ALLIANCE Open-Label Extension: Switch to B/F/TAF in People With Both HIV-1 and HBV

666

ALLIANCE

Anchalee Avihingsanon[1], Chee Loon Leong[2], Chien-Ching Hung[3], Sasisopin Kiertiburanakul[4], Man-Po Lee[5], Sharline Madera[6], Hongyuan Wang[6], Jason T Hindman[6], Taisheng Li[7]

[1]HIV-NAT, Thai Red Cross AIDS Research Centre, Bangkok, Thailand; [2]Kuala Lumpur General Hospital, Kuala Lumpur, Malaysia; [3]National Taiwan University Hospital Yunlin Branch, Yunlin, Taiwan; [4]Faculty of Medicine Ramathibodi Hospital, Mahidol University, Bangkok, Thailand; [5]Queen Elizabeth Hospital, Kowloon, Hong Kong; [6]Gilead Sciences, Inc., Foster City, CA, USA; [7]Peking Union Medical College Hospital, Beijing, China

Conclusions

- Through 48 weeks of the OLE phase, B/F/TAF maintained high rates of HIV-1 and HBV virologic suppression following switch from DTG + F/TDF
- Further improvements in key clinical outcomes, including ALT normalization, HBeAg loss and seroconversion, and HBsAg loss, were also seen following the switch
- B/F/TAF was well tolerated, with no study drug discontinuations due to TEAEs through 48 weeks of the OLE
 - — Most TEAEs were mild to moderate severity
- Median eGFR increased following switch from DTG + F/TDF to B/F/TAF, suggesting improved renal health
- Metabolic parameters remained stable during the OLE
- These results demonstrate the efficacy and safety of switching to B/F/TAF in TE people with both HIV-1 and HBV

Plain Language Summary

- The ALLIANCE study looked at how well two treatments called B/F/TAF and DTG + F/TDF work to treat adults who had both human immunodeficiency virus type 1 (HIV-1) and hepatitis B virus (HBV) infections
 - — We previously showed that after 96 weeks, both treatments lowered the levels of HIV-1 and HBV in the blood[1]
- The presence of two proteins called HBeAg and HBsAg is a sign of continuing HBV infection. A goal of treatment is to remove these proteins from the blood
 - — We previously showed that fewer people taking B/F/TAF than DTG + F/TDF had these proteins in the blood after 96 weeks of treatment[1]
- In this part of the ALLIANCE study, researchers wanted to see how safe and effective B/F/TAF is when taken for 1 year in people who had been taking DTG + F/TDF for 2 years and then switched to B/F/TAF
- After 1 year of treatment, B/F/TAF was effective at keeping HIV-1 and HBV at low levels in the blood
 - — During that time, the number of people with HBeAg and HBsAg proteins in the blood also continued to go down
- Side effects were rare
- This study shows that B/F/TAF is effective for people with both HIV-1 and HBV infections who were previously treated with DTG + F/TDF

Introduction

- An estimated 3.1 million people are living with both HIV-1 and hepatitis B virus (HBV)[2]
- Tenofovir alafenamide (TAF)– or tenofovir disoproxil fumarate (TDF)–based antiretroviral therapies are recommended for most adults and adolescents with HIV-1 and HBV[3-5]
- The ALLIANCE study showed that bictegravir/emtricitabine/tenofovir alafenamide (B/F/TAF) was noninferior for HIV-1 RNA suppression and superior for HBV DNA suppression, when compared with dolutegravir (DTG) + F/TDF at Week 48 in treatment-naïve adults with HIV-1 and HBV[1]
 - — Suppression rates were maintained through Week 96, after which participants receiving DTG + F/TDF had the option to switch to B/F/TAF for an additional 48 weeks of open-label extension (OLE)
- The efficacy and safety of B/F/TAF in treatment-experienced (TE) people with both HIV-1 and HBV have not been evaluated

Objective

- To evaluate the efficacy and safety of B/F/TAF in TE adults with HIV-1 and HBV who switched from DTG + F/TDF through 48 weeks (1 year) of treatment in the OLE

Methods

- ALLIANCE (NCT03547908) was a randomized, double-blind, active-controlled, Phase 3 clinical study of B/F/TAF versus DTG + F/TDF in adults with HIV-1 and HBV[1]
- This analysis reports data from OLE baseline to OLE Week 48 in participants who switched to B/F/TAF from DTG + F/TDF after ≥ 96 weeks of the randomized phase

