Weight Change on F/TAF Versus Placebo: Using Common F/TDF Groups to Bridge Data Across Clinical Trials

892

GS-US-311-7049

David Glidden[1], Andrew Whiteman[2], Yuan Tian[2], Andrea Marongiu[3], Joshua Gruber[2], Cal Cohen[2]

[1]University of California San Francisco, San Francisco, CA, USA; [2]Gilead Sciences, Inc., Foster City, CA, USA; [3]Gilead Sciences Ltd, Uxbridge, UK

Conclusions

- This is the first analysis to compare changes in weight among people taking F/TAF versus placebo
 - — Data from two Phase 3 randomized double-blind clinical trials in people without HIV were used, with F/TDF cohorts in each clinical trial acting as bridging cohorts
- Mean change in weight with F/TDF was consistent between the two trials, allowing comparison of the F/TAF and placebo cohorts
- Changes in weight were observed in the placebo group, providing important information on the weight increase expected over time in people without HIV assigned male at birth
- There were no statistically or clinically significant differences in mean weight gain with F/TAF compared with placebo
- Additionally, top percentiles of weight gain at Week 48 were similar with F/TAF and placebo
- Weight gain through 96 weeks in participants who switched from F/TDF to F/TAF at baseline was higher than for participants on placebo with no prior F/TDF exposure; this is consistent with a reversible weight-suppressive effect of F/TDF

Plain Language Summary

- Some people taking a human immunodeficiency virus (HIV) medication containing emtricitabine/tenofovir alafenamide (F/TAF) gain weight
 - — Sometimes this is because the HIV medication is helping them become healthier, but at other times the reasons are not fully known
- This analysis looked at weight gain in two studies (one called iPrEx and the other called DISCOVER) of people who did not have HIV but who took medication to protect themselves from getting HIV (also known as PrEP or prevention)
- In iPrEx, one group was taking placebo (a pill that does not contain any medicine but that makes people think they are taking the study medication), while in DISCOVER, one group was taking F/TAF
- In both studies, the second group of people was taking emtricitabine/tenofovir disoproxil fumarate (F/TDF; the standard of care at the time of the iPrEx study). This common group helped compare one study to another
- The amount of weight gained by people without HIV who took F/TAF compared with people who took placebo was similar. Any difference was too small to be meaningful

Introduction

- Emtricitabine/tenofovir alafenamide (F/TAF) and emtricitabine/tenofovir disoproxil fumarate (F/TDF) are nucleoside/nucleotide reverse transcriptase inhibitors commonly used as the backbones for many guideline-recommended treatments for HIV[1]
- F/TDF has previously been associated with reversible weight suppression,[2] whereas the association of F/TAF with weight gain is currently debated[3,4]
- People with HIV often gain weight during treatment, and the cause of this can be multifactorial.[5] This means that determining the direct impact of HIV medication on weight is challenging
- Comparison of F/TAF with placebo in people without HIV may give valuable insight into the effect of F/TAF on weight

Objective

- To compare weight gain with F/TAF versus placebo among people without HIV, using data from two large Phase 3 randomized double-blind pre-exposure prophylaxis (PrEP) efficacy and safety clinical trials

Methods

- This was a retrospective analysis using data from two large Phase 3 randomized clinical trials of F/TDF versus placebo (iPrEx) and F/TDF versus F/TAF (DISCOVER) as PrEP in people without HIV (**Figure 1**)
- Both trials enrolled adults assigned male at birth who had an increased likelihood of HIV acquisition
- Body weight was measured during regular clinic visits
- To analyze the mean change in weight with F/TAF compared with placebo, common F/TDF groups in iPrEx and DISCOVER were used to bridge the trials
 - — Participants in DISCOVER were stratified based on prior F/TDF use at baseline
- A linear mixed model, adjusting for baseline covariates and time-varying indicators of weight change–associated medication, was used to analyze mean weight change
 - — Restricted cubic splines were used to model how effects of treatment may change over time
- Quantile regression was used to compare top percentiles of weight gain at Week 48, adjusting for the same baseline covariates (complete case analysis)

