Efficacy and Safety of Tenofovir Alafenamide (TAF) at 2 Years in Children and Adolescents With Chronic Hepatitis B (CHB)

1414-C

Kathleen B Schwarz[1], Jorge A Bezerra[2], Byung Ho Choe[3], Chuan-Hao Lin[4], Frida Abramov[5], Hongyuan Wang[5], Yang Liu[5], John F Flaherty[5], Daniela Pacurar[6], Kyung Mo Kim[7], Ilsiyar Khaertynova[8], Dr Shalimar[9], Jia-Feng Wu[10], Manish Tandon[11], Philip Rosenthal[12], Viacheslav Morozov[13], Etienne Sokal[14], Mei Hwei Chang[15]

[1]UC San Diego School of Medicine, La Jolla, and Rady Children's Hospital-San Diego, San Diego, CA, USA; [2]University of Cincinnati, Cincinnati Children's Hospital Medical Center, Cincinnati, OH, USA; [3]Kyungpook National University College of Medicine, Daegu, Republic of Korea; [4]Children's Hospital Los Angeles, Los Angeles, CA, USA; [5]Gilead Sciences, Inc., Foster City, CA, USA; [6]Universitatea de Medicina si Farmacie "Carol Davila" din Bucuresti, Spitalul Clinic de Urgenta pentru Copii "Grigore Alexandrescu," Bucuresti, Romania; [7]University of Ulsan College of Medicine, Asan Medical Center Children's Hospital, Seoul, Republic of Korea; [8]Kazan State Medical University, Kazan, Russian Federation; [9]All India Institute of Medical Sciences, New Delhi, India; [10]National Taiwan University Hospital, Taipei City, Taiwan; [11]M. V. Hospital and Research Centre, Lucknow, India; [12]University of California San Francisco, San Francisco, CA, USA; [13]Hepatolog, Samara, Russian Federation; [14]Université Catholique de Louvain, Louvain-la-Neuve, Cliniques Universitaires Saint-Luc, Brussels, Belgium; [15]National Taiwan University Children's Hospital, Taipei City, Taiwan

Copies of this poster obtained through QR (Quick Response) and/or text key codes are for personal use only and may not be reproduced without written permission of the authors.

Key Findings

- **With longer-term TAF treatment, increasing proportions of pediatric patients achieved virologic suppression, ALT normalization, and HBeAg loss with seroconversion at Week 96**
- **Over 2 years of study treatment, no study patients discontinued due to an adverse event**
- **Changes in whole-body and spine BMD and in renal parameters were not impacted in a clinically meaningful way over 96 weeks of TAF treatment**

Conclusions

In pediatric patients with CHB, treatment responses improved with prolonged (2 years) TAF treatment

A favorable safety profile was maintained through 2 years of TAF treatment

References: **1.** EASL. *J Hepatol.* 2017;67:370-98. **2.** Jonas MM, et al. *Hepatology*. 2016;63:377-87. **3.** Murray KF, et al. *Hepatology*. 2012;56:2018-26. **4.** Terrault NA, et al. *Hepatology.* 2018;67:1560-99. **5.** Cathcart AI, et al. *Antimicrob Agents Chemother.* 2018;62(10):e01064-81.

Acknowledgments: We extend our thanks to the patients, their families, and all participating investigators: Belgium: E Sokal; Hong Kong: GLH Wong; India: A Chowdhury, R Mehta, Dr Shalimar, A Shukla, M Tandon; New Zealand: H Evans; Republic of Korea: BH Choe, YH Choe, KM Kim; Romania: D Pacurar, A Streinu-Cercel; Russian Federation: I Khaertynova, Y Lobzin, V Morozov, T Strokova; Taiwan: MH Chang, MW Lai, HH Shih, JF Wu, YJ Yang; USA: J Bezerra, J Daniel, L Gillis, JR Honegger, S Horslen, CH Lin, N Mittal, D Mogul, K Murray, MR Narkewicz, D Pan, Y Pham, G Rao, P Rosenthal, K Schwarz, M Whitworth. This study was funded by Gilead Sciences, Inc., Foster City, CA, USA. Editorial and writing support were provided by Helen Rodgers, PhD, of AlphaScientia, a Red Nucleus company, and funded by Gilead Sciences, Inc.

Disclosures: **KBS** reports research grants funded by Albireo; Gilead Sciences, Inc.; and National Institute of Diabetes and Digestive and Kidney Diseases; and consulting for Gilead Sciences, Inc.; Mirum; Sarepta; and Up to Date. **JAB** reports grant and research support from Gilead Sciences, Inc., and Shyer. **CHL** reports grant and research support from Gilead Sciences, Inc., and Mirum. **FA, HW, YL,** and **JFF** are employees of Gilead Sciences, Inc., and may own stock or stock options. **DP** reports speaking and teaching opportunities from Angellini, Bristol Myers Squibb, Dr Phyto, and Secom; and grant and research support from Gilead Sciences, Inc. **KMK** reports grant and research support from Celltrion. **PR** reports grant and research support from AbbVie; Albireo; Arrowhead; Gilead Sciences, Inc.; Merck; Mirum; Takeda; and Travere; and consulting for Albireo; Ambrys; Audentes; BioMarin; Dicerna; Encoded; Gilead Sciences, Inc.; Mirum; MedinCell; Takeda; and Travere. **BHC, IK, DrS, JFW, MT, VM, ES,** and **MHC** report no conflicts of interest.

Introduction

- Children and adolescents with chronic hepatitis B (CHB) have an increased lifetime risk for clinical complications, including progression to cirrhosis, decompensated liver disease, and liver cancer[1]
- First-line treatment options for children with CHB are generally similar to those for adults; however, there are potential safety and efficacy limitations, including reduced response rates and/or development of resistance in patients receiving entecavir with prior lamivudine experience and bone mineral density (BMD) reductions (relative to placebo [PBO]) with use of tenofovir disoproxil fumarate (TDF)[2-4]
- Based on week 24 primary endpoint results from Study GS-US-320-1092, a randomized, double-blind, PBO-controlled trial, the US Food and Drug Administration granted approval for tenofovir alafenamide (TAF), a novel prodrug of tenofovir, for the treatment of CHB in patients aged 12 and older with compensated liver disease
- Likewise, the European Medicines Agency expanded the indication for TAF to be used in children aged 6 years and older weighing ≥25 kg based on 48-week findings from GS-US-320-1092

Objective

- To evaluate the efficacy and safety of TAF 25 mg once daily (QD) when given for 96 weeks compared with PBO treatment for 24 weeks followed by TAF 25 mg QD for 72 weeks in adolescents (aged ≥12 to <18 years, weighing ≥35 kg) and children (aged ≥6 to <12 years, weighing ≥25 kg) with CHB

Methods

- Randomized (2:1), double-blind, PBO-controlled, multicenter study (GS-US-320-1092, NCT02932150) consisting of randomized and open-label extension phases