Study Design

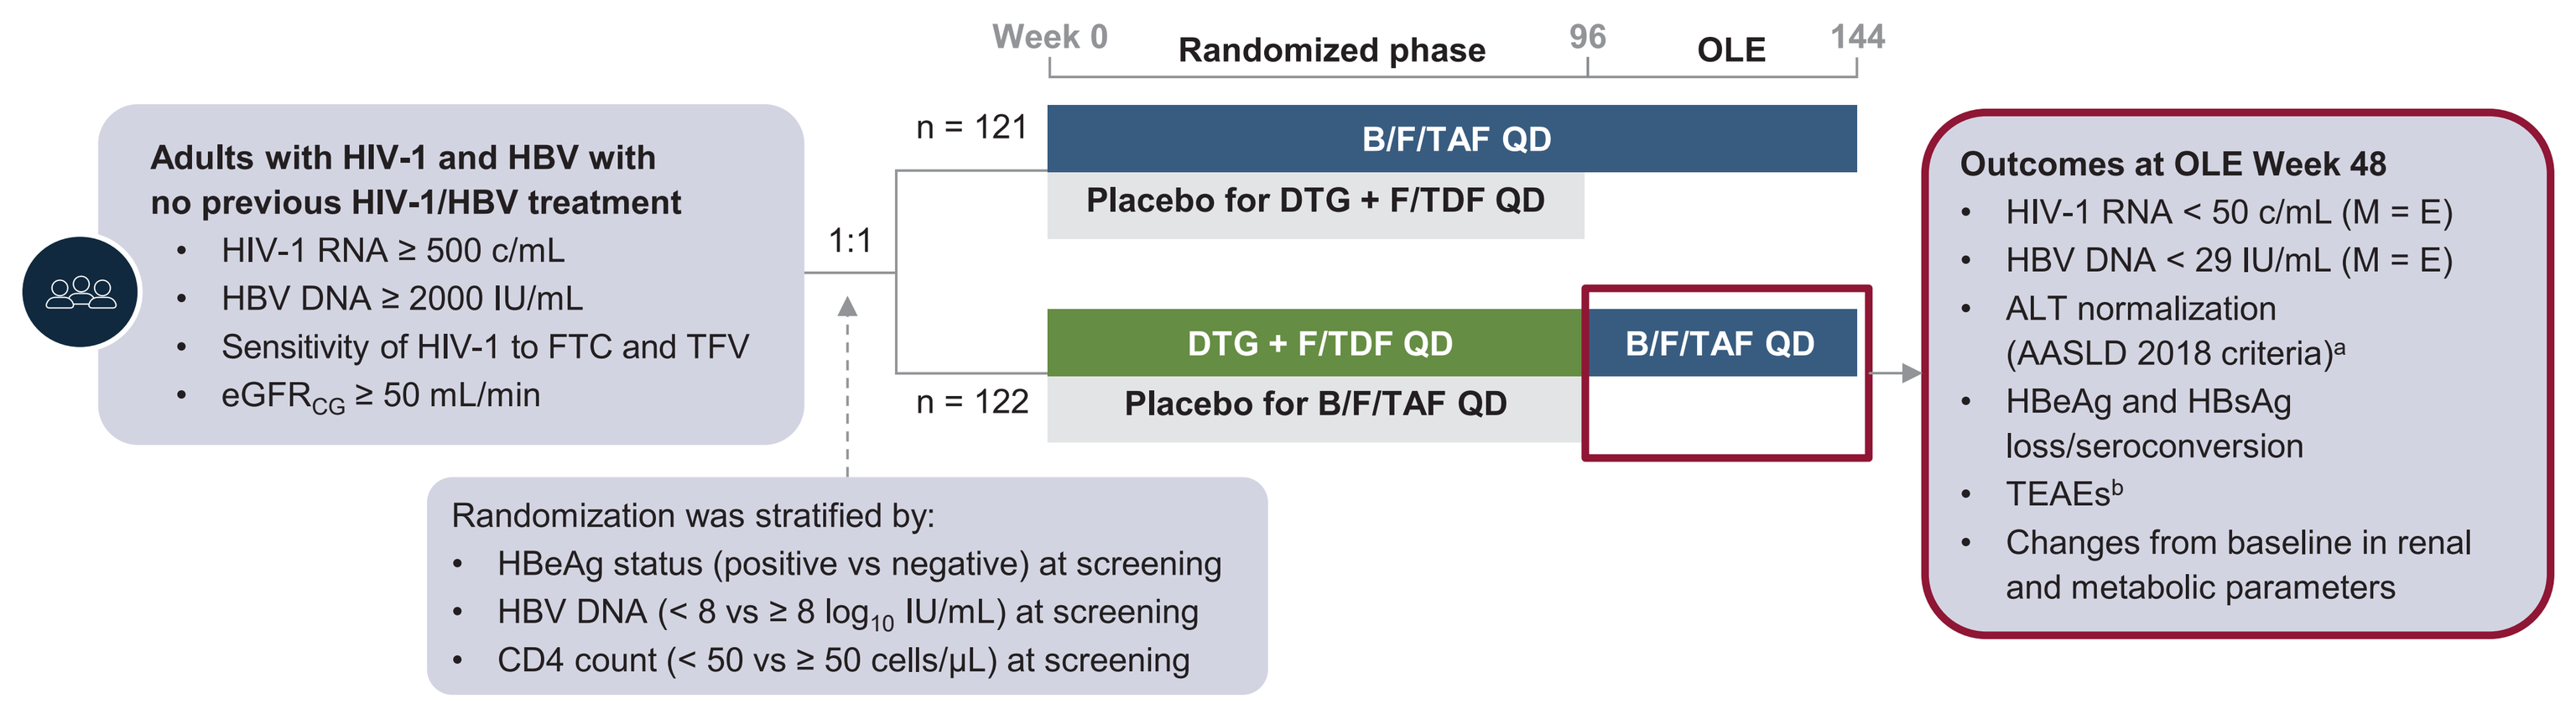
Week 0
Randomized phase
96
OLE
144
Adults with HIV-1 and HBV with no previous HIV-1/HBV treatment
• HIV-1 RNA ≥ 500 c/mL
• HBV DNA ≥ 2000 IU/mL
• Sensitivity of HIV-1 to FTC and TFV
• eGFR$_{CG}$ ≥ 50 mL/min
1:1
n = 121
B/F/TAF QD
Placebo for DTG + F/TDF QD
n = 122
DTG + F/TDF QD
Placebo for B/F/TAF QD
B/F/TAF QD
Randomization was stratified by:
• HBeAg status (positive vs negative) at screening
• HBV DNA (< 8 vs ≥ 8 log_{10} IU/mL) at screening
• CD4 count (< 50 vs ≥ 50 cells/μL) at screening
Outcomes at OLE Week 48
• HIV-1 RNA < 50 c/mL (M = E)
• HBV DNA < 29 IU/mL (M = E)
• ALT normalization (AASLD 2018 criteria)a
• HBeAg and HBsAg loss/seroconversion
• TEAEsb
• Changes from baseline in renal and metabolic parameters

[a]Change in ALT concentration from > ULN (female participants: 25 U/mL; male participants: 35 U/mL)[6] at baseline to ≤ ULN at Week 144. [b]Safety was assessed through the end of study. AASLD, American Association for the Study of Liver Diseases; ALT, alanine aminotransferase; B/F/TAF, bictegravir/emtricitabine/tenofovir alafenamide; c, copies; CD4, cluster of differentiation 4; DTG, dolutegravir; eGFR$_{CG}$, estimated glomerular filtration rate by Cockcroft-Gault equation; F/TDF, emtricitabine/tenofovir disoproxil fumarate; FTC, emtricitabine; HBeAg, hepatitis B e antigen; HBsAg, hepatitis B surface antigen; HBV, hepatitis B virus; M = E, missing = excluded; OLE, open-label extension; QD, once daily; TEAE, treatment-emergent adverse event; TFV, tenofovir; ULN, upper limit of normal.

