P-2052

ABI-1179-101

Efficacy and Virologic Analyses of GS-1179, a Novel, Oral, Long-Acting HSV Helicase-Primase Inhibitor for Recurrent Genital Herpes: Results From a Phase 1b Study

Andrew Edwards[1], Rebecca Dunsmoor-Su[2], Martha Sue Fanning[3], Claire Thurlow[4], Wayne Hayter[5], Victoria Siriett[6], James Stanley[7], King Cheung[8], Tim Humphrey[9], Cory Sellwood[10], Joseph Sasadeusz[11,12], Grace Wang[13], Jieming Liu[13], Katie Zomorodi[13], Yiannis Koullias[14], Yoshihiko Murata[14], Shuguang Chen[14], Kathryn M Kitrinos[13], Edward J Gane[15], Mark Bloch[16,17], Christine Johnston[18,19]

[1]Momentum Clinical Research Kapiti, Waikanae Beach, New Zealand; [2]Seattle Clinical Research Center, Seattle, WA, USA; [3]AMR Clinical Kansas City, Kansas City, MO, USA; [4]Pacific Clinical Research Network – Tasman, Nelson, New Zealand; [5]Momentum Clinical Research Palmerston North, Palmerston North, New Zealand; [6]Pacific Clinical Research Network – Hamilton, Hamilton, New Zealand; [7]Momentum Clinical Research Rotorua, Rotorua, New Zealand; [8]Momentum Clinical Research Sunshine, Melbourne, VIC, Australia; [9]Pacific Clinical Research Network – Wellington, Upper Hutt, New Zealand; [10]New Zealand Clinical Research, Christchurch, New Zealand; [11]University of Melbourne at the Doherty Institute for Infection and Immunity, Melbourne, VIC, Australia; [12]The Royal Melbourne Hospital at the Doherty Institute for Infection and Immunity, Melbourne, VIC, Australia; [13]Assembly Biosciences, Inc., South San Francisco, CA, USA; [14]Gilead Sciences, Inc., Foster City, CA, USA; [15]University of Auckland, Auckland, New Zealand; [16]Momentum Clinical Research Darlinghurst, Sydney, NSW, Australia; [17]Kirby Institute, University of New South Wales, Sydney, NSW, Australia; [18]University of Washington Medical Center, Seattle, WA, USA; [19]Fred Hutchinson Cancer Center, Seattle, WA, USA

Andrew Edwards
andrew.edwards@momentumclinicalresearch.co.nz
43 Te Moana Rd
Waikanae Beach, Waikanae 5036, New Zealand
Tel: +64 04 908 1001

Copies of this poster obtained through QR (Quick Response) code are for personal use only and may not be reproduced without written permission of the authors

Conclusions

- Once-weekly administration of GS-1179 50, 20, or 10 mg resulted in nominally statistically significant reductions in herpes simplex virus type 2 shedding rate, herpes simplex virus type 2 high viral load shedding rate, genital lesion rate, and virologically confirmed herpes simplex virus type 2 genital lesion rate compared with placebo
- GS-1179 50, 20, or 10 mg also led to a numerically lower duration of viral shedding compared with placebo
- Dose-dependent reductions in herpes simplex virus type 2 shedding rate, herpes simplex virus type 2 high viral load shedding rate, and duration of viral shedding were observed with increasing doses of GS-1179
- These data support the continued development of the helicase-primase inhibitor GS-1179 for suppression of recurrent genital herpes in people with herpes simplex virus type 2

Plain Language Summary

- Herpes simplex virus type 2 is a virus that causes recurrent genital herpes, a common infection that can cause sores or blisters in the genital area
- Even with daily antiviral medicine, people often experience breakthrough episodes
 - Better medicines are needed to suppress episodes in people with recurrent genital herpes
- GS-1179 is a new medicine that is being developed for recurrent genital herpes
- In this study, people who received once-weekly GS-1179 had lower viral shedding rates and less frequent lesions compared with people who received placebo
- These results suggest that GS-1179 has potential as a new drug to suppress recurrent genital herpes