Study Design

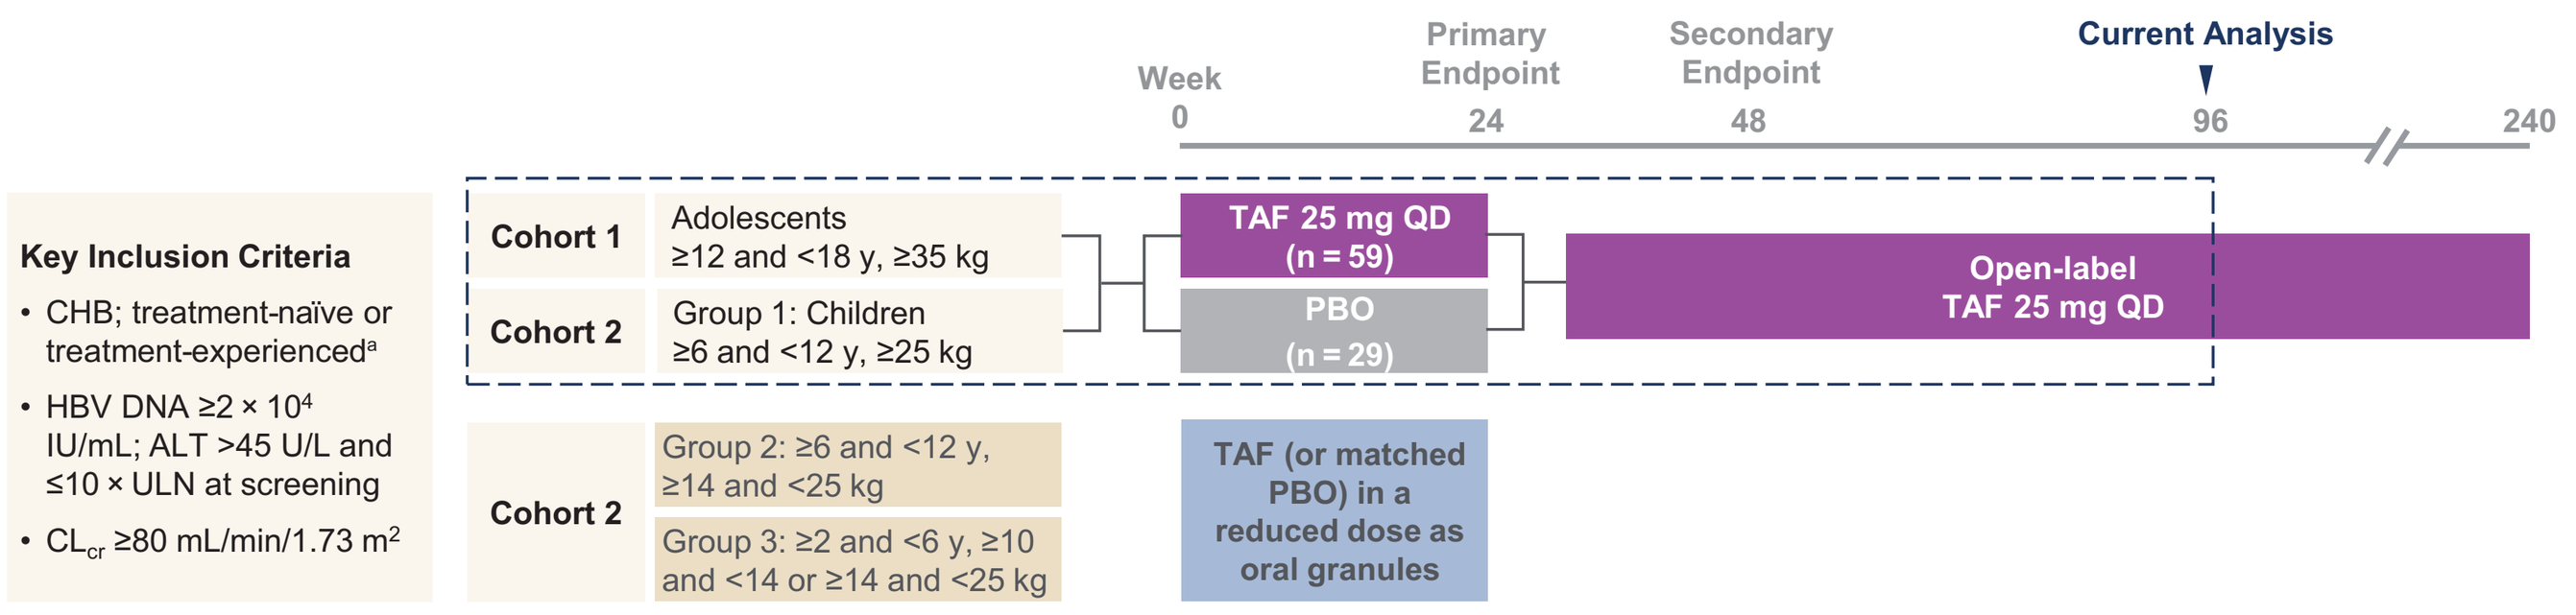
Key Inclusion Criteria
• CHB; treatment-naïve or treatment-experienced[a]
• HBV DNA ≥2 × 10⁴ IU/mL; ALT >45 U/L and ≤10 × ULN at screening
• CLcr ≥80 mL/min/1.73 m²
Week 0
Primary Endpoint 24
Secondary Endpoint 48
Current Analysis 96
240
Cohort 1
Adolescents ≥12 and <18 y, ≥35 kg
Cohort 2
Group 1: Children ≥6 and <12 y, ≥25 kg
TAF 25 mg QD (n = 59)
PBO (n = 29)
Open-label TAF 25 mg QD
Cohort 2
Group 2: ≥6 and <12 y, ≥14 and <25 kg
Group 3: ≥2 and <6 y, ≥10 and <14 or ≥14 and <25 kg
TAF (or matched PBO) in a reduced dose as oral granules

[a]Treatment naïve: <12 weeks of treatment with any oral nucleos(t)ide analogue; treatment experienced: ≥12 weeks of treatment with any oral nucleos(t)ide analogue.
ALT, alanine aminotransferase; CHB, chronic hepatitis B; CL_{cr}, creatinine clearance; HBV, hepatitis B virus; PBO, placebo; QD, once daily; TAF, tenofovir alafenamide; ULN, upper limit of normal.

Efficacy Endpoints

- Antiviral efficacy: Hepatitis B virus (HBV) DNA <20 IU/mL
 - — COBAS AmpliPrep/COBAS TaqMan HBV Test, v2.0 (Roche Diagnostics, Indianapolis, IN; lower limit of quantitation [LLOQ]: 20 IU/mL)
- Biochemical efficacy: Alanine aminotransferase (ALT) normalization by central laboratory and American Association for the Study of Liver Diseases (AASLD upper limit of normal: ≤30 U/L) criteria
- Serology: Hepatitis B e antigen (HBeAg) and hepatitis B surface antigen (HBsAg) loss/seroconversion
- Resistance: HBV polymerase/reverse transcriptase (pol/RT) deep sequencing for patients with HBV DNA ≥69 IU/mL

Safety Endpoints

- Graded adverse events (AEs), serious AEs (SAEs), and laboratory abnormalities
- Bone: changes in spine and whole-body (minus head) BMD and corresponding Z-scores by dual-energy x-ray absorptiometry
- Renal: change in estimated creatinine clearance (Schwartz formula)