Results

OLE Baseline Demographics and Characteristics

		DTG + F/TDF → B/F/TAF N = 89
Age, years, median (Q1, Q3)		34 (28, 39)
Male sex at birth, n (%)		87 (98)
Race, n (%)	Asian / Black / Other	84 (94) / 4 (4) / 1 (1)
Hispanic or Latine ethnicity, n (%)		5 (6)
HIV-1 RNA, log_{10} c/mL, median (Q1, Q3)		1.28 (1.28, 1.28)
CD4 count, cells/μL, median (Q1, Q3)		497 (320, 617)
HIV disease status: asymptomatic, n (%)		57 (64)
HBV genotype, n (%)[a]	A	7 (9)
	B	15 (19)
	C	49 (61)
	D	8 (10)
	Mixed	1 (1)
HBV DNA, log_{10} IU/mL, median (Q1, Q3)		0.95 (0.95, 1.20)
HBV DNA < 29 IU/mL, n (%)		74 (83)
HBeAg positive, n (%)		52 (58)
HBsAg positive, n (%)[b]		77 (87)
ALT, U/L, median (Q1, Q3)		26 (18, 38)
ALT > ULN (AASLD 2018 criteria), n (%)[c]		27 (30)
Time since HBV diagnosis to OLE baseline, years, median (Q1, Q3)[d]		4.0 (3.0, 5.0)

[a]HBV genotype data were missing for nine participants. [b]Not all participants were HBsAg positive at entry of the OLE as some tested negative during 96 weeks of DTG + F/TDF treatment in the blinded phase. [c]Based on the AASLD 2018 criteria (ULN is 25 U/L for females and 35 U/L for males).[6] [d]Self-reported or based on examination/medical records. AASLD, American Association for the Study of Liver Diseases; ALT, alanine aminotransferase; B/F/TAF, bictegravir/emtricitabine/tenofovir alafenamide; c, copies; CD4, cluster of differentiation 4; DTG, dolutegravir; F/TDF, emtricitabine/tenofovir disoproxil fumarate; HBV, hepatitis B virus; HBeAg, hepatitis B e antigen; HBsAg, hepatitis B surface antigen; OLE, open-label extension; Q, quartile; ULN, upper limit of normal.

- Of 110 participants who completed DTG + F/TDF treatment in the blinded phase, 89 entered the OLE
 - — 88 completed the OLE and 1 discontinued (due to loss to follow-up)
- Median (quartile [Q]1, Q3) exposure to B/F/TAF during the OLE was 48 (48, 49) weeks

HIV-1 and HBV Suppression Through 48 Weeks of OLE (M = E)