References: **1.** Gupta R, et al. *Lancet*. 2007;370:2127-37. **2.** Harfouche M, et al. *Sex Transm Infect*. 2025;101:214-23. **3.** Workowski KA, et al. *MMWR Recomm Rep*. 2021;70:1-187. **4.** VALTREX (valacyclovir) tablets, for oral use [package insert]. GSK; 2025. **5.** Corey L, et al. *N Engl J Med*. 2004;350:11-20. **6.** Bermek O, Williams RS. *Open Biol*. 2021;11:210011. **7.** Yao Q, et al. *Nat Microbiol*. 2025;10:3191-201. **8.** Wald A, et al. *JAMA*. 2016;316:2495-503. **9.** Schiffer JT, et al. *J R Soc Interface*. 2014;11:20140160.

Acknowledgments: We express our gratitude to all the participants, investigators, and site staff who participated in the study. This study was funded by Assembly Biosciences, Inc. Medical writing and editorial support were provided by Jane Abolafia, PhD, of Lumanity Communications Inc., and were funded by Gilead Sciences, Inc.

Correspondence: Andrew Edwards, andrew.edwards@momentumclinicalresearch.co.nz

Disclosures: **RD-S** provides consultative services for Bayer, Eli Lilly, and Novo Nordisk. **CS** received funding from Assembly Biosciences, Inc., and Gilead Sciences, Inc., to attend scientific meetings. **J Sasadeusz** received research funding from Aicuris, Moderna, Takeda, and Vedanta Biosciences; and served on advisory boards for Gilead Sciences, Inc., Merck, and Takeda. **GW**, **JL**, **KZ**, and **KMK** are stockholders and employees of Assembly Biosciences, Inc. **YK**, **YM**, and **SC** are stockholders and employees of Gilead Sciences, Inc. **EJG** is a member of scientific advisory boards for Aligos, Assembly Biosciences, Inc., AusperBio, Epigenics, Gilead Sciences, Inc., GSK, IntegerBio, nChroma, OrsoBio, Precision BioSciences, Tune Therapeutics, Vir Biotechnology, and Virion Therapeutics. **MB** received funding from Assembly Biosciences, Inc., Gilead Sciences, Inc., and ViiV Healthcare to attend scientific meetings; and received funding from Gilead Sciences, Inc., GSK, MSD, and ViiV Healthcare for lectureships and medical advisory boards. **CJ** is a consultant for Aicuris, Assembly Biosciences, Inc., Gilead Sciences, Inc., GSK, Oximio, and Pfizer; and received research funding from Assembly Biosciences, Inc., GSK, Moderna, and Pfizer. **AE**, **MSF**, **CT**, **WH**, **VS**, **J Stanley**, **KC**, and **TH** have no disclosures to report.

Introduction

- Herpes simplex virus type 2 (HSV-2) is the most common cause of recurrent genital herpes (RGH) and affects an estimated 520 million people aged 15 to 49 years globally[1,2]
- Nucleoside analogs are the current standard of care for HSV-2 infection and function by inhibiting the viral polymerase[3,4]
 - Nucleoside analogs can be administered episodically during recurrences or daily as suppressive therapy but are only partially effective at preventing breakthrough recurrences and transmission[4,5]
 - There remains an unmet need for more effective and potent therapies for HSV-2 suppression in individuals with RGH
- Herpes simplex virus (HSV) helicase-primase inhibitors target the viral helicase-primase complex, which is essential for viral replication[6,7]
 - Daily helicase-primase inhibitors have demonstrated clinical efficacy over nucleoside analogs in people with RGH[8]
- GS-1179 is an investigational, long-acting HSV helicase-primase inhibitor in development as a suppressive therapy for RGH
- A Phase 1b study evaluated the virologic and clinical efficacy of once-weekly (QW) GS-1179 50, 20, or 10 mg compared with placebo

Objective

- To investigate the antiviral efficacy of GS-1179 in participants who are seropositive for HSV-2 with RGH