Patient Disposition

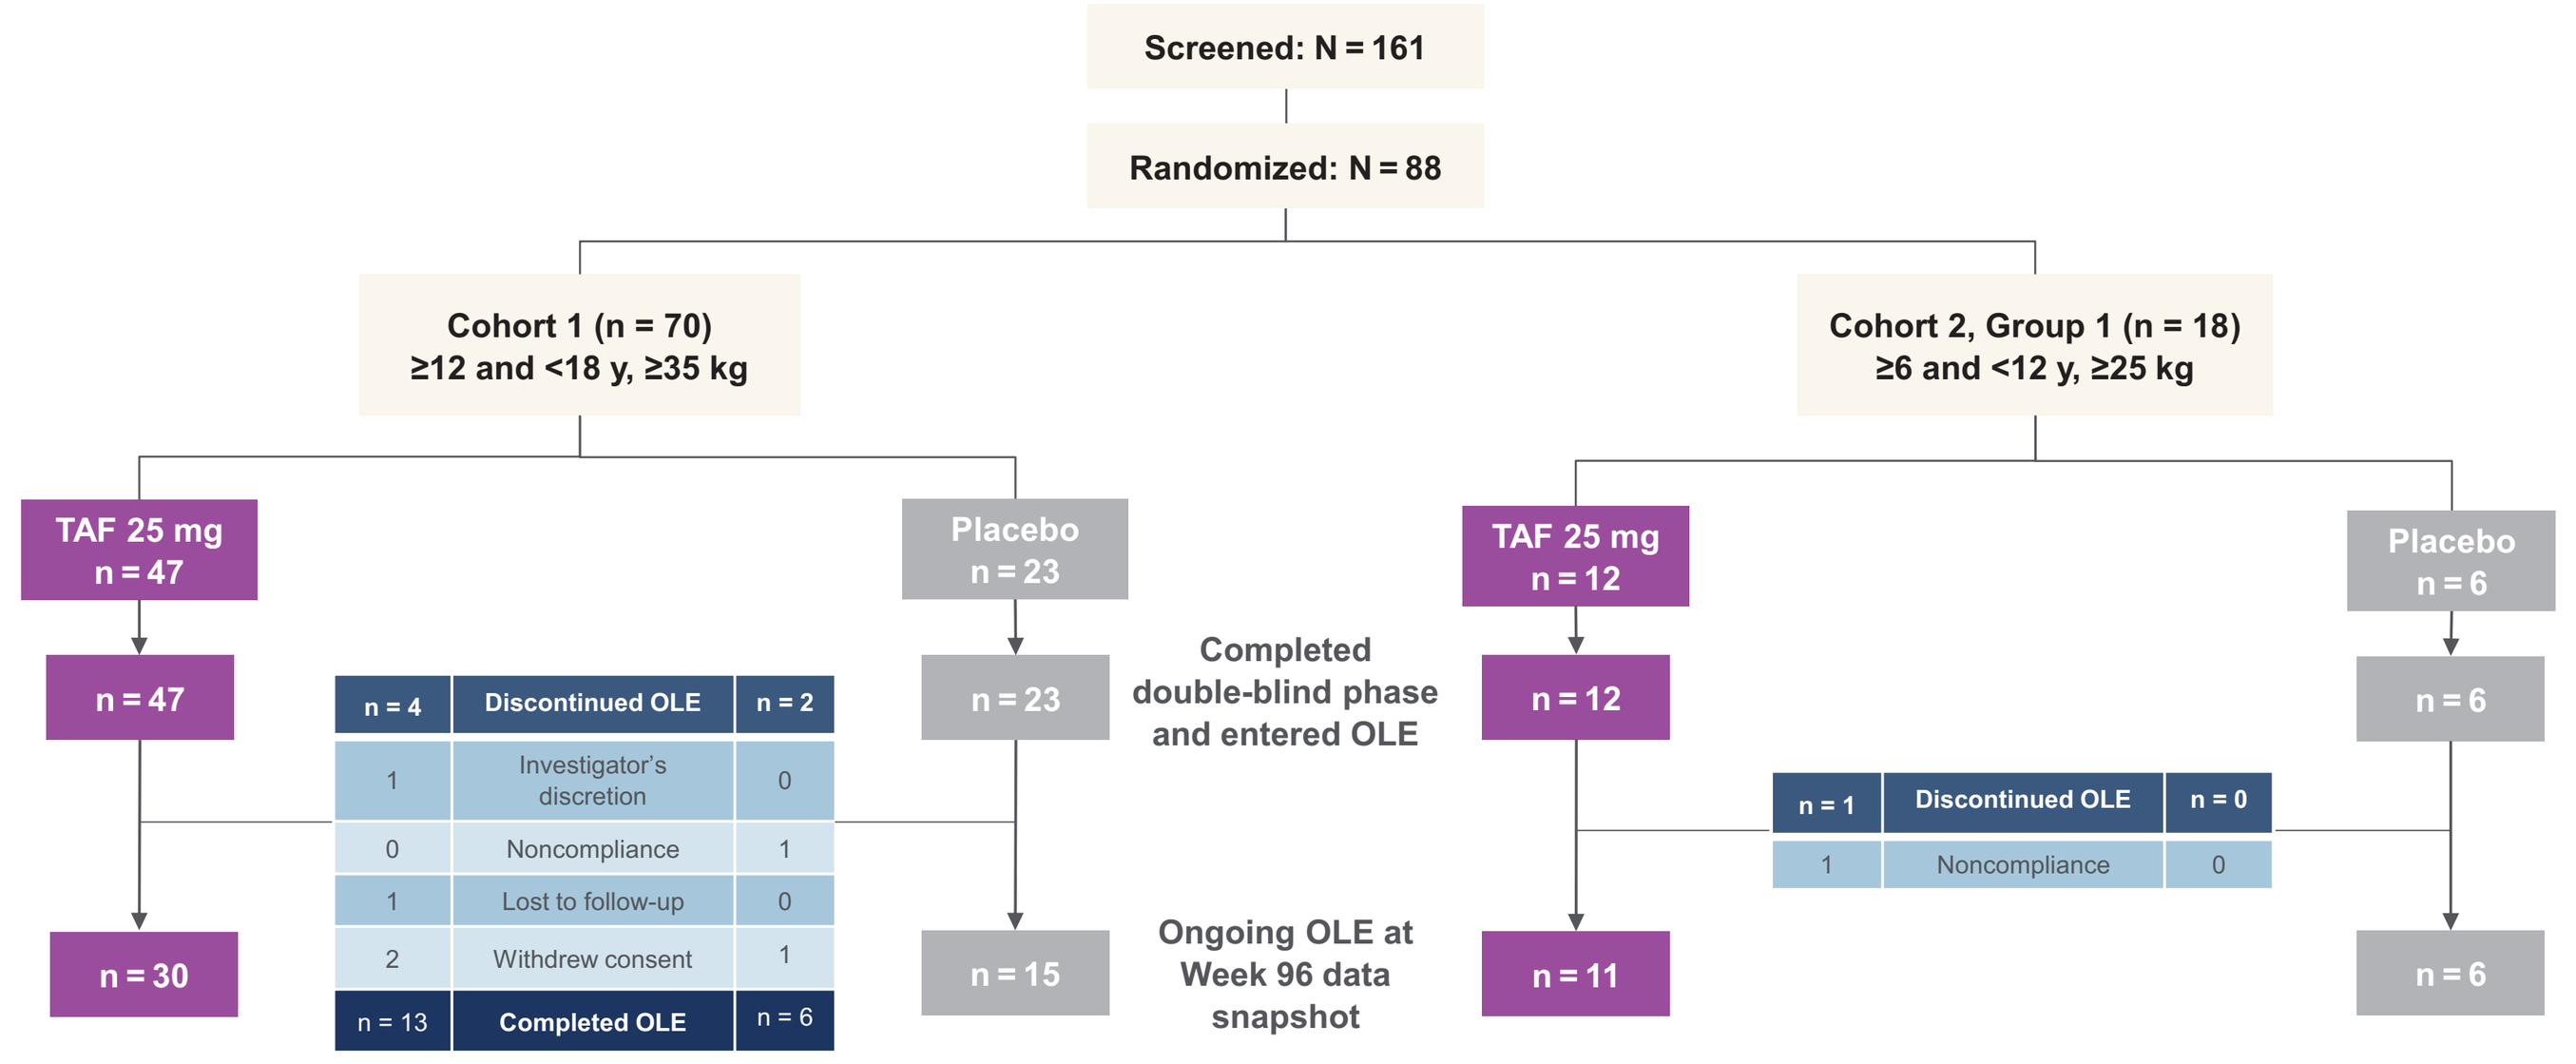
Screened: N = 161
Randomized: N = 88
Cohort 1 (n = 70) ≥12 and <18 y, ≥35 kg
Cohort 2, Group 1 (n = 18) ≥6 and <12 y, ≥25 kg
TAF 25 mg n = 47
Placebo n = 23
TAF 25 mg n = 12
Placebo n = 6
n = 47
n = 23
n = 12
n = 6
Completed double-blind phase and entered OLE
n = 4 | Discontinued OLE | n = 2
1 | Investigator's discretion | 0
0 | Noncompliance | 1
1 | Lost to follow-up | 0
2 | Withdrew consent | 1
n = 13 | Completed OLE | n = 6
n = 1 | Discontinued OLE | n = 0
1 | Noncompliance | 0
n = 30
n = 15
n = 11
n = 6
Ongoing OLE at Week 96 data snapshot

OLE, open-label extension; TAF, tenofovir alafenamide.

Results

Patient Characteristics, Total Population

	TAF (n = 59)	PBO→TAF (n = 29)
Age, years, mean (range)	14 (7–17)	13 (7–17)
Sex, male, n (%)	34 (58)	17 (59)
Asian, n (%)	37 (63)	21 (72)
White, n (%)	16 (27)	6 (21)
Asia region, n (%)[a]	18 (31)	13 (45)
BMI, kg/m², mean (SD)	20.2 (2.91)	19.8 (3.02)
Prior OAV, n (%)	15 (25)	5 (17)
Prior IFN, n (%)	11 (19)	2 (7)
HBV DNA, log_{10} IU/mL, mean (SD)	7.9 (1.12)	8.1 (0.72)
HBV DNA ≥8 log_{10} IU/mL, n (%)	39 (66)	21 (72)
HBsAg, log_{10} IU/mL, mean (SD)	4.4 (0.58)	4.6 (0.55)
HBeAg positive, n (%)	58 (98)	29 (100)
ALT, U/L, median (Q1, Q3)	65 (50, 109)	66 (54, 89)
CL_{Cr}, mL/min/1.73 m², median (Q1, Q3)	154 (137, 169)	149 (143, 180)
HBV genotype, n (%)[b]		
A	5 (9)	1 (4)
B	13 (24)	6 (22)
C	12 (22)	8 (30)
D	24 (44)	12 (44)

[a]Denotes Hong Kong, India, Republic of Korea, and Taiwan. [b]Six patients had an undetermined genotype (TAF, n = 4; placebo, n = 2).
ALT, alanine aminotransferase; BMI, body mass index; CL_{Cr}, creatinine clearance; HBeAg, hepatitis B e antigen; HBsAg, hepatitis B surface antigen; HBV, hepatitis B virus; IFN, interferon; OAV, oral antiviral; Q, quartile; TAF, tenofovir alafenamide.