Figure 1. Study Design

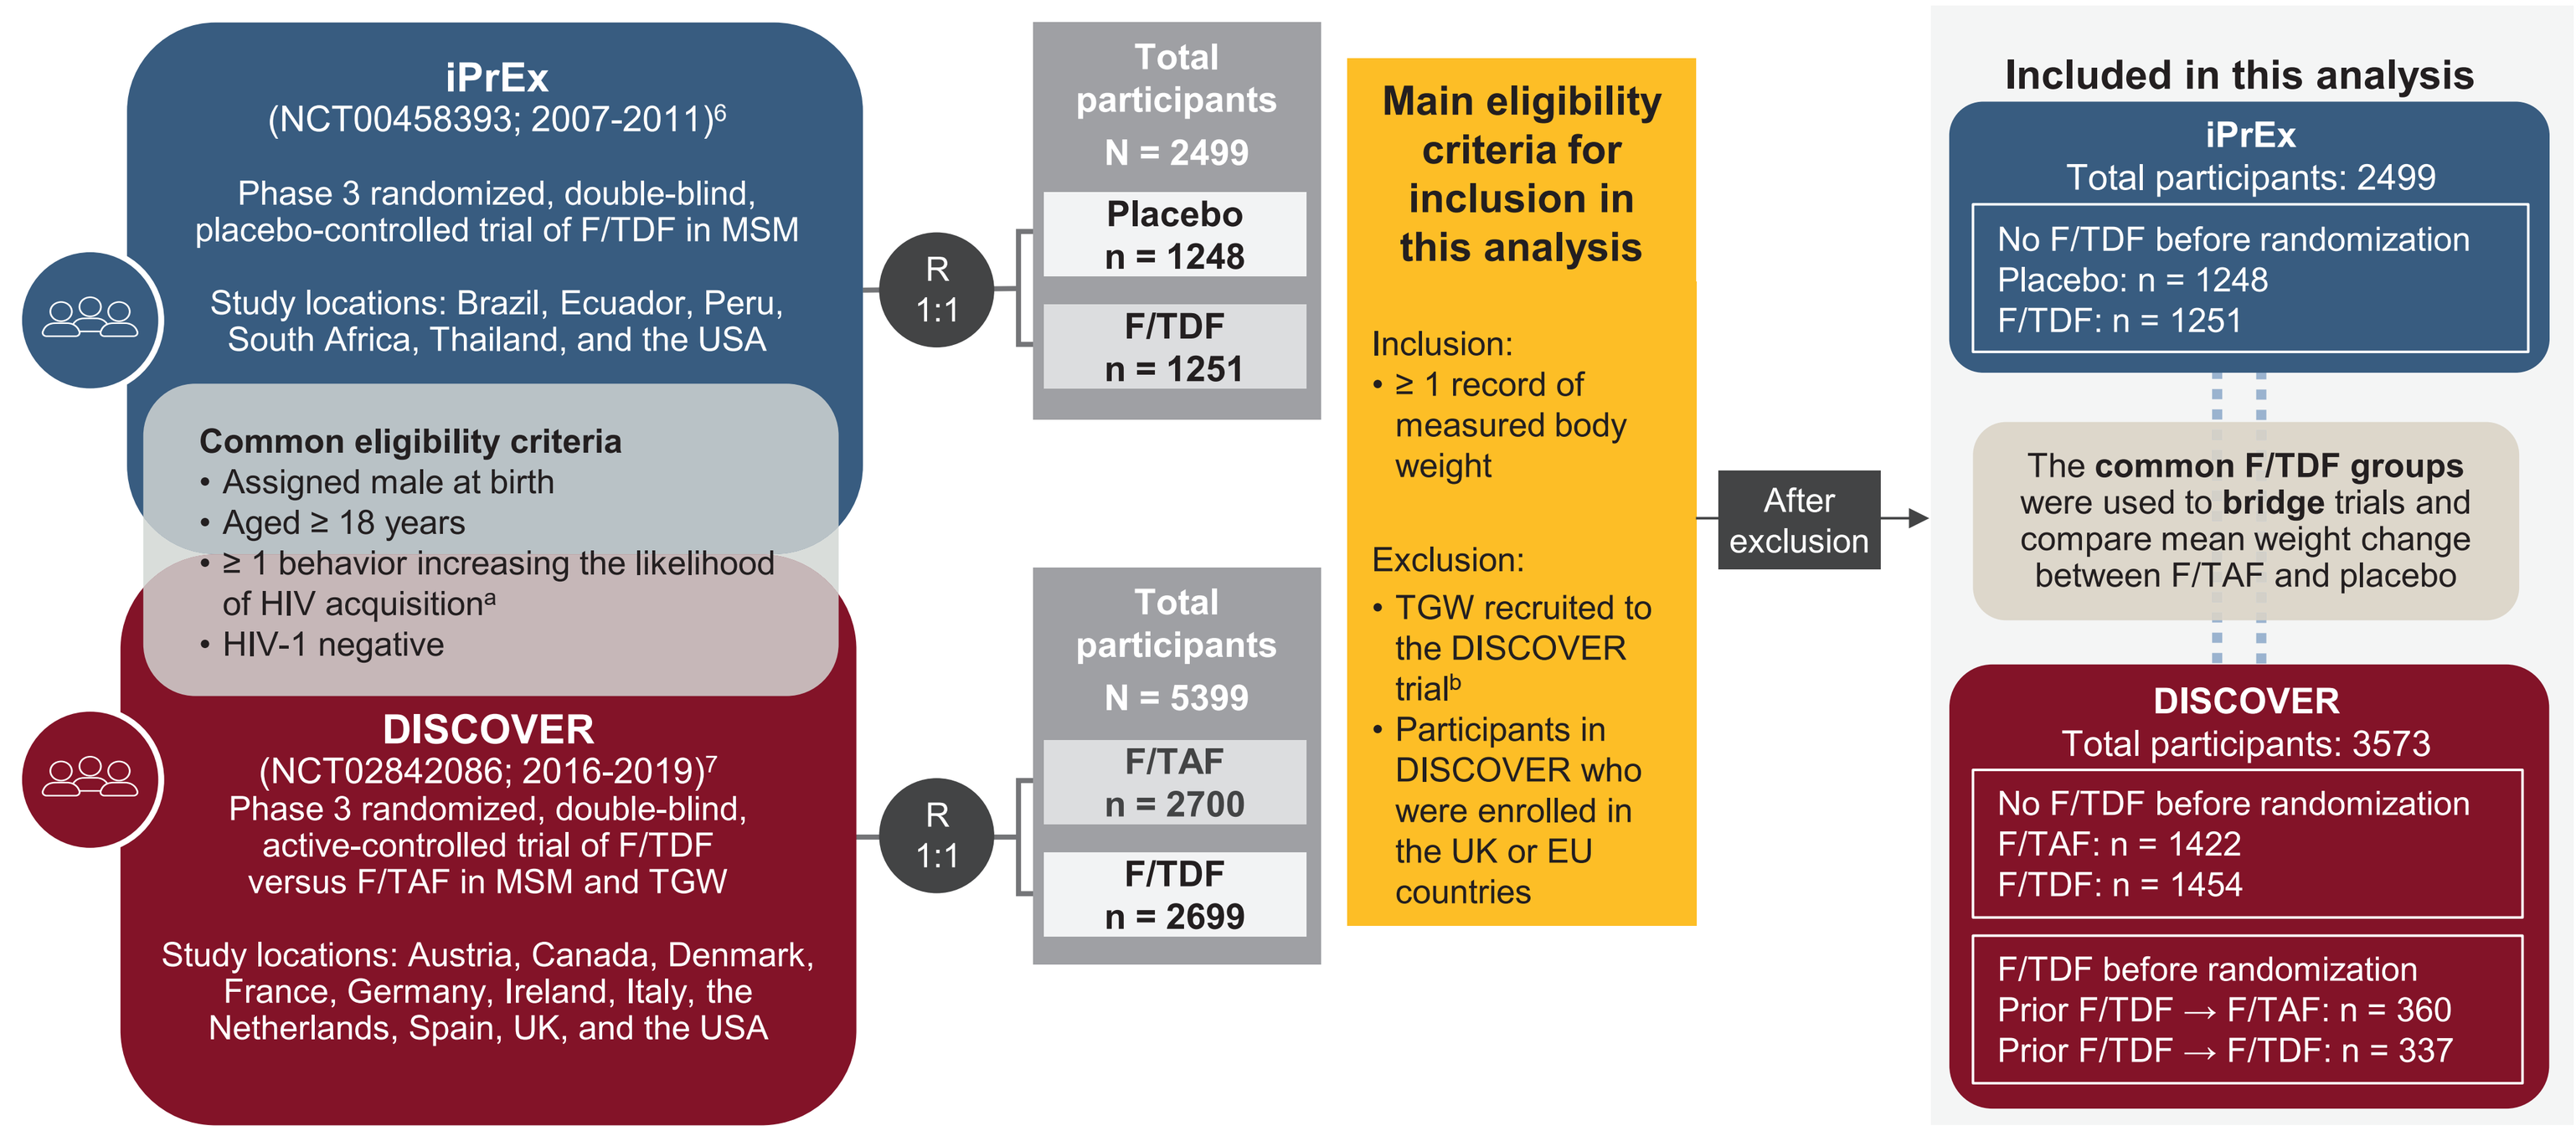
iPrEx
(NCT00458393; 2007-2011)6
Phase 3 randomized, double-blind, placebo-controlled trial of F/TDF in MSM
Study locations: Brazil, Ecuador, Peru, South Africa, Thailand, and the USA
Common eligibility criteria
• Assigned male at birth
• Aged ≥ 18 years
• ≥ 1 behavior increasing the likelihood of HIV acquisitiona
• HIV-1 negative
DISCOVER
(NCT02842086; 2016-2019)7
Phase 3 randomized, double-blind, active-controlled trial of F/TDF versus F/TAF in MSM and TGW
Study locations: Austria, Canada, Denmark, France, Germany, Ireland, Italy, the Netherlands, Spain, UK, and the USA
R 1:1
Total participants N = 2499
Placebo n = 1248
F/TDF n = 1251
Total participants N = 5399
F/TAF n = 2700
F/TDF n = 2699
Main eligibility criteria for inclusion in this analysis
Inclusion:
• ≥ 1 record of measured body weight
Exclusion:
• TGW recruited to the DISCOVER trialb
• Participants in DISCOVER who were enrolled in the UK or EU countries
After exclusion
Included in this analysis
iPrEx
Total participants: 2499
No F/TDF before randomization
Placebo: n = 1248
F/TDF: n = 1251
The common F/TDF groups were used to bridge trials and compare mean weight change between F/TAF and placebo
DISCOVER
Total participants: 3573
No F/TDF before randomization
F/TAF: n = 1422
F/TDF: n = 1454
F/TDF before randomization
Prior F/TDF → F/TAF: n = 360
Prior F/TDF → F/TDF: n = 337

