Bictegravir + Lenacapavir: Baseline and Week 48 Resistance Analyses in ARTISTRY-1 Phase 2

Nicolas Margot, Nina Pennetzdorfer, Vidula Naik, Mauricio Montezuma, Peter Sklar, Christian Callebaut

Gilead Sciences, Inc., Foster City, CA, USA

738

GS-US-621-6289

Conclusions

- The high frequency of baseline RAMs among VS participants in ARTISTRY-1 Phase 2 was consistent with the study requirement that participants be on a complex regimen
- The presence of baseline RAMs did not affect the efficacy of BIC + LEN
 - 93% treatment success overall, regardless of baseline RAMs, including baseline INSTI RAMs
- On-treatment emergence of capsid polymorphism (N74T) was observed in one participant who resuppressed on a LEN-containing regimen
 - No phenotypic loss of susceptibility to LEN
- These data underscore the potential of the use of once-daily BIC + LEN to optimize therapy in people receiving a complex ART regimen

Plain Language Summary

- At least two or more drugs (called antiretroviral drugs) are needed to treat people who have human immunodeficiency virus (HIV). Some antiretroviral drugs may not work for some people with HIV, due to side effects or because the virus has become resistant to the drugs (in other words, those specific drugs no longer kill the virus). As a result, these people sometimes need to take multiple pills each day to treat their HIV; this can be hard to do.
- Lenacapavir and bictegravir are two drugs that are approved to treat HIV infection and are each taken together with other drugs. Combining these two drugs into one pill taken once a day could help lower the number of pills that people need to take to treat their HIV infection, and make their treatment easier to take.
- People in this study took an average of three pills per day to prevent the growth of their HIV and had no HIV in their blood at the start of the study. After 48 weeks of treatment, people who switched their treatment plan to the combination of lenacapavir plus bictegravir experienced a similar result as when they were still taking many pills to treat their HIV. This showed that combining lenacapavir and bictegravir was an effective treatment.
- Even though many participants with HIV had mutations (changes) in their HIV showing resistance (some drugs may no longer kill the virus) when this study started, giving lenacapavir and bictegravir together stopped the virus from making copies of itself.

Introduction

- Once-daily single tablet regimens (STRs) are standard of care for HIV treatment[1]
- However, many people with HIV (PWH) take complex antiretroviral therapy (ART) regimens for reasons including drug resistance, intolerance, toxicity, drug-drug interactions, and contraindications to existing STRs[1-4]
- A bictegravir + lenacapavir (BIC + LEN) STR could optimize the treatment of virologically suppressed (VS) PWH who are unable to take currently available STRs
 - BIC is a global, guideline-recommended integrase strand transfer inhibitor (INSTI) with a high barrier to resistance[1,5-7]
 - LEN is a first-in-class capsid inhibitor with an orthogonal resistance profile to existing/major drug classes[8]
 - There is an absence of capsid inhibitor resistance in the HIV community[9]
- ARTISTRY-1 (NCT05502341) is a Phase 2/3 study evaluating the safety and efficacy of a complete regimen combining BIC and LEN
 - At Baseline, 43%, 19%, 11%, and 27% of participants were taking 2, 3, 4, or ≥ 5 pills/day, respectively
- In Phase 2 of ARTISTRY-1, the efficacy of BIC + LEN was compared with remaining on a complex multi-tablet ART regimen in VS participants (n = 128)
 - Arm 1: BIC 75 mg + LEN 25 mg, n = 51; Arm 2: BIC 75 mg + LEN 50 mg, n = 52; Arm 3: Complex ART regimen, n = 25
- **High rates of virologic suppression were observed across treatment groups at Week 48 (90.4% to 100%; no statistical differences between groups)[10]**

Objectives

- Characterize the baseline resistance profile of participants
- Analyze the potential emergence of resistance to study drugs after 48 weeks of treatment

Methods

- ARTISTRY-1 is a Phase 2/3, randomized, open-label, multicenter study

Study Design of Phase 2 of ARTISTRY-1 (NCT05502341)

