Factors Associated With a Lack of Viral Suppression in Chronic HBV (CHB) Patients After 8 Years of Treatment With Tenofovir Alafenamide (TAF) or Tenofovir Disoproxil Fumarate (TDF) Followed by TAF Treatment

1418-C

Edward J Gane[1], Maria Buti[2,3], Scott K Fung[4], Henry LY Chan[5], Namiki Izumi[6], Wan-Long Chuang[7], Sang Hoon Ahn[8], Rajiv Mehta[9], Selim Gurel[10], Frida Abramov[11], Leland J Yee[11], Hongyuan Wang[11], Roberto Mateo[11], John F Flaherty[11], Xiaoli Ma[12], Calvin Q Pan[13], Young-Suk Lim[14], Patrick Marcellin[15]

[1]Auckland Clinical Studies, Auckland, New Zealand; [2]Hospital Universitario Vall d'Hebron, Barcelona, Spain; [3]CIBEREHD del Instituto Carlos III, Madrid, Spain; [4]University of Toronto, Department of Medicine, Toronto, Canada; [5]Faculty of Medicine, the Chinese University of Hong Kong, HMA Office, Tai Wai, Shatin, Hong Kong; [6]Department of Gastroenterology and Hepatology, Japanese Red Cross Musashino Hospital, Tokyo, Japan; [7]Kaohsiung Medical University Hospital, Kaohsiung Medical University, Kaohsiung City, Taiwan; [8]Department of Internal Medicine, Institute for Gastroenterology, Yonsei University College of Medicine, Seoul, South Korea; [9]Nirmal Hospital, Surat, India; [10]Uludağ University, Department of Gastroenterology, Bursa, Turkey; [11]Gilead Sciences, Inc., Foster City, CA, USA; [12]Hahnemann University Hospital, Philadelphia, PA, USA; [13]NYU Langone Health, New York University Grossman School of Medicine, New York, NY, USA; [14]Asan Medical Center, University of Ulsan College of Medicine, Seoul, South Korea; [15]Hepatology Department, Hôpital Beaujon, APHP, INSERM, University of Paris, Clichy, France

Copies of this poster obtained through QR (Quick Response) and/or text key codes are for personal use only and may not be reproduced without written permission of the authors.

Key Findings

- **95% (853 of 895) of all CHB patients, regardless of HBeAg status, treated with TAF alone or TDF followed by TAF achieved HBV DNA <29 IU/mL at Year 8 (week 384)**
- **Of the 42 patients (5% total) not achieving HBV DNA <29 IU/mL, most had either a viral blip or virological breakthrough; only 4 patients (0.4% total) were persistently viremic**
- **No resistance to TAF was detected in any patient at year 8 (see Poster #1430-C)**
- **Biochemical and serological responses were lower in patients with HBV DNA ≥29 IU/mL vs those with HBV DNA <29 IU/mL at year 8**
- **By multivariate analysis, younger age (<50 years) and infection with HBV genotypes A and D were associated with HBV DNA ≥29 IU/mL at year 8; study drug adherence (by pill count) was not a significant factor**

Conclusions

Treatment with TAF or with TDF followed by TAF for up to 8 years was associated with high rates of viral suppression, and lack of suppression was uncommon (≤5%)

Patients having a lack of viral suppression at year 8 were more likely to be younger and infected with genotypes A or D

Introduction

- With an estimated all age prevalence of 4.1%, representing ~316 million people living with chronic hepatitis B (CHB) worldwide, infection with the hepatitis B virus (HBV) resulted in over 800,000 deaths in 2019, primarily from cirrhosis and hepatocellular carcinoma (HCC)[1]
- Tenofovir alafenamide (TAF), a novel prodrug of tenofovir with enhanced plasma stability and more efficient hepatic delivery, has ~90% lower circulating levels of tenofovir relative to tenofovir disoproxil fumarate (TDF) when given at a lower daily dose than TDF[2,3]
- In 2 randomized, Phase 3 studies (Studies 108 and 110), TAF showed noninferior efficacy with improved renal and bone safety vs TDF at weeks 48 and 96[4–6]
 - At 8 years, TAF-treated patients had similarly high rates of HBV DNA suppression compared to patients randomized to TDF followed by open-label TAF[7]
- Despite achieving and maintaining high rates of virologic suppression through 8 years, it is important to characterize those patients that failed to achieve an HBV DNA level below the lower limit of quantification (LLOQ) at year 8

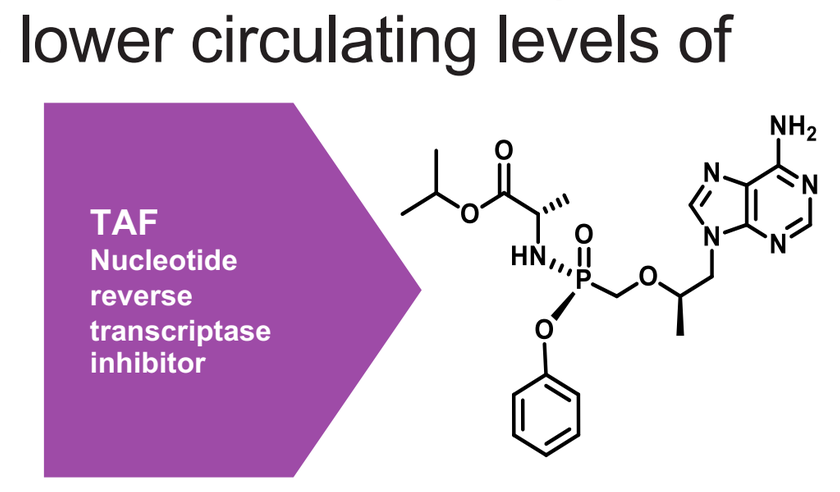
TAF
Nucleotide reverse transcriptase inhibitor
NH2
HN
O
P
N

References: **1.** Sheena BE, et al. Global Burden of Disease (GBD) 2019, Hepatitis B. *Lancet Gastroenterol Hepatol.* 2022;7:796-829. **2.** Agarwal K, et al. *J Hepatol.* 2015;62:533-40. **3.** Murakami E, et al. *Antimicrob Agents Chemother.* 2015:59:3563-9. **4.** Buti M, et al. *Lancet Gastroenterol Hepatol.* 2016;1:196-206. **5.** Chan HL, et al. *Lancet Gastroenterol Hepatol.* 2016;1:185-95. **6.** Agarwal K, et al. *J Hepatol.* 2018;68:672-81. **7.** Buti M, et al. Abstract presented at: EASL; June 23, 2023; Vienna, Austria. Presentation OS-067.

Acknowledgments: We extend our thanks to the patients, their families, and all participating investigators. This study was funded by Gilead Sciences, Inc. Medical writing support was provided by Helen Rodgers, PhD, of AlphaScientia, a Red Nucleus company, and was funded by Gilead Sciences, Inc.

