Development of Antidrug Antibodies on Bulevirtide Monotherapy in Chronic Hepatitis Delta Does Not Impact Bulevirtide Efficacy, Safety, or Pharmacokinetics

1465-C

Pietro Lampertico[1,2], Soo Aleman[3], Adrian Streinu-Cercel[4], Pavel Bogomolov[5], Viacheslav Morozov[6], Nina Mamonova[7], Dmitry Manuilov[8], Lei Ye[8], Ben L Da[8], Renee-Claude Mercier[8], Audrey H Lau[8], Grace Chee[8], Navita L Mallalieu[8], Parag Kumar[8], Tatyana Stepanova[9], Vladimir Chulanov[7], Stefan Zeuzem[10], Heiner Wedemeyer[11], Marc Bourliere[12], Fabien Zoulim[13], Tarik Asselah[14], Maurizia Rossana Brunetto[15], Markus Cornberg[11], Antje Blank[16]

[1]CRC "A. M. and A. Migliavacca" Center for Liver Disease, University of Milan, Department of Pathophysiology and Transplantation, Milan, Italy; [2]Foundation IRCCS Ca' Granda Ospedale Maggiore Policlinico, Division of Gastroenterology and Hepatology, Milan, Italy; [3]Karolinska University Hospital/Karolinska Institutet, Department of Infectious Diseases, Stockholm, Sweden; [4]Matei Bals National Institute of Infectious Diseases, Bucharest, Romania; [5]State Budgetary Institution of Health Care of Moscow Region "Moscow Regional Research Clinical Institute Named After M.F. Vladimirsky," Moscow, Russian Federation; [6]Hepatolog, Samara, Russian Federation; [7]FSBI National Research Medical Center for Phthisiopulmonology and Infectious Diseases of the Ministry of Health of the Russian Federation, Moscow, Russian Federation; [8]Gilead Sciences, Inc., Foster City, CA, USA; [9]Clinic of Modern Medicine, Moscow, Russian Federation; [10]University Hospital Frankfurt, Department of Medicine, Frankfurt am Main, Germany; [11]Medizinische Hochschule Hannover, Klinik für Gastroenterologie, Hepatologie und Endokrinologie, Hannover, Germany; [12]Hôpital Saint Joseph Marseille, Marseille, France; [13]Hospital Croix Rousee, Service Hepatologie, Lyon, France; [14]Hôpital Beaujon APHP, Université de Paris, INSERM, Clichy, France; [15]Hepatology Unit, Reference Center of the Tuscany Region for Chronic Liver Disease and Cancer, University Hospital of Pisa and Department of Clinical and Experimental Medicine, University of Pisa, Pisa, Italy; [16]Heidelberg University Hospital, Clinical Pharmacology and Pharmacoepidemiology, Heidelberg, Germany

Copies of this poster obtained through QR (Quick Response) and/or text key codes are for personal use only and may not be reproduced without written permission of the authors.

Key Findings

- Incidence of ADA development over 48 weeks of BLV treatment was 23% among the 2-mg group and 22% among the 10-mg group
- No clinically meaningful differences were observed in the combined virologic and biochemical responses at both doses based on ADA status
- BLV was safe and well tolerated regardless of ADA status

Conclusions

Development of ADAs in response to BLV monotherapy did not impact efficacy, safety, or PK throughout 48 weeks of treatment

References: 1. Stockdale AJ, et al. *J Hepatol*. 2020;73:523-32. 2. European Medicines Agency. Hepcludex (bulevirtide). Accessed August 28, 2023. Available at: https://www.ema.europa.eu/en/medicines/human/EPAR/hepcludex. 3. Bogomolov P, et al. *J Hepatol*. 2016;65:490-8.

Acknowledgments: We extend our thanks to the patients, their families, and all participating investigators. Writing and editorial support was provided by Molly Yeager, PhD, of AlphaScientia, a Red Nucleus company, and funded by Gilead Sciences, Inc.

Disclosures: **PL** reports speaking and teaching fees from and participation in advisory committees or review panels for AbbVie; Aligos; Alnylam; Antios; Arrowhead; Bristol Myers Squibb; Eiger Biopharmaceuticals; Gilead Sciences, Inc.; GSK; Janssen; Merck Sharp & Dohme; MYR GmbH; Roche; and Spring Bank Pharmaceuticals. **SA** received honoraria for lectures and educational events from AbbVie; Biogen; Gilead Sciences, Inc.; and Merck Sharp & Dohme; and reports grants from AbbVie and Gilead Sciences, Inc. **ASC, PB, VM,** and **NM** report no conflicts of interest. **DM, LY, BLD, RCM, AHL, GC, NLM,** and **PK** are current or former employees of Gilead Sciences, Inc., and may own stock in Gilead Sciences, Inc. **TS** reports no conflict of interest. **VC** reports being a consultant and giving sponsored lectures for AbbVie; AstraZeneca; Bristol Myers Squibb; Gilead Sciences, Inc.; GSK; Hepatera; Merck Sharp & Dohme; Roche; and R-Pharm. **SZ** reports speaker's bureau and/or consultancy for AbbVie; Allergan; BioMarin; Gilead Sciences, Inc.; Intercept; Janssen; Merck Sharp & Dohme; Novo Nordisk; Swedish Orphan Biovitrum; and Theratechnologies. **HW** reports honoraria for speaking or consulting from Abbott; AbbVie; Boehringer Ingelheim, Bristol Myers Squibb; Eiger Biopharmaceuticals; Gilead Sciences, Inc.; Janssen; Merck Sharp & Dohme; MYR GmbH; Novartis; Novira; Roche; Roche Diagnostics; Siemens; and Transgene; and research support from Abbott; Bristol Myers Squibb; Gilead Sciences, Inc.; Novartis, Roche; and Roche Diagnostics. **MB** reports being a board member and speaker for AbbVie; Gilead Sciences, Inc.; Intercept; and Roche. **FZ** reports consulting fees from AiCuris; Aligos; Assembly Biosciences; Blue Jay; Gilead Sciences, Inc.; and GSK; and research funding to INSERM from Assembly Biosciences; Beam; and Janssen. **TA** acted as a speaker and investigator for AbbVie; Eiger Biopharmaceuticals; Gilead Sciences, Inc.; Janssen; Merck; MYR Pharmaceuticals; and Roche. **MRB** reports speaker's bureau for AbbVie; EISAI-MSD; and Gilead Sciences, Inc. and advisory/consultancy for AbbVie; Gilead Sciences, Inc.; Janssen, and Roche. **MC** received honoraria from AbbVie; Falk; Gilead Sciences, Inc.; GSK; Janssen-Cilag; Merck Sharp & Dohme; Novartis; Roche; Spring Bank Pharmaceuticals; and Swedish Orphan Biovitrum. **AB** received research funding from MYR GmbH.