[a]iPrEx: No condom use during anal intercourse with a male HIV-positive partner or a male partner of unknown HIV status during the last 6 months; anal intercourse with > 3 male sex partners during the last 6 months; exchange of money, gifts, shelter, or drugs for anal sex with a male partner during the last 6 months; sex with a male partner and diagnosis of a sexually transmitted infection during the last 6 months or at screening; sexual partner of an HIV-positive man with whom condoms were not consistently used in the last 6 months.
DISCOVER: Condomless anal intercourse with ≥ 2 unique male partners in the past 12 weeks (partners must be either HIV-positive or unknown HIV status); documented history of syphilis or rectal gonorrhea or chlamydia in the past 24 weeks. [b]TGW were not excluded from iPrEx in this study; further analyses of this subpopulation are ongoing.
F/TAF, emtricitabine/tenofovir alafenamide; F/TDF, emtricitabine/tenofovir disoproxil fumarate; MSM, men who have sex with men; R, randomization; TGW, transgender women.

Results

Baseline Characteristics

- Participants in DISCOVER (and the subgroup of participants in DISCOVER with baseline F/TDF) were more likely to be White, aged ≥ 36 years, with a body mass index of ≥ 27 kg/m² and higher use of medications associated with weight gain, compared with participants in iPrEx (**Table 1**)
 - — Mean alanine aminotransferase levels, estimated glomerular filtration rate, and blood glucose levels (non-fasting) were similar in both trials

Table 1. Baseline Demographics and Clinical Characteristics

	F/TDF vs Placebo (iPrEx)	F/TAF vs F/TDF (DISCOVER)	
	All Participants N = 2499	All Participants N = 3573[a]	Receiving F/TDF Before Baseline n = 697[a]
Age, years, mean (SD)	27.1 (9)	36.1 (11)	37.5 (11)
Race or ethnicity, n (%)			
White	431 (17)	2825 (79)	579 (83)
Black	214 (9)	405 (11)	55 (8)
Other[b]	1854 (74)[c]	343 (10)	63 (9)
Hispanic or Latine	1806 (72)	871 (24)	121 (17)
Medications associated with weight change,[d] n (%)			
Gain	63 (3)	516 (14)	131 (19)
Loss	28 (1)	287 (8)	62 (9)
Unknown	31 (1)	246 (7)	72 (10)
Body weight, kg, mean (SD)	68.1 (14)	85.0 (18)	84.6 (18)
BMI, kg/m², mean (SD)	23.8 (4)	27.0 (5)	26.7 (5)
Alanine aminotransferase level, U/L, mean (SD)	26.5 (17)	28.34 (15)	29.9 (15)
$eGFR_{CG}$, mL/min, mean (SD)	118.6 (23)	129.8 (37)	124.2 (36)
Blood glucose level (non-fasting), mg/dL, mean (SD)	90.4 (14)	94.9 (19)	92.8 (18)

[a]Participants in DISCOVER receiving F/TDF before baseline are counted in both DISCOVER columns and were subsequently randomized to continue with F/TDF or switch to F/TAF. [b]Includes American Indian or Alaska Native; Asian; Native Hawaiian or Other Pacific Islander; other race; and participants with Not Permitted designation (when local regulators did not allow collection of race/ethnicity information). [c]The majority of participants in the iPrEx study self-reported as "mixed race or other". [d]Antidepressants/psychoanaleptics, antidiabetics, antiepileptics, antihistamines, antipsychotics, contraceptives, corticosteroids, beta-androgenic blockers, insulin.
BMI, body mass index; eGFR, estimated glomerular filtration rate by Cockroft-Gault equation; F/TAF, emtricitabine/tenofovir alafenamide; F/TDF, emtricitabine/tenofovir disoproxil fumarate.