Disclosures: **EJG** served as an advisor for AbbVie; Aligos Therapeutics; Arbutus; Gilead Sciences, Inc.; Janssen; Roche; Vir Biotechnology; and Virion Therapeutics. **MB** reports speaker fees, research support, and consulting fees from AbbVie; Gilead Sciences, Inc.; and Janssen. **SKF** served as an advisor for AbbVie; Gilead Sciences, Inc.; Novo Nordisk; and Pfizer; reports speaker fees from AbbVie; Gilead Sciences, Inc.; and Lupin; and received research support from Gilead Sciences, Inc. **HLYC** served as an advisor for Aligos Therapeutics; GSK; Gilead Sciences, Inc.; Roche; Vaccitech; Vir Biotechnology, Inc.; and Virion Therapeutics, and reports speaker fees from Gilead Sciences, Inc.; Roche; and Viatris. **SG** served as an investigator and speaker for AbbVie and Gilead Sciences, Inc. **FA, LJY, HW, RM,** and **JFF** are employees and stockholders of Gilead Sciences, Inc. **XM** served as a consultant and speaker for Gilead Sciences, Inc. **CQP** received research support from Gilead Sciences, Inc. **PM** received grants from AbbVie; Aligos Therapeutics; Assembly Biosciences; Bristol Myers Squibb; Gilead Sciences, Inc.; Humedics; Madrigal; Novo Nordisk; Pfizer; Roche; and Intercept. **NI, WLC, SHA, RM,** and **YSL** report no conflicts of interest.

Objective

- Characterize factors associated with the lack of virologic suppression after 8 years of treatment among patients with CHB treated with TAF or with TDF followed by TAF

Methods

Study Design

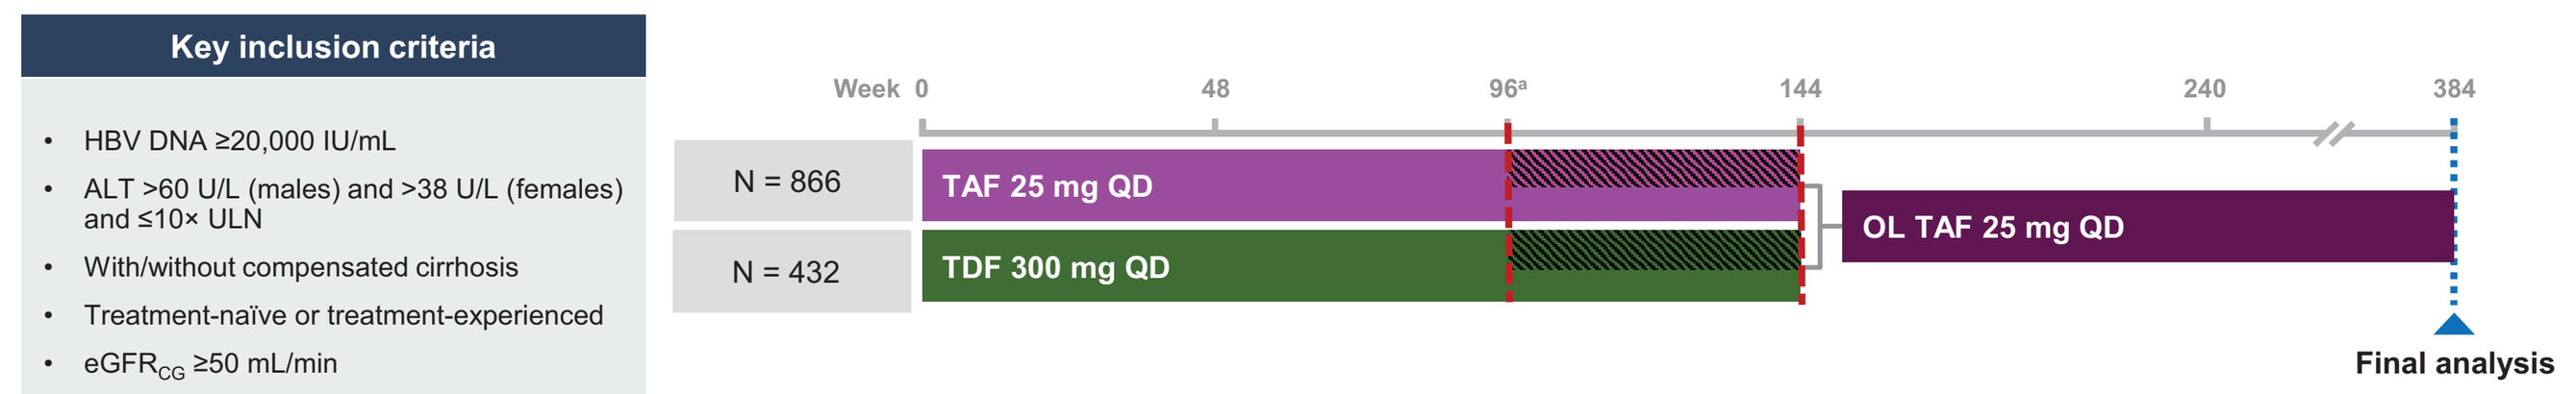
Key inclusion criteria
HBV DNA ≥20,000 IU/mL
ALT >60 U/L (males) and >38 U/L (females) and ≤10× ULN
With/without compensated cirrhosis
Treatment-naïve or treatment-experienced
eGFRCG ≥50 mL/min
Week 0 48 96a 144 240 384
N = 866
TAF 25 mg QD
N = 432
TDF 300 mg QD
OL TAF 25 mg QD
Final analysis

[a]Amendment 3 enacted to extend DB to week 144 and OL to week 384 (year 8). Shaded/slashed areas represent patients who rolled over to OL TAF at week 96.
ALT, alanine aminotransferase; DB, double-blind; $eGFR_{CG}$, estimated glomerular filtration rate by Cockcroft-Gault; HBV, hepatitis B virus; OL, open-label; QD, once daily; TAF, tenofovir alafenamide; TDF, tenofovir disoproxil fumarate; ULN, upper limit of normal.

- Two Phase 3, randomized, double-blind studies in patients with chronic hepatitis B who were hepatitis B e antigen (HBeAg)-negative (Study 108, NCT01940341)[6,7] and HBeAg-positive (Study 110, NCT01940471)[7,8] were conducted
 - **Double-blind (DB) phase:** randomized 2:1 (TAF 25 mg:TDF 300 mg once daily) and stratified by HBV DNA level and treatment status (naïve/experienced)
 - **Open-label (OL) phase:** TAF 25 mg once daily in patients who received TAF or who received TDF for 2 or 3 years
- HBV DNA determined in plasma by Roche COBAS Taqman HBV Test for Use with the High Pure System (baseline → week 144; LLOQ 29 IU/mL) and by COBAS/ AmpliPrep/ COBAS TaqMan HBV Test, v2.0 (week 168 → week 384; LLOQ 20 IU/mL), including HBV DNA <LLOQ with target not detected (TND; ie, undetectable level)
 - For consistency of reporting across the 8-year (384-week) study period, 29 IU/mL was applied as the LLOQ
- Data from Studies 108 and 110 were pooled and evaluated according to patients with or without HBV DNA <29 IU/mL at week 384 using a missing equals excluded analysis
 - Study drug adherence (%) was determined by pill count (ie, [number of tablets taken/ tablets prescribed at each dispensing visit during the DB and OL phases] × 100)
 - Univariate and multivariate logistic regression analyses were performed to identify factors associated with HBV DNA ≥29 IU/mL at week 384
 - Individual cases with HBV DNA ≥29 IU/mL at week 384 were evaluated and assigned to 3 categories: 1) viral blip[a], 2) virologic breakthrough[b], and 3) persistent viremia[c]
- Biochemical and serological responses were assessed in patients with HBV DNA <29 IU/mL vs HBV DNA ≥29 IU/mL at week 384

[a]Viral blip was defined as a single (unconfirmed) increase in HBV DNA at or above 29 IU/mL at week 384 in a patient achieving HBV DNA <29 IU/mL for at least 2 consecutive visits; [b]Virologic breakthrough was defined as a confirmed increase in HBV DNA ≥29 IU/mL after achieving HBV DNA <29 IU/mL for at least 2 consecutive visits; [c]Persistent viremia was defined as HBV DNA ≥29 IU/mL at all study visits over 384 weeks.