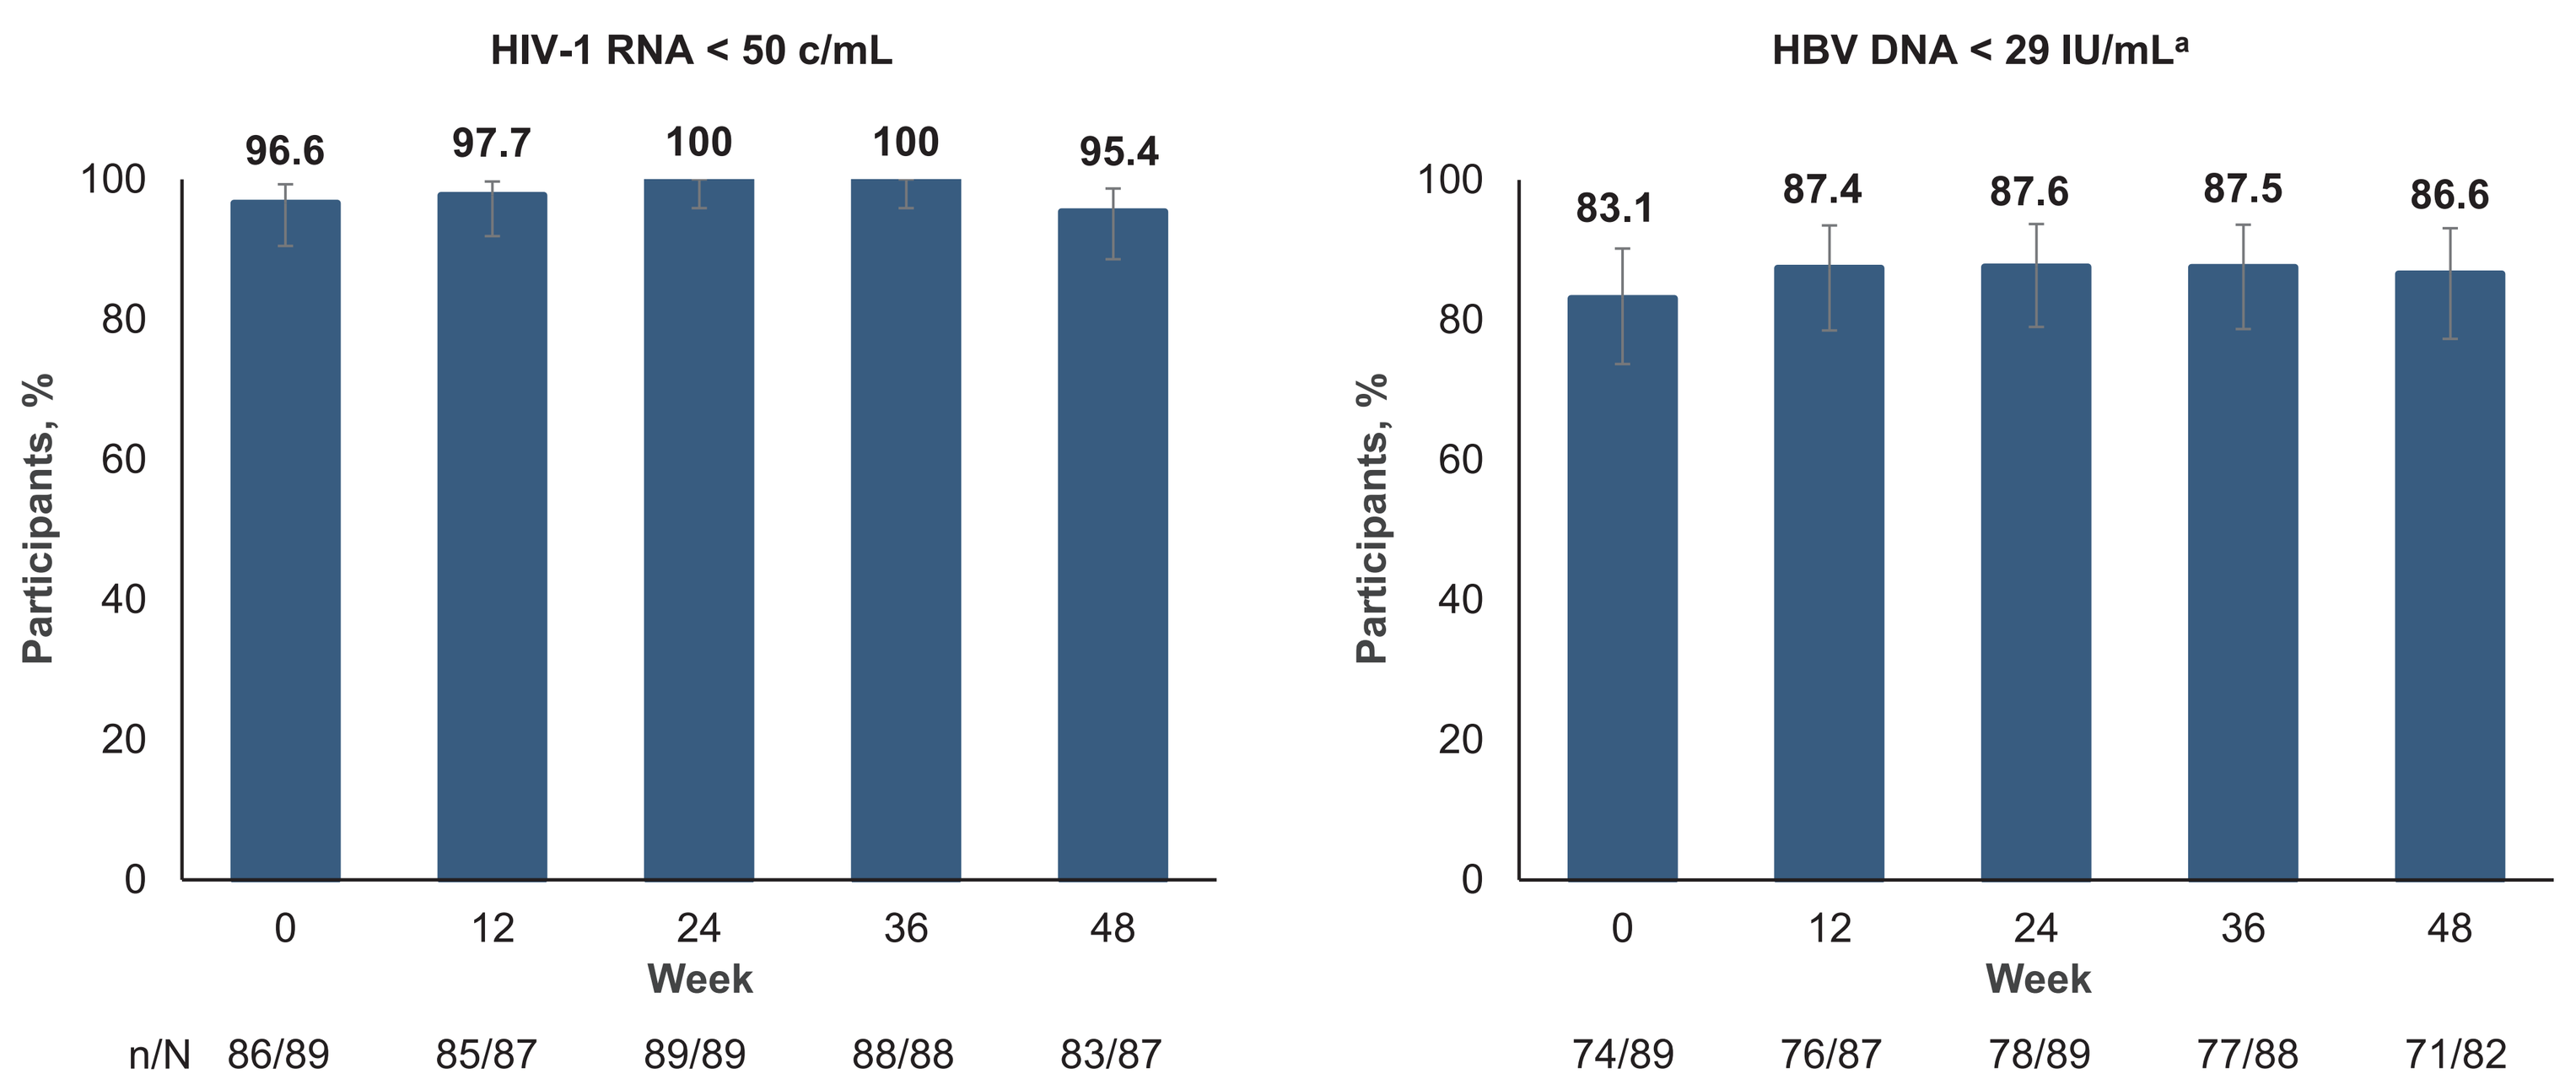
HIV-1 RNA < 50 c/mL
Participants, %
96.6
97.7
100
100
95.4
0
12
24
36
48
Week
n/N 86/89 85/87 89/89 88/88 83/87
HBV DNA < 29 IU/mLa
Participants, %
83.1
87.4
87.6
87.5
86.6
0
12
24
36
48
Week
74/89 76/87 78/89 77/88 71/82

Outcomes in the all B/F/TAF full analysis set (N = 89). The denominator is the number of participants with non-missing data for the endpoint at each visit. Error bars represent 95% CIs. [a]Includes all data collected up to 1 day after permanent discontinuation of B/F/TAF. B/F/TAF, bictegravir/emtricitabine/tenofovir alafenamide; c, copies; HBV, hepatitis B virus; M = E, missing = excluded; OLE, open-label extension.