Proportion With HBV DNA <20 IU/mL Through Week 96

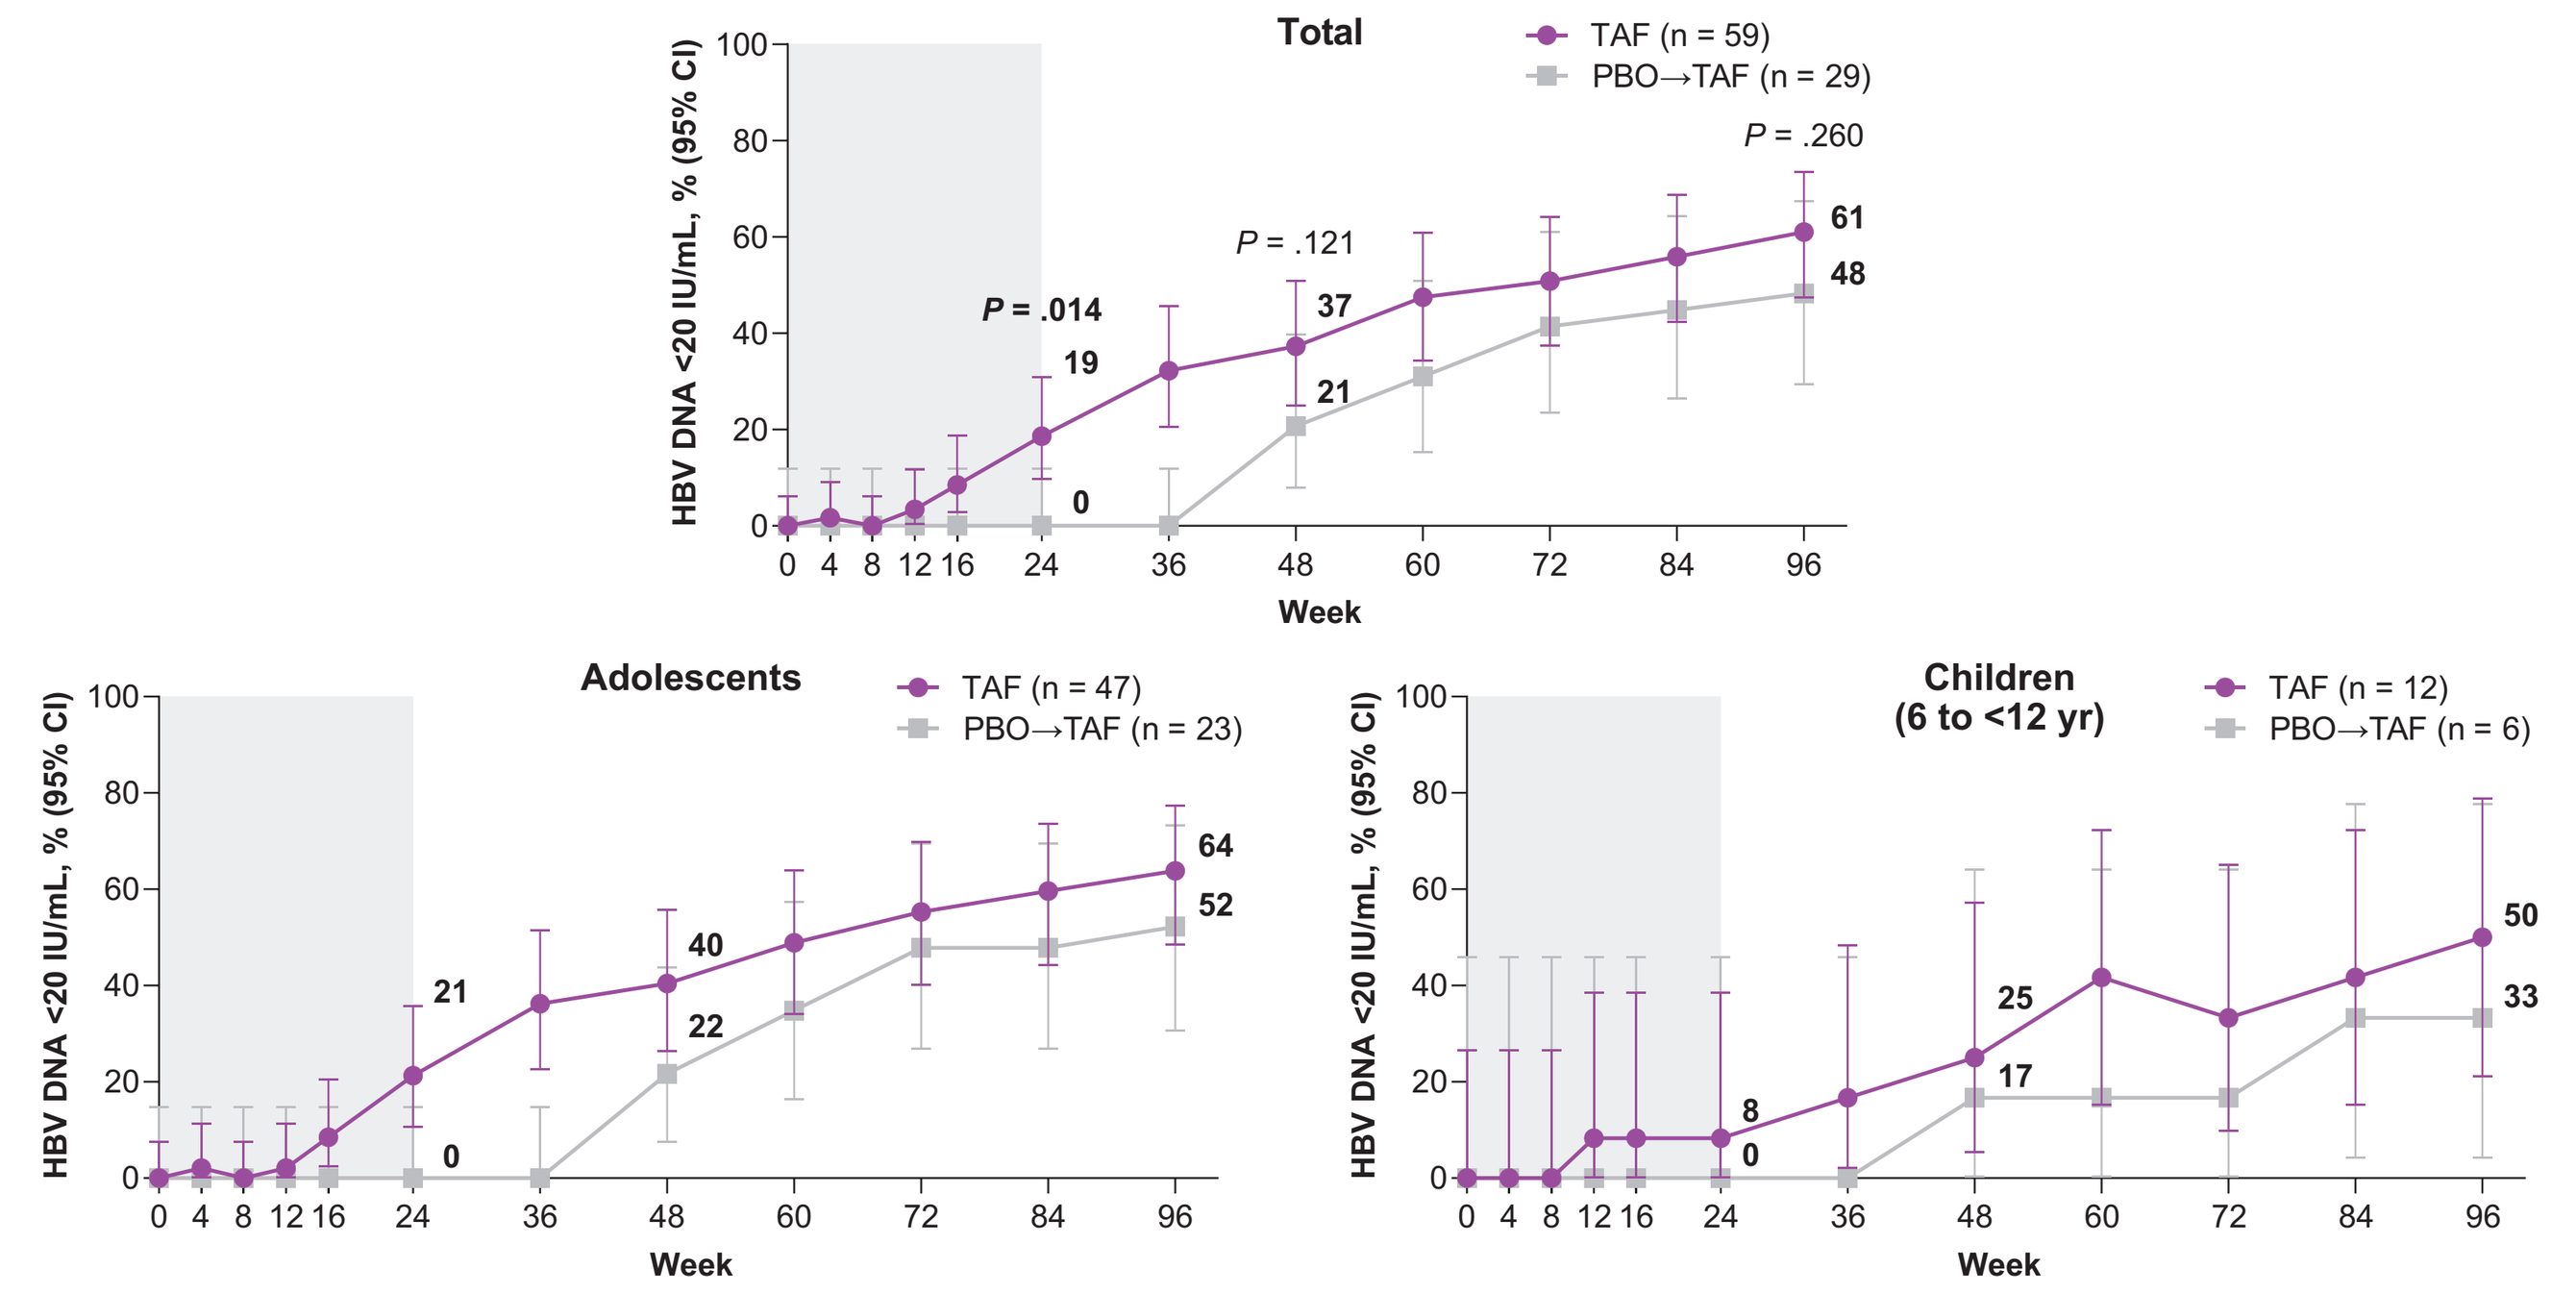
Total
TAF (n = 59)
PBO→TAF (n = 29)
P = .014
P = .121
P = .260
19
0
37
21
61
48
HBV DNA <20 IU/mL, % (95% CI)
Week
Adolescents
TAF (n = 47)
PBO→TAF (n = 23)
21
0
40
22
64
52
Children (6 to <12 yr)
TAF (n = 12)
PBO→TAF (n = 6)
8
0
25
17
50
33

