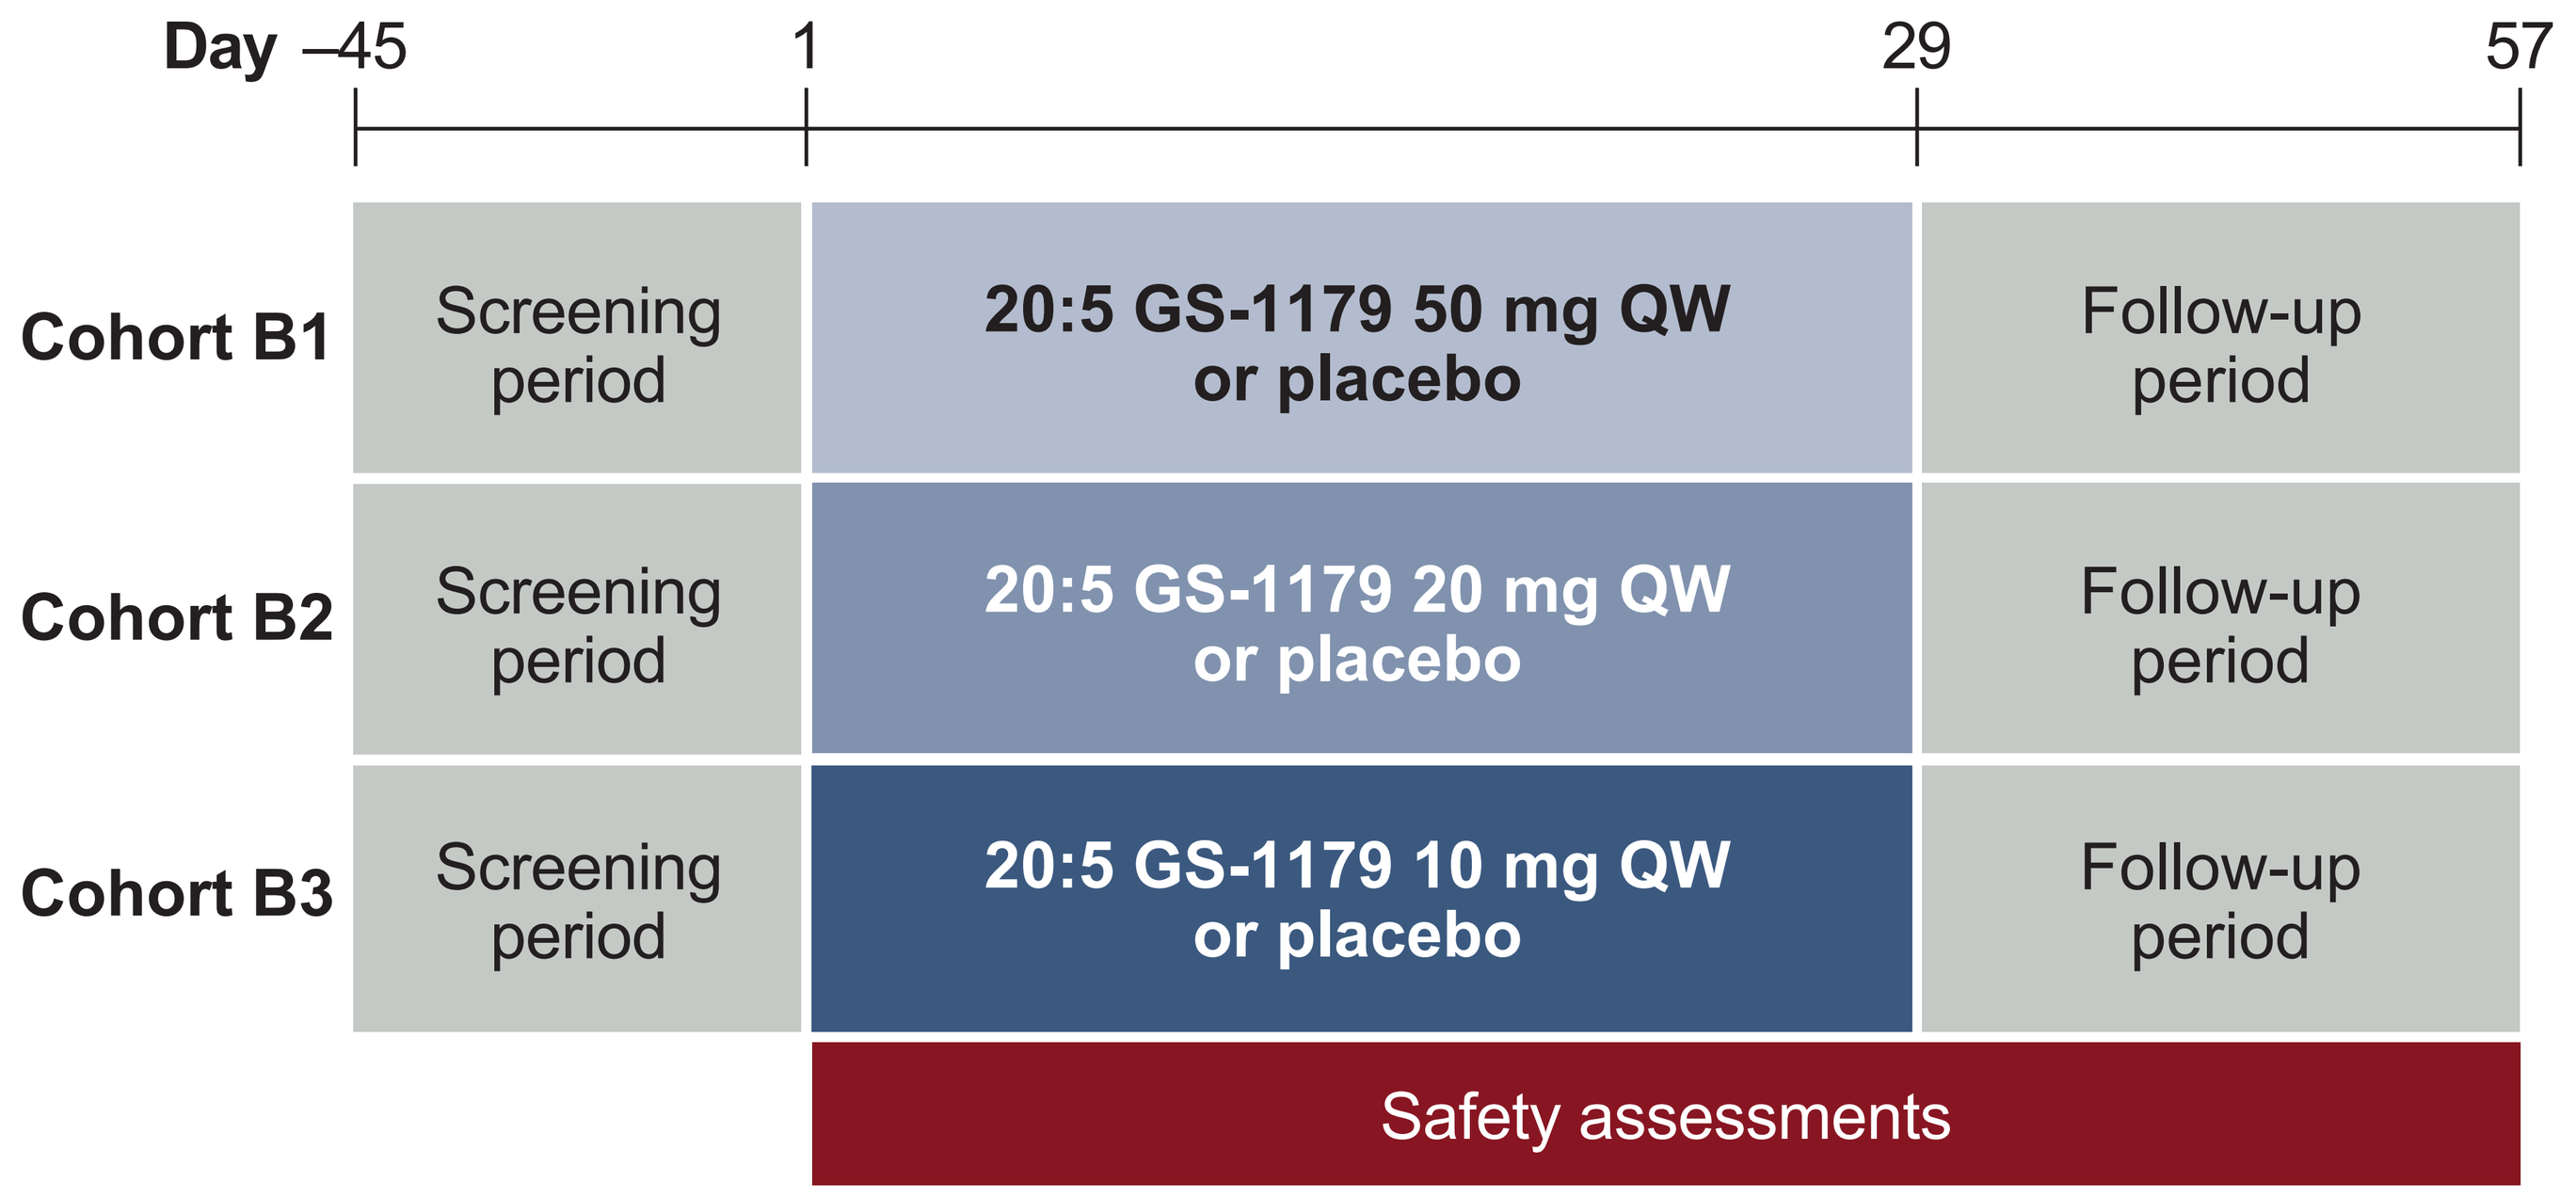

Day –45
1
29
57
Cohort B1
Screening period
20:5 GS-1179 50 mg QW or placebo
Follow-up period
Cohort B2
Screening period
20:5 GS-1179 20 mg QW or placebo
Follow-up period
Cohort B3
Screening period
20:5 GS-1179 10 mg QW or placebo
Follow-up period
Safety assessments

QW, once weekly.

Results

Participants

- Overall, 73 participants were enrolled in 3 dosing cohorts with placebo controls
 - One participant was lost to follow-up, and 1 participant discontinued the study for reasons not related to adverse events or study treatments
 - One participant was assigned to receive placebo within cohort B2 but did not receive treatment
 - All participants who received treatment (n = 72) were evaluated for safety outcomes (**Table 1**)