- High rates of HIV-1 RNA and HBV DNA suppression were maintained through 48 weeks after a switch to B/F/TAF

ALT Normal[a] and ALT Normalization[b] by AASLD Criteria Through 48 Weeks of OLE (M = E)

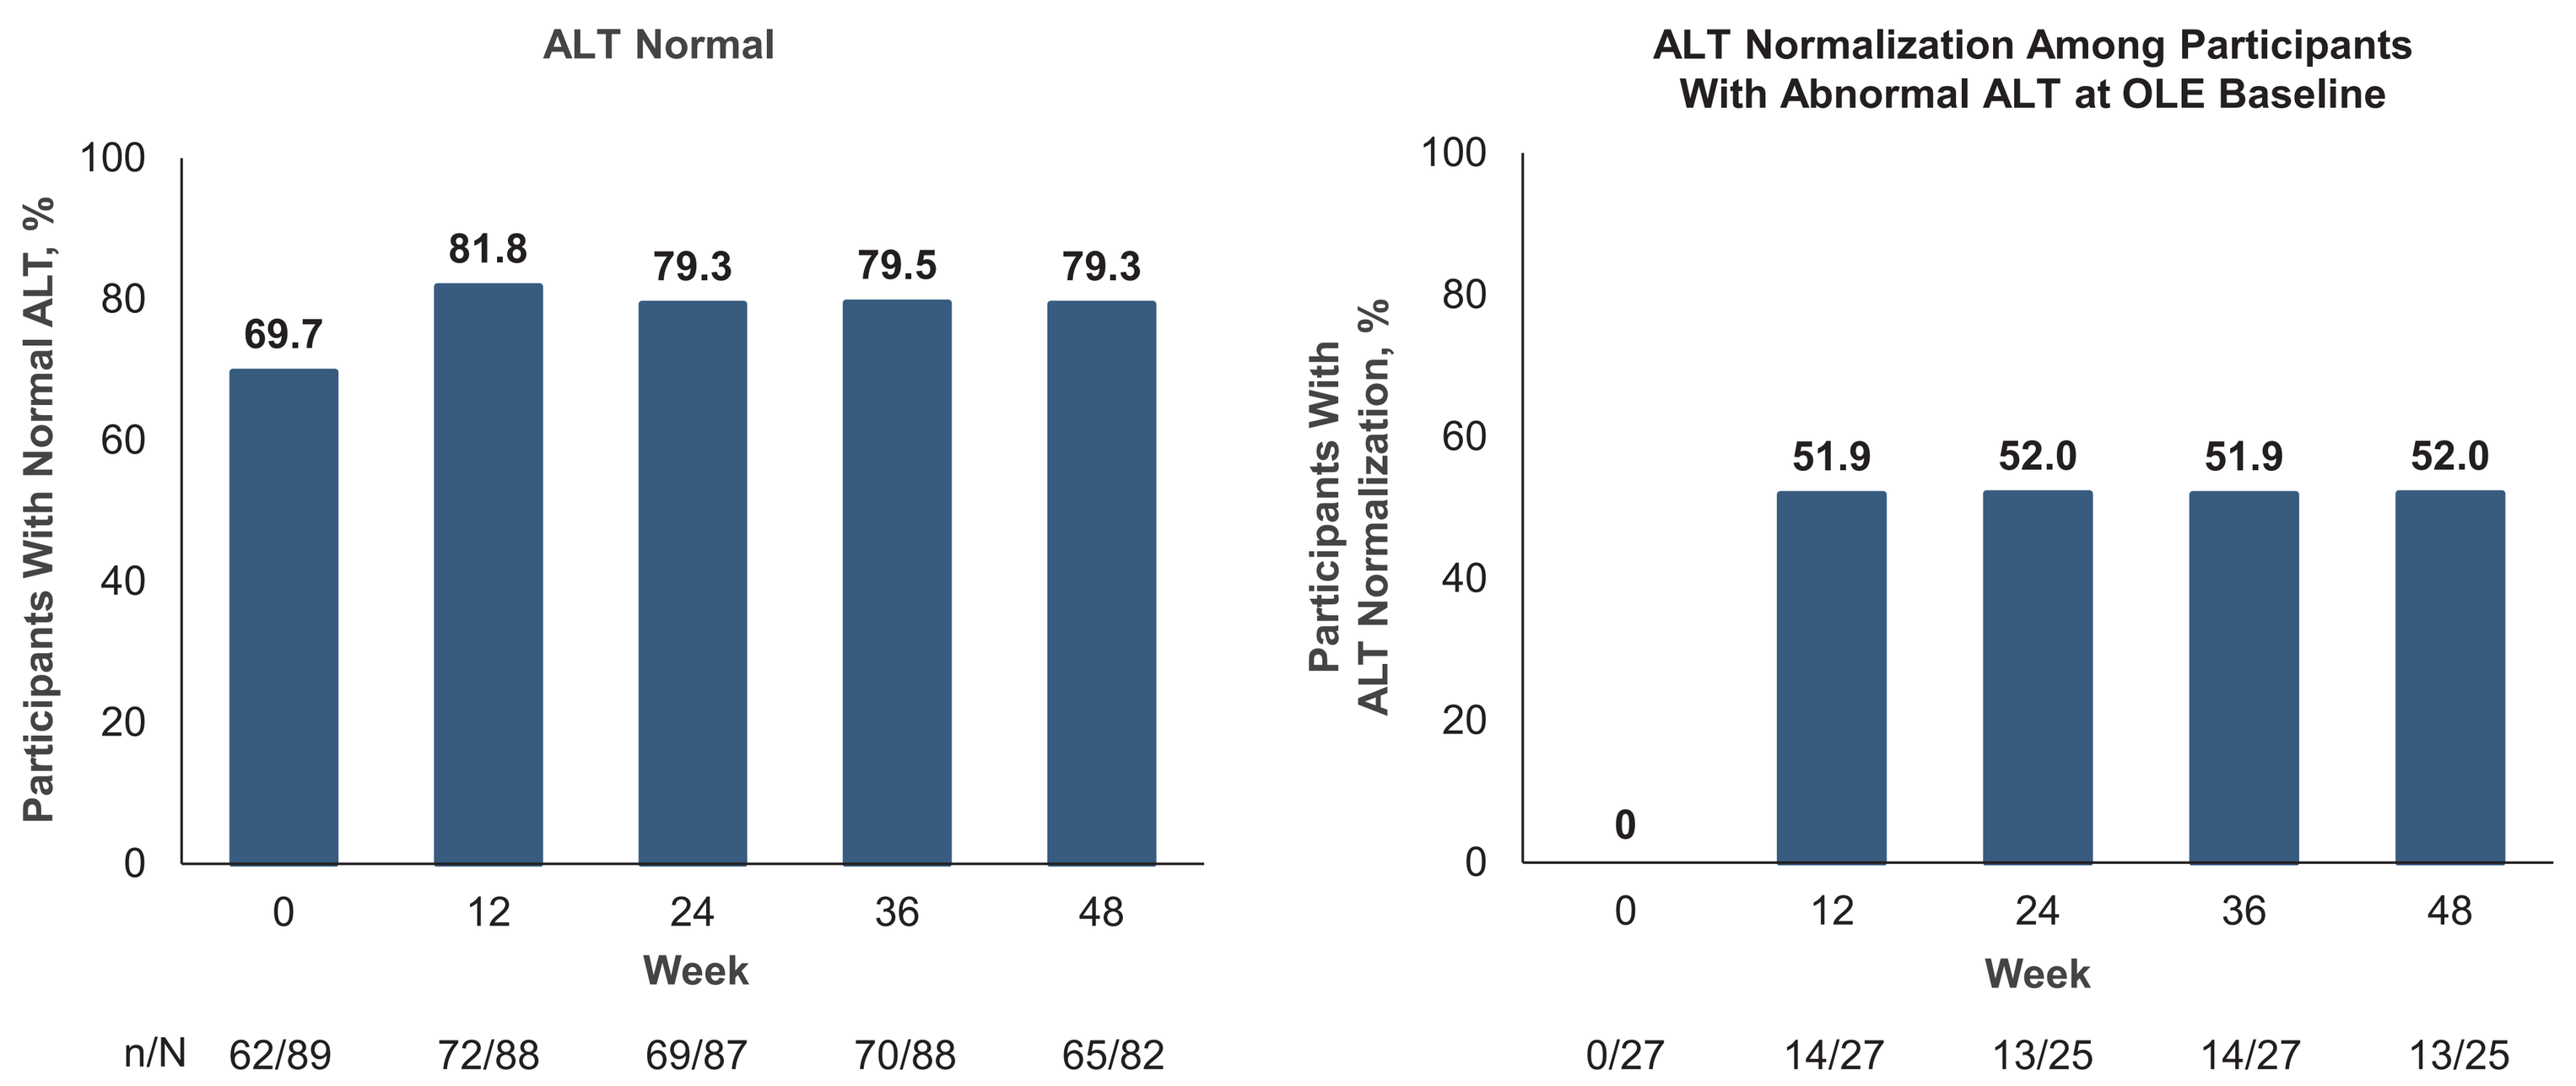
ALT Normal
Participants With Normal ALT, %
69.7
81.8
79.3
79.5
79.3
0
12
24
36
48
Week
n/N 62/89 72/88 69/87 70/88 65/82
ALT Normalization Among Participants With Abnormal ALT at OLE Baseline
Participants With ALT Normalization, %
0
51.9
52.0
51.9
52.0
0
12
24
36
48
Week
0/27 14/27 13/25 14/27 13/25