Results

Patient Disposition

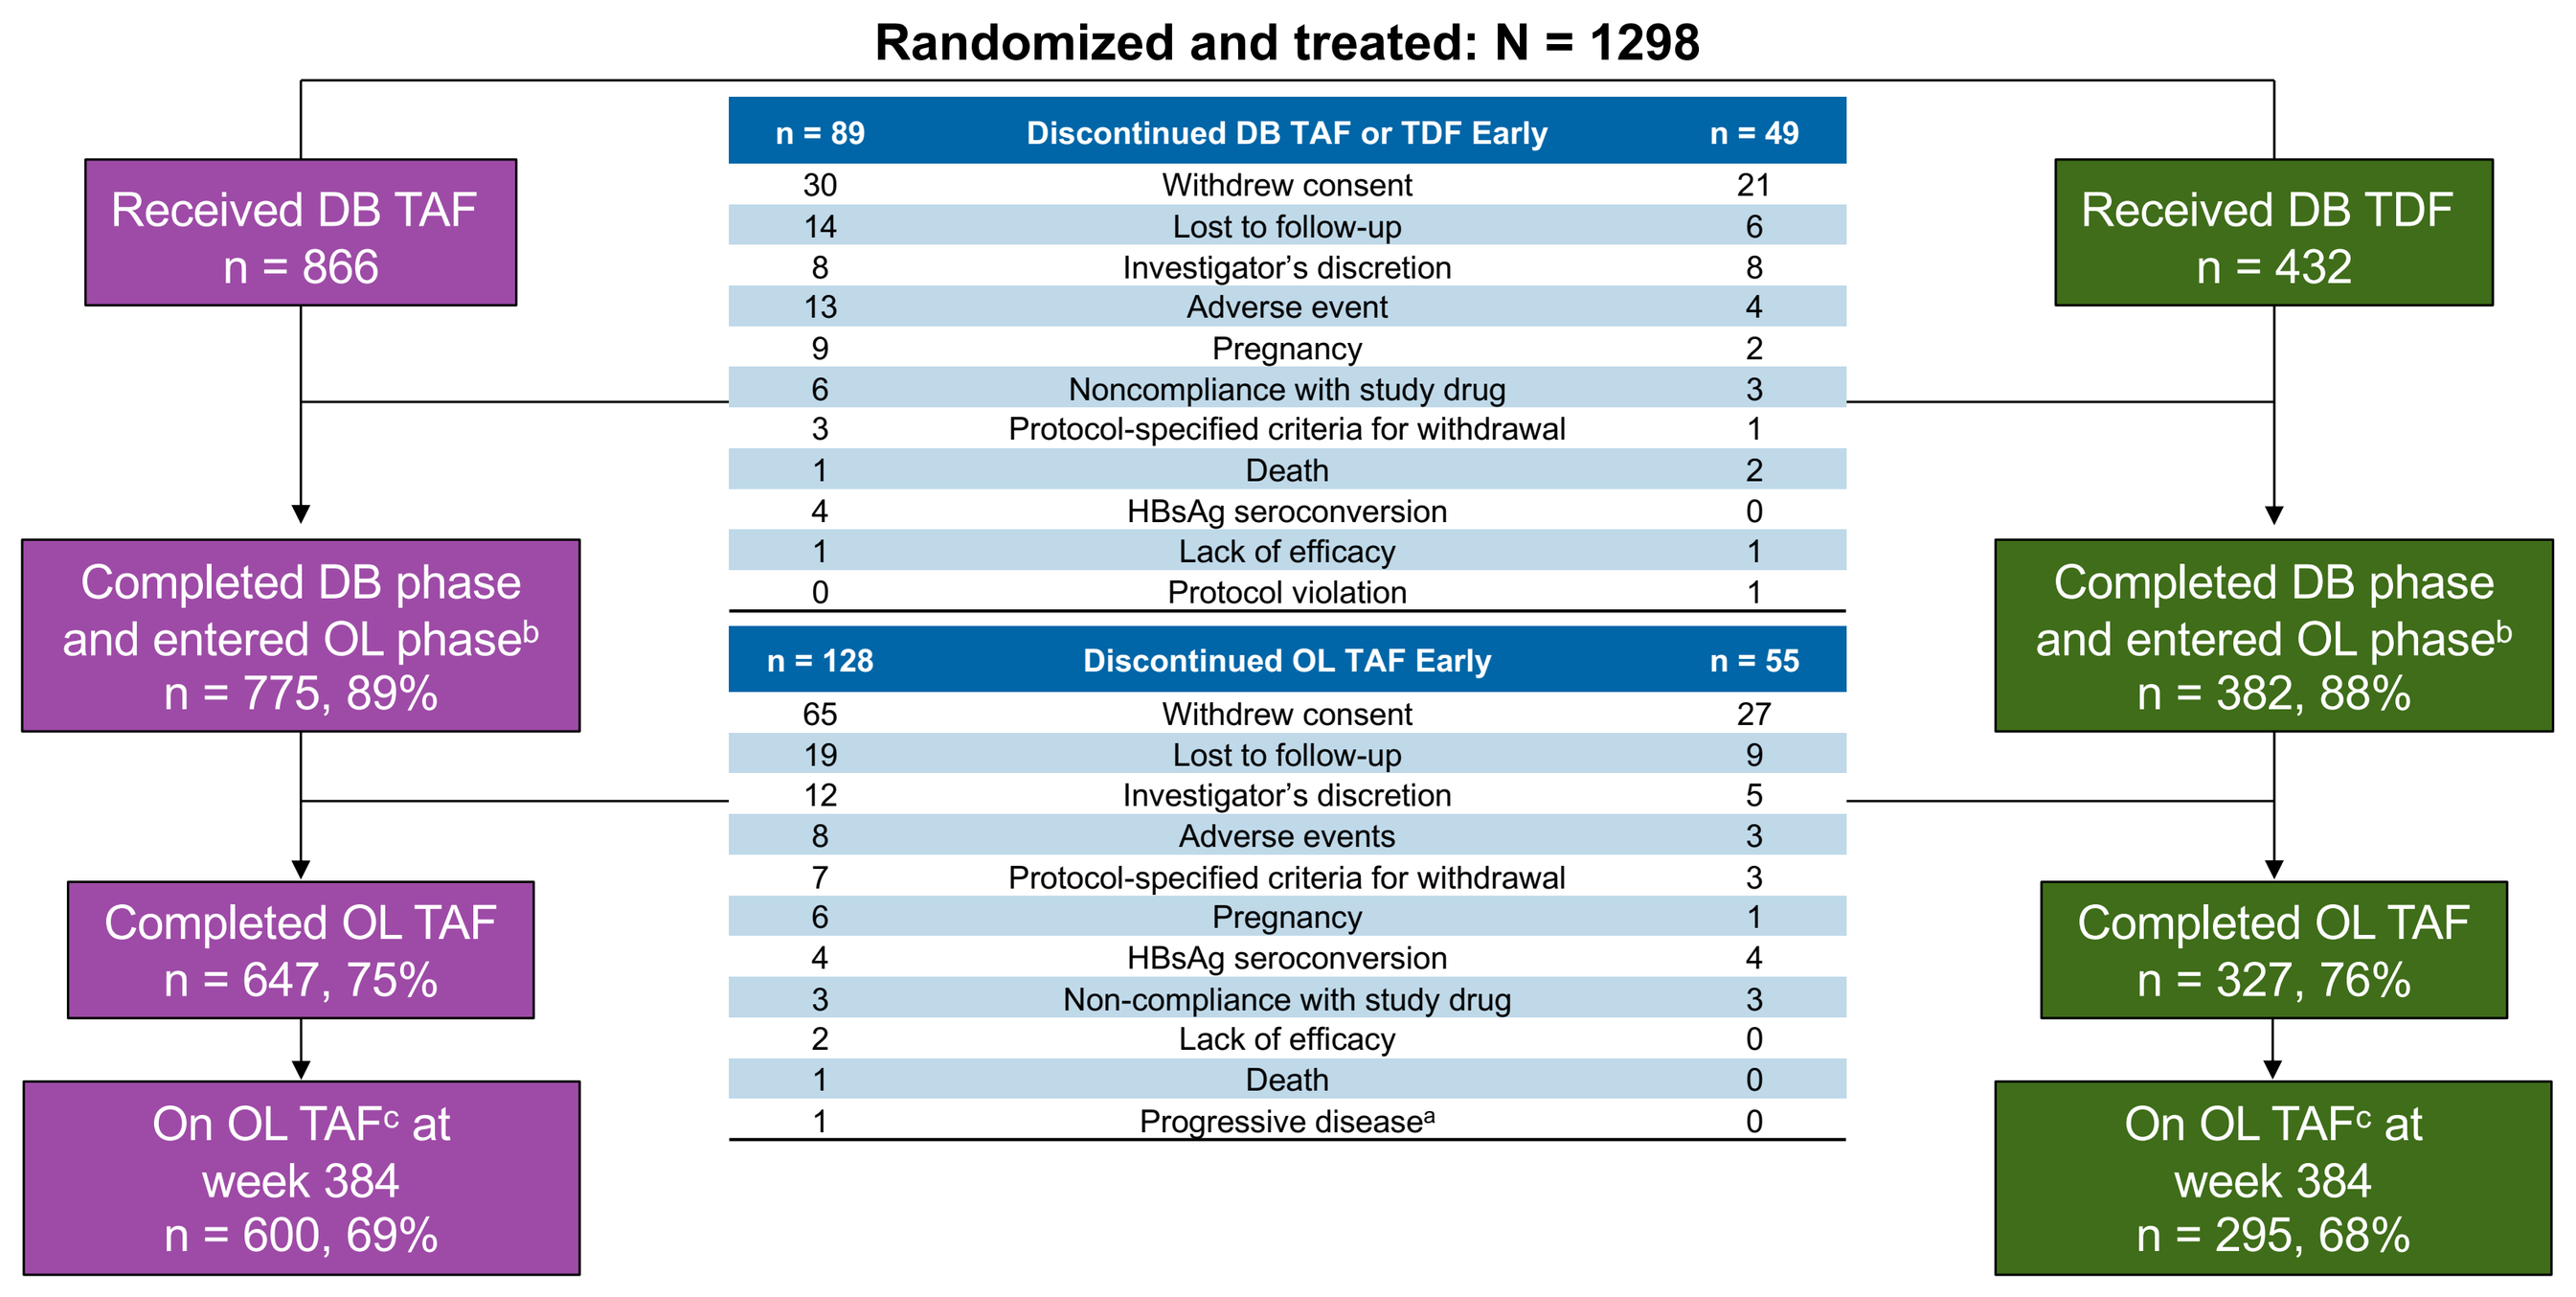
Randomized and treated: N = 1298
Received DB TAF
n = 866
Received DB TDF
n = 432
n = 89 | Discontinued DB TAF or TDF Early | n = 49
30 | Withdrew consent | 21
14 | Lost to follow-up | 6
8 | Investigator's discretion | 8
13 | Adverse event | 4
9 | Pregnancy | 2
6 | Noncompliance with study drug | 3
3 | Protocol-specified criteria for withdrawal | 1
1 | Death | 2
4 | HBsAg seroconversion | 0
1 | Lack of efficacy | 1
0 | Protocol violation | 1
Completed DB phase and entered OL phase[b]
n = 775, 89%
Completed DB phase and entered OL phase[b]
n = 382, 88%
n = 128 | Discontinued OL TAF Early | n = 55
65 | Withdrew consent | 27
19 | Lost to follow-up | 9
12 | Investigator's discretion | 5
8 | Adverse events | 3
7 | Protocol-specified criteria for withdrawal | 3
6 | Pregnancy | 1
4 | HBsAg seroconversion | 4
3 | Non-compliance with study drug | 3
2 | Lack of efficacy | 0
1 | Death | 0
1 | Progressive disease[a] | 0
Completed OL TAF
n = 647, 75%
Completed OL TAF
n = 327, 76%
On OL TAF[c] at week 384
n = 600, 69%
On OL TAF[c] at week 384
n = 295, 68%