Methods

- This randomized, blinded, placebo-controlled study (ClinicalTrials.gov Identifier: NCT06698575) enrolled participants who were seropositive for HSV-2 with RGH (**Figure 1**)
 - Eligible participants had 4 to 9 clinical RGH episodes within the past 12 months if not currently on suppressive therapy, or within the 12 months prior to initiation of suppressive therapy
- Participants were randomized 20:5 to receive QW GS-1179 (50 mg [cohort B1], 20 mg [cohort B2], or 10 mg [cohort B3]) or placebo for 29 days, with follow-up through Day 57
- To evaluate HSV-2 shedding, anogenital swabs were self-collected twice daily (BID) from Day 8 to Day 35
 - HSV DNA was quantified using a validated quantitative real-time polymerase chain reaction assay
 - High viral load shedding was defined as HSV DNA >4 $\log_{10}$ copies/mL, as HSV-2 transmission risk below this threshold is predicted to be low based on modeling of serodiscordant couples[9]
- Participants recorded RGH symptoms BID from Day 8 to Day 35
- HSV-2 shedding and genital lesion rates were compared between GS-1179 and placebo cohorts using Poisson regression models
 - *P* <0.05 was considered the threshold for nominal statistical significance

Figure 1. Study Design

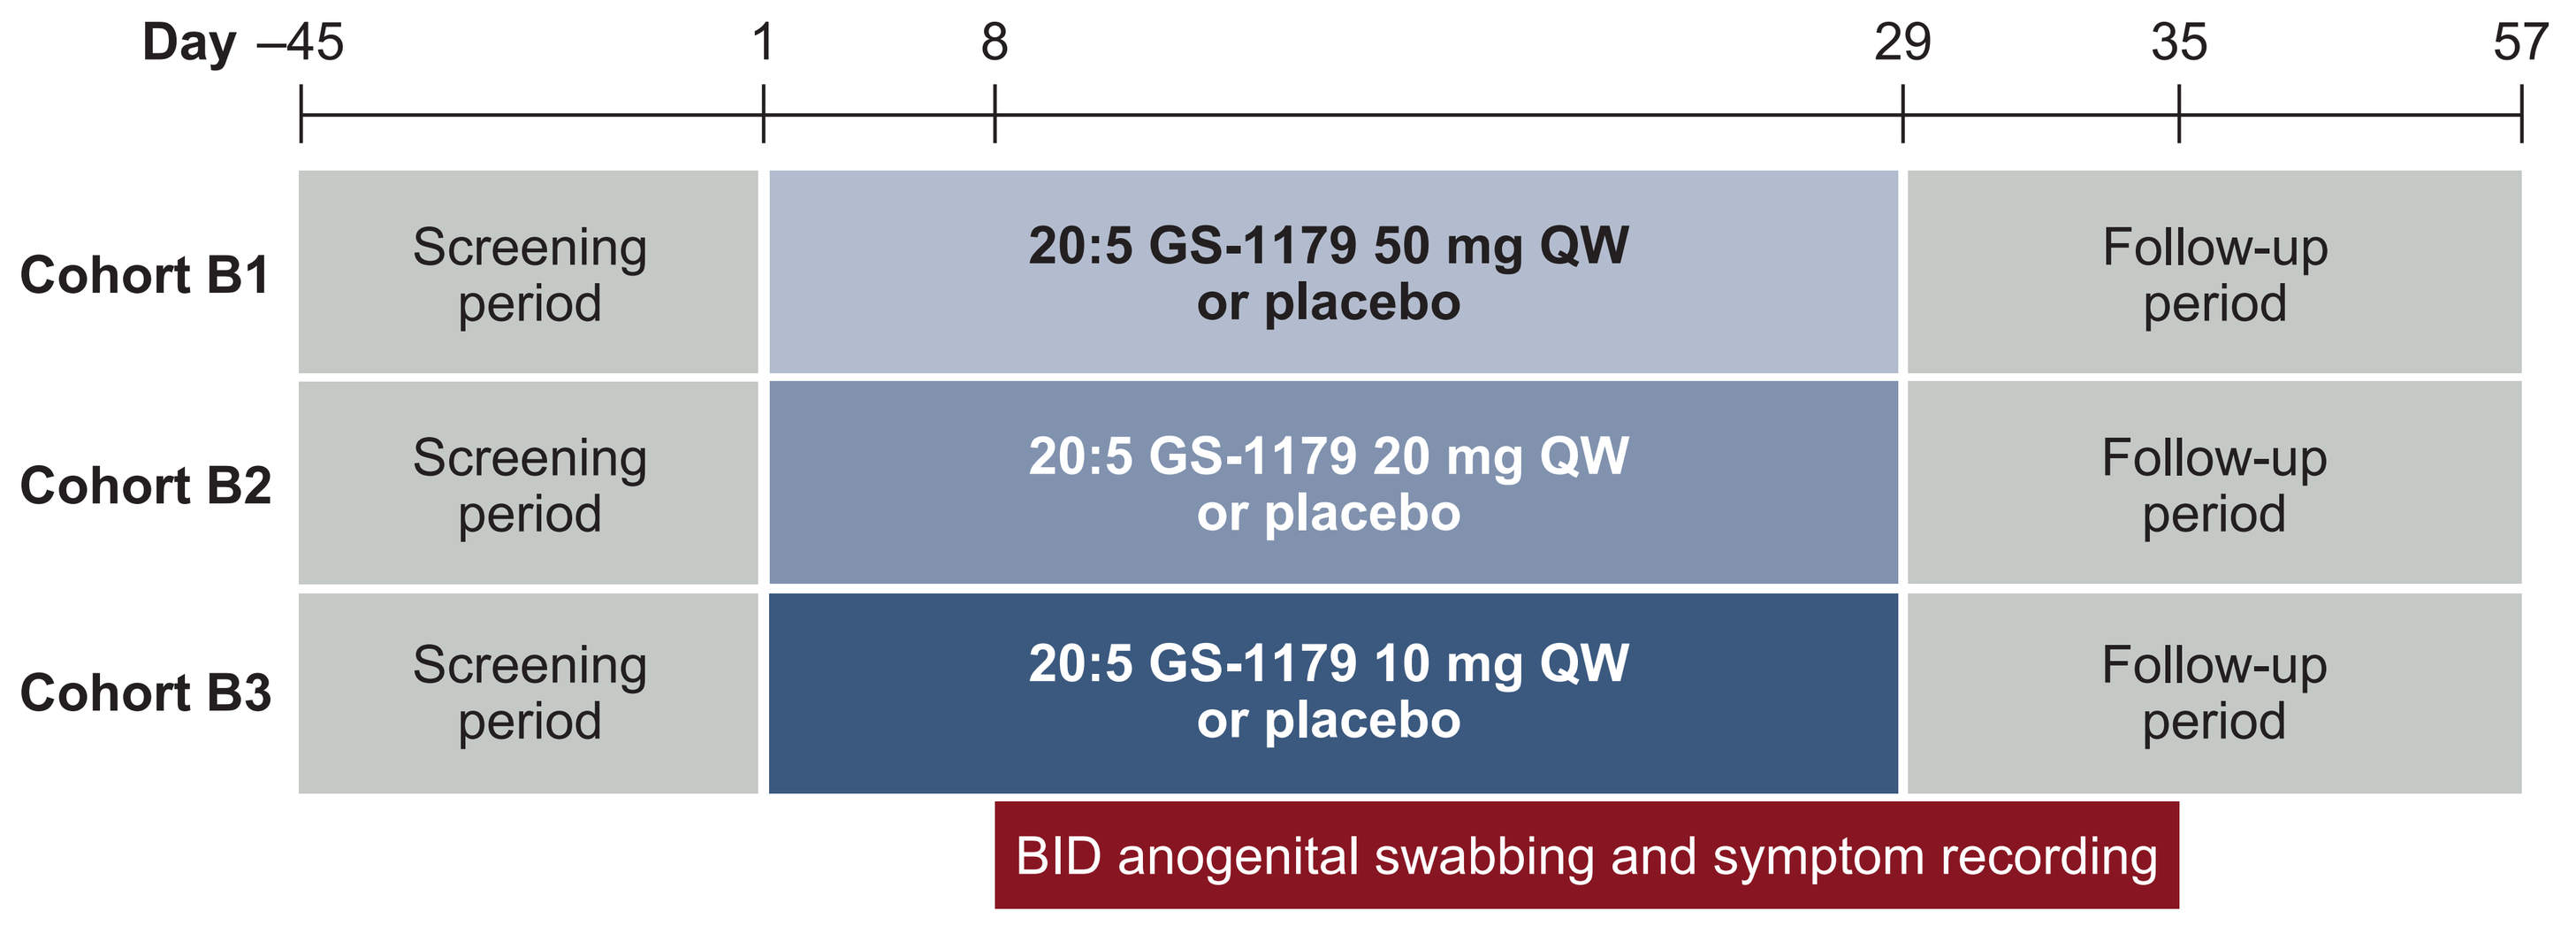

Day –45
1
8
29
35
57
Cohort B1
Screening period