Introduction

- Hepatitis delta virus (HDV) represents the most severe form of chronic viral hepatitis and is estimated to affect between 10 and 20 million people worldwide[1]
- Bulevirtide (BLV), a novel entry inhibitor of HDV, is approved in the European Union at 2 mg/day for the treatment of adults with chronic hepatitis delta (CHD) and compensated liver disease[2]
- BLV is a linear 47–amino acid chemically synthesized lipopeptide
- Early results from a Phase 1/2 study (MYR201) reported no correlation between antidrug antibody (ADA) development and the efficacy, safety, or pharmacokinetic (PK) profile of BLV after 24 weeks[3]

Objective

- To evaluate the impact of ADA development on the efficacy, safety, and PK of 48-week BLV monotherapy during treatment for CHD

Methods

Figure 1. Study Design

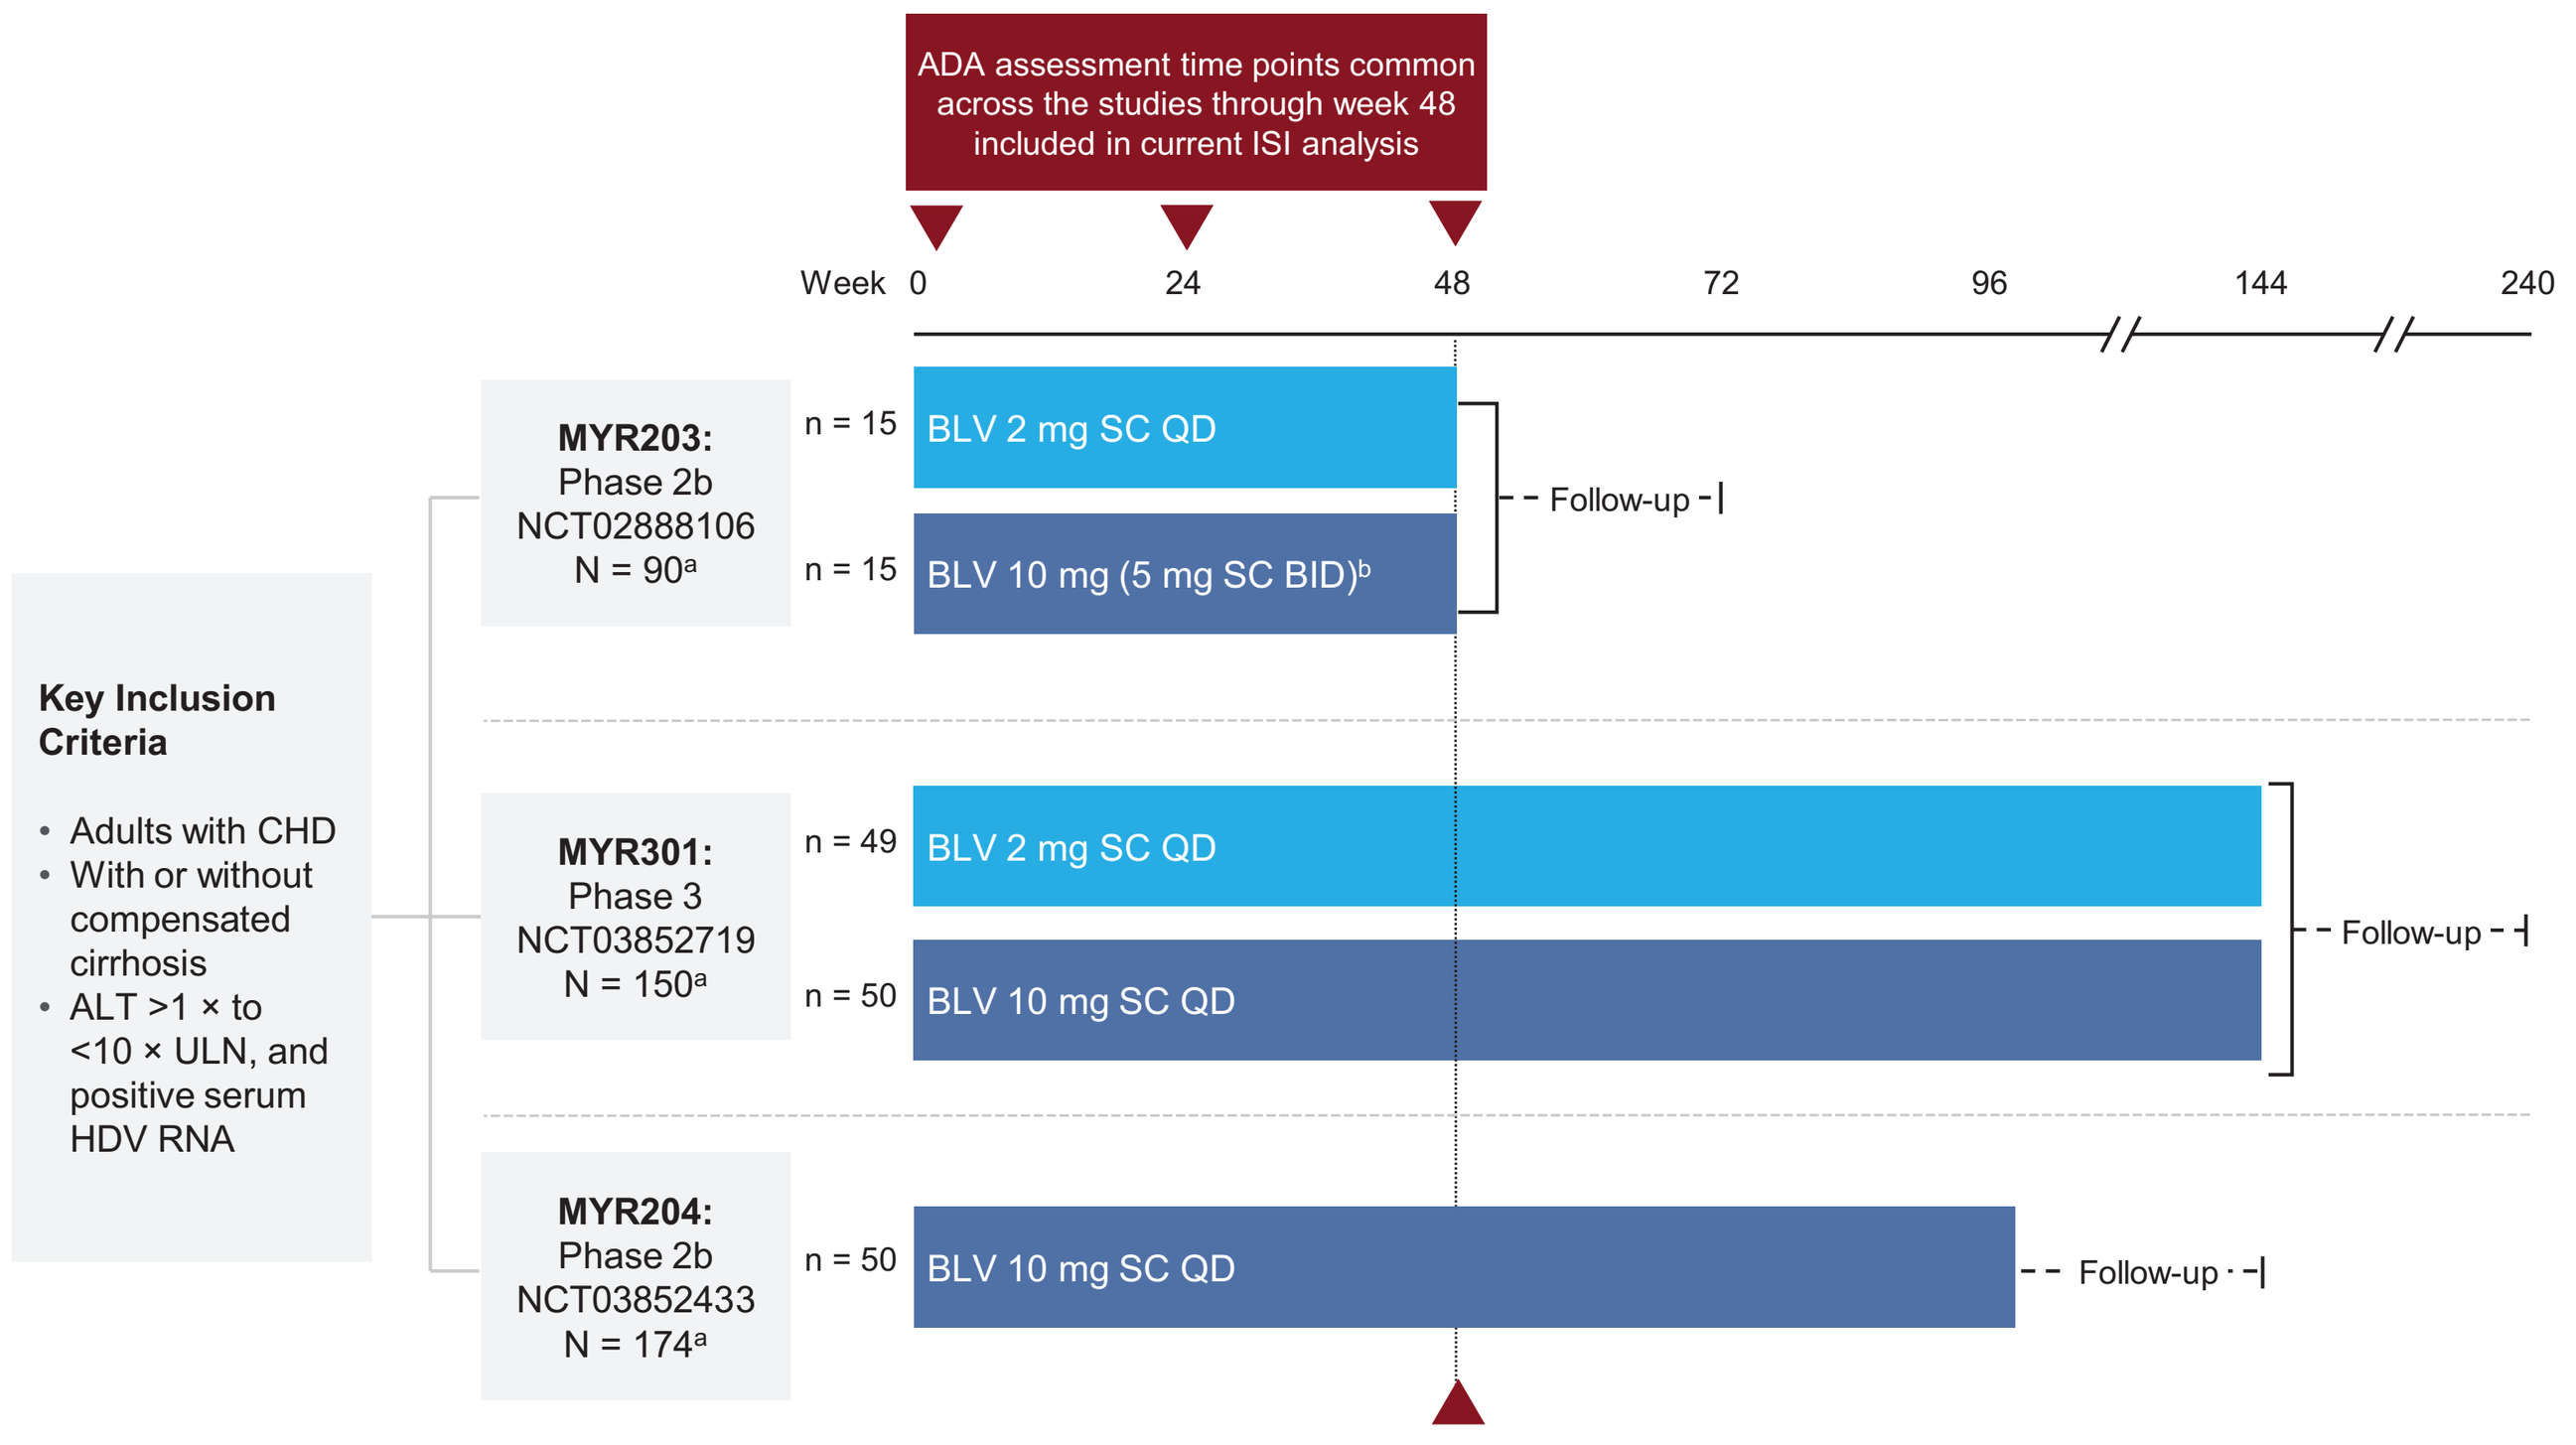
ADA assessment time points common across the studies through week 48 included in current ISI analysis
Week 0 24 48 72 96 144 240
Key Inclusion Criteria
• Adults with CHD
• With or without compensated cirrhosis
• ALT >1 × to <10 × ULN, and positive serum HDV RNA
MYR203: Phase 2b NCT02888106 N = 90a
n = 15 BLV 2 mg SC QD
n = 15 BLV 10 mg (5 mg SC BID)b
Follow-up
MYR301: Phase 3 NCT03852719 N = 150a
n = 49 BLV 2 mg SC QD
n = 50 BLV 10 mg SC QD
Follow-up
MYR204: Phase 2b NCT03852433 N = 174a
n = 50 BLV 10 mg SC QD
Follow-up