[a]Metastatic HCC. [b]2 patients in the TAF group and 1 patient in the TDF to TAF group completed the DB phase and did not enter the OL phase. [c]47 TAF and 32 TDF→TAF patients completed OL treatment at week 144 under protocol amendment 2 or did not have HBV DNA data at week 384 and were excluded from the present analysis.
DB, double-blind; HBsAg, hepatitis B surface antigen; HBV, hepatitis B virus; HCC, hepatocellular carcinoma; OL, open-label; TAF, tenofovir alafenamide; TDF, tenofovir disoproxil fumarate.

HBV DNA <29 IU/mL Over 384 Weeks

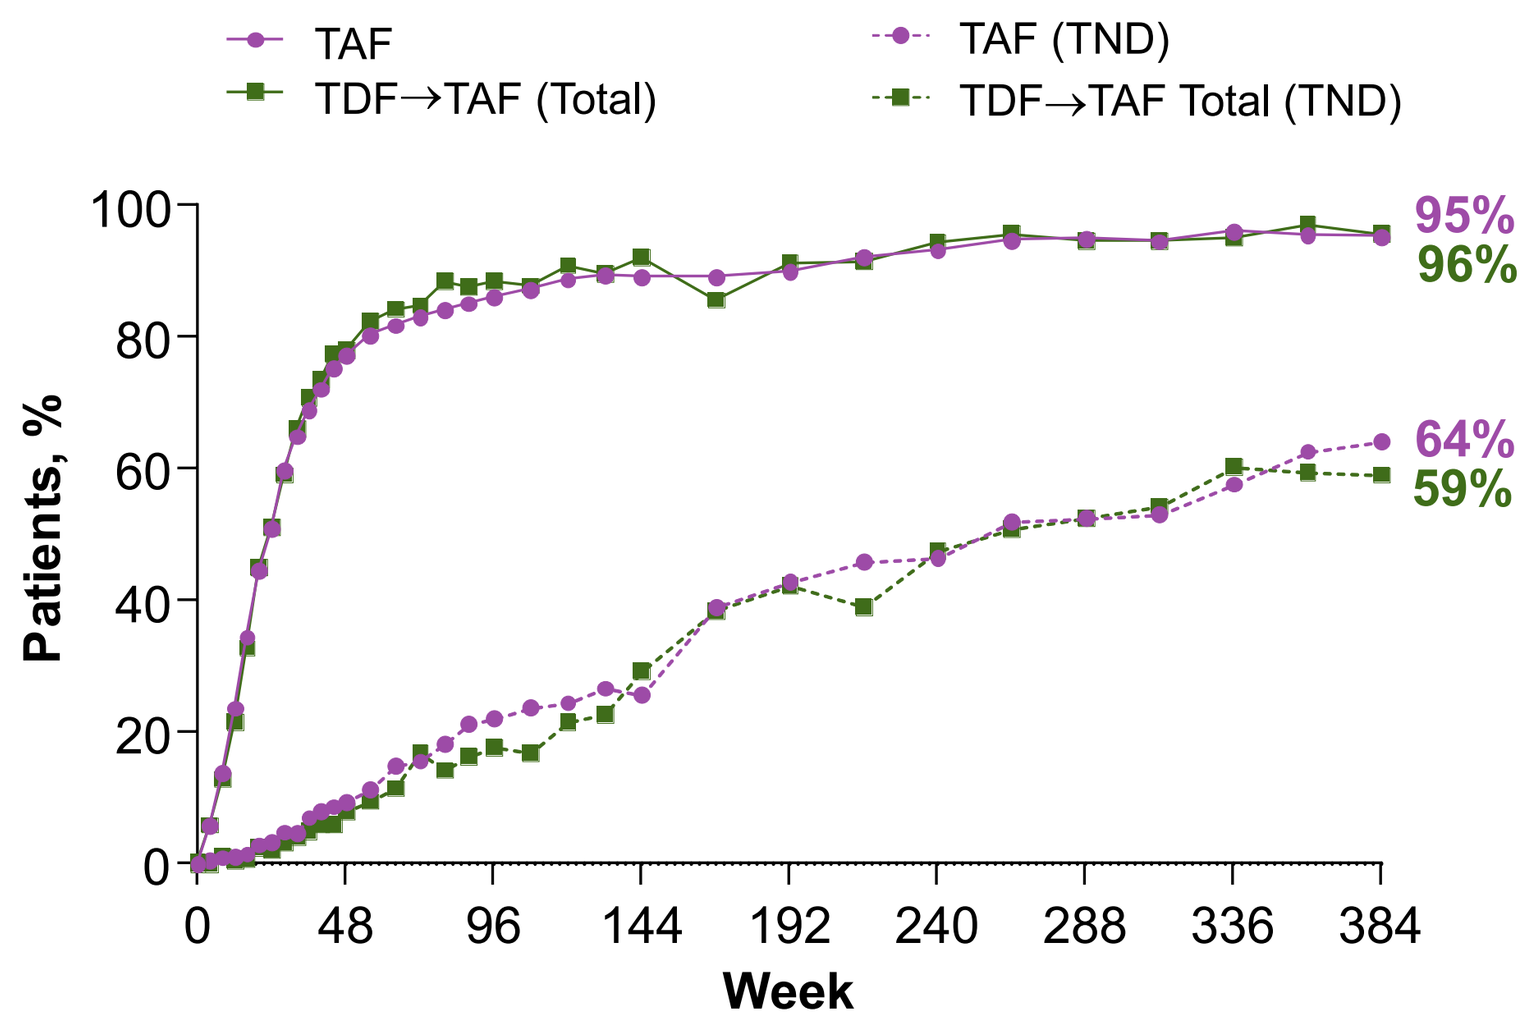
TAF
TDF→TAF (Total)
TAF (TND)
TDF→TAF Total (TND)
Patients, %
Week
95%
96%
64%
59%

HBV DNA Levels at Week 384 (N = 895)

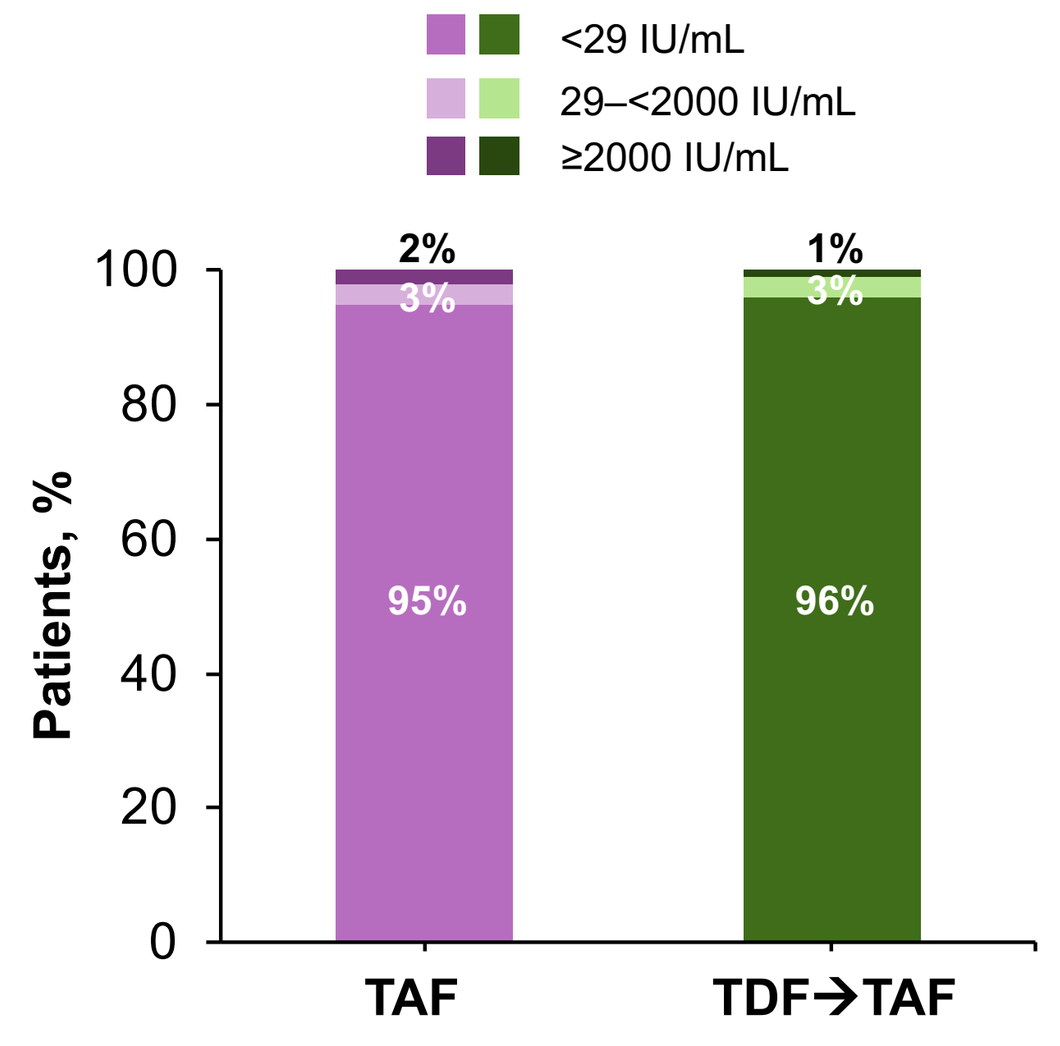
<29 IU/mL
29–<2000 IU/mL
≥2000 IU/mL
Patients, %
2%
3%
95%
1%
3%
96%
TAF
TDF→TAF