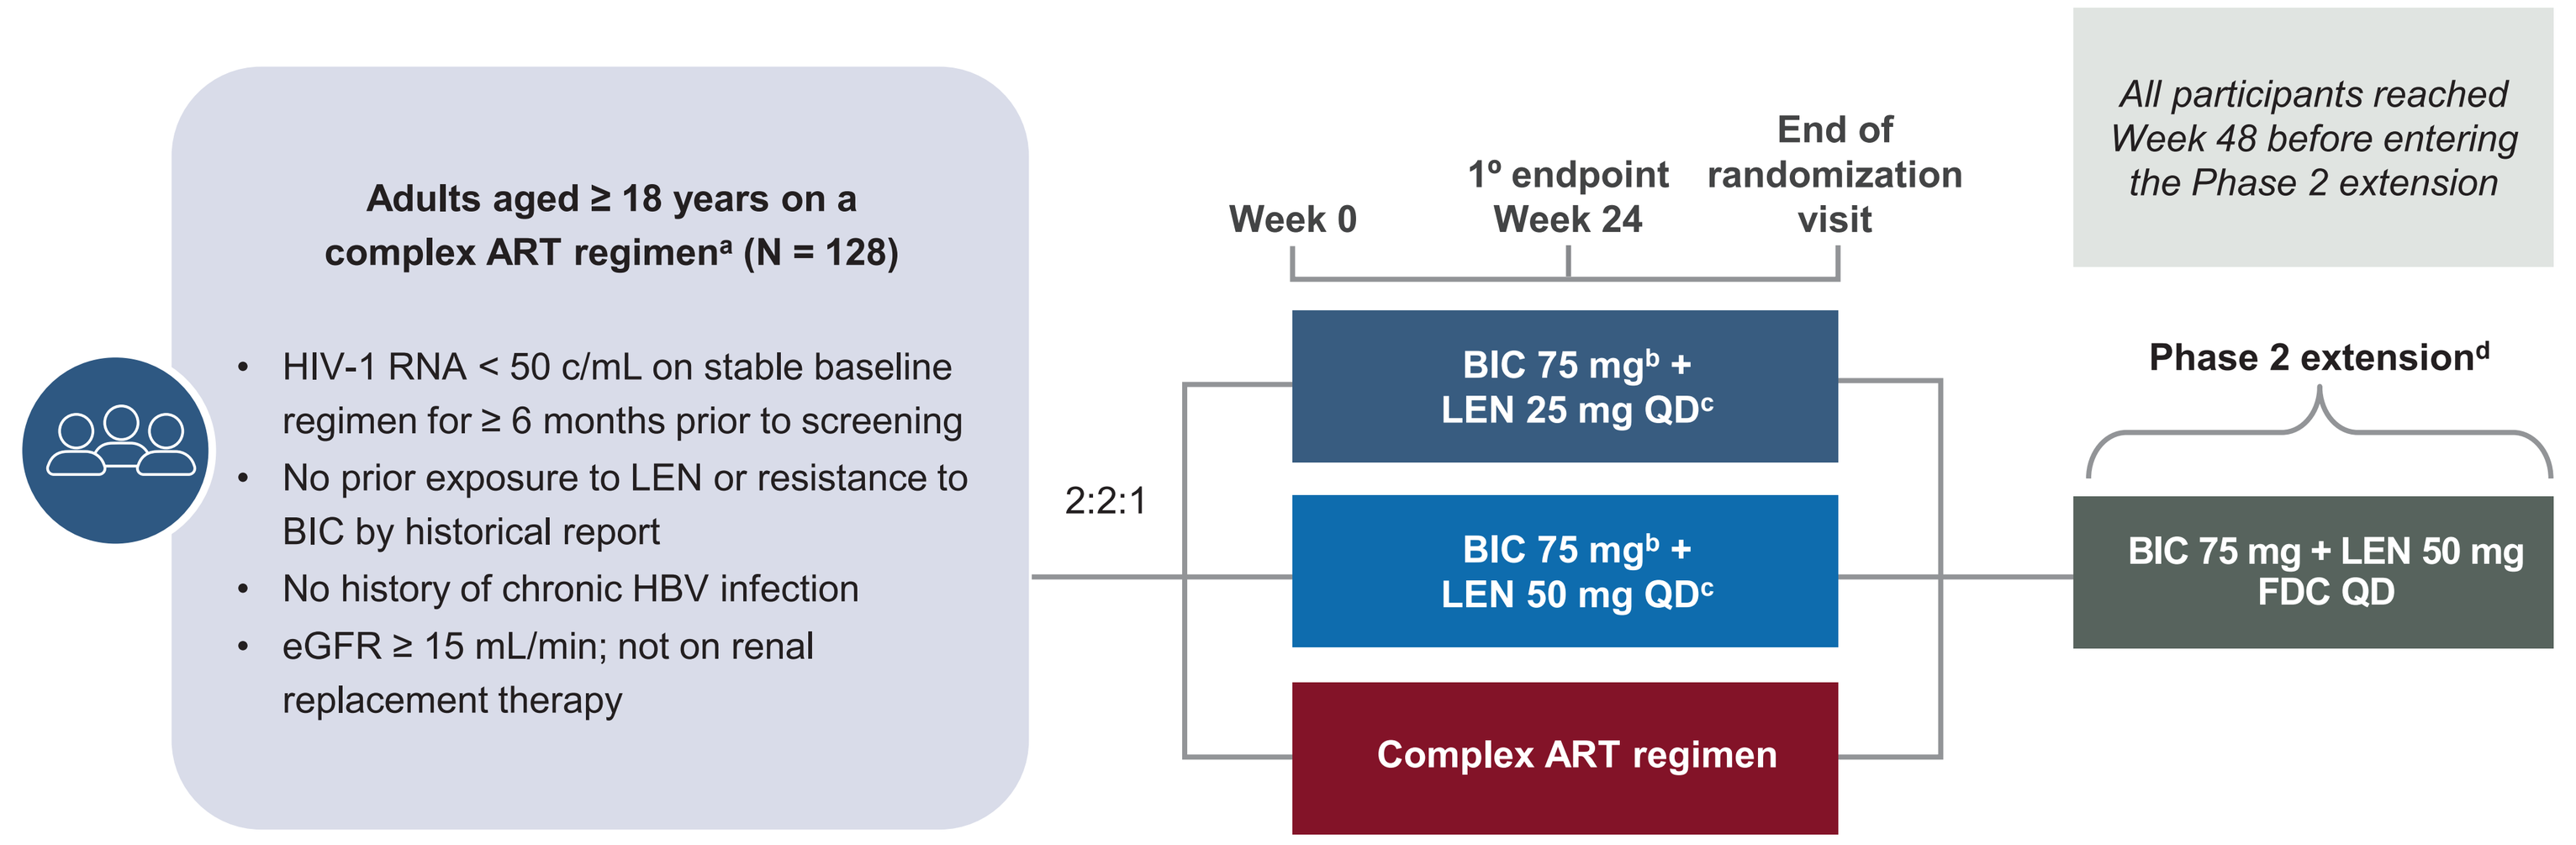

Adults aged ≥ 18 years on a complex ART regimen[a] (N = 128)
HIV-1 RNA < 50 c/mL on stable baseline regimen for ≥ 6 months prior to screening
No prior exposure to LEN or resistance to BIC by historical report
No history of chronic HBV infection
eGFR ≥ 15 mL/min; not on renal replacement therapy
2:2:1
Week 0
1° endpoint Week 24
End of randomization visit
BIC 75 mg[b] + LEN 25 mg QD[c]
BIC 75 mg[b] + LEN 50 mg QD[c]
Complex ART regimen
All participants reached Week 48 before entering the Phase 2 extension
Phase 2 extension[d]
BIC 75 mg + LEN 50 mg FDC QD

A **complex ART regimen** was defined as:
- A regimen containing a boosted PI or NNRTI plus ≥ 1 other third agent from a class other than NRTI, or
- A regimen of ≥ 2 pills/day, or a regimen requiring dosing more than QD, or
- A regimen containing parenteral agent(s) (excluding a complete long-acting injectable regimen), as well as oral agents