[a]Total number of treated study patients; only arms pooled for integrated analyses are shown; groups treated with the combination of BLV and pegylated interferon are not included in this analysis. [b]In combination with TDF.
ADA, antidrug antibody; ALT, alanine aminotransferase; BID, twice daily; BLV, bulevirtide; CHD, chronic hepatitis delta; HDV, hepatitis delta virus; ISI, integrated summary of immunogenicity; QD, once daily; SC, subcutaneous; TDF, tenofovir disoproxil fumarate; ULN, upper limit of normal.

- Completed and ongoing Phase 2 and 3 studies that include BLV monotherapy up to week 48 are included in this integrated summary of immunogenicity analysis
 - The efficacy analysis included studies MYR203 and MYR301. Efficacy parameters assessed included virologic and biochemical response rates
 - Safety and PK analysis included studies MYR203, MYR301, and MYR204. Safety parameters assessed included graded treatment-emergent adverse events (TEAEs) and hypersensitivity or immune-mediated TEAEs. TEAEs were coded according to MedDRA version 24.0
 - ADA assessment time points that were evaluated across the 3 studies through week 48 are baseline, week 24, and week 48
- ADA detection was performed via an enzyme-linked immunosorbent assay and was developed and validated at Prolytic GmbH (Frankfurt, Germany), with a calibrated range of 500 to 10,000 ng/mL and a sensitivity of 250 ng/mL
- The incidence of ADA development (ADA+) was defined as ADA negativity or missing at baseline with subsequent ADA positivity at any postbaseline visit through week 48. Patients with at least one available ADA measurement postbaseline were included
- ADA assessments through week 48 were done at the following time points:
 - MYR203: day 1, week 12, week 24, and week 48
 - MYR204 and MYR301: day 1, week 16, week 24, and week 48

Results

Table 1. Baseline Characteristics by Dose and ADA Incidence by Week 48

	BLV 2 mg		BLV 10 mg	
	ADA+ (n = 15)	ADA− (n = 49)	ADA+ (n = 25)	ADA− (n = 88)
Age, years, mean (SD)	41 (7)	44 (10)	40 (9)	40 (8)
Male sex, n (%)	11 (73)	30 (61)	16 (64)	63 (72)
White race, n (%)	13 (87)	43 (88)	23 (92)	77 (88)
Cirrhosis, n (%)	6 (40)	20 (41)	8 (32)	32 (36)
Concomitant HBV NUC treatment, n (%)	7 (47)	24 (49)	9 (36)	40 (45)
ALT, U/L, mean (SD)	121.7 (97.5)	107.7 (64.7)	107.0 (65.2)	116.9 (96.8)
Platelet count, 10^9/L, mean (SD)	164.2 (57.3)	158.0 (53.2)	193.6 (46.3)	168.8 (51.1)
HDV RNA, $\log_{10}$ IU/mL, mean (SD)	4.8 (1.5)	5.4 (1.2)	5.5 (1.7)	5.2 (1.3)
HBsAg, $\log_{10}$ IU/mL, mean (SD)	3.7 (0.5)	3.8 (0.5)	3.8 (0.7)	3.7 (0.7)
LSM, kPa, mean (SD)	14.9 (10.9)	13.7 (6.7)	12.5 (5.5)	13.5 (8.4)
Total bile acid, μmol/L, mean (SD)	16.1 (17.5)	13.3 (11.8)	10.6 (6.5)	14.1 (11.6)

Demographics of total cohort include MYR203, MYR301, MYR204.
ADA, antidrug antibody; ALT, alanine aminotransferase; BLV, bulevirtide; HBsAg, hepatitis B surface antigen; HBV, hepatitis B virus; HDV, hepatitis delta virus; LSM, liver stiffness measurement; NUC, nucleos(t)ide analogue.