P-values were based on a 2-sided Cochran-Mantel-Haenszel test adjusted for age at baseline. Missing = failure analysis. Shaded area indicates DB phase; unshaded area indicates OL extension phase.
DB, double-blind; HBV, hepatitis B virus; OL, open-label; PBO, placebo; TAF, tenofovir alafenamide.

- Increasing proportions of patients achieved HBV DNA <20 IU/mL over time in both cohorts
- High proportions of children and adolescents receiving TAF exhibited genotype D infection (58% vs 40%, respectively) and high BL viral load of ≥8 log_{10} IU/mL (75% vs 64%, respectively). Both of these factors are associated with a delayed response in adults treated with TDF or TAF[5]

ALT Normalization Over 96 Weeks

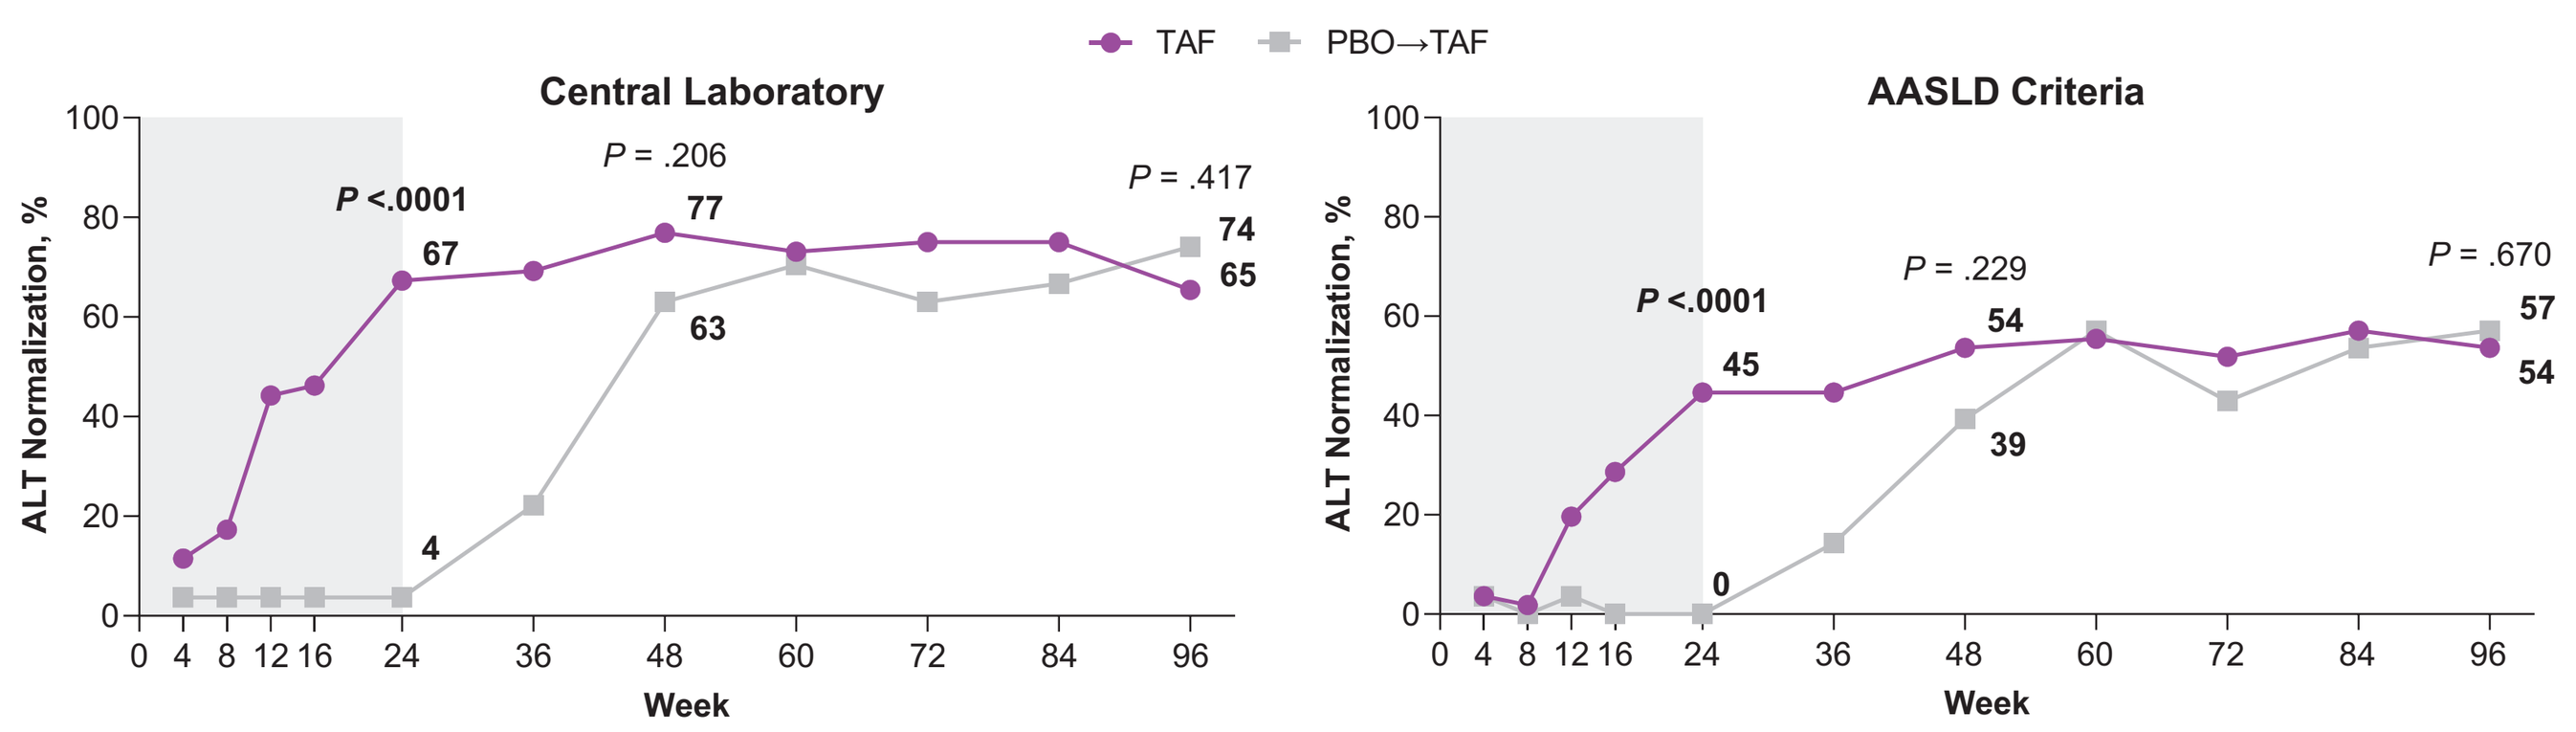
TAF
PBO→TAF
Central Laboratory
P <.0001
P = .206
P = .417
67
4
77
63
74
65
AASLD Criteria
P <.0001
P = .229
P = .670
45
0
54
39
57
54
ALT Normalization, %
Week

The population used for analysis of ALT normalization included only patients with ALT >ULN at baseline; central laboratory ULN: 34 U/L for females aged ≥2 y and males aged 1–9 y, and 43 U/L for males aged ≥9 y; 2018 AASLD ULN: 30 U/L for pediatric males and females. Shaded area of the plot indicates DB phase; unshaded area indicates OL extension phase. *P*-values were based on a 2-sided Cochran-Mantel-Haenszel test adjusted for age at baseline. AASLD, American Association for the Study of Liver Disease; ALT, alanine aminotransferase; DB, double-blind; OL, open-label; PBO, placebo; TAF, tenofovir alafenamide; ULN, upper limit of normal.