Endpoints at Week 48
- Proportions of participants with HIV-1 RNA < 50 c/mL, determined using US FDA Snapshot analysis
- Change from baseline in CD4 cell count
- Proportion of participants with AEs

[a]Due to viral resistance, intolerance, or contraindication to existing STRs. [b]BIC 75-mg single agent provides exposure consistent with BIC 50 mg as part of B/F/TAF. [c]All participants receiving BIC + LEN received an oral loading dose of LEN 600 mg on Days 1 and 2 of treatment. [d]Participants who switch from a complex ART regimen in the extension phase will receive the oral loading doses of LEN.
AE, adverse event; ART, antiretroviral therapy; B/F/TAF, bictegravir/emtricitabine/tenofovir alafenamide; BIC, bictegravir; c, copies; CD4, cluster of differentiation 4; eGFR, estimated glomerular filtration rate; FDA, Food and Drug Administration; FDC, fixed-dose combination; HBV, hepatitis B virus; LEN, lenacapavir; NNRTI, non-nucleoside reverse transcriptase inhibitor; NRTI, nucleoside/nucleotide reverse transcriptase inhibitor; PI, protease inhibitor; QD, once daily; STR, single tablet regimen.

- The presence of resistance-associated mutations (RAMs) at baseline was assessed using historical genotypic reports and retrospective proviral DNA analysis (GenoSure Archive; Monogram Biosciences, South San Francisco, CA, USA)
- RAMs are as follows:
 - **Nucleoside/nucleotide reverse transcriptase inhibitors (NRTIs):** M41L, K65R/E/N, D67N, T69 insertion, K70E/R, L74V/I, Y115F, Q151M, M184V/I, L210W, T215Y/F, K219E/Q/N/R in reverse transcriptase (RT)
 - **Non-nucleoside reverse transcriptase inhibitors (NNRTIs):** L100I, K101E/P, K103N/S, V106M/A, V108I, E138A/G/K/Q/R, V179L, Y181C/I/V, Y188C/H/L, G190A/Q/S, H221Y, P225H, F227C/L, M230L/I in RT
 - **Protease inhibitors (PIs):** D30N, V32I, M46I/L, I47V/A, G48V, I50V/L, I54M/L/V, Q58E, T74P, L76V, V82A/F/L/S/T, N83D, I84V, N88S, L90M in protease (PR)
 - **INSTIs:** T66I/A/K, E92Q/G/V, G118R, F121C/Y, G140R, Y143R/H/C, S147G, Q148H/K/R, N155H/S, R263K in integrase (IN)
- The impact of baseline RAMs on treatment response at Week 48 was evaluated using the Fisher exact test
- Post-baseline resistance analysis was conducted for participants experiencing virologic rebound (VR)
 - VR: Confirmed HIV-1 RNA ≥ 50 copies (c)/mL or last visit HIV-1 RNA ≥ 50 c/mL
 - Analyzed if HIV-1 RNA ≥ 200 c/mL
 - Samples analyzed at Monogram Biosciences
- Phenotypic analyses were conducted at Monogram Biosciences and Gilead Sciences (Foster City, CA, USA)
 - Mutations of interest were introduced in pXXLAI molecular clones
 - Cloned into wild-type XXLAI or XXLAI containing diverse polymorphisms in capsid
 - Susceptibility to BIC and LEN were determined in single-cycle (Monogram Gag-Pro assay) and multi-cycle assays (MT-2 and RevLun assays[8,11])