Table 2. Onset and Duration of On-Treatment ADA Incidence by Week 48

	BLV 2 mg	BLV 10 mg	Total
ADA onset, weeks, mean (SD)	26 (14.1) n = 15	21 (11.3) n = 25	23 (12.5) n = 40
ADA duration, weeks, mean (SD)	11 (12.5) n = 11	7 (8.3) n = 22	9 (9.9) n = 33

ADA, antidrug antibody; BLV, bulevirtide.

- Incidence rates for ADA development were similar with BLV 2 and 10 mg (23% and 22%, respectively). For this analysis, only patients who were evaluable for ADA incidence were included
- At baseline, 3 patients were ADA+ in the BLV 2- and 10-mg groups each

Figure 2. Efficacy by Week 48 of BLV Monotherapy by ADA Incidence

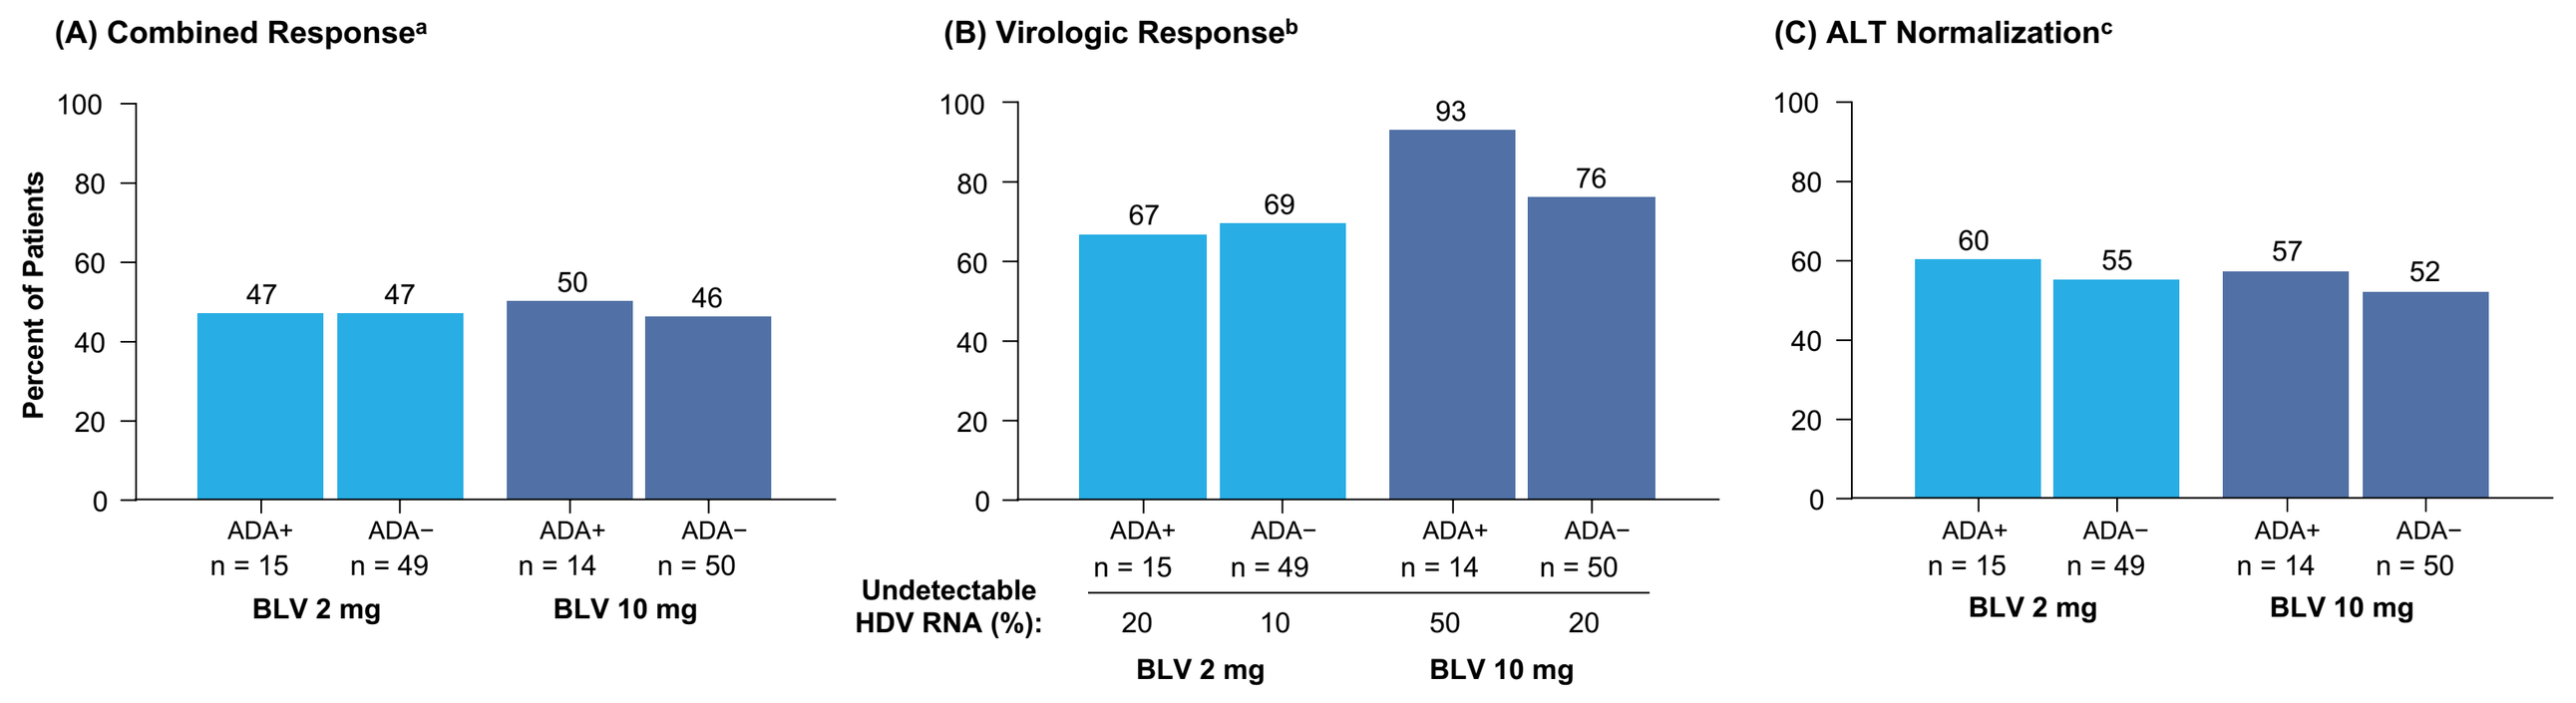
(A) Combined Response[a]
Percent of Patients
47 47 50 46
ADA+ n = 15 ADA− n = 49 BLV 2 mg
ADA+ n = 14 ADA− n = 50 BLV 10 mg
(B) Virologic Response[b]
67 69 93 76
Undetectable HDV RNA (%): 20 10 50 20
(C) ALT Normalization[c]
60 55 57 52

[a]Combined response determined as undetectable HDV RNA or ≥2 $\log_{10}$ IU/mL decrease from baseline and ALT normalization. [b]Virologic response determined as undetectable HDV RNA or ≥2 $\log_{10}$ IU/mL decrease from baseline. [c]ALT normalization was defined at Russian sites as ≤31 U/L for women and ≤41 U/L for men, and at all other sites as ≤34 U/L for women and ≤49 U/L for men. Efficacy ISI analysis includes MYR203 and MYR301.
ADA, antidrug antibody; ALT, alanine aminotransferase; BLV, bulevirtide; HDV, hepatitis delta virus; ISI, integrated summary of immunogenicity.