20:5 GS-1179 50 mg QW or placebo
Follow-up period
Cohort B2
Screening period
20:5 GS-1179 20 mg QW or placebo
Follow-up period
Cohort B3
Screening period
20:5 GS-1179 10 mg QW or placebo
Follow-up period
BID anogenital swabbing and symptom recording

BID, twice daily; QW, once weekly.

Results

Participants

- Overall, 73 participants were enrolled in 3 dosing cohorts with placebo controls
 - One participant was lost to follow-up, and 1 participant discontinued the study for reasons not related to adverse events or study treatments
 - One participant was assigned to receive placebo within cohort B2 but did not receive treatment
 - All participants who received treatment and had ≥1 evaluable postdose efficacy assessment (n = 72) were evaluated for efficacy outcomes (**Table 1**)

Table 1. Participant Demographic and Baseline Characteristics

Characteristic	Placebo (n = 14)[a]	Cohort B1: GS-1179 50 mg QW (n = 20)	Cohort B2: GS-1179 20 mg QW (n = 20)	Cohort B3: GS-1179 10 mg QW (n = 18)
Age, years, mean (SD)	41.6 (10.9)	38.4 (12.4)	40.5 (9.9)	42.0 (10.9)
Sex at birth, male, n (%)	5 (36)	5 (25)	12 (60)	4 (22)
Race, White, n (%)	11 (79)	19 (95)	18 (90)	14 (78)
BMI, kg/m², mean (SD)	25.9 (3.8)	25.7 (4.0)	25.3 (3.0)	27.3 (3.8)
Time since RGH diagnosis, years, mean (SD)	13.1 (9.5)	10.2 (9.6)	11.3 (9.9)	12.9 (10.0)