Full analysis set. Missing = excluded analysis.
HBV, hepatitis B virus; TAF, tenofovir alafenamide; TDF, tenofovir disoproxil fumarate; TND; proportion with target not detected.

- Similar high rates of viral suppression were achieved and maintained across treatment groups; 95%–96% of patients had HBV DNA <29 IU/mL at week 384 (year 8)
- The proportions with HBV DNA <29 IU/mL and target not detected (ie, undetectable HBV DNA) increased progressively with similar results by treatment group at week 384 (year 8)

Baseline Characteristics in Patients with HBV DNA <29 vs ≥29 IU/mL at Week 384

	HBV DNA <29 IU/mL n = 853	HBV DNA ≥29 IU/mL n = 42
Treatment group, n (%)		
TAF	571 (67)	29 (69)
TDF→TAF (total)	282 (33)	13 (31)
Age, years, median (Q1, Q3)	42 (34, 52)	33 (25, 38)
Age < 50 years, n (%)	610 (72)	39 (93)
Male sex, n (%)	540 (63)	35 (83)
Race/ethnicity, n (%)[a]		
White	157 (18)	14 (33)
Asian	683 (80)	27 (64)
East Asian[b]	576/683 (84)	13/27 (48)
Asian, other[c]	107/683 (16)	14/27 (52)
HBV DNA, log_{10} IU/mL, median (Q1, Q3)	7.4 (5.7, 8.3)	8.0 (5.9, 8.6)
HBV DNA ≥8 log_{10} IU/mL, n (%)	279 (33)	21 (50)
HBsAg, log_{10} IU/mL, median (Q1, Q3)	3.82 (3.36, 4.38)	4.36 (3.95, 4.72)
ALT, U/L, median (Q1, Q3)	80 (54, 125)	104 (66, 159)
ALT >5× ULN AASLD, n (%)	145 (17)	10 (24)
HBeAg-positive, n (%)	543 (64)	33 (79)
HBV genotype, n (%)		
A or D	243 (28)	26 (62)
B or C	600 (70)	16 (38)
Other (E, F, or H)	10 (1)	0
CL_{CR}, mL/min, median (Q1, Q3)	104 (89, 124)	123 (107, 140)
Oral antiviral treatment experienced, n (%)	219 (26)	19 (45)
FibroTest score ≥0.75 (Metavir F4/cirrhosis), n (%)	77/834 (9)	3/42 (7)
Adherence rate (%), median (Q1, Q3)		
DB phase	99 (98, 100)	99 (98, 100)
OL phase	99 (97, 100)	98 (97, 100)

Full analysis set. [a]Race/ethnicity was by self report. [b]East Asian includes Chinese, Japanese, Korean, Taiwanese, and Vietnamese. [c]Other includes Indian.
AASLD, American Association for the Study of Liver Disease; ALT, alanine aminotransferase; DB, double-blind; CL_{Cr}, creatinine clearance by the Cockcroft-Gault method; HBeAg, hepatitis B e antigen; HBsAg, hepatitis B surface antigen; HBV, hepatitis B virus; OL, open-label; Q, quartile; TAF, tenofovir alafenamide; TDF, tenofovir disoproxil fumarate; ULN, upper limit of normal.

- Of 895 patients included in this analysis, 42 (5%; TAF, n = 29; TDF→TAF, n = 13) had an HBV DNA level ≥29 IU/mL at week 384
 - Review of individual HBV DNA profiles showed the following viral response categories:
 - HBeAg-negative patients (Study 108; n = 9): viral blip (n = 3), virologic breakthrough (n = 6), persistent viremia (n = 0)
 - HBeAg-positive patients (Study 110; n = 33): viral blip (n = 17), virologic breakthrough (n = 12), persistent viremia (n = 4)
 - Total (N = 42): viral blip (n = 20), virologic breakthrough (n = 18), persistent viremia (n = 4)
 - Representative patient examples of HBV DNA and ALT profiles over 384 weeks are provided in the following figures