Figure 3. HDV RNA Decline Over 48 Weeks of BLV Monotherapy

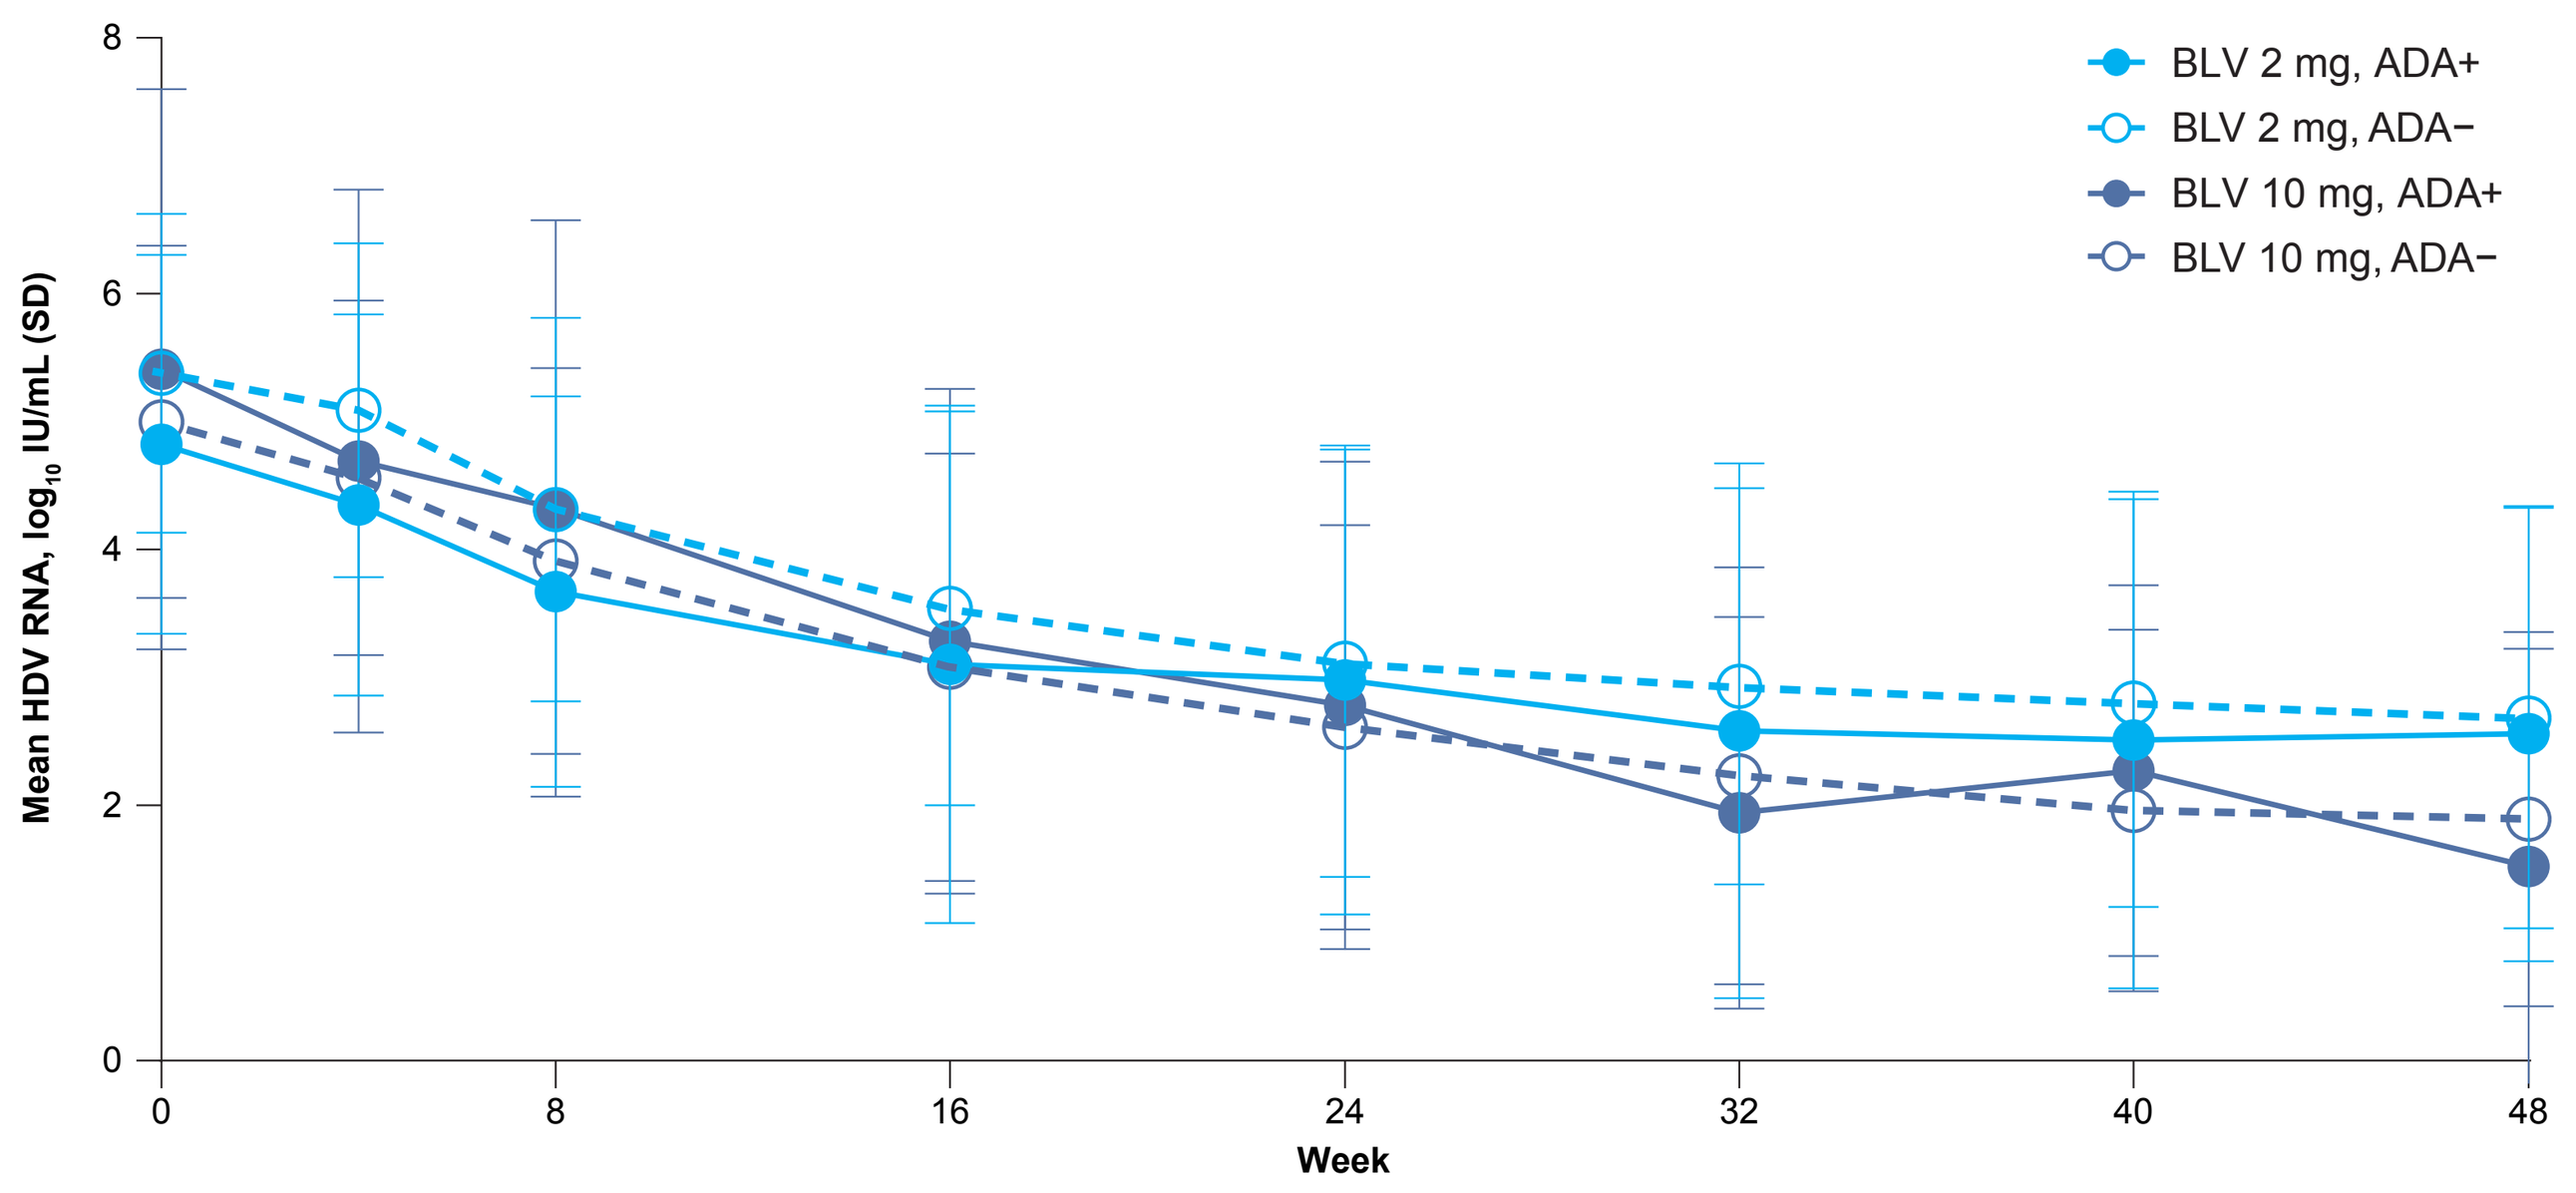
Mean HDV RNA, log10 IU/mL (SD)
Week
BLV 2 mg, ADA+
BLV 2 mg, ADA−
BLV 10 mg, ADA+
BLV 10 mg, ADA−