Results

Baseline Pre-Existing Resistance Mutations Per Treatment Group

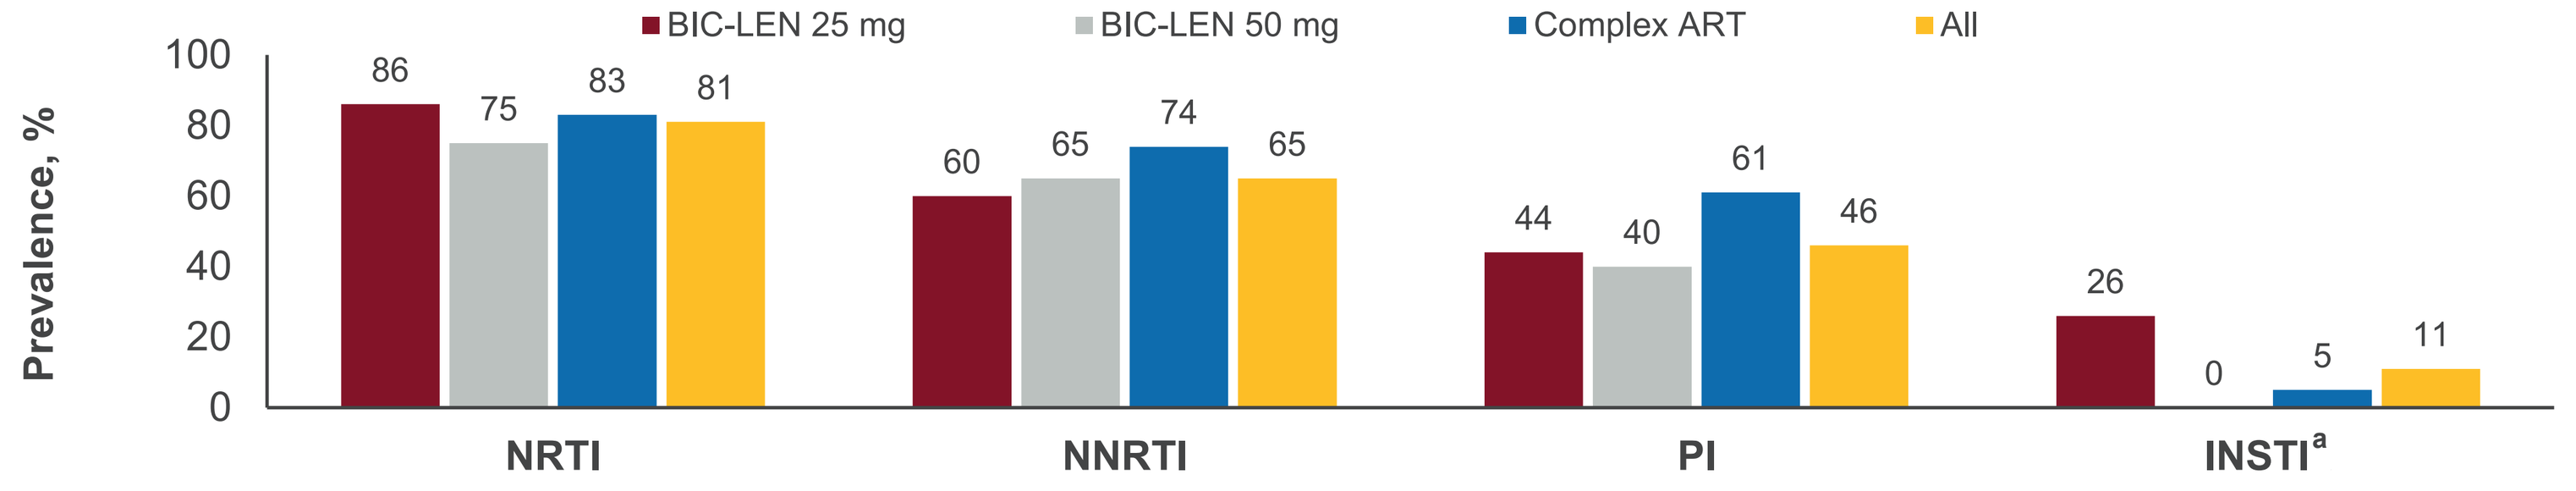

BIC-LEN 25 mg
BIC-LEN 50 mg
Complex ART
All
Prevalence, %
100
80
60
40
20
0
86 75 83 81
60 65 74 65
44 40 61 46
26 0 5 11
NRTI
NNRTI
PI
INSTI[a]

[a]Proviral DNA analysis only for INSTI class (n with data = 94). For all three other mutation categories, n with data = 124. ART, antiretroviral therapy; BIC, bictegravir; INSTI, integrase strand transfer inhibitor; LEN, lenacapavir; NNRTI, non-nucleoside reverse transcriptase inhibitor; NRTI, nucleoside/nucleotide reverse transcriptase inhibitor; PI, protease inhibitor.