No Significant Difference in Estimated Mean Weight Change From Baseline Between F/TAF and Placebo in Participants With No Prior Use of F/TDF

- Estimated mean weight change from baseline with F/TDF was similar in DISCOVER and iPrEX (**Figure 2**)
- There was no statistically significant difference across the 96-week period in mean weight change from baseline between participants receiving F/TAF or placebo at any time (*P* = 0.10) (**Figure 2**), including at Week 24, 48, 72 and Week 96 (**Table 2**)
- The results of sensitivity assessments (not shown) were highly consistent with primary findings. These assessments included per protocol–type analyses and robust estimation strategies

Figure 2. Estimated Mean Weight Change From Baseline in iPrEx and DISCOVER Participants With No Prior Use of F/TDF

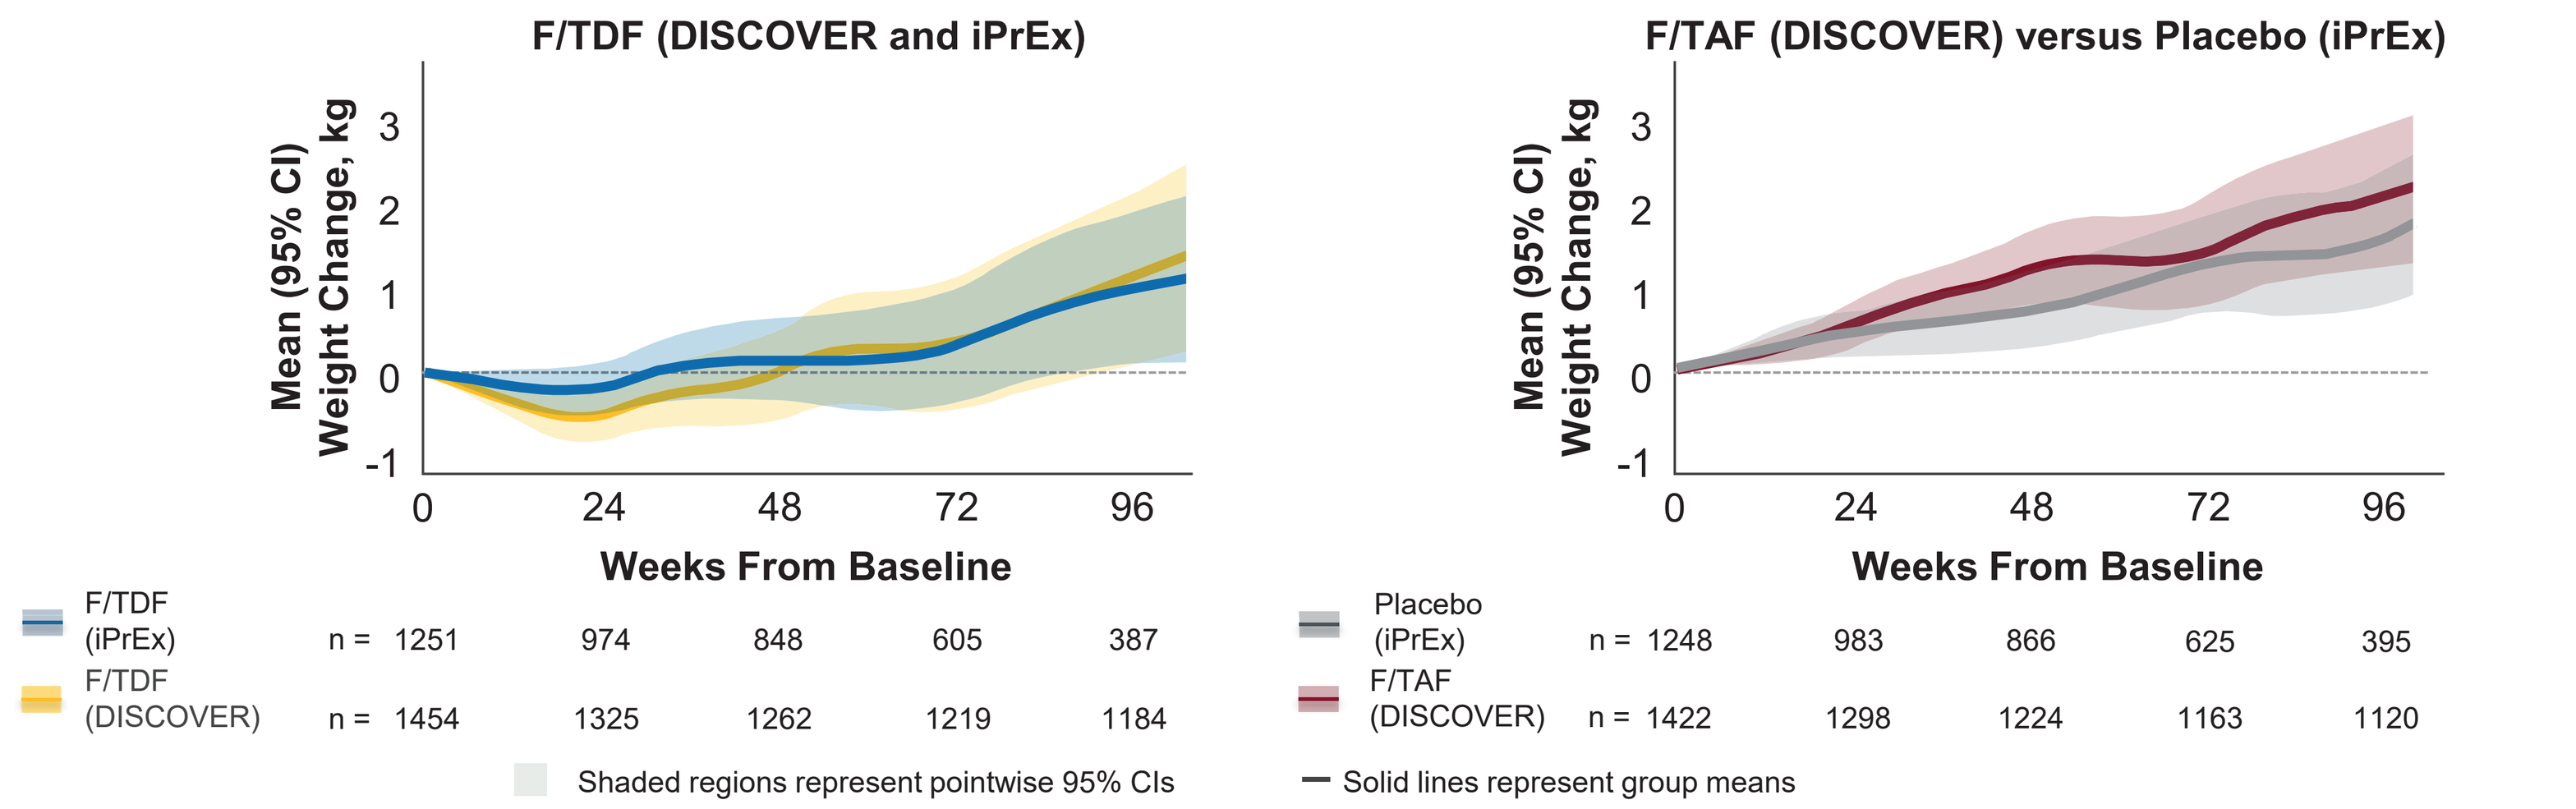
F/TDF (DISCOVER and iPrEx)
F/TAF (DISCOVER) versus Placebo (iPrEx)
Mean (95% CI) Weight Change, kg
Weeks From Baseline
0 24 48 72 96
F/TDF (iPrEx) n = 1251 974 848 605 387
F/TDF (DISCOVER) n = 1454 1325 1262 1219 1184
Placebo (iPrEx) n = 1248 983 866 625 395
F/TAF (DISCOVER) n = 1422 1298 1224 1163 1120
Shaded regions represent pointwise 95% CIs
Solid lines represent group means