ADA, antidrug antibody; BLV, bulevirtide; HDV, hepatitis delta virus.

- ADA status did not have a clinically meaningful impact on HDV RNA decline with either BLV treatment group
- Combined, virologic, and biochemical response rates were similar regardless of ADA status in both BLV treatment groups

Table 3. Safety Outcomes by Dose and ADA Incidence by Week 48

	BLV 2 mg		BLV 10 mg	
Preferred Term, n (%)	ADA+ (n = 15)	ADA− (n = 49)	ADA+ (n = 25)	ADA− (n = 88)
TEAEs	15 (100)	40 (82)	22 (88)	76 (86)
TEAEs related to BLV	11 (73)	27 (55)	16 (64)	55 (63)
TEAEs of Grade ≥3	0	7 (14)	0	13 (15)
TE SAEs	0	2 (4)	1 (4)	1 (1)
TEAEs leading to premature BLV discontinuation	0	0	0	0
All deaths	0	0	0	0
TEAEs, ISR[a]	6 (40)	4 (8)	5 (20)	18 (20)
TEAEs, hypersensitivity[b]	0	2 (4)	0	5 (6)
TEAEs, immune-mediated/autoimmune[c]	1 (7)	0	0	1 (1)

[a]ISR terms included erythema, reaction, pruritus, pain, hematoma, swelling, dermatitis, induration, rash, coldness, and irritation. [b]Hypersensitivity terms included rash, injection-site dermatitis, injection-site rash, angioedema, rash macular, and toxic skin eruption. [c]Immune-mediated/autoimmune terms included chronic gastritis and psoriasis.
ADA, antidrug antibody; BLV, bulevirtide; ISR, injection-site reaction; SAE, serious adverse event; TE, treatment-emergent; TEAE, treatment-emergent adverse event.

- Rates of TEAEs, including those of Grade ≥3 and serious AEs, were comparable between subgroups
- No correlation was observed between ADA development and hypersensitivity or immune-mediated TEAEs