Number of recurrences,[b] mean (SD)	6.0 (1.4)	6.1 (1.3)	5.3 (0.9)	5.8 (1.4)
On suppressive therapy at baseline,[c] n (%)	10 (71)	17 (85)	16 (80)	12 (67)

[a]One participant assigned to receive placebo in cohort B2 did not receive treatment and was not evaluated.
[b]Number of recurrences was defined as the number of lesions in the past 12 months or prior to suppressive therapy.
[c]Participants discontinued suppressive therapies for RGH beginning 7 days prior to Day 1 dosing through Day 35.
BMI, body mass index; QW, once weekly; RGH, recurrent genital herpes.

Antiviral Efficacy Outcomes

- Compared with placebo, GS-1179 50, 20, and 10 mg led to nominally statistically significant reductions in:
 - HSV-2 shedding rate (97%, 91%, and 62%, respectively; **Table 2** and **Figure 2**)
 - HSV-2 high viral load shedding rate (100%, 95%, and 82%, respectively)
 - Genital lesion rate (74%, 82%, and 80%, respectively)
 - Virologically confirmed HSV-2 genital lesion rate (92%, 100%, and 83%, respectively)
- Compared with placebo, participants receiving GS-1179 50, 20, and 10 mg experienced a numerically lower mean (SD) duration of viral shedding (0.5 [0.0], 1.0 [0.8], and 2.6 [2.0] days, respectively, vs 4.1 [3.7] days with placebo)

Table 2. Summary Statistics and Statistical Comparison of Antiviral Activity and Clinical Outcomes Across Treatments