- ALT normalization rates increased through week 48 and then remained relatively stable thereafter in the TAF group
- During the open-label phase, ALT normalization in the PBO→TAF group showed improvements comparable to the TAF group by week 60

Serology at Week 96, Total Population

Patients, n/N (%)		TAF (n = 59)	PBO→TAF (n = 29)
HBeAg[a]	Loss	14/58 (24)	5/29 (17)[b]
	Seroconversion	14/58 (24)	5/29 (17)[c]
HBsAg	Loss	1/59 (2)	0
	Seroconversion	0	0

[a]The population used for HBeAg analysis included only patients who were HBeAg positive and HBeAb negative or missing at baseline. HBsAg analysis included all available data. [b]*P* = .48 (2-sided Wilcoxon rank sum test). [c]*P* = .48 (2-sided Wilcoxon rank sum test). *P*-values were based on a 2-sided Cochran-Mantel-Haenszel test adjusted for age at baseline.
HBeAb, hepatitis B e antibody; HBeAg, hepatitis B e antigen; HBsAg, hepatitis B surface antigen; PBO, placebo; TAF, tenofovir alafenamide.

- At week 96, increasing proportions of patients experienced HBeAg loss and seroconversion with TAF treatment compared with earlier analyses

Resistance Analysis at Week 96

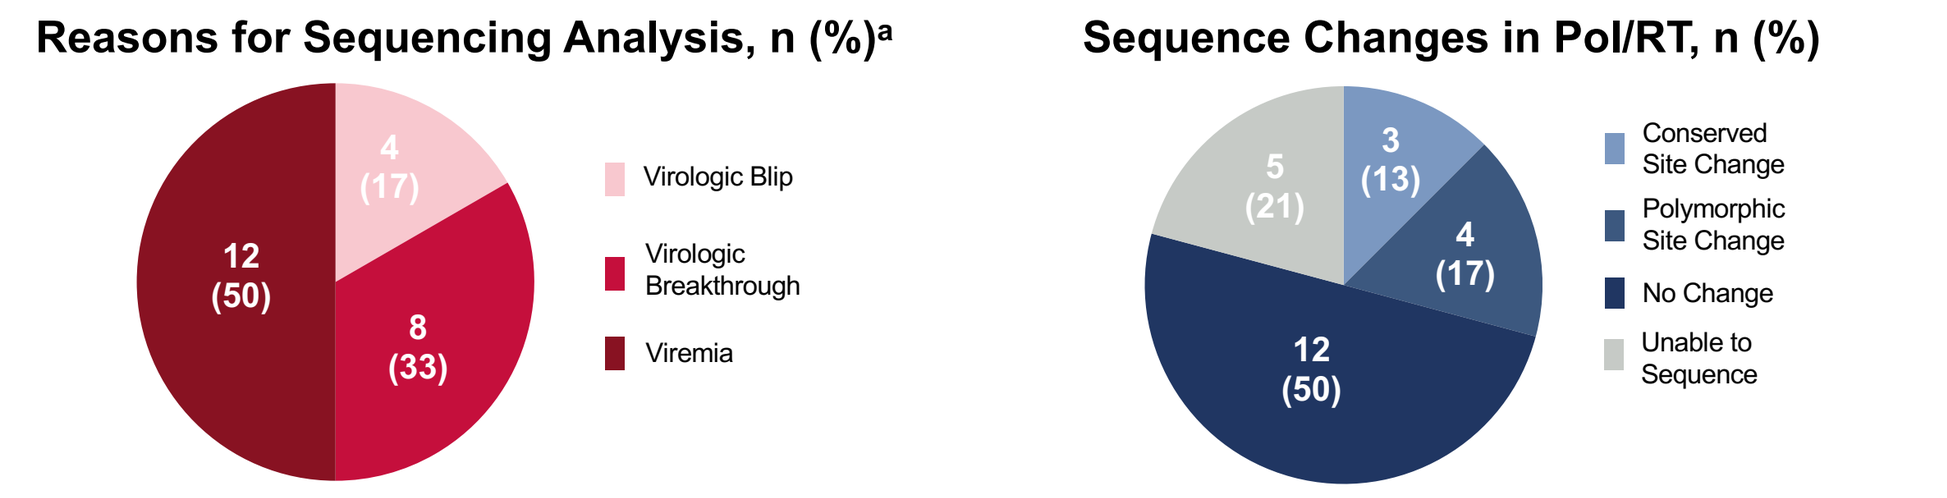
Reasons for Sequencing Analysis, n (%)[a]
4 (17)
8 (33)
12 (50)
Virologic Blip
Virologic Breakthrough
Viremia
Sequence Changes in Pol/RT, n (%)
3 (13)
4 (17)
12 (50)
5 (21)
Conserved Site Change
Polymorphic Site Change
No Change
Unable to Sequence

Phenotypic evaluation was performed for any patient who developed substitutions at conserved sites or developed changes at polymorphic residues in the HBV pol/RT if the changes were observed in more than 1 patient, or who experienced virologic breakthrough with any amino acid change developed at HBV pol/RT.
[a]Virologic breakthrough: HBV DNA ≥1 log_{10} IU/mL increase from nadir or confirmed ≥69 IU/mL if previously <69 IU/mL for 2 consecutive visits; virologic blip: met 1 virologic breakthrough criterion at only 1 visit; viremia: persistent HBV DNA ≥20 IU/mL over the treatment course. HBV, hepatitis B virus; pol/RT, polymerase/reverse transcriptase.

- Of 24 patients that qualified for sequencing analysis, 12 (50%) were due to persistent viremia
- 17 of 24 (71%) patients undergoing testing showed either no sequence change in pol/RT from baseline or were unable to be sequenced
- Five patients qualified for phenotypic analysis due to virologic breakthrough with any amino acid change (n = 2) or due to a conserved site change (n = 3)
 - — Week-96 isolates remained sensitive to TAF in vitro (fold-change in half-maximal effective concentration [EC_{50}] <2 from BL)
- No HBV amino acid substitutions associated with resistance to TAF were detected through 96 weeks of treatment

96-week resistance data from this study are being presented in detail by Rosenthal et al, poster 1429C (November 10, 2023)

Safety, Open-Label Phase

Adverse Events		
Patients, n (%)	TAF (n = 59)	PBO→TAF (n = 29)
Any AE	41 (70)	22 (76)
Any study drug–related AE	12 (20)	4 (14)
Grade 3 or 4 AE	3 (5)[a]	3 (3)
Serious AE	1 (2)[b]	0
D/C due to AE	0	0
Death	0	0

Grade 3 or 4 Laboratory Abnormalities		
Patients, n (%)	TAF (n = 59)	PBO→TAF (n = 29)
Any Grade 3 or 4 abnormality	13 (22)	5 (17)
ALT (increased)	1 (2)	1 (3)
Creatine kinase (increased)	2 (3)	1 (3)
Hemoglobin (decreased)	1 (2)	0
Neutrophils (decreased)	1 (2)	0
Platelets (decreased)	1 (2)	0
Hyperkalemia	1 (2)	0
Lymphocytes (decreased)	0	1 (3)
Occult blood in urine	5 (9)	2 (7)

[a]Grade 3: mononucleosis, tibia fracture; Grade 4: suicidal ideation in a patient who also experienced Grade 3 affective disorder and antisocial behavior. [b]Suicidal ideation (not related to treatment).
AE, adverse event; ALT, alanine aminotransferase; D/C, discontinuation; PBO, placebo; TAF, tenofovir alafenamide.