Figure 4. Total Bile Acid Levels Over 48 Weeks of BLV Monotherapy

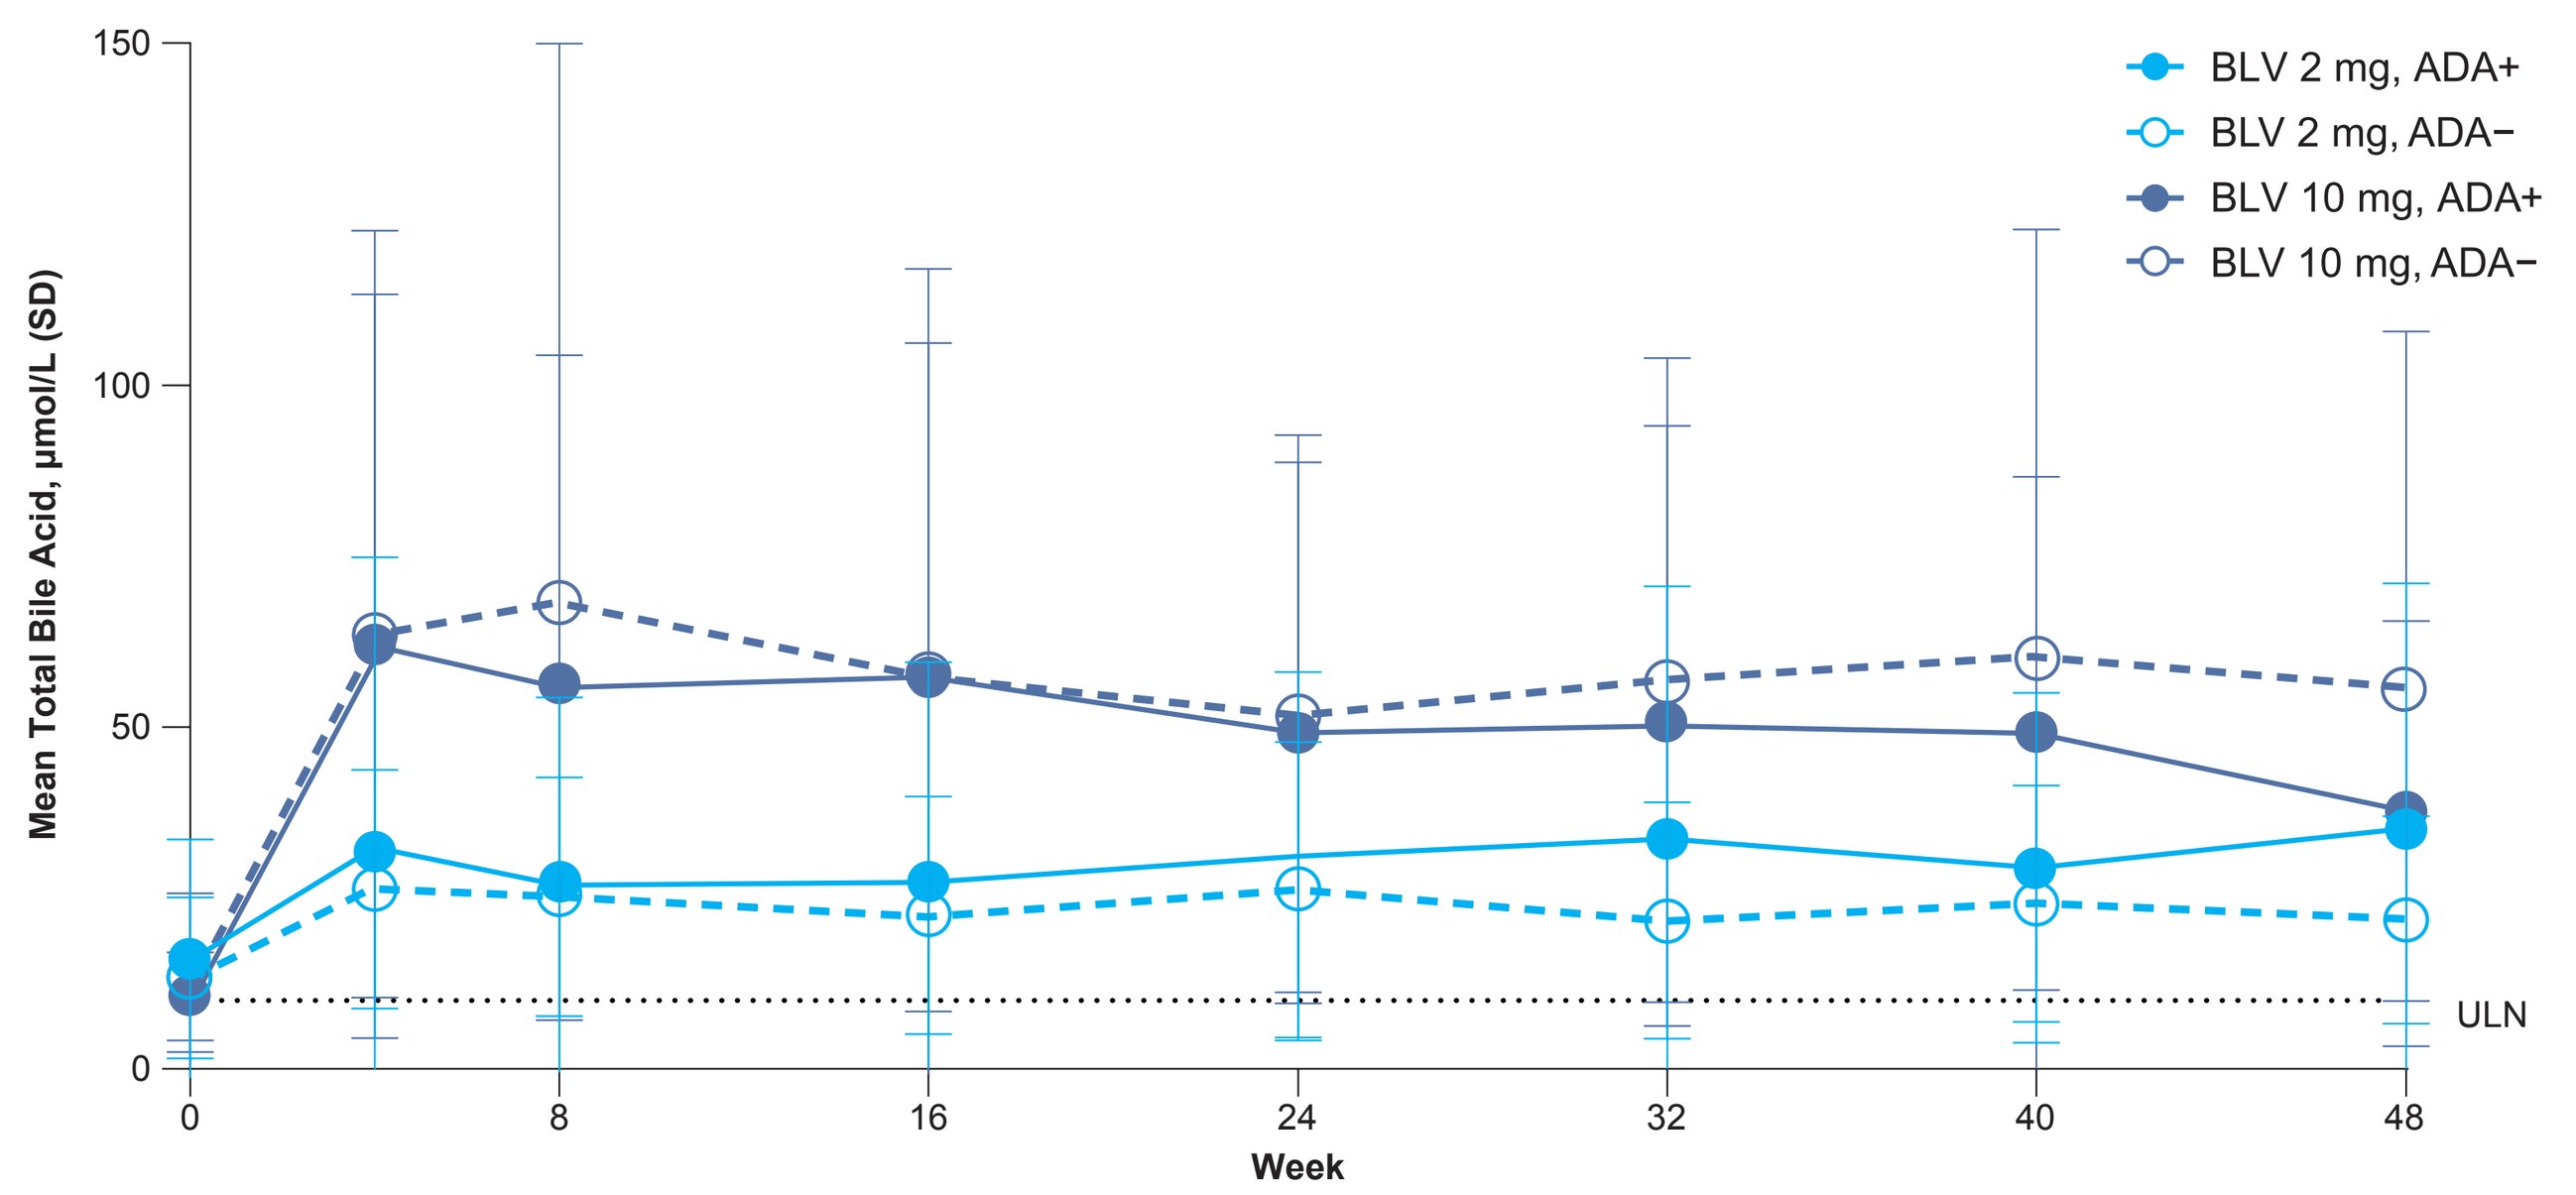
Mean Total Bile Acid, μmol/L (SD)
Week
ULN
BLV 2 mg, ADA+
BLV 2 mg, ADA−
BLV 10 mg, ADA+
BLV 10 mg, ADA−

ADA, antidrug antibody; BLV, bulevirtide; ULN, upper limit of normal.

- Dose-dependent asymptomatic elevations in total bile acids were not impacted by ADA incidence