- High level of resistance to NRTI, NNRTI, and PI classes at baseline

Baseline Pre-Existing NRTI Mutations Per Treatment Group

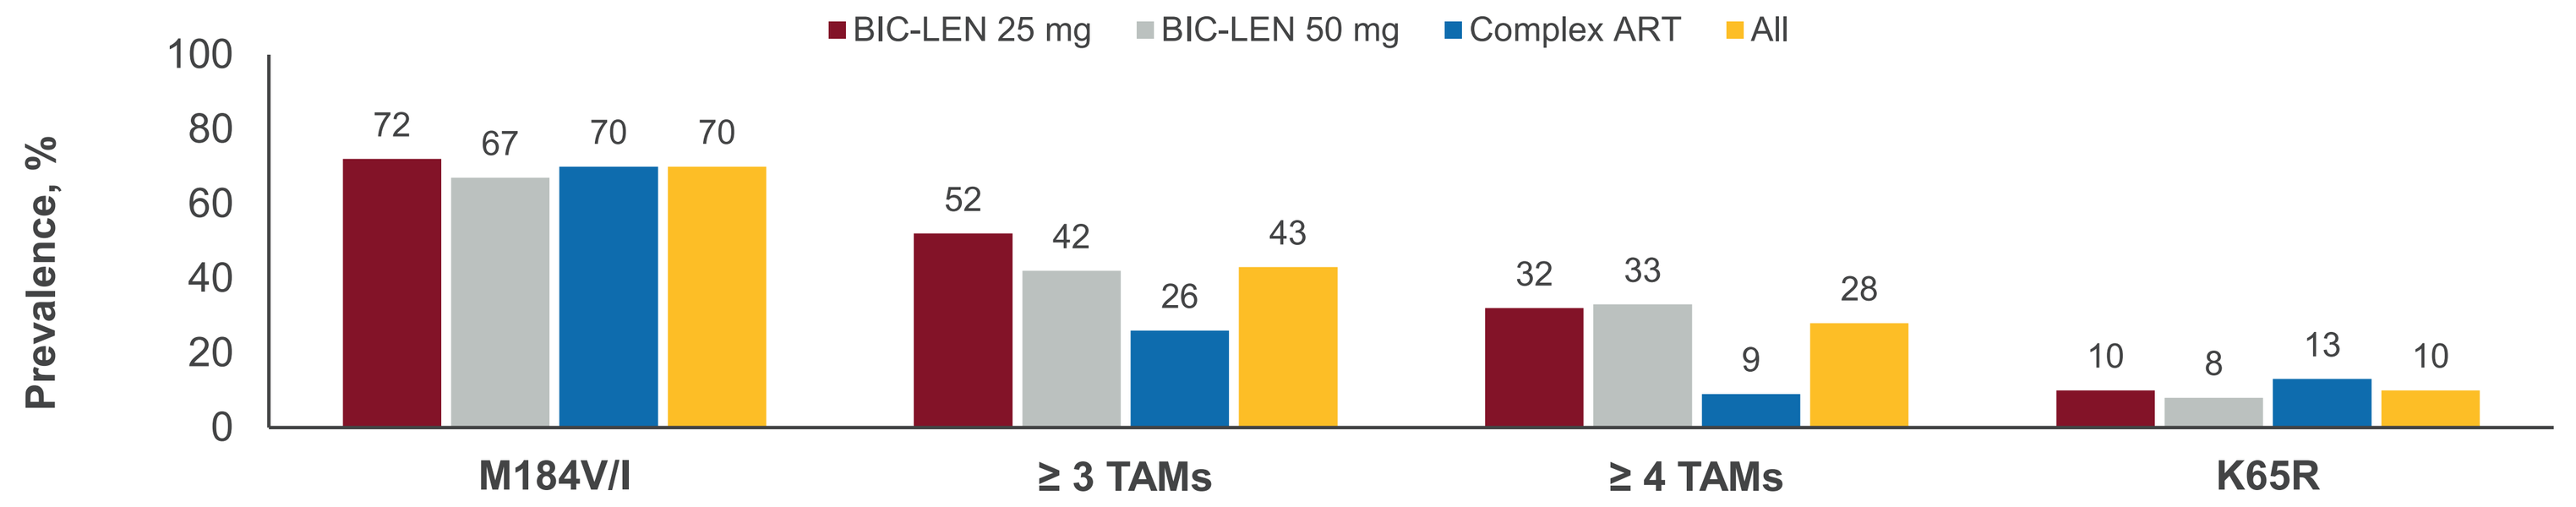

BIC-LEN 25 mg
BIC-LEN 50 mg
Complex ART
All
Prevalence, %
100
80
60
40
20
0
72 67 70 70
52 42 26 43
32 33 9 28
10 8 13 10
M184V/I
≥ 3 TAMs
≥ 4 TAMs
K65R

TAMs: M41L, D67N, K70R, L210W, T215Y/F, and K219E/N/Q/R in RT; N with data = 124.
ART, antiretroviral therapy; BIC, bictegravir; LEN, lenacapavir; RT, reverse transcriptase; TAM, thymidine analog mutation.

- High prevalence of M184V/I and thymidine analog mutation (TAMs) at baseline
 - Only 11% of participants with M184V/I alone

Response to Treatment Per Baseline Resistance Category

	Participants With Virologic Suppression, % (n/N)[a]			
	BIC-LEN 25 mg	BIC-LEN 50 mg	Complex ART	All
All participants	92.2 (47/51)	90.4 (47/52)	100 (25/25)	93.0 (119/128)
With NRTI-RAMs	92.5 (37/40)	91.9 (34/37)	100 (18/18)	93.7 (89/95)
Without NRTI-RAMs	88.9 (8/9)	86.7 (13/15)	100 (5/5)	89.7 (26/29)
P value[b]	0.57	0.62	1.0	0.44
With NNRTI-RAMs	92.6 (25/27)	93.5 (29/31)	100 (16/16)	94.6 (70/74)
Without NNRTI-RAMs	90.9 (20/22)	85.7 (18/21)	100 (7/7)	90.0 (45/50)
P value[b]	1.0	0.38	1.0	0.48
With PI-RAMs	100 (22/22)	88.2 (15/17)	100 (12/12)	96.1 (49/51)
Without PI-RAMs	85.2 (23/27)	91.4 (32/35)	100 (11/11)	90.4 (66/73)
P value[b]	0.12	1.0	1.0	0.31
With INSTI-RAMs[c]	88.9 (8/9)	0	100 (1/1)	90.0 (9/10)
Without INSTI-RAMs[c]	88.5 (23/26)	89.7 (35/39)	100 (19/19)	91.7 (77/84)
P value[b]	1.0	NA	1.0	1.0

[a]US FDA-defined Snapshot algorithm: treatment success if HIV-1 RNA < 50 c/mL. [b]Fisher exact test. [c]Proviral DNA analysis only (n with data = 94). For all three other mutation categories, n with data = 124. ART, antiretroviral therapy; BIC, bictegravir; c, copies; FDA, Food and Drug Administration; INSTI, integrase strand transfer inhibitor; LEN, lenacapavir; NA, not available; NNRTI, non-nucleoside reverse transcriptase inhibitor; NRTI, nucleoside/nucleotide reverse transcriptase inhibitor; PI, protease inhibitor; RAM, resistance-associated mutation.

- Similarly high virologic suppression rates across groups
- Presence of baseline RAMs did not affect treatment response

Response in Participants With Baseline INSTI RAMs

ID	INSTI RAMs	Treatment Group	Treatment Outcome at Week 48
1	T66T/A E92E/Q S147S/G Q148Q/R N155N/H	BIC-LEN 25 mg	Virologic suppression
2	R263R/K	BIC-LEN 25 mg	Virologic suppression
3	T66T/A	BIC-LEN 25 mg	Virologic suppression
4	G140G/S Q148Q/H	BIC-LEN 25 mg	Virologic suppression
5	Q148Q/H	BIC-LEN 25 mg	Virologic suppression
6	Q148Q/R	Complex ART	Virologic suppression
7	N155N/H	BIC-LEN 25 mg	Participant's decision to discontinue (virologic suppression at last visit)
8	N155N/H	BIC-LEN 25 mg	Virologic suppression
9	N155N/H	BIC-LEN 25 mg	Virologic suppression
10	G140G/R	BIC-LEN 25 mg	Virologic suppression

INSTI, integrase strand transfer inhibitor; RAM, resistance-associated mutation.

- None of the participants with baseline INSTI RAMs experienced treatment failure with BIC + LEN