Data were adjusted for baseline age, country of enrollment, diabetes status, glucose (non-fasting), kidney function (estimated glomerular filtration rate), liver function (alanine aminotransferase), race (White/Black/Other), height, use of medications associated with weight change (both baseline and trial-incident), and weight.
F/TAF, emtricitabine/tenofovir alafenamide; F/TDF, emtricitabine/tenofovir disoproxil fumarate.

Table 2. Mean Weight Change Contrasts (F/TAF > Placebo) at Landmark Timepoints (With No Prior Use of F/TDF)

	Difference in Mean (95% CI) Weight Change, kg
Week 24	+0.07 (-0.25, 0.39)
Week 48	+0.44 (-0.04, 0.92)
Week 72	+0.13 (-0.51, 0.77)
Week 96	+0.53 (-0.28, 1.34)

Data were adjusted for baseline age, country of enrollment, diabetes status, glucose level (non-fasting), kidney function (estimated glomerular filtration rate), liver function (alanine aminotransferase level), race (White/Black/other), height, use of medications associated with weight change (both baseline and trial-incident), and body weight.
F/TAF, emtricitabine/tenofovir alafenamide; F/TDF, emtricitabine/tenofovir disoproxil fumarate.

- Proportions of individuals experiencing large increases in body weight over 48 weeks were similar with F/TAF versus placebo (**Table 3**)

Table 3. Top Percentiles[a] (95% CI) of Weight Change From Baseline at Week 48 in Participants With No Prior Use of F/TDF Who Were Receiving Placebo (iPrEx) or F/TAF (DISCOVER)

	Placebo (iPrEx), kg	F/TAF (DISCOVER), kg
80th	4.2 (3.7, 4.7)	4.1 (3.8, 4.4)
85th	5.0 (4.4, 5.7)	4.6 (4.2, 5.1)
90th	5.7 (5.1, 6.3)	5.8 (5.3, 6.2)
95th	6.9 (5.8, 8.1)	7.9 (7.0, 8.7)
99th	11.6 (9.0, 14.2)	11.1 (10.1, 12.2)

[a]Estimated percentiles were adjusted for the same baseline covariates as in the primary analysis.
F/TAF, emtricitabine/tenofovir alafenamide; F/TDF, emtricitabine/tenofovir disoproxil fumarate.