Figure 5. BLV PK by ADA Incidence

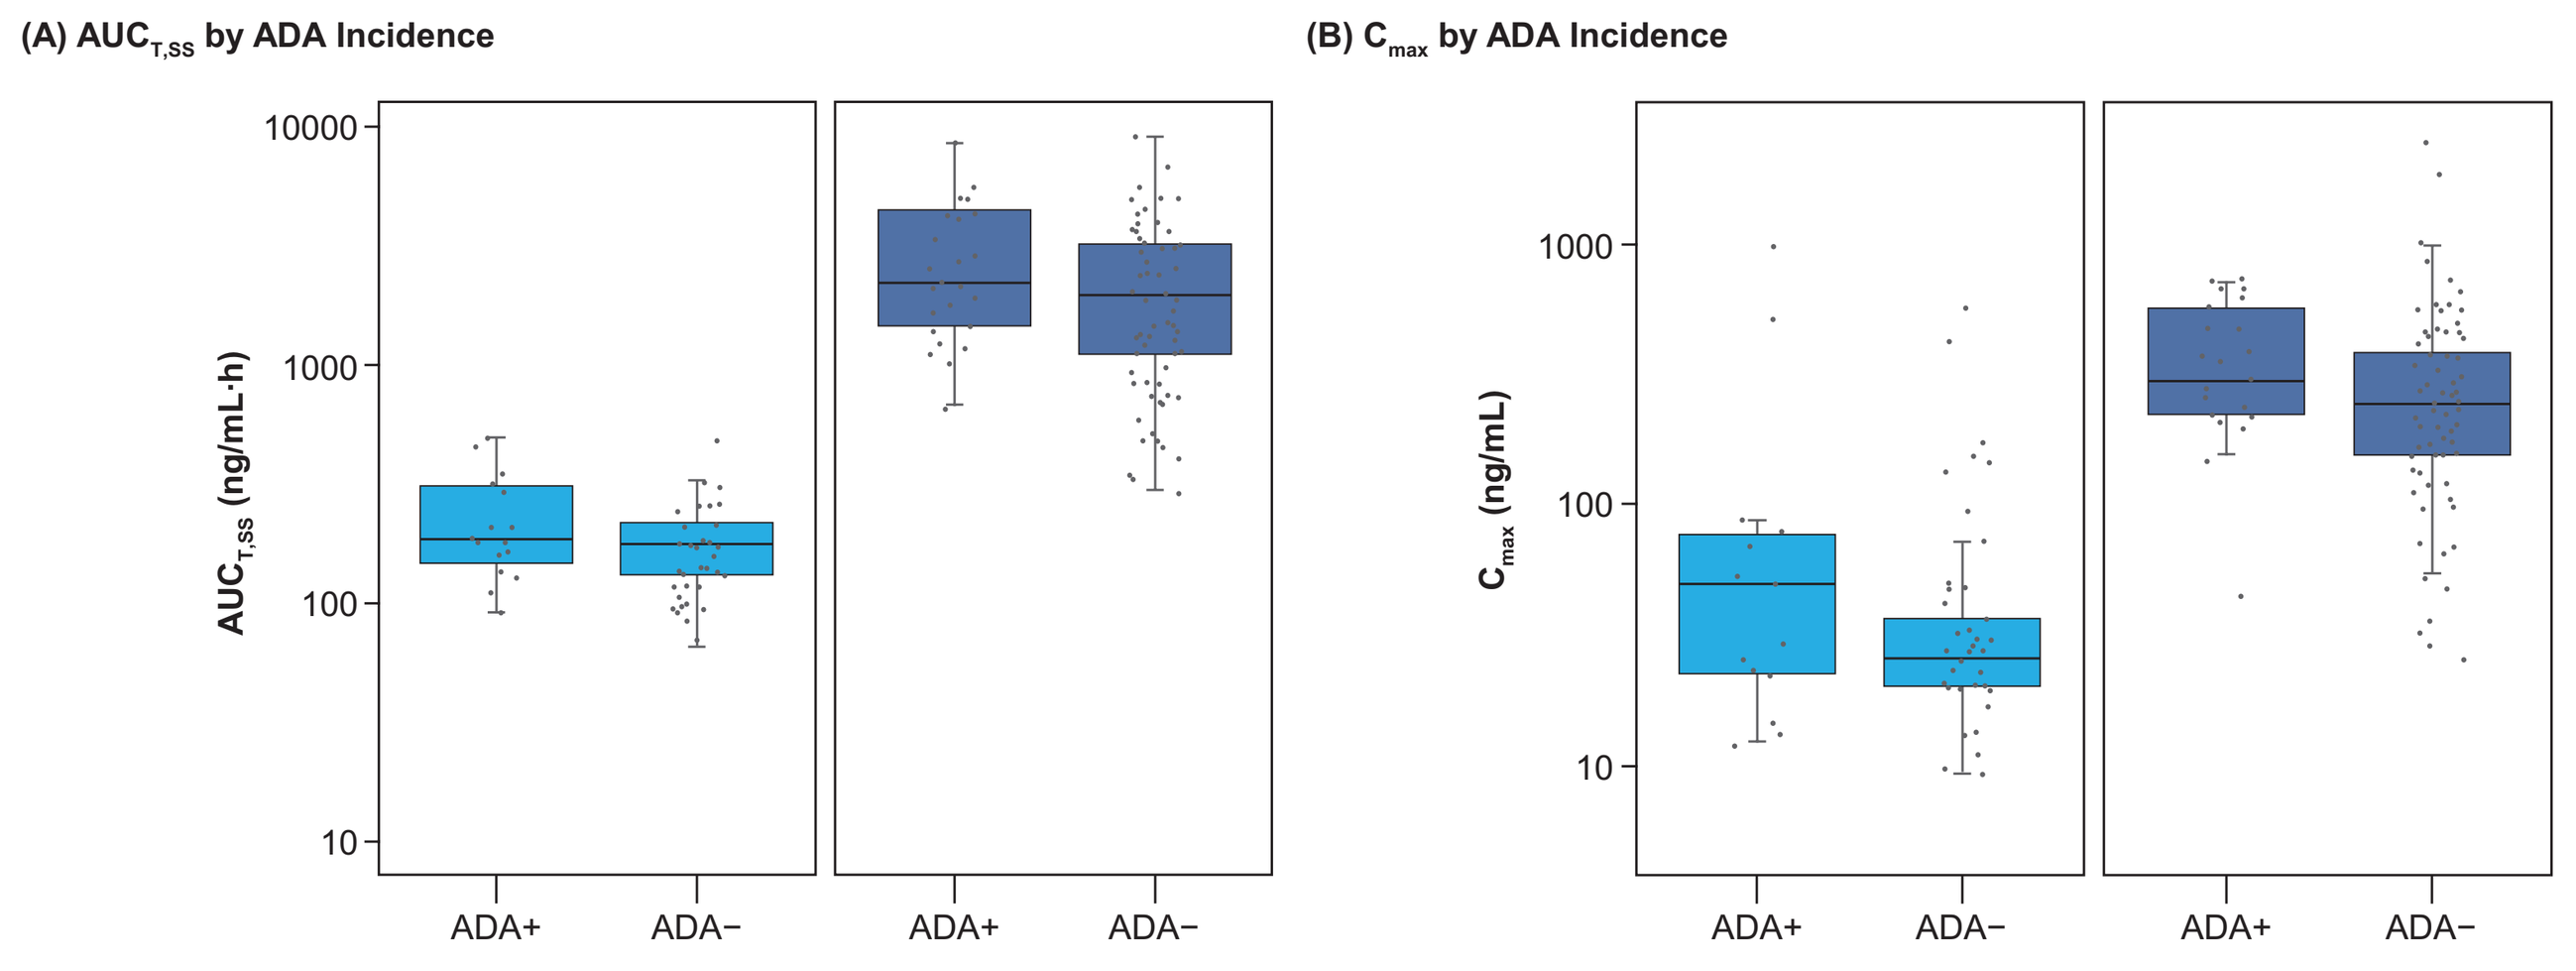
(A) AUCτ,ss by ADA Incidence
AUCτ,ss (ng/mL·h)
(B) Cmax by ADA Incidence
Cmax (ng/mL)
ADA+ ADA− ADA+ ADA−

	2 mg		10 mg	
	ADA+ (n = 15)	ADA− (n = 49)	ADA+ (n = 25)	ADA− (n = 88)
$AUC_{\tau,SS}$, ng/mL·h, geometric mean	202	165	2330	1760
GMR ADA+/ADA− (90% CI)	1.22 (1.0–1.6)		1.32 (1.0–1.7)	

	2 mg		10 mg	
	ADA+ (n = 15)	ADA− (n = 49)	ADA+ (n = 25)	ADA− (n = 88)
C_{max}, ng/mL, geometric mean	51.9	31.5	329	237
GMR ADA+/ADA− (90% CI)	1.65 (0.9–3.0)		1.39 (1.1–1.8)	

Boxplots depict the median, 25th percentile, and 75th percentile of $AUC_{\tau,ss}$ and C_{max} for patients receiving 2 or 10 mg bulevirtide from population PK modeling; gray dots represent individual patient data points.
ADA, antidrug antibody; $AUC_{\tau,ss}$, area under the plasma concentration-time curve over a dosing interval at steady state; BLV, bulevirtide; C_{max}, maximum observed concentration of drug; GMR, geometric mean ratio; PK, pharmacokinetic.

- Development of ADAs did not meaningfully influence BLV PK exposure

AASLD: The Liver Meeting; November 10–14, 2023; Boston, MA, USA