Participant With HIV-1 RNA ≥ 50 c/mL at Week 48 – 1 of 3

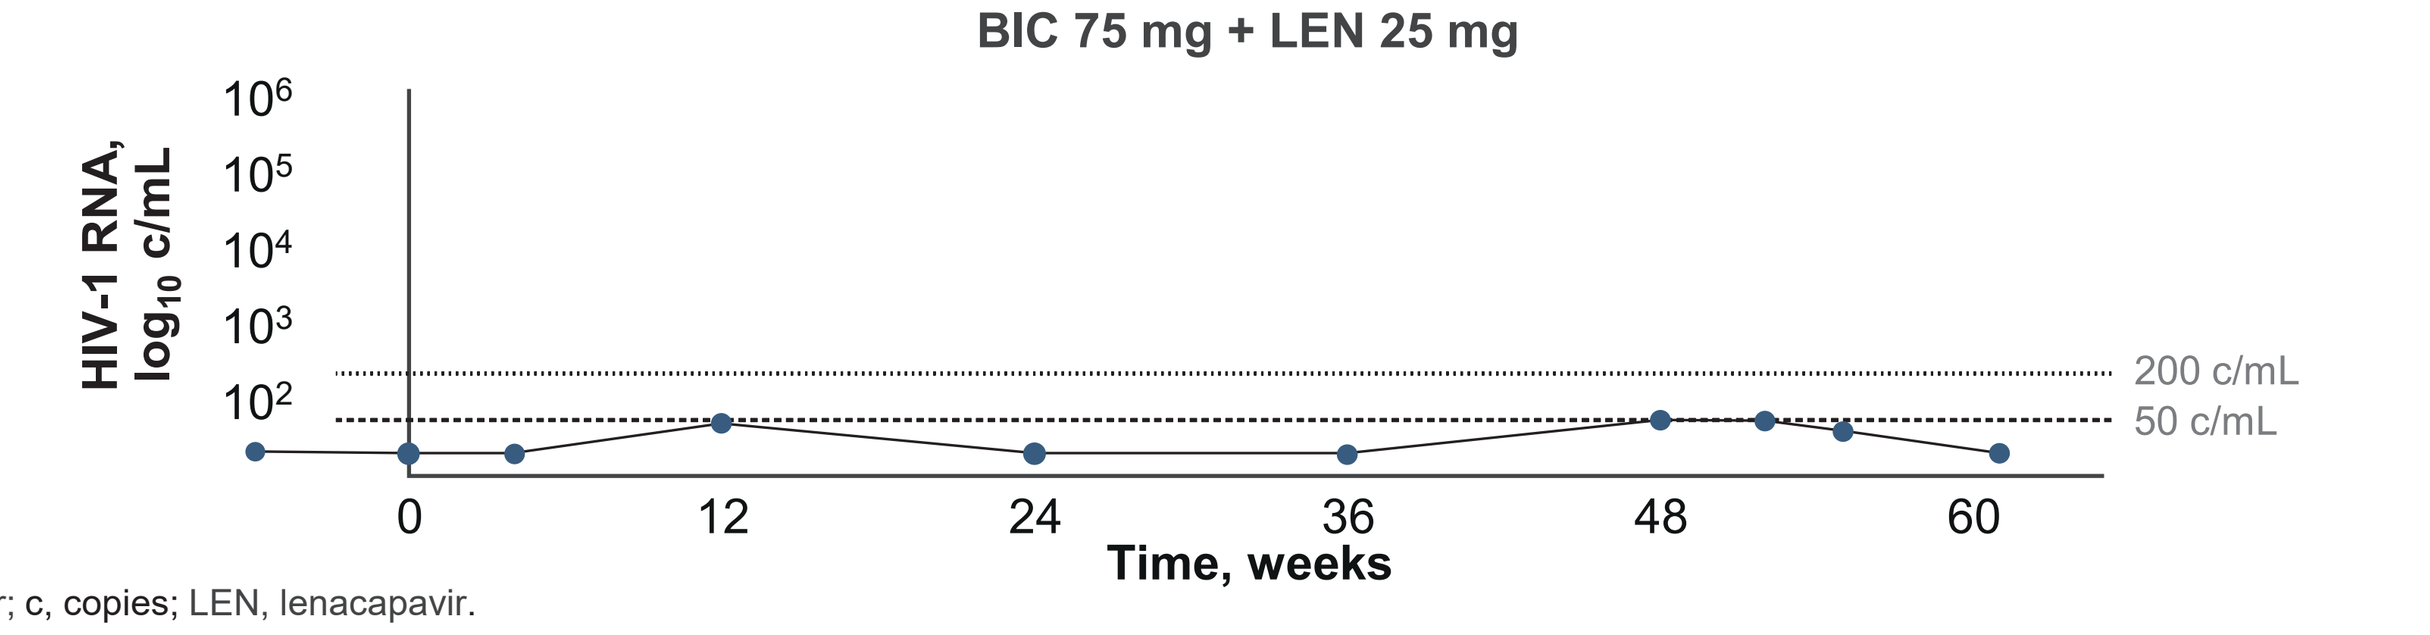

BIC 75 mg + LEN 25 mg
HIV-1 RNA, log10 c/mL
10^6
10^5
10^4
10^3
10^2
200 c/mL
50 c/mL
0
12
24
36
48
60
Time, weeks

BIC, bictegravir; c, copies; LEN, lenacapavir.

- FDA Snapshot HIV-1 RNA ≥ 50 c/mL at Week 48: HIV-1 RNA = 50 c/mL
- Participant did not meet threshold for resistance analysis (viral load [VL] ≥ 200 c/mL) and resuppressed at next visit