TEAEs

- TEAEs were reported in 18/20 (90%), 15/20 (75%), and 12/18 (67%) participants receiving GS-1179 50, 20, and 10 mg, respectively, and in 12/14 (86%) participants receiving placebo (**Table 2**)
- Most TEAEs were Grade 1 or 2
 - One participant receiving GS-1179 10 mg experienced a Grade 3 TEAE of headache, and 1 participant receiving GS-1179 10 mg experienced a Grade 3 serious TEAE of abdominal pain that was considered not related to the study drug by investigators
 - One participant receiving placebo experienced a Grade 3 TEAE of migraine
 - No Grade 4 TEAEs were reported
- The most common TEAEs were headache (in 45%, 10%, and 39% of participants receiving GS-1179 50, 20, and 10 mg, respectively, and in 7% of participants receiving placebo) and upper respiratory tract infection (in 10%, 5%, and 11% of participants receiving GS-1179 50, 20, and 10 mg, respectively, and in 29% of participants receiving placebo)
- Treatment-related TEAEs occurred in 8/20 (40%), 7/20 (35%), and 5/18 (28%) participants receiving GS-1179 50, 20, and 10 mg, respectively, and in 4/14 (29%) participants receiving placebo
 - The most common treatment-related TEAE was headache (in 30%, 10%, and 22% of participants receiving GS-1179 50, 20, and 10 mg, respectively, and in 0 participants receiving placebo)
 - Most treatment-related headache TEAEs resolved by the end of study follow-up
- No TEAEs leading to premature discontinuation of treatment or death occurred