	Placebo (n = 14)[a]	Cohort B1: GS-1179 50 mg QW (n = 20)			Cohort B2: GS-1179 20 mg QW (n = 20)			Cohort B3: GS-1179 10 mg QW (n = 18)		
Outcome	Summary Statistic	Summary Statistic	Reduction vs Placebo, %	*P* Value	Summary Statistic	Reduction vs Placebo, %	*P* Value	Summary Statistic	Reduction vs Placebo, %	*P* Value
HSV-2 shedding rate,[b] %	14.8	0.4	97	0.002	1.4	91	<0.001	5.6	62	0.011
HSV-2 high viral load shedding rate,[c] %	10.1	0.0	100	–[d]	0.5	95	0.001	1.9	82	0.002
Genital lesion rate,[e] %	10.0	2.6	74	0.044	1.8	82	0.026	2.0	80	0.038
Virologically confirmed HSV-2 genital lesion rate,[f] %	9.5	0.7	92	0.004	0.0	100	–[d]	1.6	83	0.007
Duration of viral shedding, days, mean (SD)	4.1 (3.7)	0.5 (0.0)	NA	NA	1.0 (0.8)	NA	NA	2.6 (2.0)	NA	NA
Duration of genital lesions, days, mean (SD)	4.3 (1.9)	4.7 (0.6)	NA	NA	3.3 (1.2)	NA	NA	2.5 (1.7)	NA	NA

[a]One participant assigned to receive placebo in cohort B2 did not receive treatment and was not evaluated.
[b]HSV-2 shedding rate was calculated as the number of positive HSV-2 anogenital swabs divided by the total number of swabs collected.
[c]HSV-2 high viral load shedding rate was calculated as the number of positive HSV-2 anogenital swabs with >10^4 HSV-2 copies/mL divided by the total number of swabs collected.
[d]There were no events reported in the dosing cohort, which led to unstable model estimation and an unreliable *P* value. The observed difference (100% reduction compared with placebo) was consistent with a highly significant effect.
[e]Genital lesion rate was calculated as the number of days with genital lesions of any kind present divided by the total number of days assessed.
[f]Virologically confirmed HSV-2 genital lesion rate was calculated as the number of days with genital lesions with HSV-2 detected divided by the total number of days assessed.
HSV-2, herpes simplex virus type 2; NA, not applicable; QW, once weekly.

Figure 2. Reduction Versus Placebo in Rates of (A) HSV-2 Shedding, (B) HSV-2 High Viral Load Shedding, (C) Genital Lesions, and (D) Virologically Confirmed HSV-2 Genital Lesions Across Treatments