Participant With HIV-1 RNA ≥ 50 c/mL at Week 48 – 2 of 3

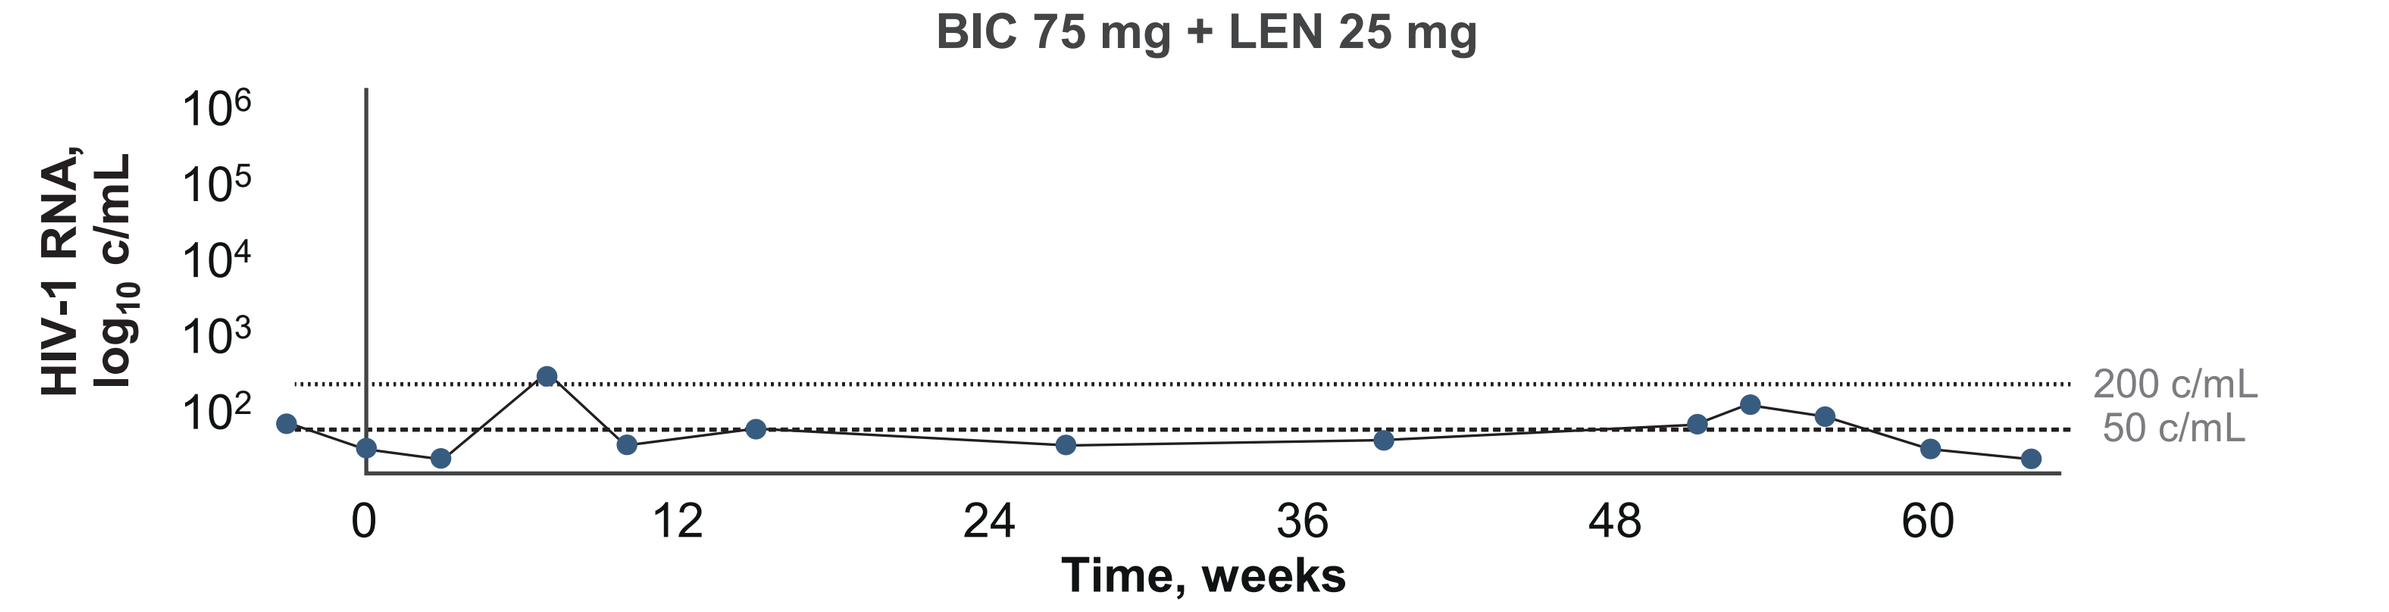

BIC 75 mg + LEN 25 mg
HIV-1 RNA, log10 c/mL
10^6
10^5
10^4
10^3
10^2
200 c/mL
50 c/mL
0
12
24
36
48
60
Time, weeks

BIC, bictegravir; c, copies; LEN, lenacapavir.

- FDA Snapshot HIV-1 RNA ≥ 50 c/mL at Week 48: HIV-1 RNA = 98 c/mL
- Participant did not meet threshold for resistance analysis (VL ≥ 200 c/mL) and resuppressed at next visit