Outcomes in the all B/F/TAF full analysis set (N = 89), and includes all data collected up to 1 day after permanent discontinuation of B/F/TAF. The denominator is the number of participants with non-missing data for the endpoint at each visit. [a]Proportion of participants with normal ALT level (by AASLD 2018 criteria). [b]Reduction in ALT level to ≤ ULN for participants with ALT > ULN at baseline based on AASLD 2018 criteria, where ULN is 25 U/L for females and 35 U/L for males.[6] AASLD, American Association for the Study of Liver Diseases; ALT, alanine aminotransferase; B/F/TAF, bictegravir/emtricitabine/tenofovir alafenamide; M = E, missing = excluded; OLE, open-label extension; ULN, upper limit of normal.

- 30.3% (27/89) of participants switching to B/F/TAF had abnormal alanine aminotransferase (ALT) levels at OLE baseline
 - — Of these participants, over half achieved ALT normalization after 12 weeks of B/F/TAF treatment, with this proportion remaining consistent through Week 48
 - — A similar trend was observed for ALT normal, with an increasing proportion of participants achieving normal ALT levels

New HBeAg/HBsAg Loss and Seroconversion at OLE Week 48 (M = E)

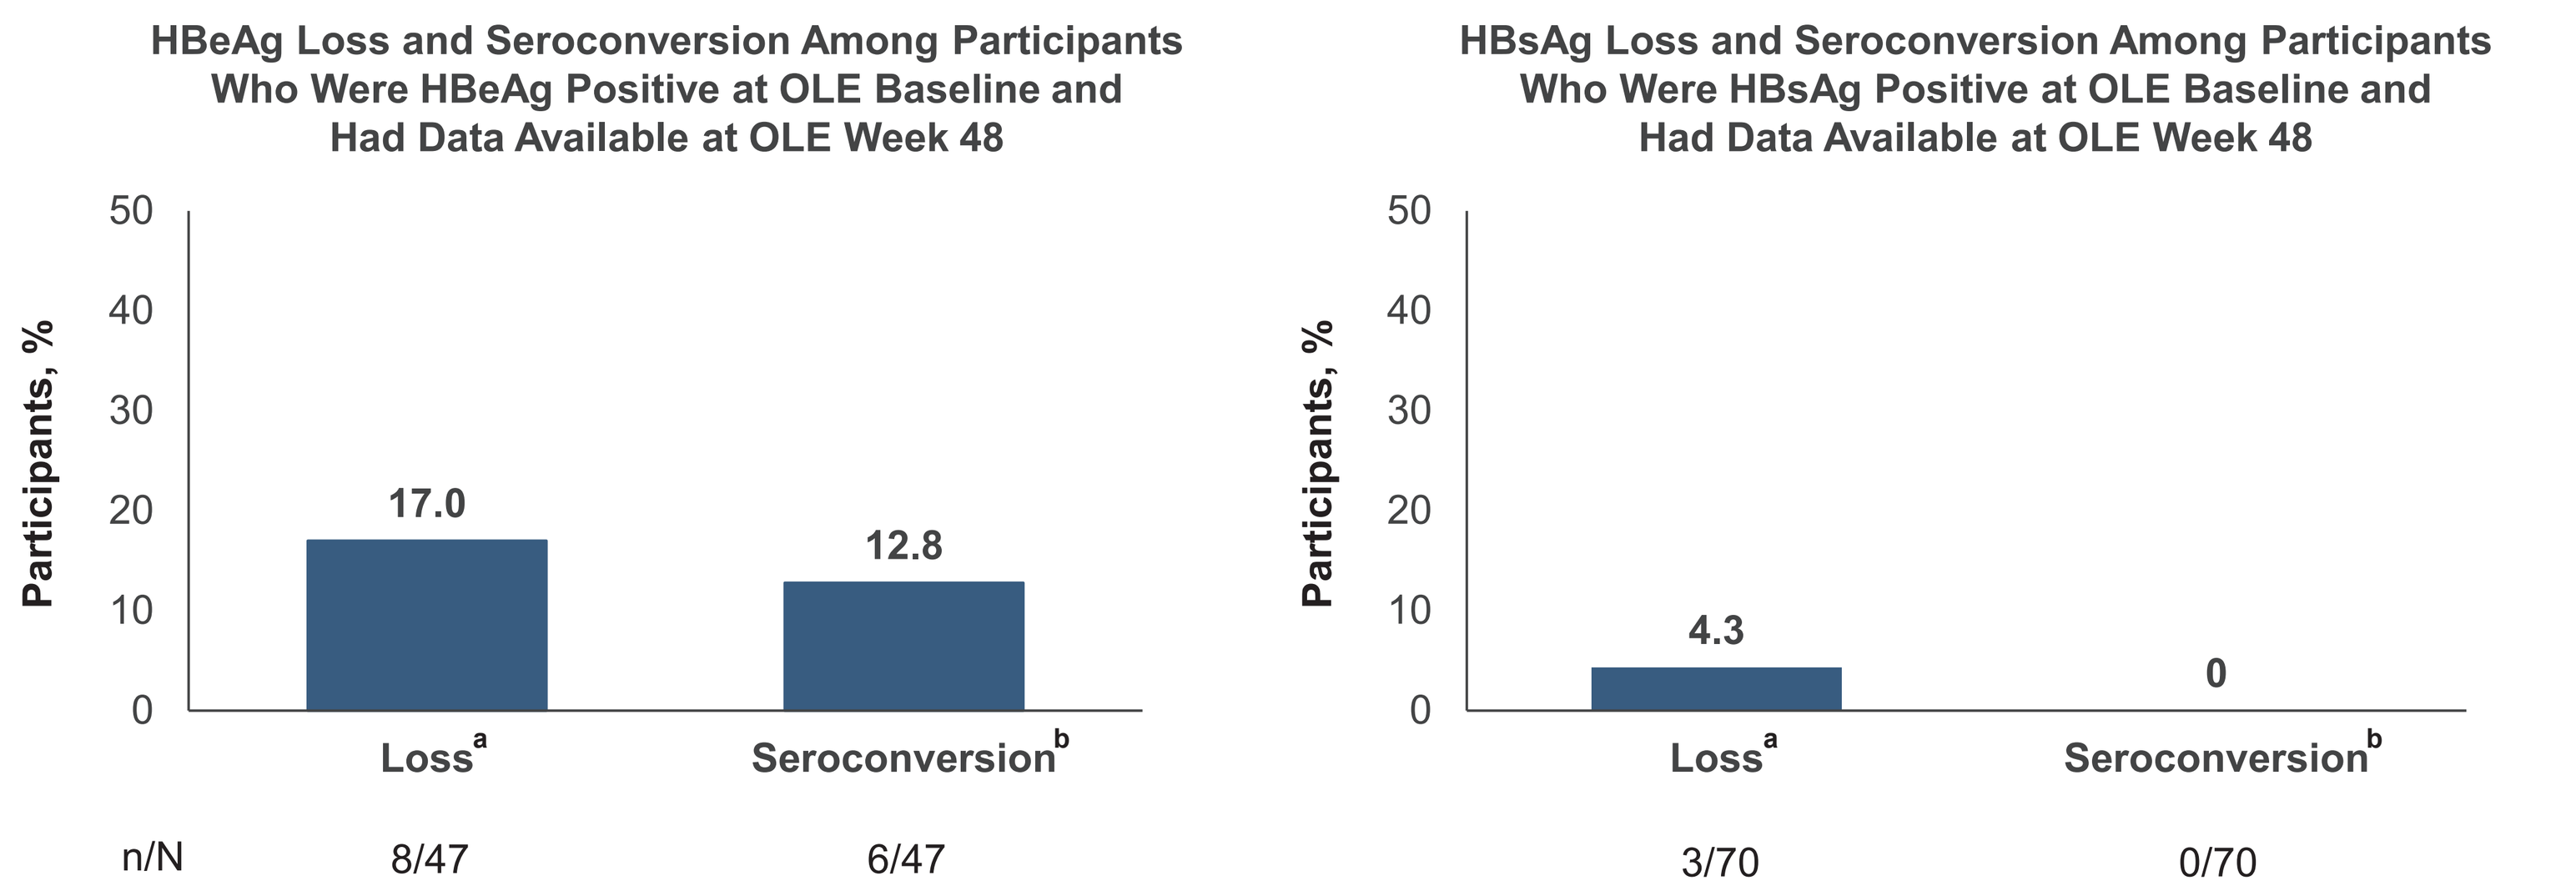