Example Cases

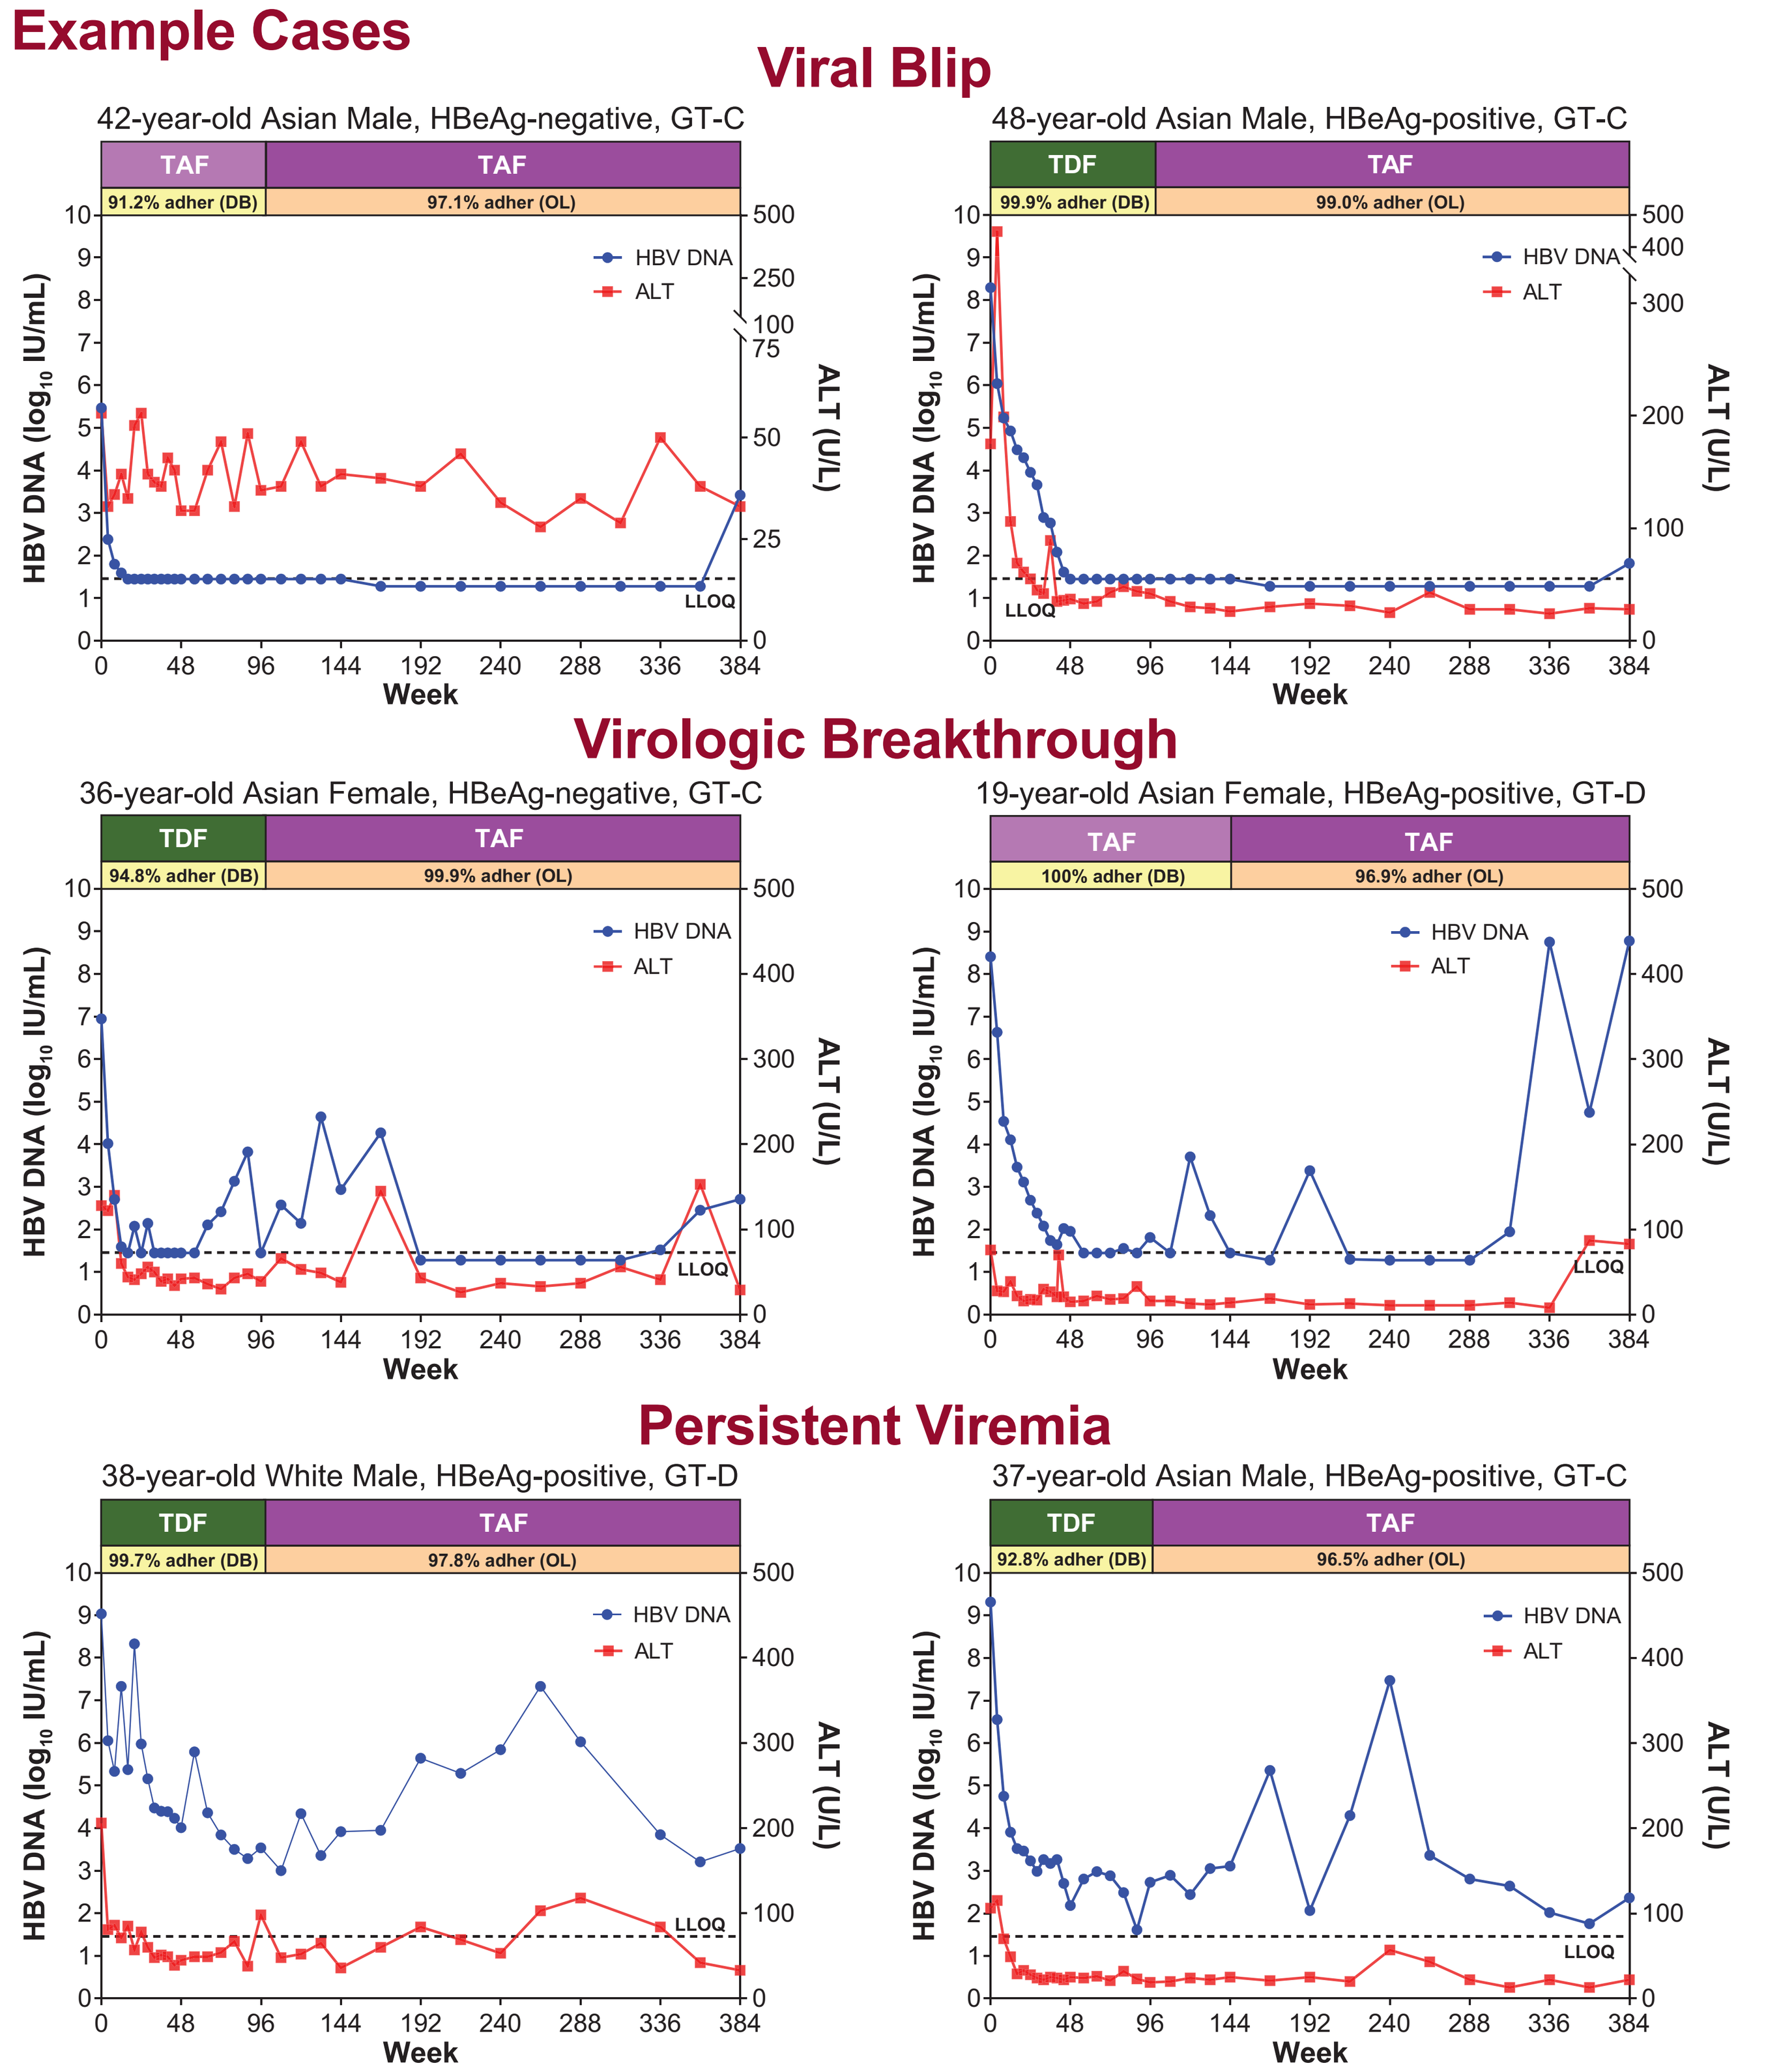
Viral Blip
42-year-old Asian Male, HBeAg-negative, GT-C
TAF
TAF
91.2% adher (DB)
97.1% adher (OL)
48-year-old Asian Male, HBeAg-positive, GT-C
TDF
TAF
99.9% adher (DB)
99.0% adher (OL)
Virologic Breakthrough
36-year-old Asian Female, HBeAg-negative, GT-C
TDF
TAF
94.8% adher (DB)
99.9% adher (OL)
19-year-old Asian Female, HBeAg-positive, GT-D
TAF
TAF
100% adher (DB)
96.9% adher (OL)
Persistent Viremia
38-year-old White Male, HBeAg-positive, GT-D
TDF
TAF
99.7% adher (DB)
97.8% adher (OL)
37-year-old Asian Male, HBeAg-positive, GT-C
TDF
TAF
92.8% adher (DB)
96.5% adher (OL)
HBV DNA
ALT
LLOQ
HBV DNA (log_{10} IU/mL)
ALT (U/L)
Week