Average Weight Gain (Relative to Placebo) in Participants Taking F/TAF With Prior Use of F/TDF at Baseline Was Consistent With a Reversible Weight-Suppressive Effect of F/TDF

- Participants switching from F/TDF to F/TAF at baseline in DISCOVER experienced a general increase in mean (95% CI) body weight compared with those receiving placebo who had not been on F/TDF at baseline (**Figure 3**)

Figure 3. Estimated Mean Weight Change in Participants Taking F/TDF at Baseline Who Switched to F/TAF

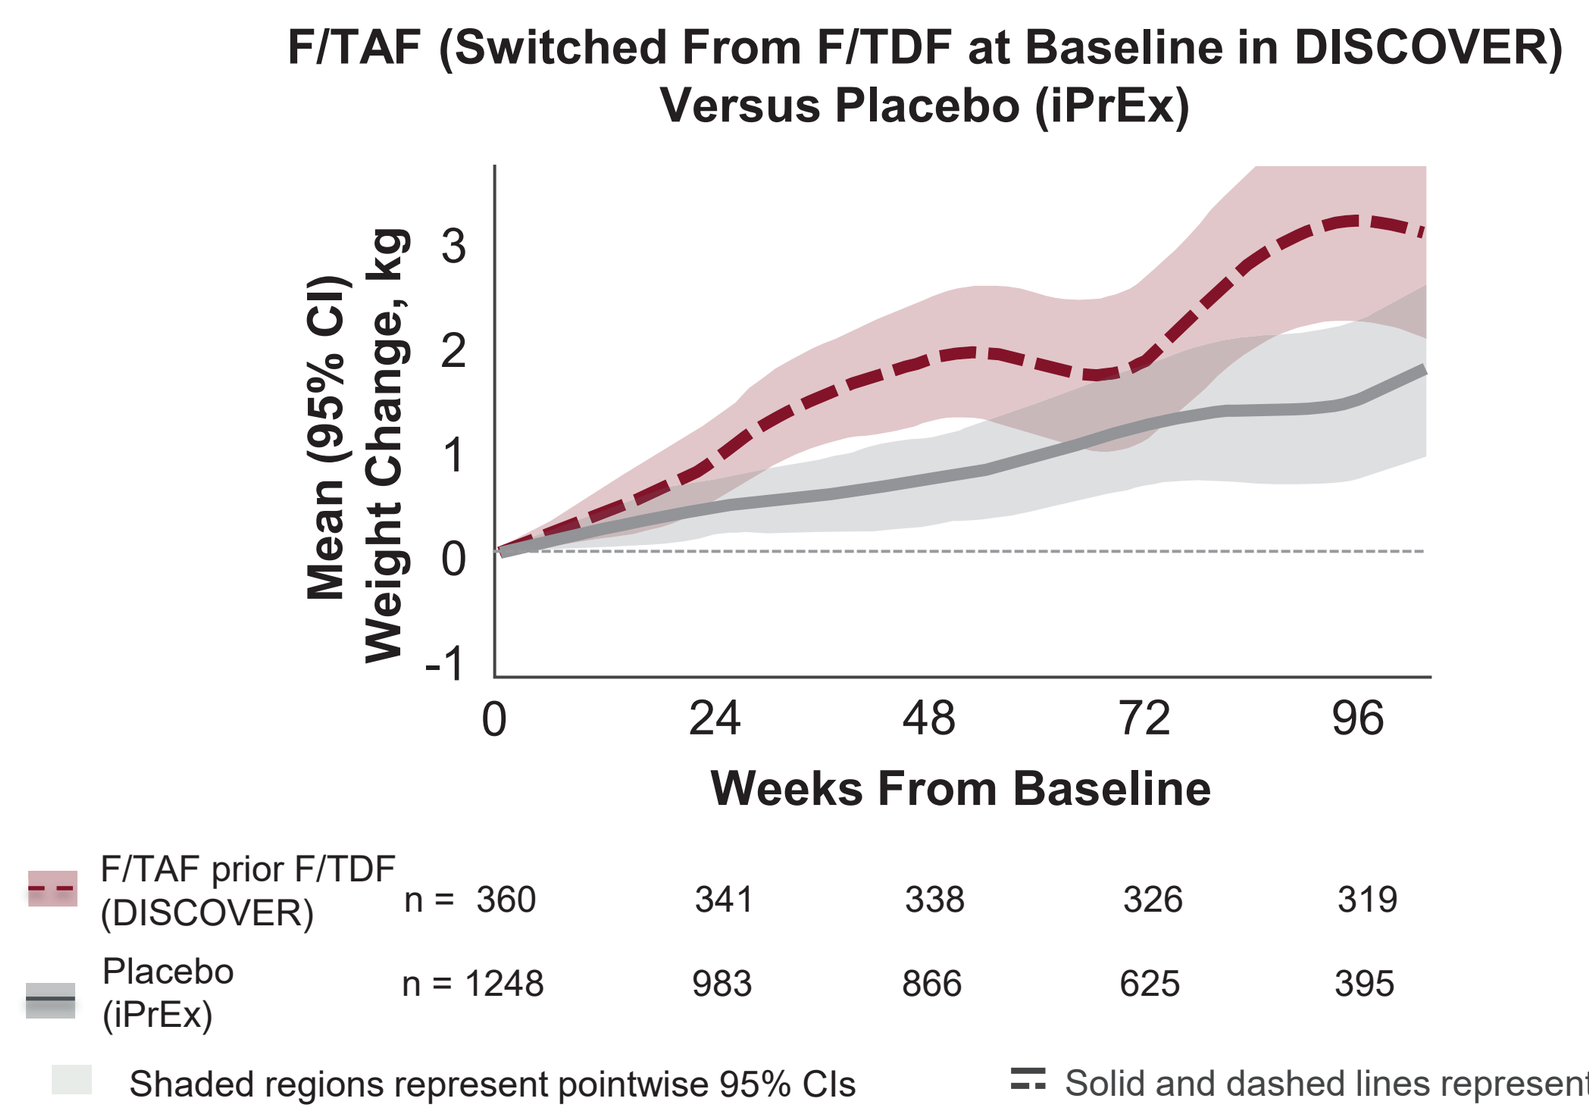
F/TAF (Switched From F/TDF at Baseline in DISCOVER) Versus Placebo (iPrEx)
Mean (95% CI) Weight Change, kg
Weeks From Baseline
0 24 48 72 96
F/TAF prior F/TDF (DISCOVER) n = 360 341 338 326 319
Placebo (iPrEx) n = 1248 983 866 625 395
Shaded regions represent pointwise 95% CIs
Solid and dashed lines represent group means