HBeAg Loss and Seroconversion Among Participants Who Were HBeAg Positive at OLE Baseline and Had Data Available at OLE Week 48
Participants, %
17.0
12.8
Lossa
Seroconversionb
n/N 8/47 6/47
HBsAg Loss and Seroconversion Among Participants Who Were HBsAg Positive at OLE Baseline and Had Data Available at OLE Week 48
Participants, %
4.3
0
Lossa
Seroconversionb
3/70 0/70

The all B/F/TAF serologically evaluable full analysis set, defined as the number of participants in the all B/F/TAF full analysis set who were HBe/sAg positive and HBe/sAb negative or had missing data at baseline, was used for assessment of HBeAg and HBsAg loss/seroconversion, respectively (n = 52 and n = 77). [a]Defined as change in serum HBeAg/HBsAg status from positive at baseline to negative at a post-baseline visit, with baseline HBeAb/HBsAb status negative or missing. [b]Defined as loss of serum HBeAg/HBsAg and serum HBeAb/HBsAb status change from negative or missing at baseline to positive at a post-baseline visit. B/F/TAF, bictegravir/emtricitabine/tenofovir alafenamide; HBeAb, hepatitis B e antibody; HBeAg, hepatitis B e antigen; HBsAb, hepatitis B surface antibody; HBsAg, hepatitis B surface antigen; M = E, missing = excluded; OLE, open-label extension.