Adher, adherence; ALT, alanine aminotransferase; DB, double-blind; GT, genotype; HBeAg, hepatitis B e antigen; HBV, hepatitis B virus; LLOQ, lower limit of quantification; OL, open-label; TAF, tenofovir alafenamide; TDF, tenofovir disoproxil fumarate.

- By univariate analysis (UV), baseline factors significantly (P <.05) associated with HBV DNA ≥29 IU/mL at week 384 included age (Odds Ratio, 95% Confidence Interval 0.92 [0.89, 0.95]; P <0.0001), creatinine clearance (1.02 [1.01, 1.03]; P <.0001), non-genotype D infection (0.29 [0.16, 0.54]; P = .0001), non-East Asia study region (3.93 [1.86, 8.30]; P = .0003), hip bone mineral density (BMD) (30.52 [3.34, 278.54]; P = 0.0024), spine BMD (7.01 [1.14, 42.23]; P = .036), female sex (0.35 [0.15, 0.79]; P = .011), Non-Asian race (2.23 [1.16, 4.29]; P = 0.016), HBV DNA <8 log_{10} IU/mL (0.49 [0.26, 0.91]; P = .023), Vitamin D (1.02, [1.00, 1.04]; P = 0.018), and absence (vs presence) of proteinuria (0.39 [0.16, 0.90]; P = .028). Treatment assignment and study drug adherence were not significant by UV analysis
- Results for the multivariate logistic regression analysis are as follows:

Factor	Comparison	Odds Ratio	95% CI	*P*-value
HBV genotype D	Yes vs No	4.03	2.06, 7.91	<.001
Age group	<50 vs ≥50 years	4.78	1.45, 15.71	.01
HBV genotype A	Yes vs No	3.24	1.13, 9.26	.028

- By multivariate analysis, HBV genotype A or D infection and age <50 years were the only factors found to be significant predictors of HBV DNA ≥29 IU/mL at week 384

HBV Outcomes at Week 384 in Patients HBV DNA <29 vs ≥29 IU/mL

Outcome, n/N (%)[a]	HBV DNA <29 IU/mL N = 853	HBV DNA ≥29 IU/mL N = 42
ALT normalization[b]		
Central laboratory criteria	662/748 (89)	18/38 (47)
AASLD criteria[c]	638/807 (79)	15/40 (37.5)
HBeAg loss/seroconversion[d]	247/520 (47.5)/164/520 (32)	6/32 (19)/6/32 (19)
HBsAg loss/seroconversion	25/823 (3)/19/823 (2)	0/41 (0%)/0/41 (0%)
Median change in HBsAg, log_{10} IU/mL, (Q1, Q3)	−0.56 (−1.21, −0.18)	−0.37 (−0.85, −0.01)

[a]Among patients in the Full Analysis Set; all analyses are missing = excluded; the denominators are patients with available data at week 384. [b]Includes only patients with ALT >ULN at baseline; [c]2018 criteria: 35 U/L males and 25 U/L females. [d]Includes only patients who were HBeAg-positive and HBeAb negative/missing at baseline. AASLD, American Association for the Study of Liver Disease; ALT, alanine aminotransferase; HBeAg, hepatitis B e antigen; HBsAg, hepatitis B surface antigen; HBV, hepatitis B virus; Q, quartile; ULN, upper limit of normal.

- Treatment responses (biochemical and serological) were lower in patients with HBV DNA ≥29 IU/mL at week 384

AASLD: The Liver Meeting; November 10–14, 2023; Boston, MA, USA