- At week 96, most AEs were mild to moderate in severity; no patient had a Grade 3 or 4 AE or SAE related to study treatment, and no patients discontinued OL TAF due to an AE

Renal Safety Over 96 Weeks

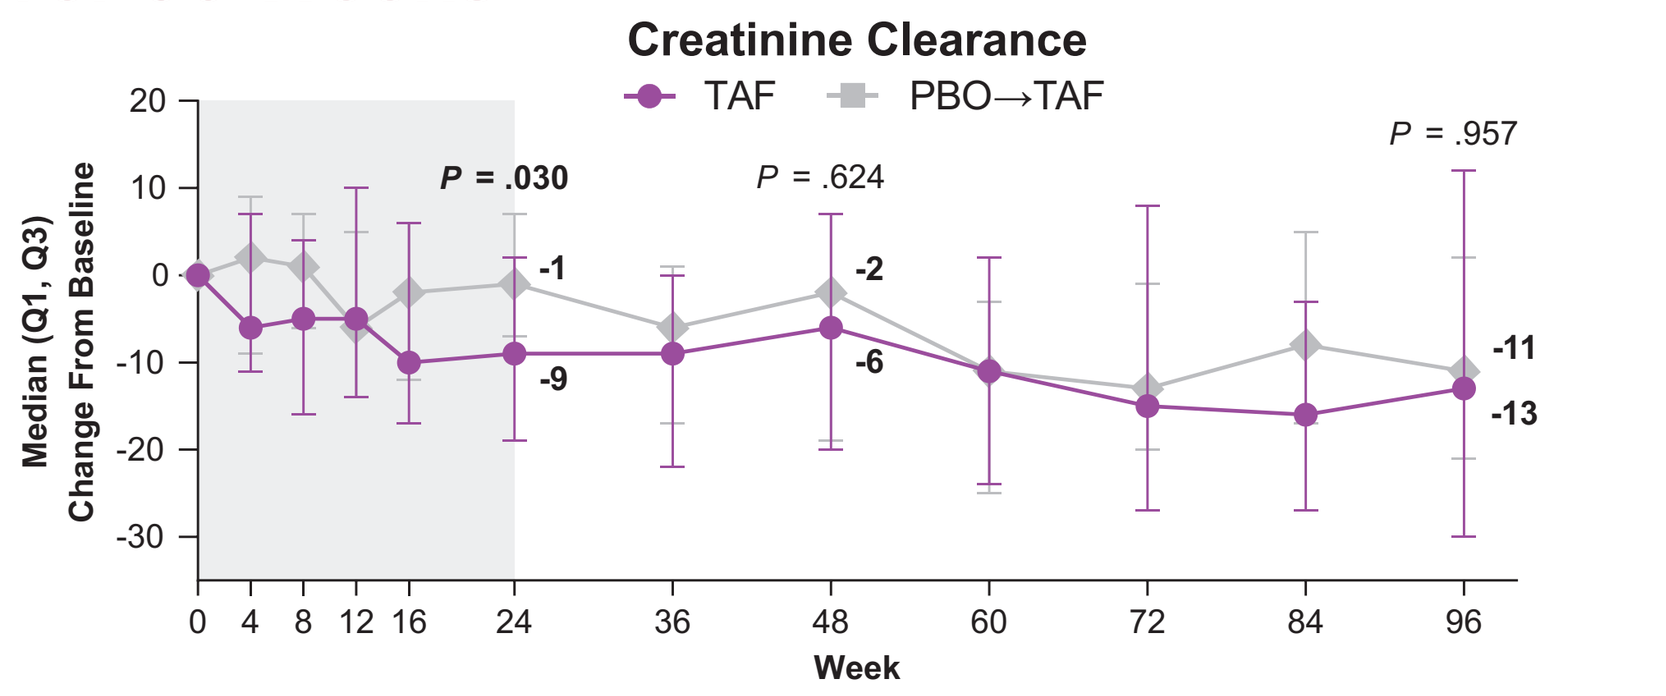
Creatinine Clearance
TAF
PBO→TAF
P = .030
P = .624
P = .957
-1
-9
-2
-6
-11
-13
Median (Q1, Q3) Change From Baseline
Week

Continuous baseline value data are expressed as median (Q1, Q3). Shaded area of the plot indicates DB phase; unshaded area indicates OL extension phase. *P*-values were based on a 2-sided Wilcoxon rank sum test. DB, double-blind; OL, open-label; PBO, placebo; Q, quartile; TAF, tenofovir alafenamide.

- Fluctuations in creatinine clearance were observed over the study period in both groups
- At week 96, changes from BL in creatinine clearance for both groups were similar, and no patient had creatinine clearance <90 mL/min/1.73 m²