- At OLE baseline, 58.4% (52/89) of participants were hepatitis B e antigen (HBeAg) positive and 86.5% (77/89) of participants were hepatitis B surface antigen (HBsAg) positive
- HBeAg loss and seroconversion and HBsAg loss continued during 48 weeks following the switch to B/F/TAF

Safety During 48 Weeks of OLE

n (%)	DTG + F/TDF → B/F/TAF N = 89
Any TEAE	66 (74)
Study drug–related TEAEs	17 (19)
Any Grade 3 or 4 TEAEs	6 (7)
Study drug–related Grade 3 or 4 TEAEs[a]	3 (3)
Any serious TEAEs	2 (2)
Study drug–related serious TEAEs	0
Study drug discontinuation due to TEAE	0
Death	0
Any Grade 3 or 4 laboratory abnormalities[b]	7 (8)
Grade 3 or 4 abnormalities occurring in > 1 participant	
Hypercholesterolemia, fasting	3 (3)

- B/F/TAF was well tolerated, with no study drug discontinuations due to treatment-emergent adverse events (TEAEs)
- The most commonly reported study drug–related TEAEs were weight gain (n = 8; 9%)[c] and low-density lipoprotein (LDL) cholesterol increased (n = 3; 3%)

Safety outcomes, except laboratory abnormalities, were assessed in the all B/F/TAF safety analysis set (N = 89), which included all randomly assigned participants who received ≥ 1 dose of study drug. Treatment-emergent laboratory abnormalities were assessed in the all B/F/TAF safety analysis set (N = 89) with ≥ 1 post-baseline laboratory value. [a]Abnormal weight gain (n = 2) and hyperlipidemia (n = 1). [b]Grade 3: n = 5; Grade 4: n = 2. [c]Includes weight increased (n = 4) and abnormal weight gain (n = 4). B/F/TAF, bictegravir/emtricitabine/tenofovir alafenamide; DTG, dolutegravir; F/TDF, emtricitabine/tenofovir disoproxil fumarate; OLE, open-label extension; TEAE, treatment-emergent adverse event.

Change From OLE Baseline in Renal and Metabolic Parameters

Median (Q1, Q3)[a]	DTG + F/TDF → B/F/TAF N = 89	
	At OLE Baseline	Change at OLE Week 48
eGFR$_{CG}$, mL/min	94 (80, 113)	+6.6 (-2.4, +13.2)[b]
Glucose, mg/dL	90 (86, 96)	-2 (-7, +3)[b]
Total cholesterol, mg/dL	168 (143, 201)	+17 (+3, +38)[c]
HDL cholesterol, mg/dL	44 (38, 50)	+5 (0, +10)[c]
LDL cholesterol, mg/dL	101 (82, 124)	+19 (-1, +34)[c]
Triglycerides, mg/dL	97 (70, 132)	+3 (-15, +38)[c]
Total:HDL cholesterol	3.7 (3.3, 4.5)	+0.1 (-0.3, +0.4)[c]
Body weight, kg	66.5 (59.0, 75.4)	+2.0 (+0.1, +4.0)[b]

- At OLE Week 48, following the switch to B/F/TAF:
 - — There was a small increase in estimated glomerular filtration rate (eGFR)
 - — Fasting glucose concentration, lipid parameters, and weight remained stable, except for a small increase in total and LDL cholesterol level
 - — Three participants (3.4%) initiated lipid-modifying agents
 - — Most participants had clinically insignificant changes in lipid levels; observed increases for some lipids were consistent with those in other TDF to TAF switch studies[7-9]

[a]All metabolic parameters, except body weight, were measured under fasting conditions. [b]n = 87. [c]n = 86. B/F/TAF, bictegravir/emtricitabine/tenofovir alafenamide; DTG, dolutegravir; eGFR$_{CG}$, estimated glomerular filtration rate by Cockcroft-Gault equation; F/TDF, emtricitabine/tenofovir disoproxil fumarate; HDL, high-density lipoprotein; LDL, low-density lipoprotein; OLE, open-label extension; Q, quartile.

References: **1.** Avihingsanon A, et al. *Lancet HIV*. 2023;10:e640-52. **2.** Leumi S, et al. *Clin Infect Dis*. 2020;71:2799-806. **3.** European AIDS Clinical Society. https://eacs.sanfordguide.com/ (accessed Nov. 18, 2024). **4.** Department of Health and Human Services. https://clinicalinfo.hiv.gov/sites/default/files/guidelines/documents/adult-adolescent-arv/guidelines-adult-adolescent-arv.pdf (accessed Nov. 18, 2024). **5.** Gandhi RT, et al. *JAMA*. 2023;329:63-84. **6.** Terrault NA, et al. *Hepatology*. 2018;67:1560-99. **7.** Brunet L, et al. *Clin Drug Investig*. 2021;41:955-65. **8.** Mallon PWG, et al. *Open Forum Infect Dis*. 2021;9:ofab621. **9.** Suzuki K. *PLOS ONE*. 2022;17:e0261760.

Acknowledgments: We thank all study participants, investigators, and staff. We also thank Khuanchai Supparatpiny (Chiang Mai University) for his contributions to abstract development and Frida Abramov (Gilead Sciences, Inc.) for HBV scientific expertise. This study was sponsored by Gilead Sciences, Inc. Medical writing support was provided by Joanna Nikitorowicz-Buniak, PhD (Aspire Scientific Ltd, UK), and was funded by Gilead Sciences, Inc.

Disclosures: **AA** reports research/grant support from Gilead Sciences, Inc., GSK/ViiV Healthcare, Janssen, MSD, and Roche; travel support from Gilead Sciences, Inc.; and membership with the Committee of Thai AIDS Society, the Strategic and Technical Advisory Group on HIV, Hepatitis and STI for WHO SEARO, and the Thailand National Committee on ART, Hepatitis and TB/TPT. **C-CH** reports support for the present study, consulting fees, and speaker honoraria from Gilead Sciences, Inc. **SM**, **HW**, and **JTH** are employees of, and own stocks/shares in, Gilead Sciences, Inc. **TL** reports support for the present study from Gilead Sciences, Inc.; speaker honoraria from Gilead Sciences, Inc., GSK Pharmaceuticals, and PerkinElmer; travel support from Gilead Sciences, Inc., GSK Pharmaceuticals, and Sansure Biotech; and is the Chair of the Chinese Society of Infectious Diseases. **CLL**, **SK**, and **M-PL** have no conflicts of interest to report.

Correspondence: Anchalee Avihingsanon, anchaleea2009@gmail.com.

Conference on Retroviruses and Opportunistic Infections (CROI); March 9–12, 2025; San Francisco, CA, USA