Treatment-Emergent Laboratory Abnormalities

- Treatment-emergent laboratory abnormalities were reported in 7/20 (35%), 7/20 (35%), and 9/18 (50%) participants receiving GS-1179 50, 20, and 10 mg, respectively, and in 5/14 (36%) participants receiving placebo
 - All treatment-emergent laboratory abnormalities were Grade 1 or 2

Table 1. Participant Demographic and Baseline Characteristics

Characteristic	Placebo (n = 14)[a]	Cohort B1: GS-1179 50 mg QW (n = 20)	Cohort B2: GS-1179 20 mg QW (n = 20)	Cohort B3: GS-1179 10 mg QW (n = 18)
Age, years, mean (SD)	41.6 (10.9)	38.4 (12.4)	40.5 (9.9)	42.0 (10.9)
Sex at birth, male, n (%)	5 (36)	5 (25)	12 (60)	4 (22)
Race, White, n (%)	11 (79)	19 (95)	18 (90)	14 (78)
BMI, kg/m^2, mean (SD)	25.9 (3.8)	25.7 (4.0)	25.3 (3.0)	27.3 (3.8)
Time since RGH diagnosis, years, mean (SD)	13.1 (9.5)	10.2 (9.6)	11.3 (9.9)	12.9 (10.0)
Number of recurrences,[b] mean (SD)	6.0 (1.4)	6.1 (1.3)	5.3 (0.9)	5.8 (1.4)
On suppressive therapy at baseline,[c] n (%)	10 (71)	17 (85)	16 (80)	12 (67)
≥1 medical history condition at baseline, n (%)	13 (93)	18 (90)	20 (100)	18 (100)

[a]One participant assigned to receive placebo in cohort B2 did not receive treatment and was not evaluated.
[b]Number of recurrences was defined as the number of lesions in the past 12 months or prior to suppressive therapy.
[c]Participants discontinued suppressive therapies for RGH beginning 7 days prior to Day 1 dosing through Day 35.
BMI, body mass index; QW, once weekly; RGH, recurrent genital herpes.

Table 2. Summary of TEAEs

Participants With TEAE,[a] n (%)	Placebo (n = 14)[b]	Cohort B1: GS-1179 50 mg QW (n = 20)	Cohort B2: GS-1179 20 mg QW (n = 20)	Cohort B3: GS-1179 10 mg QW (n = 18)
Any TEAE	12 (86)	18 (90)	15 (75)	12 (67)
Grade ≥3 TEAE	1 (7)	0	0	2 (11)
Abdominal pain	0	0	0	1 (6)
Headache	0	0	0	1 (6)
Migraine	1 (7)	0	0	0
TEAE by preferred term occurring in ≥5% of participants overall				
Headache	1 (7)	9 (45)	2 (10)	7 (39)
Upper respiratory tract infection	4 (29)	2 (10)	1 (5)	2 (11)
Fatigue	0	2 (10)	3 (15)	0
Rash	1 (7)	3 (15)	0	0
Abdominal pain	1 (7)	0	2 (10)	1 (6)
Any treatment-related TEAE	4 (29)	8 (40)	7 (35)	5 (28)
Treatment-related TEAE by preferred term occurring in ≥5% of participants overall				
Headache	0	6 (30)	2 (10)	4 (22)
Any serious TEAE	0	0	0	1 (6)
TEAE leading to premature discontinuation of treatment	0	0	0	0
Death	0	0	0	0

[a]TEAEs were coded according to MedDRA Version 27.1.
[b]One participant assigned to receive placebo in cohort B2 did not receive treatment and was not evaluated.
MedDRA, *Medical Dictionary for Regulatory Activities*; QW, once weekly; TEAE, treatment-emergent adverse event.

IDWeek; October 21–24, 2026; Washington, DC, USA