Change in BMD and BMD Z-scores Over 96 Weeks

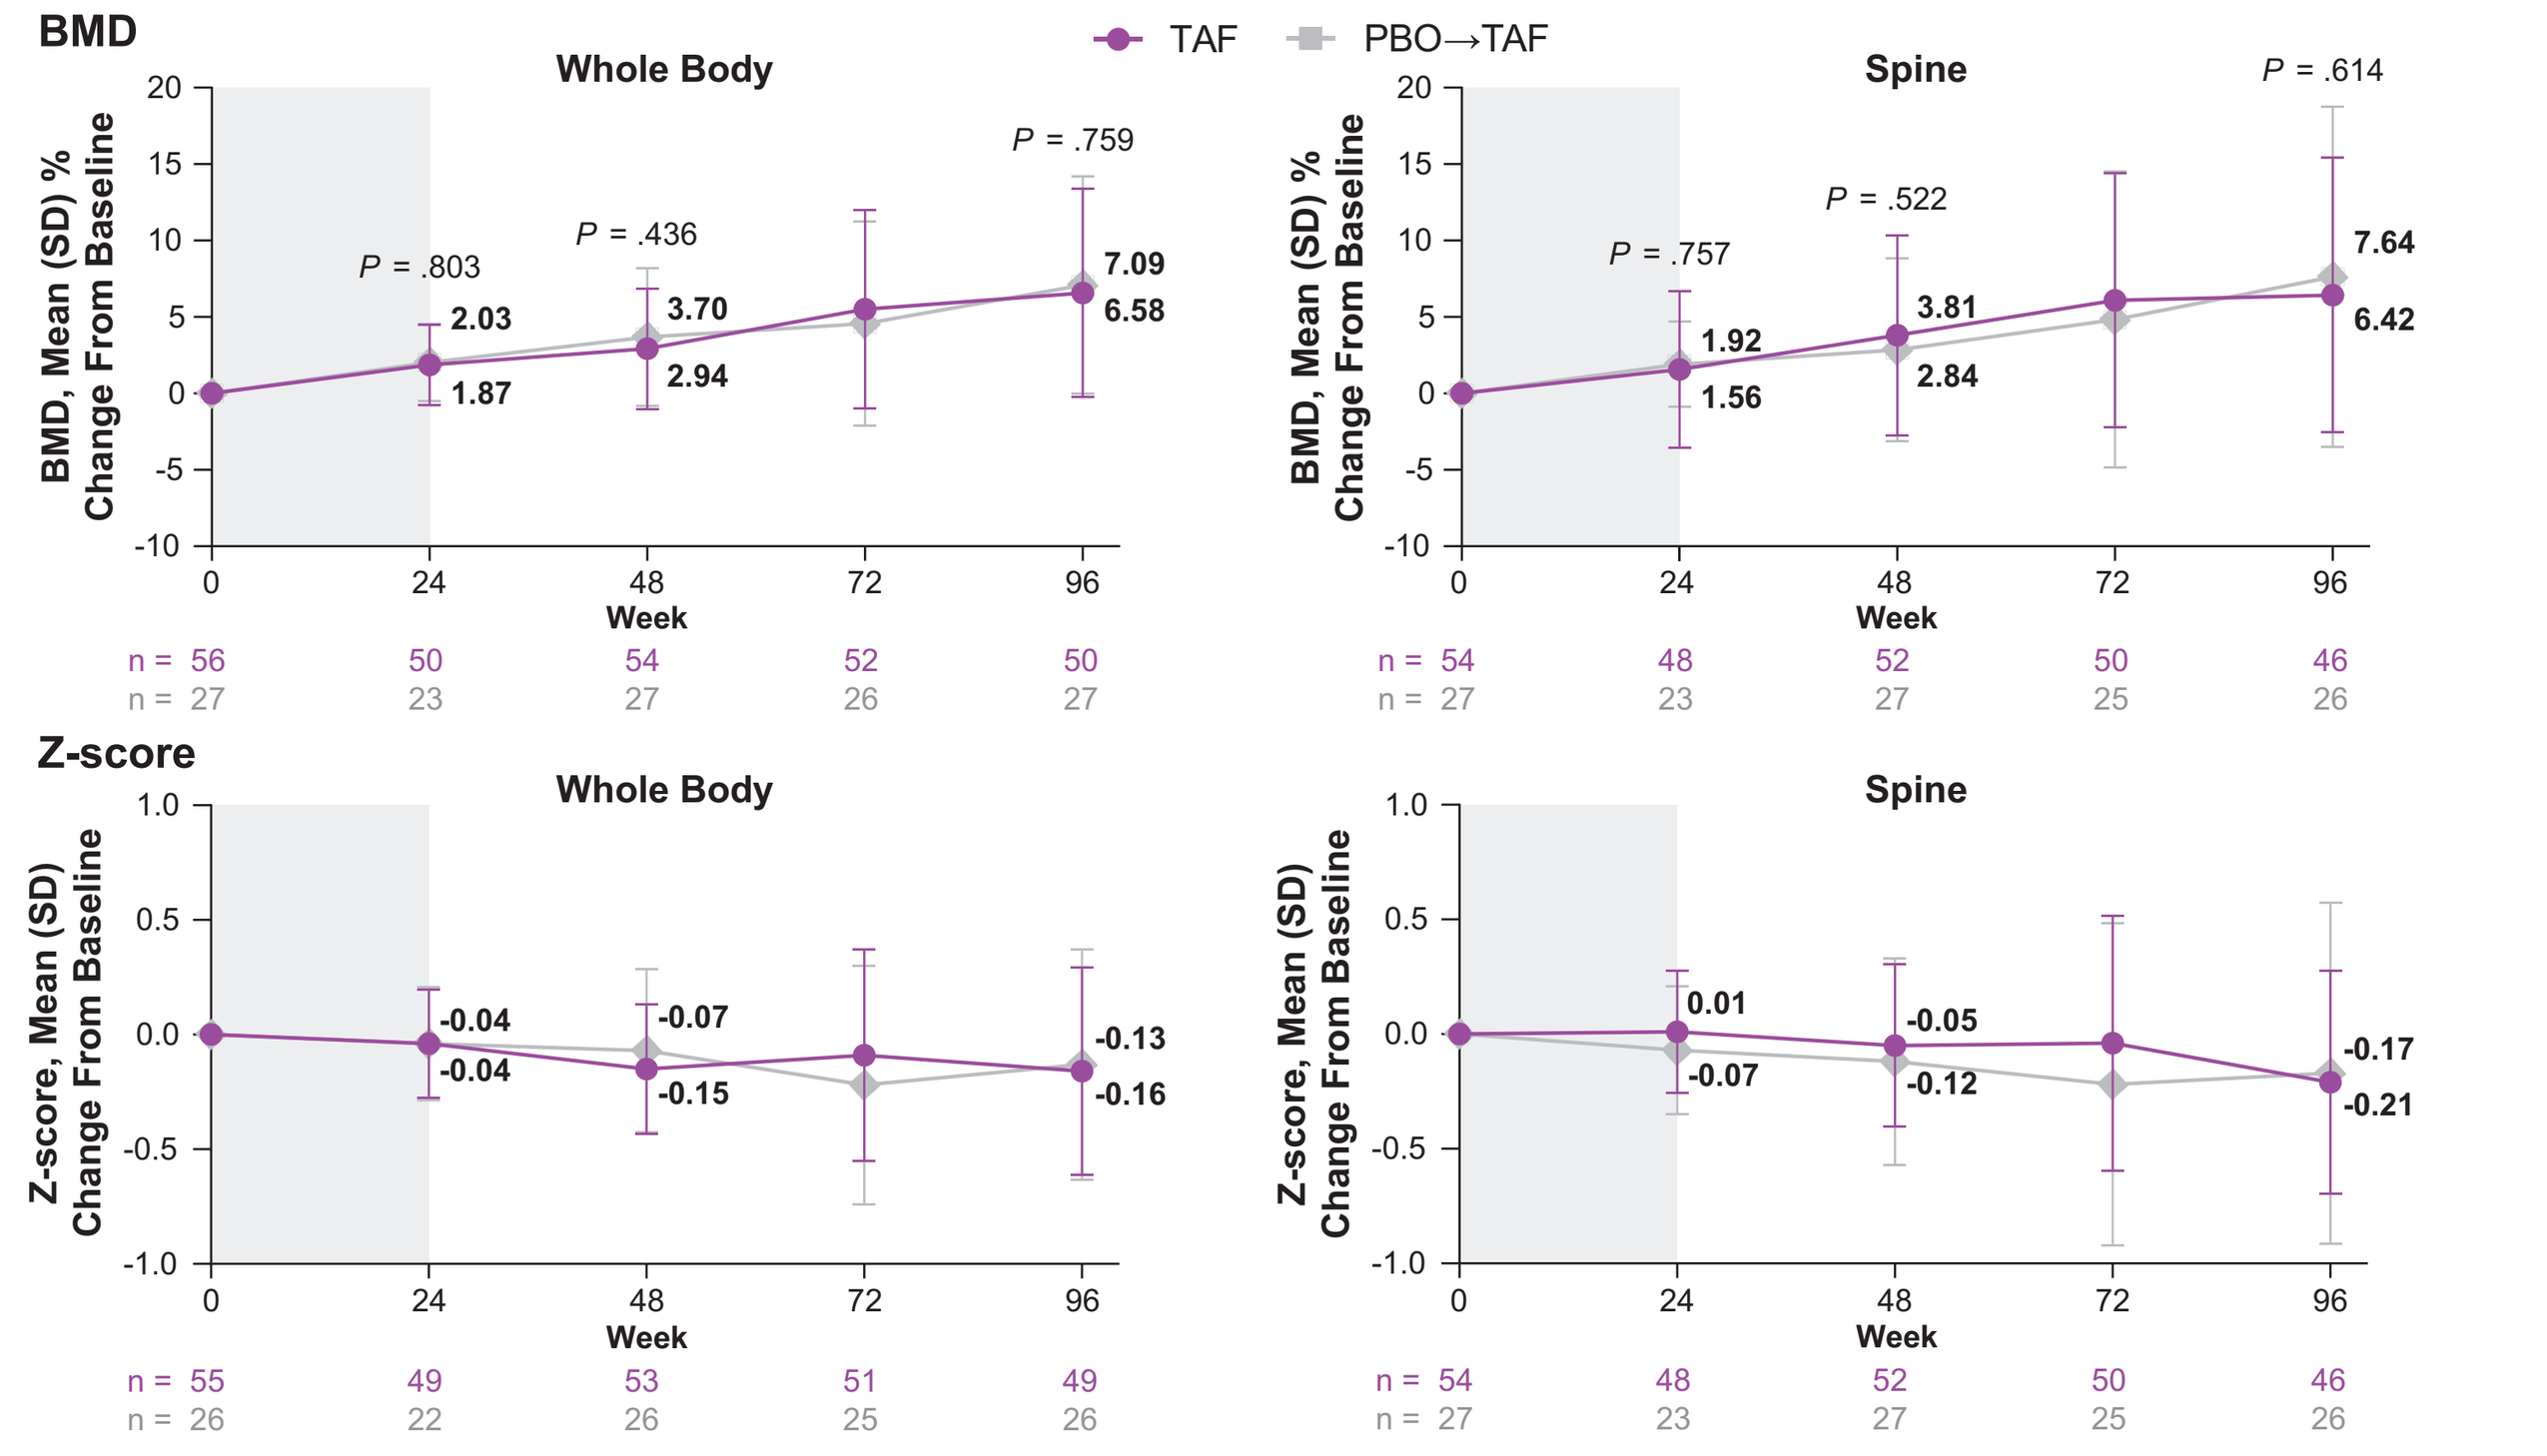
BMD
TAF
PBO→TAF
Whole Body
P = .803
P = .436
P = .759
2.03
1.87
3.70
2.94
7.09
6.58
BMD, Mean (SD) % Change From Baseline
n = 56 50 54 52 50
n = 27 23 27 26 27
Spine
P = .757
P = .522
P = .614
1.92
1.56
3.81
2.84
7.64
6.42
n = 54 48 52 50 46
n = 27 23 27 25 26
Z-score
Whole Body
-0.04
-0.04
-0.07
-0.15
-0.13
-0.16
Z-score, Mean (SD) Change From Baseline
n = 55 49 53 51 49
n = 26 22 26 25 26
Spine
0.01
-0.07
-0.05
-0.12
-0.17
-0.21
n = 54 48 52 50 46
n = 27 23 27 25 26
Week

P-values for mean change from baseline were based on an ANOVA model, including treatment as a fixed effect. Shaded area of the plot indicates DB phase; unshaded area indicates OL extension phase. ANOVA, analysis of variance; BMD, bone mineral density; DB, double-blind; OL, open-label; PBO, placebo; TAF, tenofovir alafenamide.

- At week 96, progressive mean % increases in BMD and small changes from BL in BMD z-scores were observed for both groups

AASLD: The Liver Meeting; November 10–14, 2023; Boston, MA, USA