	Difference in Mean (95% CI) Weight Change From Baseline Between F/TAF (With F/TDF Before Baseline) and Placebo, kg
Week 24	+0.46 (0.02, 0.90)
Week 48	+1.15 (0.53, 1.77)
Week 72	+0.63 (-0.16, 1.42)
Week 96	+1.74 (0.78, 2.70)

Data were adjusted for baseline age, country of enrollment, diabetes status, glucose level (non-fasting), kidney function (estimated glomerular filtration rate), liver function (alanine aminotransferase level), race (White/Black/other), height, use of medications associated with weight change (both baseline and trial-incident), and body weight.
F/TAF, emtricitabine/tenofovir alafenamide; F/TDF, emtricitabine/tenofovir disoproxil fumarate.

References: **1.** Panel on Antiretroviral Guidelines for Adults and Adolescents. https://clinicalinfo.hiv.gov/sites/default/files/guidelines/documents/adult-adolescent-arv/what-to-start-adult-adolescent-arv.pdf (accessed Dec. 2, 2024). **2.** Shah S, et al. *AIDS*. 2021;35(suppl 2):S189-95. **3.** Ogbuagu O, et al. Oral 2940 presented at: Conference on Retroviruses and Opportunistic Infections (CROI); March 8-11, 2020; Boston, MA. **4.** Erlandson KM, et al. *Clin Infect Dis*. 2021;73:1440-51. **5.** Sax PE, et al. *Clin Infect Dis*. 2020;71:1379-89. **6.** Grant RM, et al. *N Engl J Med*. 2010;363:2587-99. **7.** Mayer KH, et al. *Lancet*. 2020;396:239-54.

Acknowledgments: We thank all study participants, investigators, and staff. This study was sponsored by Gilead Sciences, Inc. Medical writing support was provided by Flint Stevenson-Jones, PhD (Aspire Scientific Ltd, UK), and was funded by Gilead Sciences, Inc.

Disclosures: **DG** reports receiving consulting/advisor fees from Gilead Sciences, Inc. Merck, and ViiV Healthcare. **AW** is an employee of Gilead Sciences, Inc., and owns stocks/shares in Gilead Sciences, Inc., Johnson & Johnson, and Viatris. **YT**, **AM**, **JG**, and **CC** are employees of, and own stocks/shares in, Gilead Sciences, Inc.

Correspondence: David Glidden, david.glidden@ucsf.edu.

Conference on Retroviruses and Opportunistic Infections (CROI); March 9–12, 2025; San Francisco, CA, USA