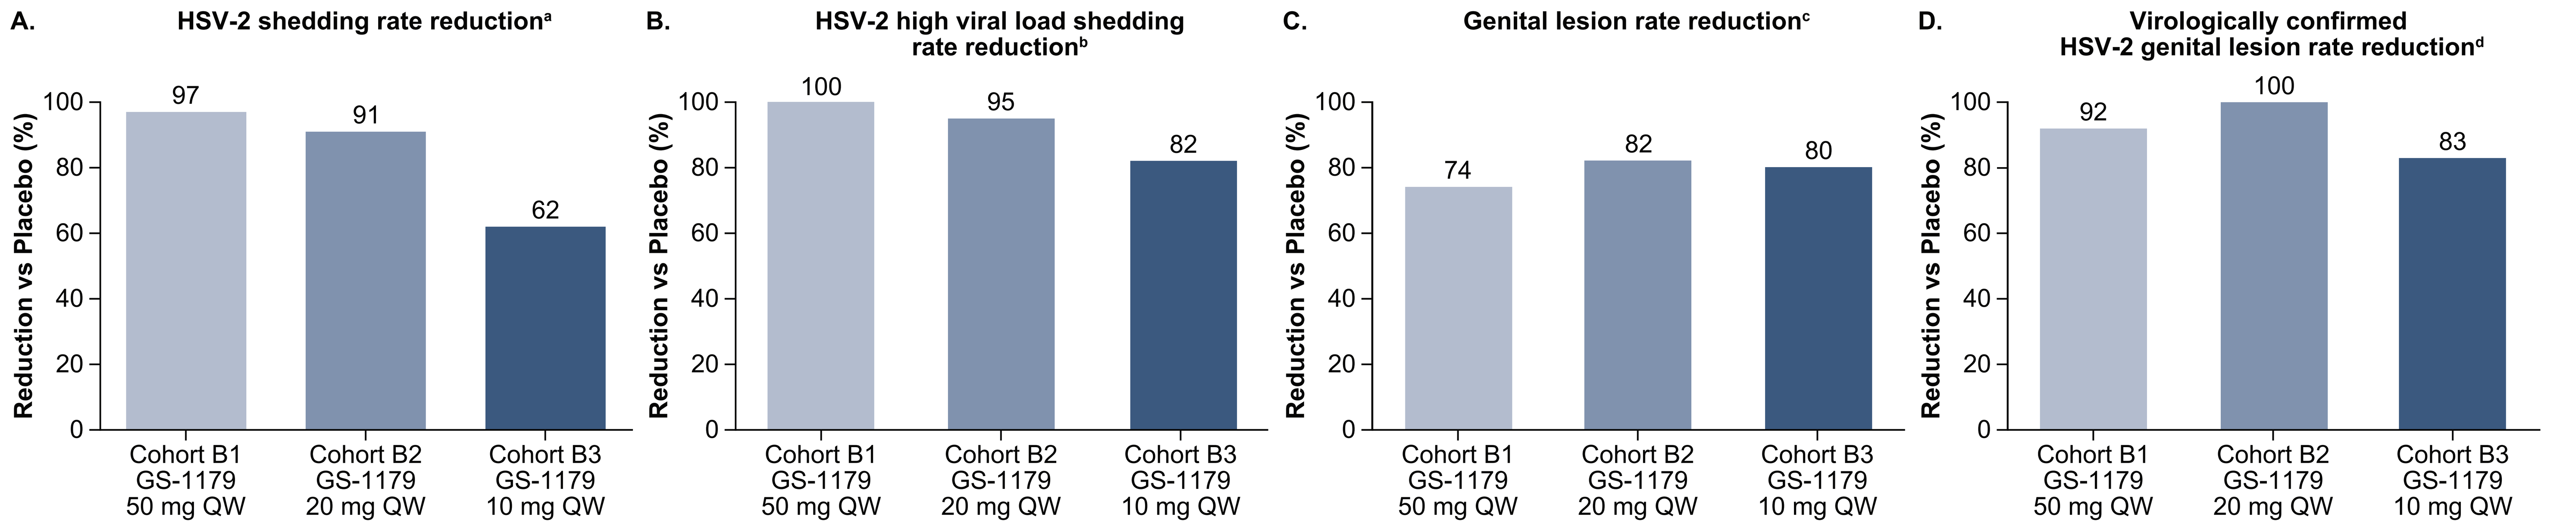

A. HSV-2 shedding rate reduction[a]
97
91
62
B. HSV-2 high viral load shedding rate reduction[b]
100
95
82
C. Genital lesion rate reduction[c]
74
82
80
D. Virologically confirmed HSV-2 genital lesion rate reduction[d]
92
100
83
Reduction vs Placebo (%)
0
20
40
60
80
100
Cohort B1 GS-1179 50 mg QW
Cohort B2 GS-1179 20 mg QW
Cohort B3 GS-1179 10 mg QW

[a]HSV-2 shedding rate was calculated as the number of positive HSV-2 anogenital swabs divided by the total number of swabs collected.
[b]HSV-2 high viral load shedding rate was calculated as the number of positive HSV-2 anogenital swabs with >10^4 HSV-2 copies/mL divided by the total number of swabs collected.
[c]Genital lesion rate was calculated as the number of days with genital lesions of any kind present divided by the total number of days assessed.
[d]Virologically confirmed HSV-2 genital lesion rate was calculated as the number of days with genital lesions with HSV-2 detected divided by the total number of days assessed.
HSV-2, herpes simplex virus type 2; QW, once weekly.

IDWeek; October 21–24, 2026; Washington, DC, USA