Participant With HIV-1 RNA ≥ 50 c/mL at Week 48 – 3 of 3

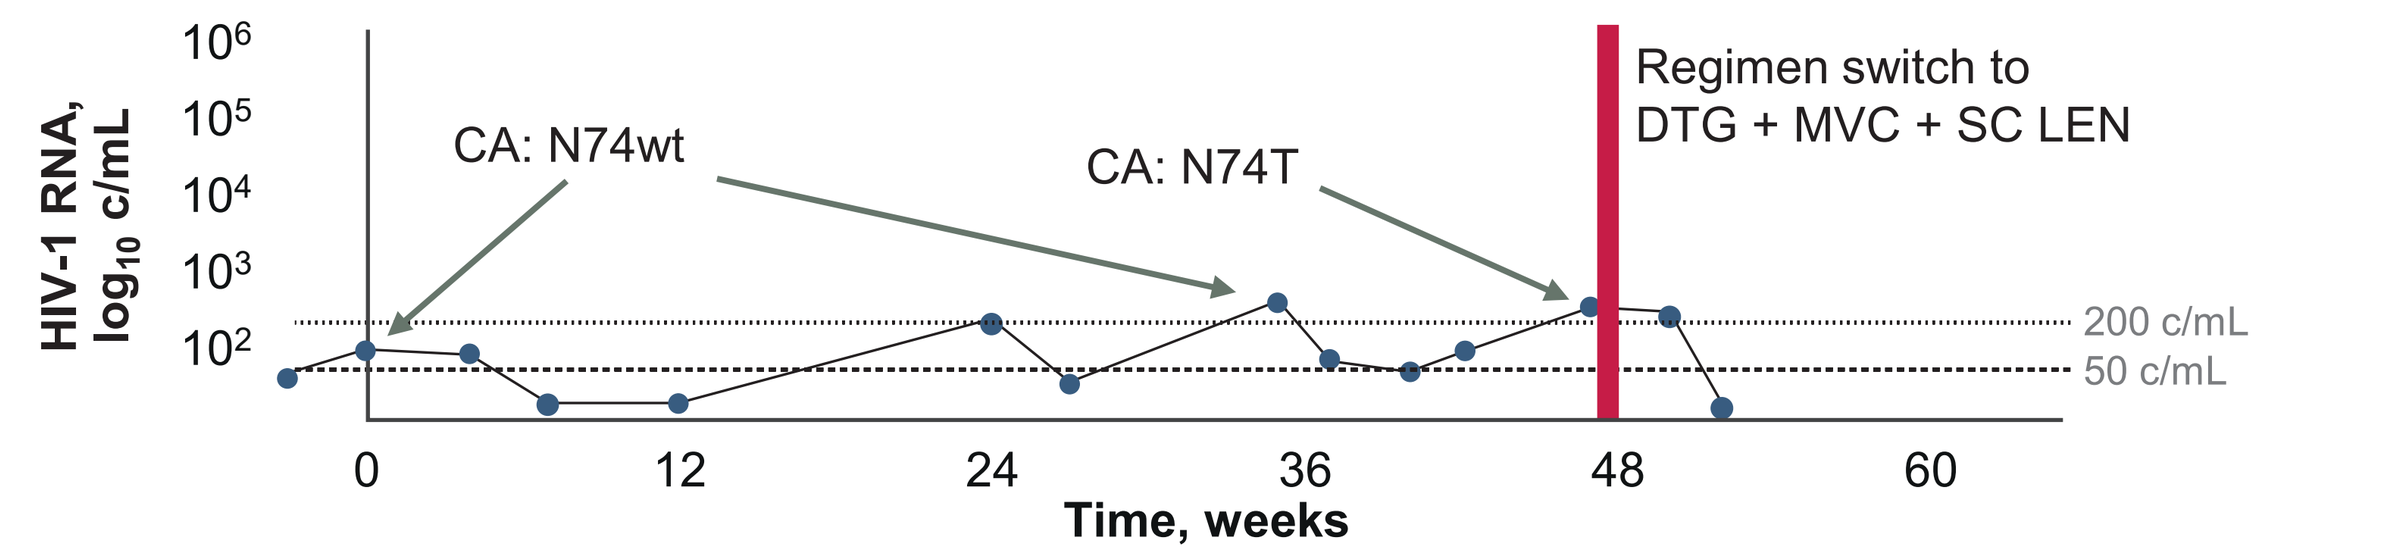

BIC 75 mg + LEN 50 mg
HIV-1 RNA, log10 c/mL
10^6
10^5
10^4
10^3
10^2
CA: N74wt
CA: N74T
Regimen switch to DTG + MVC + SC LEN
200 c/mL
50 c/mL
0
12
24
36
48
60
Time, weeks

BIC, bictegravir; c, copies; CA, capsid; DTG, dolutegravir; LEN, lenacapavir; MVC, maraviroc; SC, subcutaneous.

- FDA Snapshot HIV-1 RNA ≥ 50 c/mL at Week 48: HIV-1 RNA = 305 c/mL
- Resistance analysis showed emergence of N74T in capsid with PhenoSense assay failure; participant resuppressed subsequently with new regimen of dolutegravir + maraviroc + subcutaneous LEN

Phenotypic Characterization of N74T Capsid Mutation

- Capsid mutants containing N74T were generated by site-directed mutagenesis
 - Laboratory strain (HIV-1XXLAI) and three clinical samples with diverse capsid sequence
- Mutants were tested in single-cycle (Monogram Biosciences) and multicycle (MT-2 and RevLun) assays

Mutant	n[a]	Mean LEN EC_{50} Fold Change from WT		
		MT-2	RevLun	Monogram
N74wt	3	1.0	1.0	1.0
N74T	3	1.4	1.2	1.1
N74D	3	24.0	12.5	14.1

[a]Each mutant was tested in three to four to different genetic constructs; repeated at least three times per construct.
EC_{50}, effective concentration to inhibit 50% of viral replication; LEN, lenacapavir; WT, wild type.

- N74T does not confer resistance to LEN
 - In contrast to N74D control mutants
- N74T is a novel capsid polymorphism
- No change in susceptibility to BIC

References: **1.** Department of Health and Human Services. https://clinicalinfo.hiv.gov/sites/default/files/guidelines/documents/adult-adolescent-arv/guidelines-adult-adolescent-arv.pdf (accessed April 23, 2024). **2.** Chang H-M, et al. *BMC Infect Dis.* 2022;22:2. **3.** Rolle C-P, et al. *J Virus Erad.* 2020;6:100021. **4.** Colloty J, et al. *Br J Hosp Med (Lond).* 2023;84:1-9. **5.** Acosta RK, et al. *Antimicrob Agents Chemother.* 2019;63:e02533-18. **6.** European AIDS Clinical Society. https://www.eacsociety.org/media/guidelines-12.0.pdf (accessed July 10, 2024). **7.** Gandhi RT, et al. *JAMA.* 2023;329:63-84. **8.** Margot N, et al. *Antimicrob Agents Chemother.* 2021;65:e02057-20. **9.** Marcelin A-G, et al. *J Antimicrob Chemother.* 2020;75:1588-90. **10.** Mounzer K, et al. *Clin Infect Dis.* November 2024 [ePub]. doi:10.1093/cid/ciae522. **11.** Demirdjian S, et al. Poster 681 presented at: Conference on Retroviruses and Opportunistic Infections (CROI); March 3-6, 2024; Denver, CO.

Acknowledgments: We wish to thank the participants who chose to enroll into the study, as well as the investigators and staff who facilitated the advancement of the study. This study was sponsored by Gilead Sciences, Inc. Production assistance was provided by Aspire Scientific Ltd (Bollington, UK), and was funded by Gilead Sciences, Inc.

Disclosures: All authors are employees and stock holders of Gilead Sciences, Inc.

Correspondence: Nicolas Margot, Nicolas.Margot@gilead.com.

Conference on Retroviruses and Opportunistic Infections (CROI); March 9–12, 2025; San Francisco, CA, USA